I-152

Presented at the
20th Annual NPWH Premier Women's Healthcare Conference
October 11–14, 2017 • Seattle, WA

Development of a Patient-Centric Exit Study to Contextualize and Assess Meaningfulness of a Potential Treatment for Hypoactive Sexual Desire Disorder (HSDD)

Patricia E. Koochaki,[1] Dennis A. Revicki,[2] Hilary Wilson,[3] Robin Pokrzywinski,[2] Robert Jordan,[4] Johna Lucas[4]
[1]PEK Market Research, Inc, Loveland, OH; [2]Evidera, Bethesda, MD; [3]Evidera, Seattle, WA; [4]Palatin Technologies, Inc, Cranbury, NJ

Introduction

- Female sexual dysfunctions (FSD) include a group of conditions characterized by physiological, psychological, and social components[1]
- The most common sexual concern expressed by women with FSD is diminished or lack of desire for sexual activity. When accompanied by distress, this may be diagnosed as hypoactive sexual desire disorder (HSDD)[2]
- Bremelanotide (BMT; PT-141) is an investigational drug being developed as a treatment for HSDD in premenopausal women[3]

Study Design

- The RECONNECT studies comprise 2 identical, randomized, Phase 3, placebo-controlled, multicenter trials (NCT02333071 [Study 301] and NCT02338960 [Study 302]) designed to assess the efficacy and safety of BMT administered via an autoinjector device on an as-desired basis to treat HSDD in premenopausal women
- The Core phase of the trials includes a 1-month no-intervention qualification period, a 1-month single-blind placebo treatment period, and a 24-week double-blind treatment period. Both trials also have an ongoing 52-week open-label extension
- Co-primary efficacy endpoints are:
 - —Change from baseline to end-of-study in desire-domain score of the Female Sexual Function Index (FSFI)[4,5]
 - —Change in score for "feeling bothered by low sexual desire" as measured by Item 13 of the Female Sexual Distress Scale–Desire/Arousal/Orgasm (FSDS-DAO)[6,7]

RECONNECT Study Population

- Healthy, premenopausal, nonpregnant women, ≥18 years of age, currently in a stable (≥6 months) relationship, and diagnosed with HSDD (with or without decreased arousal) for ≥6 months
- Participants had to have experienced "normal" sexual function in the past for ≥2 years and be willing to engage in sexual activities ≥1×/month during the study
- Participants were required to have met minimum thresholds on the FSFI and FSDS-DAO
- An exit study with a subset of participants was prespecified for both studies

RECONNECT Exit Study Objectives

- Understand participants' motivations to seek treatment and their treatment goals
- Compare experiences and the impact of HSDD before the study, experiences during the study, and perceptions of/experiences with the autoinjector device
- Assess whether patients experienced meaningful benefits from BMT and, if so, what these benefits were
- Assess the experiences and differences between women who did and did not report experiencing meaningful benefit
- Determine the proportion of women who experienced meaningful benefits and whether there were significant differences between those taking BMT and those taking placebo
- Add context and meaning to the clinical study data to create a holistic, patient-centric understanding of women's experiences and meaningfulness of treatment benefits with BMT (**Figure 1**)

Figure 1. Framework for Developing a Holistic Understanding of Patient Experiences

