Poster presented at the
International Society for the Study of Women's Sexual Health Annual Meeting
February 23–26, 2017 • Atlanta, GA

Reliability and Validity of the Elements of Desire Questionnaire in the Bremelanotide RECONNECT Study

Dennis A. Revicki,[1] Stanley Althof,[2] Leonard R. DeRogatis,[3] Hilary Wilson,[4] Robert Jordan,[5] Johna Lucas[5]

[1]Evidera, Bethesda, MD, USA; [2]Case Western Reserve University School of Medicine, Cleveland, OH, USA; [3]Maryland Center for Sexual Health, Lutherville, MD, USA; [4]Evidera, Seattle, WA, USA; [5]Palatin Technologies, Inc, Cranbury, NJ, USA

Background

Female sexual dysfunction (FSD) comprises a group of conditions with physiological, psychological, and social components. In the United States and European Union, ~43% of adult women experience some form of FSD; about 22% of these women report feeling distressed by their sexual dysfunction. The most frequent concerns expressed by these women are decreased desire and/or arousal.[1]

Bremelanotide (BMT) is a novel, cyclic, 7-aminoacid, melanocortin-receptor agonist with high affinity for the type-4 receptor,[2] giving it a potential to modulate brain pathways involved in sexual response.[3] It is currently being evaluated in phase 3 trials for the treatment of hypoactive sexual desire disorder (HSDD)[a] in premenopausal women. The Elements of Desire Questionnaire (EDQ), a newly developed measure of desire, was included in these trials as an exploratory endpoint.

The EDQ is a patient-reported outcome questionnaire intended for clinical trials evaluating treatments that may improve sexual desire in women with FSD. The EDQ was developed based on input from clinical experts, and the content validity was explored through qualitative research with women with HSDD. Monthly recall and 24-hour recall daily-diary versions are available. The monthly EDQ includes 9 items scored on a Likert-type scale that ranges from 1 (never/absent/very dissatisfied) to 5 (always/very high/very satisfied). In the 24-hour version, 2 of the 9 questions have a "Yes" or "No" response. Both versions of the EDQ were used in the BMT phase 3 clinical trials (NCT02333071; NCT02338960) as an exploratory endpoint.

*In the *Diagnostic and Statistical Manual of Mental Disorders (DSM), 4th Edition, Text Revision,* HSDD was classified as discrete diagnosis. In the 5th edition, *DSM-5*, it was merged with female sexual arousal disorder into a single diagnosis of female sexual interest/arousal disorder (FSIAD). The study used to perform this evaluation predates that change, thus the term HSDD is used throughout this report.

Objective

The primary objective of this secondary analysis was to evaluate the measurement properties of both versions of the EDQ in premenopausal women with HSDD. This included examining the factor structure and item characteristics, developing a scoring algorithm, exploring the relationship between the 2 versions, examining the instrument's construct validity (convergent and known groups) and ability to detect change, and exploring interpretation thresholds.

Materials and Method

The RECONNECT study comprises 2 phase 3, multicenter, randomized, double-blind trials of BMT, each with an open-label extension. The trials include a 1-month, no-drug, qualification period (to confirm the diagnosis), followed by a monthly single-blind placebo-treatment period (to establish baseline), and a monthly double-blind–treatment period (core phase). A 52-week open-label extension is ongoing. Subjects were randomized (1:1) to either placebo or BMT 1.75 mg. Participants self-administered BMT or placebo subcutaneously via auto-injector, as-desired, prior to sexual activity. The study met both of its coprimary endpoints: change in the desire domain of the Female Sexual Function Index (FSFI-D) and the Female Sexual Distress Scale-Desire/Arousal/Orgasm (FSDS-DAO) Item 13 score (data on file).

For the EDQ psychometric analysis, data from both phase 3 BMT trials were combined and analyzed together in a single dataset that included select data from visit 1 (week 0) through the end-of-study visit, visit 9 (week 32). Participants completed the EDQ daily for 7 consecutive days prior to visits 2, 3, 6, 9, and also monthly at every clinic visit.

Analysis Population

The analysis population included participants in the modified intent-to-treat population, as defined in the statistical-analysis plan, who had FSFI data at baseline and ≥1 follow-up visits. All data were pooled across the 2 studies, and data were masked to treatment group. All analyses were predefined in the statistical-analysis plan.

