Presented at:
ARVO 2021
Association for Research in Vision and Ophthalmology
Virtual Annual Meeting
May 1–7, 2021

Protective Effects of 2 Melanocortin Agonists Delivered by Intravitreal Injection in Mouse Models of Retinopathy

John Dodd,[1] Marie Makhlina,[1] Wei H. Yang,[1] Andrew W. Taylor,[2] Tat Fong Ng,[2] Carl Spana[1]

[1]Palatin Technologies, Inc, Cranbury, NJ, USA; [2]Boston University School of Medicine, Boston, MA

Background

- Melanocortins have a wide range of activities, including inhibition of leukocyte activation, inhibition of inflammation, and protection of tissues[1-3]
- The melanocortin system plays a key role in promoting resolution of the inflammatory process[3]
- Melanocortin agonists have demonstrated anti-inflammatory effects in various experimental models of inflammatory disease[4-6]
- Targeting this pathway may protect against ocular disease
 - PL8177 is a potent and selective melanocortin 1 receptor agonist that has been granted orphan drug status by the US Food and Drug Administration for the treatment of noninfectious intermediate, posterior, pan and chronic anterior uveitis
 - PL8331 and PL9654 are melanocortin 1 receptor agonists being evaluated preclinically for the treatment of various ocular diseases

Objective

- To investigate the effects of two melanocortin receptor pan-agonists, PL8331 and PL9654, administered via intravitreal injection in mouse models of ocular disease

Methods

Study 1

- In study 1, the effect of monthly administered PL8331 was investigated in a mouse model of diabetic retinopathy using C57BL/6 mice (N=16; **Figure 1**)

Figure 1. Study 1 Design

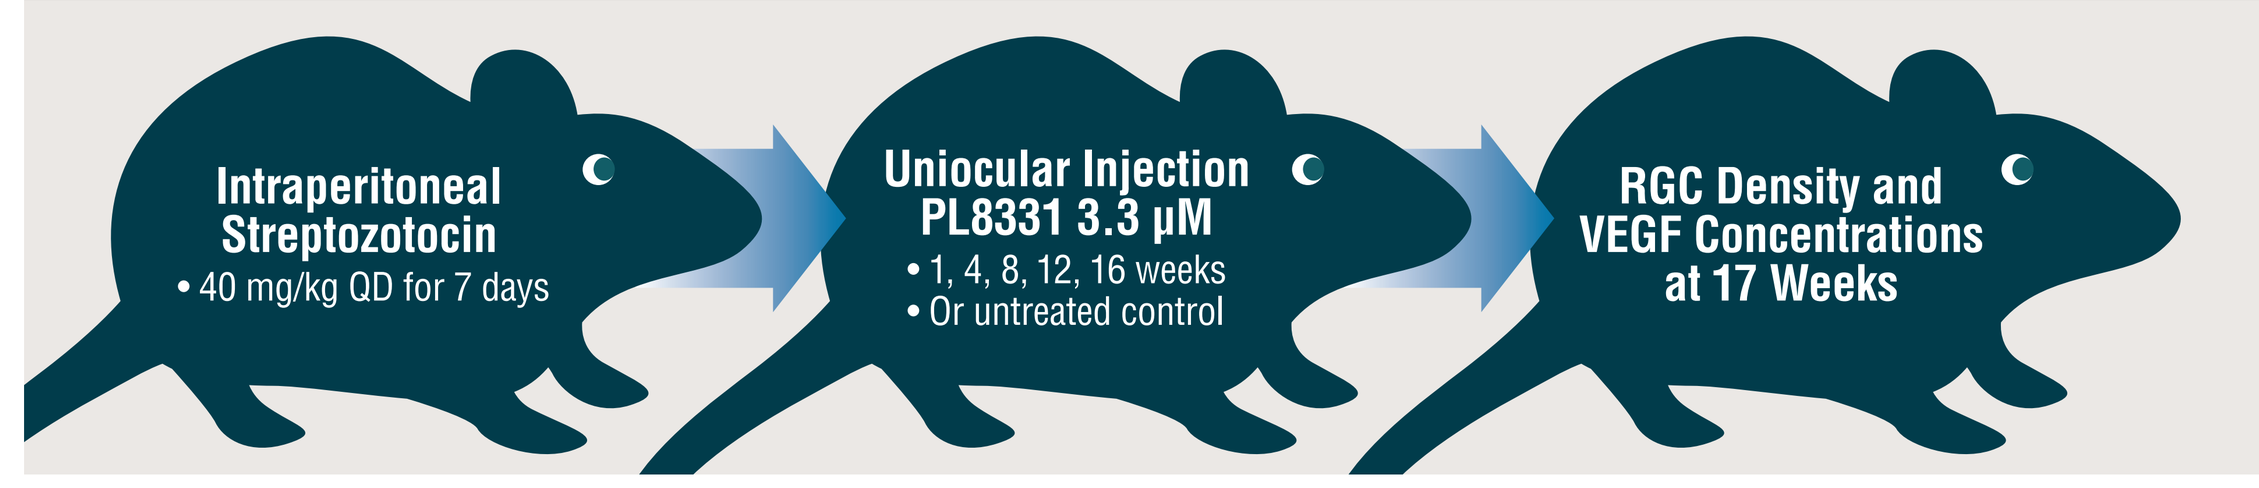

Intraperitoneal Streptozotocin
• 40 mg/kg QD for 7 days
Uniocular Injection PL8331 3.3 µM
• 1, 4, 8, 12, 16 weeks
• Or untreated control
RGC Density and VEGF Concentrations at 17 Weeks

QD, daily; RGC, retinal ganglion cell; VEGF, vascular endothelial growth factor.