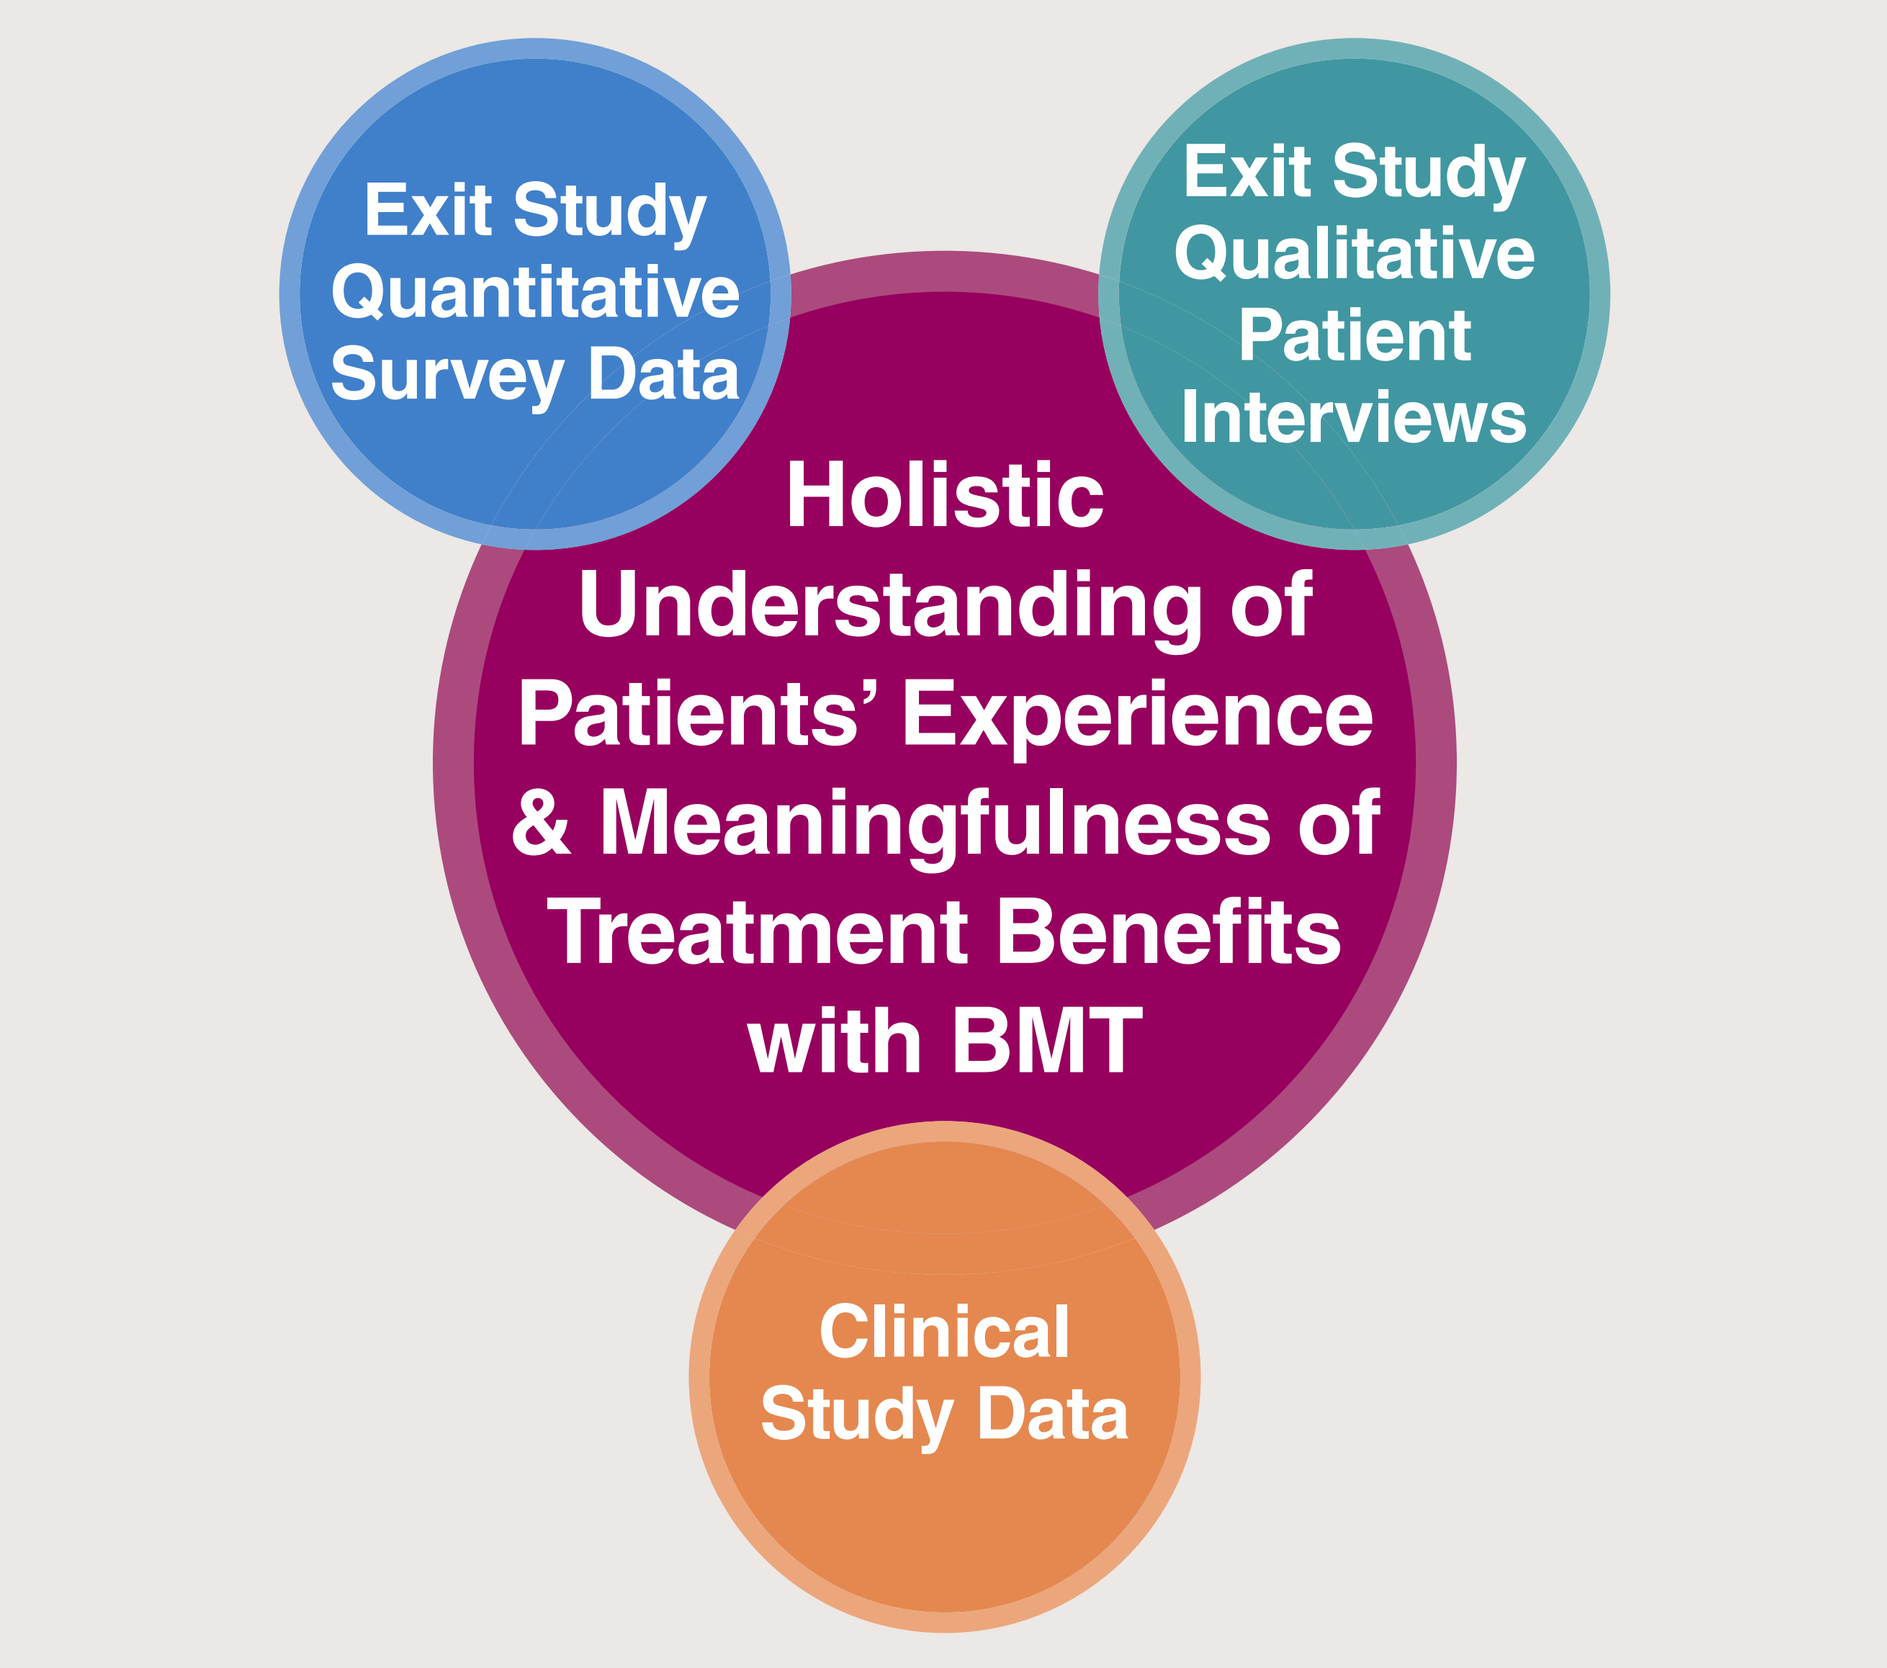
Exit Study Quantitative Survey Data
Exit Study Qualitative Patient Interviews
Holistic Understanding of Patients' Experience & Meaningfulness of Treatment Benefits with BMT
Clinical Study Data