Factor Analysis

For the factor analyses, the pooled data were randomly split into 2 samples. The exploratory factor analyses were conducted with the first-split half, and then confirmatory analyses were conducted with the second-split half.

Item Characteristics

Individual item descriptive characteristics on the EDQ item scores were calculated, including item response frequencies and item mean, median, standard deviation, and range at visits 2, 3, 6, and 9. Item-to-item correlations (Spearman's) were calculated for the EDQ item scores using visits 2 and 6 data for both the monthly recall scores and single-day 24-hour recall scores.

Scoring Algorithm

Descriptive statistics and results of the factor analysis and item-response theory (IRT) analysis were used to select the final items for the EDQ, determine the final conceptual framework, and develop scoring algorithms. The EDQ Scale scores were calculated based on the final item content and scoring algorithm for both the monthly and daily-diary versions, including both single-day and 7-day average EDQ scores for visits 2, 3, 6 and 9.

Item-to-Total Correlations

Item-to-total and corrected item-to-total correlations (Spearman's) were calculated for the EDQ Scale scores to evaluate the relative contribution of each of the item scores to the respective total scores.

Relationship Between the Monthly and 24-Hour Recall Versions

Spearman's coefficients and intraclass correlation coefficients (ICCs) were calculated between the monthly recall EDQ scores and the 7-day weekly average daily-diary EDQ scores from visits 2, 3, 6, and 9 to inform the agreement between these 2 different recall periods.

Reliability

Internal Consistency Reliability

Cronbach's formula for coefficient alpha[4] was used to examine the item-to-item correlations for the EDQ. Alpha coefficients between 0.7 and 0.9 indicate good internal consistency, between 0.4 and <0.7 indicate moderate internal consistency, and <0.4 indicate low internal consistency reliability.[4,5]

Test-Retest Reliability

Test-retest reliability was assessed by an ICC of the monthly recall EDQ scores at visit 1 and visit 2. ICC values >0.7 are generally considered acceptable for establishing test-retest reliability.[6]

Validity

Convergent and divergent validity were explored through examination of the pattern of correlations between the EDQ scores and the scores on the other measures of female sexual function that were used in the study: the FSFI, Female Sexual Encounter Profile–Revised (FSEP-R), FSDS-DAO, General Assessment Questions (GAQ), and 9-Item Women's Inventory of Treatment Satisfaction from visits 3 and 6. Correlations were classified according to their Spearman's measures as small (<0.3), moderate (0.3–0.6), or large (>0.6).[7]

Known-groups validity was evaluated by comparing the mean EDQ score(s) at visits 3, 6, and 9 in women with varying degrees of HSDD severity as defined by the FSDS-DAO (<11 vs ≥11), GAQ desire item (≥5 vs <5), and number of sexually satisfying events (SSEs; <2 vs ≥2).

Ability to Detect Change and Interpretation Thresholds

The ability to detect change was evaluated by comparing the mean change in EDQ scores from visit 2 (baseline) to visit 9 among women who had achieved the following definitions of response at visit 9: FSDS-DAO (<11 responder vs ≥11 nonresponder), GAQ desire item (≥5 responder vs <5 nonresponder), and number of SSEs (<2 nonresponder vs ≥2 responder events).

Results

Analysis Population

1267 participants were randomized, and 1198 were included in the evaluable population. The sample was predominantly white (86.5%), with a mean age of 38.7 years; the majority (98.2%) had a male partner (**Table 1**).

Table 1. Demographics Characteristics

Variable		n=1198
Age, years	Mean (SD) Median Range	38.7 (7.05) 39 (19–56)
Gender of partner, n (%)	Male Female	1176 (98.2) 22 (1.8)
Race, n (%)	White Black or African American Asian Multiple	1025 (86.5) 138 (11.6) 13 (1.1) 9 (0.8)

SD, standard deviation.