Exit Study Methodology

The Exit Study was developed using a mixed-method design that included both quantitative and qualitative assessments:

The ***Quantitative Survey*** was used to assess:

- Meaningfulness of treatment benefits
- Whether expectations were met
- Satisfaction with autoinjector device

Qualitative Interviews were conducted to:

- Contextualize the clinical and survey data through understanding the study participants' experiences and the impact of treatment on their lives
- Explore why treatment was meaningful and whether expectations for therapy were met
- Gain an understanding of study participants' experiences using the autoinjector device

Women who completed the Exit Study were paid a small stipend to participate.

Development of the Quantitative Survey

- Concepts and items were proposed by the research team and clinical experts (physicians and sex therapists)
- Cognitive debriefing interviews were conducted with 15 women with HSDD who were not participating in the clinical trial to ensure:
 - —Instructions for completing the survey were understandable
 - —Wording of each item was clear and had a singular meaning
 - —Response options were sufficient and appropriate for each question
- Survey was refined and finalized prior to the start of the Exit Study

Administration of Quantitative Survey

- Clinic site staff recruited participants at Visit 8 of the trial and administered the survey on-site at Visit 9 (Week 24), or within 7 days following the participant's last study visit, but prior to patient's initiation into an open-label trial extension
- The survey took 10 to 15 minutes to complete
- Written informed consent was obtained prior to participation
- All participants, clinic staff, and interviewers were blinded to treatment

Administration of Qualitative Semi-Structured Interviews

- Telephone interviews were conducted with a subset of participants who completed the ***Quantitative Survey***
- The semi-structured interviews were conducted by trained interviewers while the participant was at the clinical site, or at home, whichever was more convenient to the participant
- Interviews lasted approximately 60 minutes and were recorded for transcription and analysis

Measures

The ***Quantitative Survey*** consisted of 4 parts:

1. Treatment Experience: A series of questions on whether participants received meaningful benefit from the study medication. They were asked about the meaningfulness of the medication benefit overall and the meaningfulness of the benefit of the medication for treating specific issues associated with HSDD. A sample of these questions is shown below:

> *Thinking about your experiences overall during the clinical study, would you say you did or did not **BENEFIT** from the study medication?*
>
> *Thinking about your experiences while using the study medication, would you say you did or did not experience an increase in sexual **DESIRE**?*

2. Overall Experience: Questions on the participant's relationship with her partner, whether the study medication met her expectations, and whether she would recommend the study medication to others
3. Device Experience: A series of questions to rate performance of the autoinjector device and the importance of specific characteristics (eg, ease of use, convenience, or ability to use just when wanted)
4. Additional Learning: Included a question on how important it was to the participants that the medication delivered specific benefits (eg, increasing sexual thoughts, increasing interest in initiating sexual activity)

The ***Qualitative Interviews*** were conducted using a 3-part semi-structured interview guide; interviewers were blinded to treatment assignment:

1. Participant's in-depth experiences (physical, emotional, and mental) with HSDD, any measures taken to address her HSDD, her motivations for entering the study, and her expectations for the study medication
2. Participant's in-depth experience with the study medication, including physical and emotional changes, as well as any impact on her relationship with her partner, her self-confidence and self-image
3. Discussion about the trial intervention, any difficulties experienced during the trials, whether the study medication met the participant's expectations or provided meaningful benefit, and her experience with using the autoinjector device

Data Analysis

- The survey was formatted for optical character-recognition software
 - —DataFax (DF/Net Research, Inc, Seattle, WA) was used for data collected during study visits
 - —Survey data were analyzed using SAS (V9.4; SAS, Cary, NC)
- All interviews were audio recorded and transcribed for analysis using ATLAS.ti software (Scientific Software Development GmbH, Berlin; V7.1 or higher)

Quantitative Survey Analysis

- Descriptive statistics (mean, standard deviation, frequency) were used to characterize the sample in terms of the sociodemographic characteristics of the survey+interview group, survey-only group, and overall study group's sociodemographic characteristics
- Sociodemographic and clinical characteristics were compared between the BMT 1.75 mg group and the placebo group, between the survey+interview group and survey-only group, and between responders and nonresponders
- The characteristics of each survey item were described by the BMT 1.75 mg group and placebo group and the relationship between these groups was explored by survey item
- Chi-square tests were used to compare categorical variables and t-tests or analysis of variance models were used to compare continuous variables
- Logistic regression models were also conducted to identify predictors of response to treatment

Qualitative Interview Analysis

- ATLAS.ti is software designed for the qualitative analysis of textual, graphical, audio, and video data. It permits researchers to create and enter names of concepts, or "codes," that can be used for conceptualizing large amounts of qualitative data. The program allows the analyst to organize and relate these codes to each other to evaluate the underlying structure of the qualitative data
- The analysis proceeded in 5 steps:

1. A coding dictionary was developed based on the semi-structured-interview guide
2. Words and phrases provided by interview participants were selected using the coding dictionary and grouped into key themes, attributes, concepts, and relationships
3. The concepts and definitions were refined as needed, via team discussion and revision of the coding scheme
4. Using the coding dictionary, 2 staff members independently coded the first structured interview transcript. This was followed by a postcoding comparison and reconciliation with a senior scientific reviewer. All codes were compared, discussed, and reconciled wherever differences occurred
5. When agreement between the 2 coders was sufficient, they each went on to independently code the remaining transcripts with review from a senior scientific staff member. This process was repeated when new coders were added

- All qualitative coding was completed prior to treatment group unblinding. Once treatment assignments were unblinded, the treatment variable (BMT or Placebo) was assigned to the participant's ID number

Key Results

Study Population (Table 1)

- Surveys were administered to 248 participants; 6 of these were unusable because an earlier draft version of the survey was incorrectly administered, resulting in 242 surveys usable for data analysis
- Qualitative telephone interviews were conducted with 85 participants; 5 were unusable due to technical difficulties with the recording devices, leaving 80 interviews available for analysis

Table 1. Patient Demographic and Clinical Characteristics by Post-Trial Study Group (Survey+Interview vs Survey Only)

Variable	Survey + Interview[a] (n=77)	Survey Only (n=165)	Overall (N=242)	*P* value[b]
Age (years)				
Mean (SD)	37.8 (7.23)	39.4 (6.53)	38.9 (6.79)	0.0861
Race (n, % yes)				
White	66 (85.7%)	147 (89.1%)	213 (88.0%)	
Black or African American	8 (10.4%)	15 (9.1%)	23 (9.5%)	
Asian	1 (1.3%)	1 (0.6%)	2 (0.8%)	
Multiple	2 (2.6%)	2 (1.2%)	4 (1.7%)	0.7830
Diagnosis				
HSDD with decreased arousal	57 (74.0%)	94 (57.0%)	151 (62.4%)	
HSDD without decreased arousal	20 (26.0%)	71 (43.0%)	91 (37.6%)	0.0107
Duration of Diagnosis				
Mean, months (SD)	47.3 (42.12)	58.9 (50.33)	55.2 (48.09)	0.0107
Reproductive Status				
Childbearing potential	61 (79.2%)	130 (78.8%)	191 (78.9%)	
Surgically sterile	16 (20.8%)	33 (20.0%)	49 (20.2%)	
Missing	0 (0.0%)	2 (1.2%)	2 (0.8%)	0.9237
Body Mass Index				
Mean (SD)	28.6 (6.54)	28.3 (6.76)	28.4 (6.68)	0.7049
Length of Time in Relationship with Partner (months)				
Mean (SD)	128.4 (85.17)	155.5 (91.66)	146.9 (90.36)	0.0295
Baseline FSFI				
Mean (SD)	17.6 (5.02)	17.9 (4.76)	17.8 (4.83)	0.7200
Baseline FSDS-DAO				
Mean (SD)	39.3 (10.41)	40.8 (10.29)	40.3 (10.33)	0.2957
Baseline SSEs				
Mean (SD)	0.9 (1.07)	0.8 (1.07)	0.8 (1.07)	0.6688

[a]3 of the 80 interview participants completed invalid surveys and thus, were not included in the quantitative data analysis; [b]Chi-square test for categorical variables and t-test for continuous variables.