Factor Analysis

- Exploratory factor analyses in the first-split half of the sample supported a unidimensional-factor model for both versions of the EDQ
- Confirmatory factor analyses in the second-split half of the sample confirmed a unidimentional-factor structure (**Table 2**)

Table 2. EDQ Confirmatory Factor Analysis, Monthly Recall: Visit 3

EDQ Item	Standardized Coefficient n=600
Over the past 4 weeks, how often did you feel sexual desire or interest?	0.937
Over the past 4 weeks, what was the intensity of your sexual desire or interest?	0.915
Over the past 4 weeks, how often did you have thoughts or fantasies about sexual activities?	0.801
Over the past 4 weeks, how would you rate your interest in engaging in sexual activity?	0.932
Over the past 4 weeks, how often did you want to have sexual activity?	0.918
Over the past 4 weeks, how would you rate your receptivity to your partner's sexual requests?	0.704
Over the past 4 weeks, how often did you initiate a sexual activity?	0.746
Over the past 4 weeks, how would you rate your interest in initiating sexual activity?	0.899
Over the past 4 weeks, how would you rate your satisfaction with your level of desire or interest in sexual activity?	0.822

Confirmatory Fit Index=0.990, Root Mean Square Error of Approximation=0.113, and Weighted Root Mean Residual=1.342.
EDQ, Elements of Desire Questionnaire.

Item Characteristics

- Mean item scores for the monthly recall EDQ scores increased (improved) over time and ranged from 1.3 to 2.0 at visit 2, 1.7 to 2.2 at visit 3, 1.9 to 2.5 at visit 6, and 2.0 to 2.4 at visit 9. Mean item scores for the 24-hour recall version of the EDQ visits also increased (improved) over time but were generally lower (more severe) relative to the monthly recall scores
- No evidence of ceiling effects for any item at any study visit for either version
- Some evidence for a floor effect (ie, between 40% and 75% of participants endorsed the lowest [most severe] category) for a few items at visit 2 in the monthly version. These were more profound in the 24-hour version
- Item-to-item Spearman's correlations for the monthly recall version of the EDQ at visit 2 ranged from 0.22 (items 6 and 7) to 0.63 (items 1 and 2). Overall, item-to-item correlations increased across time points from visit 3 to visit 6 with correlations at visit 6 ranging from 0.52 (items 6 and 7) to 0.83 (items 4 and 5). The pattern of correlations was comparable with the item-to-item correlations for the monthly recall version

Scoring Algorithm

- Although the IRT analysis indicated potential item redundancy, the decision was made to not alter or remove any items at this time
- Since the factor analyses for both the monthly and 24-hour recall versions of the EDQ supported a unidimensional factor, a single total score was proposed
 - The monthly recall was calculated as the mean of all 9 EDQ items
 - For the 24-hour recall version the mean daily-summary score was calculated using Items 1, 2, 3, 4, 6, 8, and 9 in the 7-day period leading up to a respective study visit. Items 5 and 7 were omitted from the total weekly score as they were assessed using a Yes/No response
- The 24-hour recall scores were consistently lower (less desire) relative to the monthly recall scores. All scores increased (improved) across study visits
- Compared with the monthly version, the 24-hour recall version had substantially greater missing data (ie, the sample size for the EDQ weekly recall score was 40% to 54% smaller, relative to the monthly recall EDQ)

Item-to-Total Correlations

Item-to-total correlations for the monthly recall version of the EDQ ranged between 0.66 (item 7) and 0.85 (item 4). Item-to-total correlations for the 24-hour recall version of the EDQ were comparable with the exception of the 2 Yes/No items, which resulted in lower item-to-total correlations (item 5=0.62; item 7=0.39).

Relationship Between the Monthly and 24-Hour Recall Versions

- The relationship between the monthly and 24-hour recall versions of the EDQ was examined through Spearman's correlation coefficients and ICCs at visits 2, 3, 6, and 9. All correlation coefficients were ≥0.76; ICCs ranged between 0.64 (visit 3) and 0.83 (visit 9; **Table 3**)
- Mean 24-hour weekly EDQ scores were consistently lower (less desire) relative to the monthly recall scores

Table 3. EDQ Monthly Recall vs 24-Hour Recall, Spearman's Correlations and ICCs

	Monthly Recall, Mean (SD)	7-Day Weekly Average, Mean (SD)	Spearman's Correlation Coefficient[a]	ICC
Visit 2	1.6 (0.42)	1.5 (0.37)	0.76	0.68
Visit 3	1.9 (0.60)	1.6 (0.49)	0.77	0.64
Visit 6	2.2 (0.78)	1.9 (0.71)	0.86	0.77
Visit 9	2.2 (0.82)	1.9 (0.75)	0.89	0.83

[a]Indicates significant (*P*<.05) correlation.
EDQ, Elements of Desire Questionnaire; ICC, intraclass correlation coefficient; SD, standard deviation.