FSDS-DAO, Female Sexual Distress Scale–Desire/Arousal/Orgasm; FSFI, Female Sexual Function Index; HSDD, hypoactive sexual desire disorder; SD, standard deviation; SSEs, Sexually Satisfying Events.

Survey Results

- Women who completed the survey+interview were more likely to be diagnosed with HSDD with decreased arousal, relative to women who completed the survey only ($P<0.05$) and had a slightly shorter duration of diagnosis ($P<0.05$) and a slightly shorter length of time in relationship with their partner ($P<0.05$), relative to women who completed the survey only
- Participants in the BMT 1.75 group and responders to therapy were significantly (58.8% vs 27.1%; $P<0.0001$) more likely to report meaningful treatment benefits relative to participants in the placebo group
- Generally, the autoinjector device was well tolerated and well received. Although some women had reservations about an injectable therapy before they used the study medication, after initiating therapy, they found the autoinjector device easy to use and experienced little apprehension

Interview Results

- Participants shared how HSDD negatively impacted their relationships with their partners, particularly their closeness with their partner
- Some of the major impacts of HSDD reported by these women were emotional (eg, frustration, feelings of decreased sexual self-confidence)
- Most women said their HSDD developed gradually, rather than having a sudden onset
- Participants reported the quality of their sexual pleasure and quality of their sexual sensation was poor before entering the clinical trial. They reported ways they tried to seek help and coping techniques, but said that none of these provided sustained benefits
- Motivations for enrolling in the RECONNECT studies included "wanting to have a closer relationship with her partner" and "wanting an increase in sexual desire and arousal"
- Many women randomized to BMT reported experiencing positive changes, including increased sexual desire, increased sexual arousal and lubrication, and increased self-confidence

Conclusions

- A mixed-methods, patient-centric exit study, consisting of a quantitative survey and qualitative interviews, has been designed that further explores the impact of HSDD on premenopausal women, contextualizes experiences with BMT and the autoinjector device, and provides deep understanding of the meaningfulness of treatment benefits experienced by women in clinical studies with BMT
- The survey provides a quantitative perspective on how many, and which, participants received benefits and found them to be meaningful, and on participants' acceptance of the autoinjector delivery device
- The qualitative interviews provide insight on how and why benefits were meaningful, the impact of the benefits beyond the clinical trials, and how participants perceive the medication working to provide benefits
- Combining the information derived from the clinical trials, quantitative exit survey, and qualitative exit interviews will provide a unique, holistic approach to understanding the efficacy of BMT for treating premenopausal women with HSDD

Support This study was funded by Palatin Technologies, Inc. Editorial assistance was provided by The Curry Rockefeller Group, LLC, which was funded by Palatin Technologies, Inc.

Disclosures **PEK** received consulting fees from Palatin Technologies, Inc. **DAR**, **HW**, and **RP** are salaried employees of Evidera. Evidera received research support from Palatin Technologies, Inc. **RJ** and **JL** are employees and stockholders of Palatin Technologies, Inc.

References **1.** Basson R. *CMAJ.* 2005;172(10):1327–1333. **2.** Shifren JL, et al. *Obstet Gynecol.* 2008;112(5):970–978. **3.** Molinoff PB, et al. *Ann N Y Acad Sci.* 2003;994:96–102. **4.** Meston CM. *J Sex Marital Ther.* 2003;29(1):39–46. **5.** Rosen R, et al. *J Sex Marital Ther.* 2000;26(2):191–208. **6.** DeRogatis LR, et al. Presented at: 167th Annual Meeting of the American Psychiatric Association. New York, New York, USA, May 3–7, 2014. Abstract 159. **7.** DeRogatis LR, et al. *J Sex Marital Ther.* 2002;28(4):317–330.