Reliability

Internal Consistency Reliability

Internal consistency reliability for the monthly recall and 24-hour recall versions of the EDQ was calculated for visits 2, 3, 6, and 9. For the monthly recall, Cronbach's α ranged from 0.86 (visit 2) to 0.96 (visit 9). For the 24-hour recall, Cronbach's α ranged from 0.91 (visit 2) to 0.97 (visit 9).

Test-Retest Reliability

Test-retest reliability was measured in all subjects from visit 2 (baseline) to visit 3 (randomization) for the monthly recall version, given there was no intervention during this period. The ICC was 0.64 for the 24-hour recall EDQ, day-1 scores were compared with day-7 scores in the week leading up to visit 2; the ICC was 0.45.

Validity

- Correlation coefficients between the EDQ and other measures of desire were moderate to large, providing support for the convergent validity of the EDQ: FSFI desire (visit 3=0.79; visit 6=0.87), FSEP-R desire item (visit 3=0.60; visit 6=0.64), GAQ item 2: satisfaction with desire (visit 3=0.51; visit 6=0.66; **Table 4**)
- As expected, the correlation coefficient between the EDQ and the more distally related concept of pain (FSFI) was small (visit 3=0.07; visit 6=0.15). Results for the 24-hour recall version were comparable
- Participants in the less severe groups based on each anchor variable (FSDS-DAO, GAQ, SSEs) and at each study visit (3, 6, 9) consistently reported higher (less severe) scores on the EDQ relative to patients in the more severe groups (all *P*<.0001), providing support that the EDQ differentiates between patients with varying degrees of HSDD severity (**Figures 1a** to **1c**)

Table 4. EDQ Construct Validity, Correlations With FSFI Scores, Monthly Recall Version

	EDQ Scale Score	
	Visit 3	Visit 6
FSFI Total	r=0.63*	r=0.75*
FSFI Desire	r=0.79*	r=0.87*
FSFI Arousal	r=0.68*	r=0.75*
FSFI Lubrication	r=0.32*	r=0.38*
FSFI Orgasm	r=0.37*	r=0.46*
FSFI Satisfaction	r=0.62*	r=0.74*
FSFI Pain	r=0.07†	r=0.15*
FSEP-R Desire Item	r=0.60*	r=0.64*
FSDS-DAO Item 13	r=–.51*	r=–.60*
FSDS-DAO Total Score	r=–.54*	r=–.61*
GAQ Item 1. Satisfaction with arousal	r=0.49*	r=0.65*
GAQ Item 2. Satisfaction with desire	r=0.51*	r=0.66*
GAQ Item 3. Degree of benefit	r=0.51*	r=0.64*
GAQ Item 4. Change in relationship with partner	r=0.41*	r=0.59*
WITS-9 Total Score	r=0.65*	r=0.76*

*P<.0001; †P<.05.
EDQ, Elements of Desire Questionnaire; FSDS-DAO, Female Sexual Distress Scale-Desire/Arousal/Orgasm; FSEP-R, Female Sexual Encounter Profile–Revised; FSFI, Female Sexual Function Index; GAQ, General Assessment Questions; WITS-9, 9-item Women's Inventory of Treatment Satisfaction.

Figure 1. EDQ Known-Groups Validity Monthly Recall

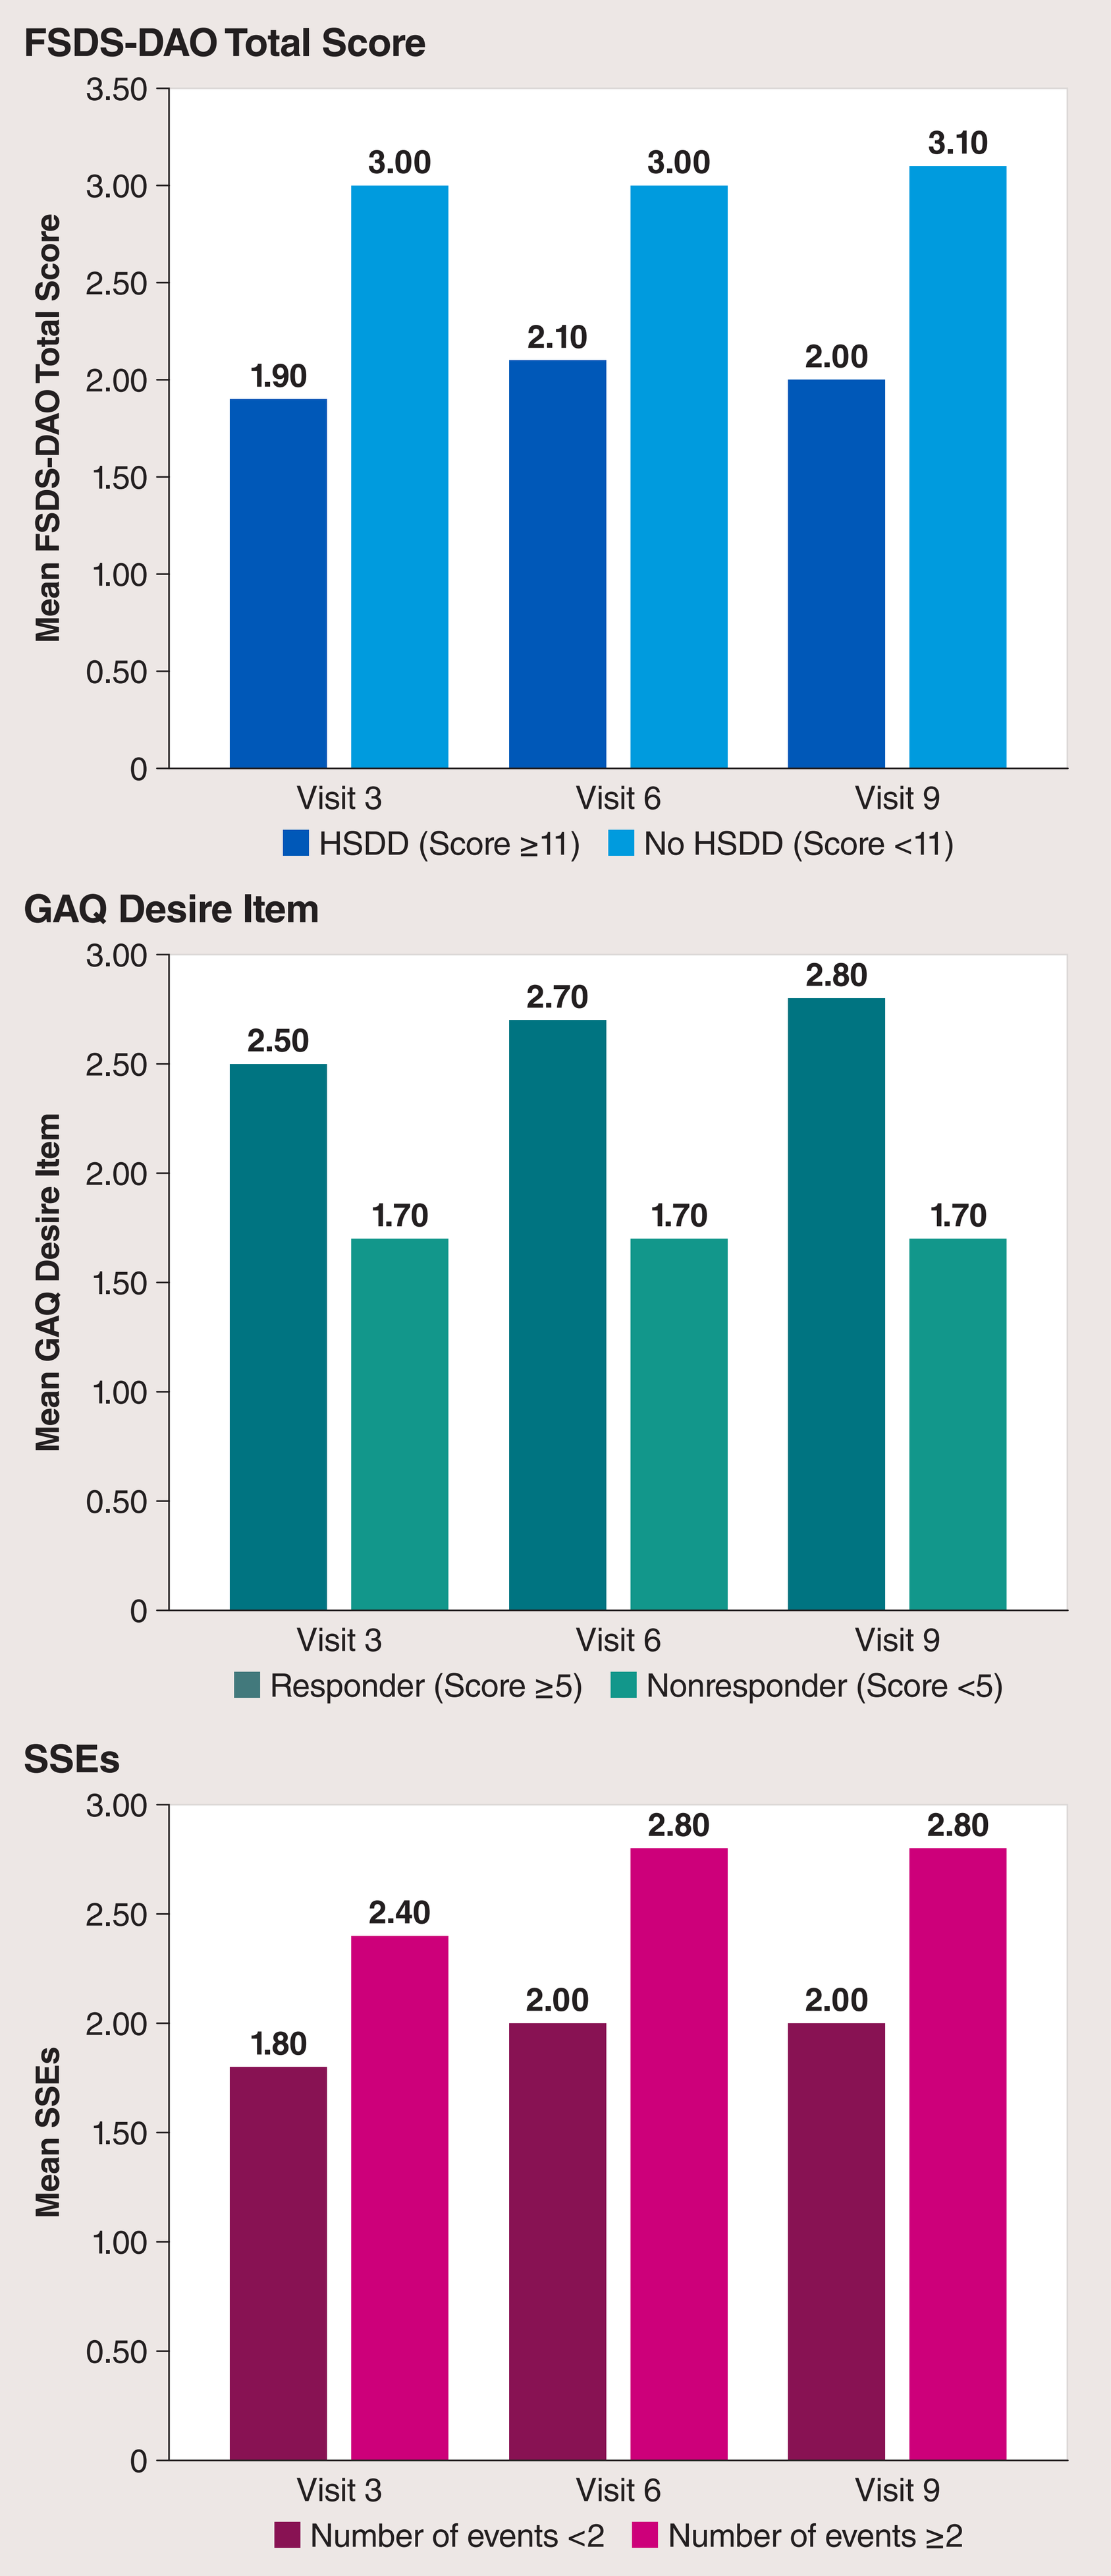

FSDS-DAO Total Score
Mean FSDS-DAO Total Score
3.50
3.00
2.50
2.00
1.50
1.00
0.50
0
1.90
3.00
2.10
3.00
2.00
3.10
Visit 3
Visit 6
Visit 9
HSDD (Score ≥11)
No HSDD (Score <11)
GAQ Desire Item
Mean GAQ Desire Item
2.50
1.70
2.70
1.70
2.80
1.70
Responder (Score ≥5)
Nonresponder (Score <5)
SSEs
Mean SSEs
1.80
2.40
2.00
2.80
2.00
2.80
Number of events <2
Number of events ≥2

FSDS-DAO, Female Sexual Distress Scale-Desire/Arousal/Orgasm; GAQ, General Assessment Questions; HSDD, hypoactive sexual desire disorder; SSEs, sexually satisfying events.

Ability to Detect Change and Interpretation Thresholds

- Responders consistently reported higher (less severe) EDQ scores relative to nonresponders for all 3 responder criteria (all *P*<.0001), providing support for the ability of the EDQ to detect change among participants known to have experienced a change in clinical status. Mean change for responders ranged between 1.1 and 1.4 for the monthly recall and between 0.6 and 1.1 for the 24-hour recall version of the EDQ
- Receiver-operating-characteristic analysis shows the probability of detecting a true signal (sensitivity) for the EDQ. Subjects were grouped as responders and nonresponders based on their response to the GAQ desire item at visit 9 (≥5 is a responder, <5 is nonresponder). The area under the curve for the monthly recall version was 0.85, and for the 24-hour recall version was 0.78, suggesting both versions of the EDQ provide good discrimination between responders and nonresponders as defined by the GAQ desire item
- An improvement of 0.6 maximized the sensitivity (74%) and specificity (86%) for the monthly recall version, and an improvement of 0.5 maximized the sensitivity (53%) and specificity (89%) for the 24-hour recall version, supporting a preliminary responder definition of between 0.5 and 0.6. The sensitivity was decreased for the 24-hour recall version relative to the monthly recall version

Conclusion

The EDQ measures different elements of desire, which along with distress, is a defining clinical issue for a diagnosis of and evaluation of treatments for HSDD. Based on this initial psychometric analysis, there is evidence supporting the use of the EDQ in clinical trials evaluating women with HSDD. The results demonstrate that the factor structure of the EDQ is confirmed in this sample of women with HSDD, and the internal consistency reliability, construct validity, and ability to detect change are adequate for use in this patient population. Additional research is needed to further evaluate the test-retest reliability of the EDQ and inform the development of interpretation thresholds. Results from previous qualitative interviews with women with HSDD indicated that given the infrequency of their sexual activity and the lack of day-to-day variations experienced with sexual desire, they objected to completing a daily diary. Given these consistent sentiments and the high rate of missing data for the daily-diary version of the EDQ, the monthly recall version of the EDQ may be more appropriate for this population.

References **1.** Shifren JL, Monz BU, Russo PA, Segreti A, Johannes CB. Sexual problems and distress in United States women: prevalence and correlates. *Obstet Gynecol.* 2008;112(5):970–978. **2.** Molinoff PB, Shadiack AM, Earle D, Diamond LE, Quon CY. PT-141: a melanocortin agonist for the treatment of sexual dysfunction. *Ann N Y Acad Sci.* 2003;994:96–102. **3.** Wikberg JE, Muceniece R, Mandrika I, et al. New aspects on the melanocortins and their receptors. *Pharmacol Res.* 2000;42(5):393–420. **4.** Cronbach L. Coefficient alpha and the internal structure of tests. *Psychometrika.* 1951;16:294–334. **5.** Nunnally JC, Bernstein IH. *Psychometric Theory. 3rd ed.* New York: McGraw-Hill; 1994. **6.** Hays RD, Revicki DA. Assessing quality of life in clinical trials. In: Fayers P, Hays RD, eds. *Reliability and Validity, Including Responsiveness. 2nd ed.* New York, NY: Oxford University Press; 2005. **7.** Hinkle D, Wiersma W, Jurs S. *Applied Statistics for the Behavioral Sciences.* Boston: Houghton-Mifflin; 1988.

Support This study was funded by Palatin Technologies, Inc. Editorial assistance was provided by The Curry Rockefeller Group, LLC, which was funded by Palatin Technologies, Inc.

Disclosures **DAR** has received research support or consulting fees from Palatin Technologies, Inc. **SA** has received research support or consulting fees from Abbott, Allergan, Bayer, Eli Lilly, Palatin Technologies, Inc., Promescent, Sprout Pharmaceuticals, and Trimel Pharmaceuticals, Inc. **LRD** has received research support or consulting fees from Palatin Technologies, Inc. **HW** has received research support or consulting fees from Palatin Technologies, Inc. **RJ** and **JL** are employees and stockholders of Palatin Technologies, Inc.