Study 2

- In study 2, the effects of monthly administered PL8331 or PL9654 were investigated in a mouse model of age-related macular degeneration using C57BL/6 mice (N=48; **Figure 2**)

Figure 2. Study 2 Design

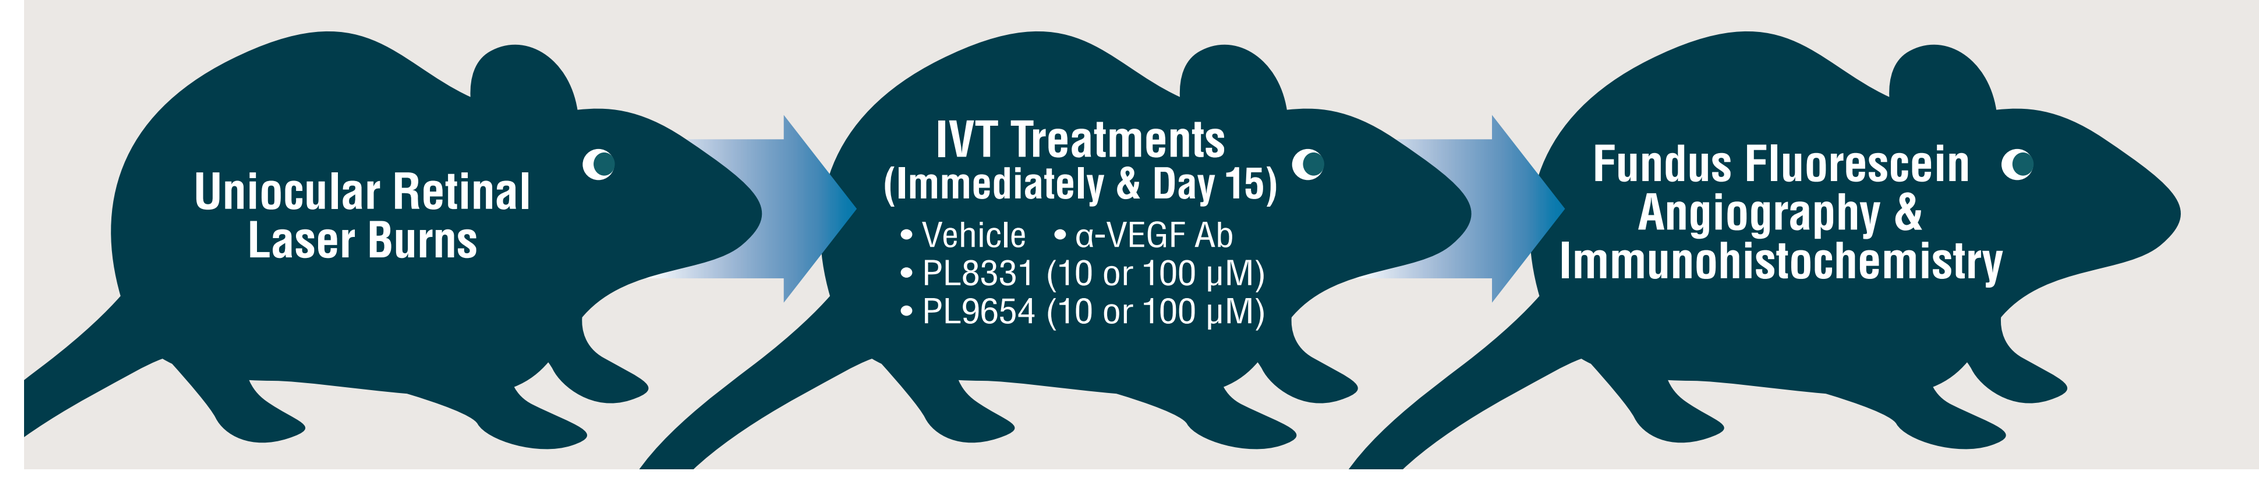

Uniocular Retinal Laser Burns
IVT Treatments (Immediately & Day 15)
• Vehicle • α-VEGF Ab
• PL8331 (10 or 100 µM)
• PL9654 (10 or 100 µM)
Fundus Fluorescein Angiography & Immunohistochemistry

Ab, antibody; IVT, intravitreal; VEGF, vascular endothelial growth factor.

Results

Study 1

Figure 3. Retinal Sections from Healthy, PL8331-Treated Diabetic Mice, and Untreated Diabetic Mice

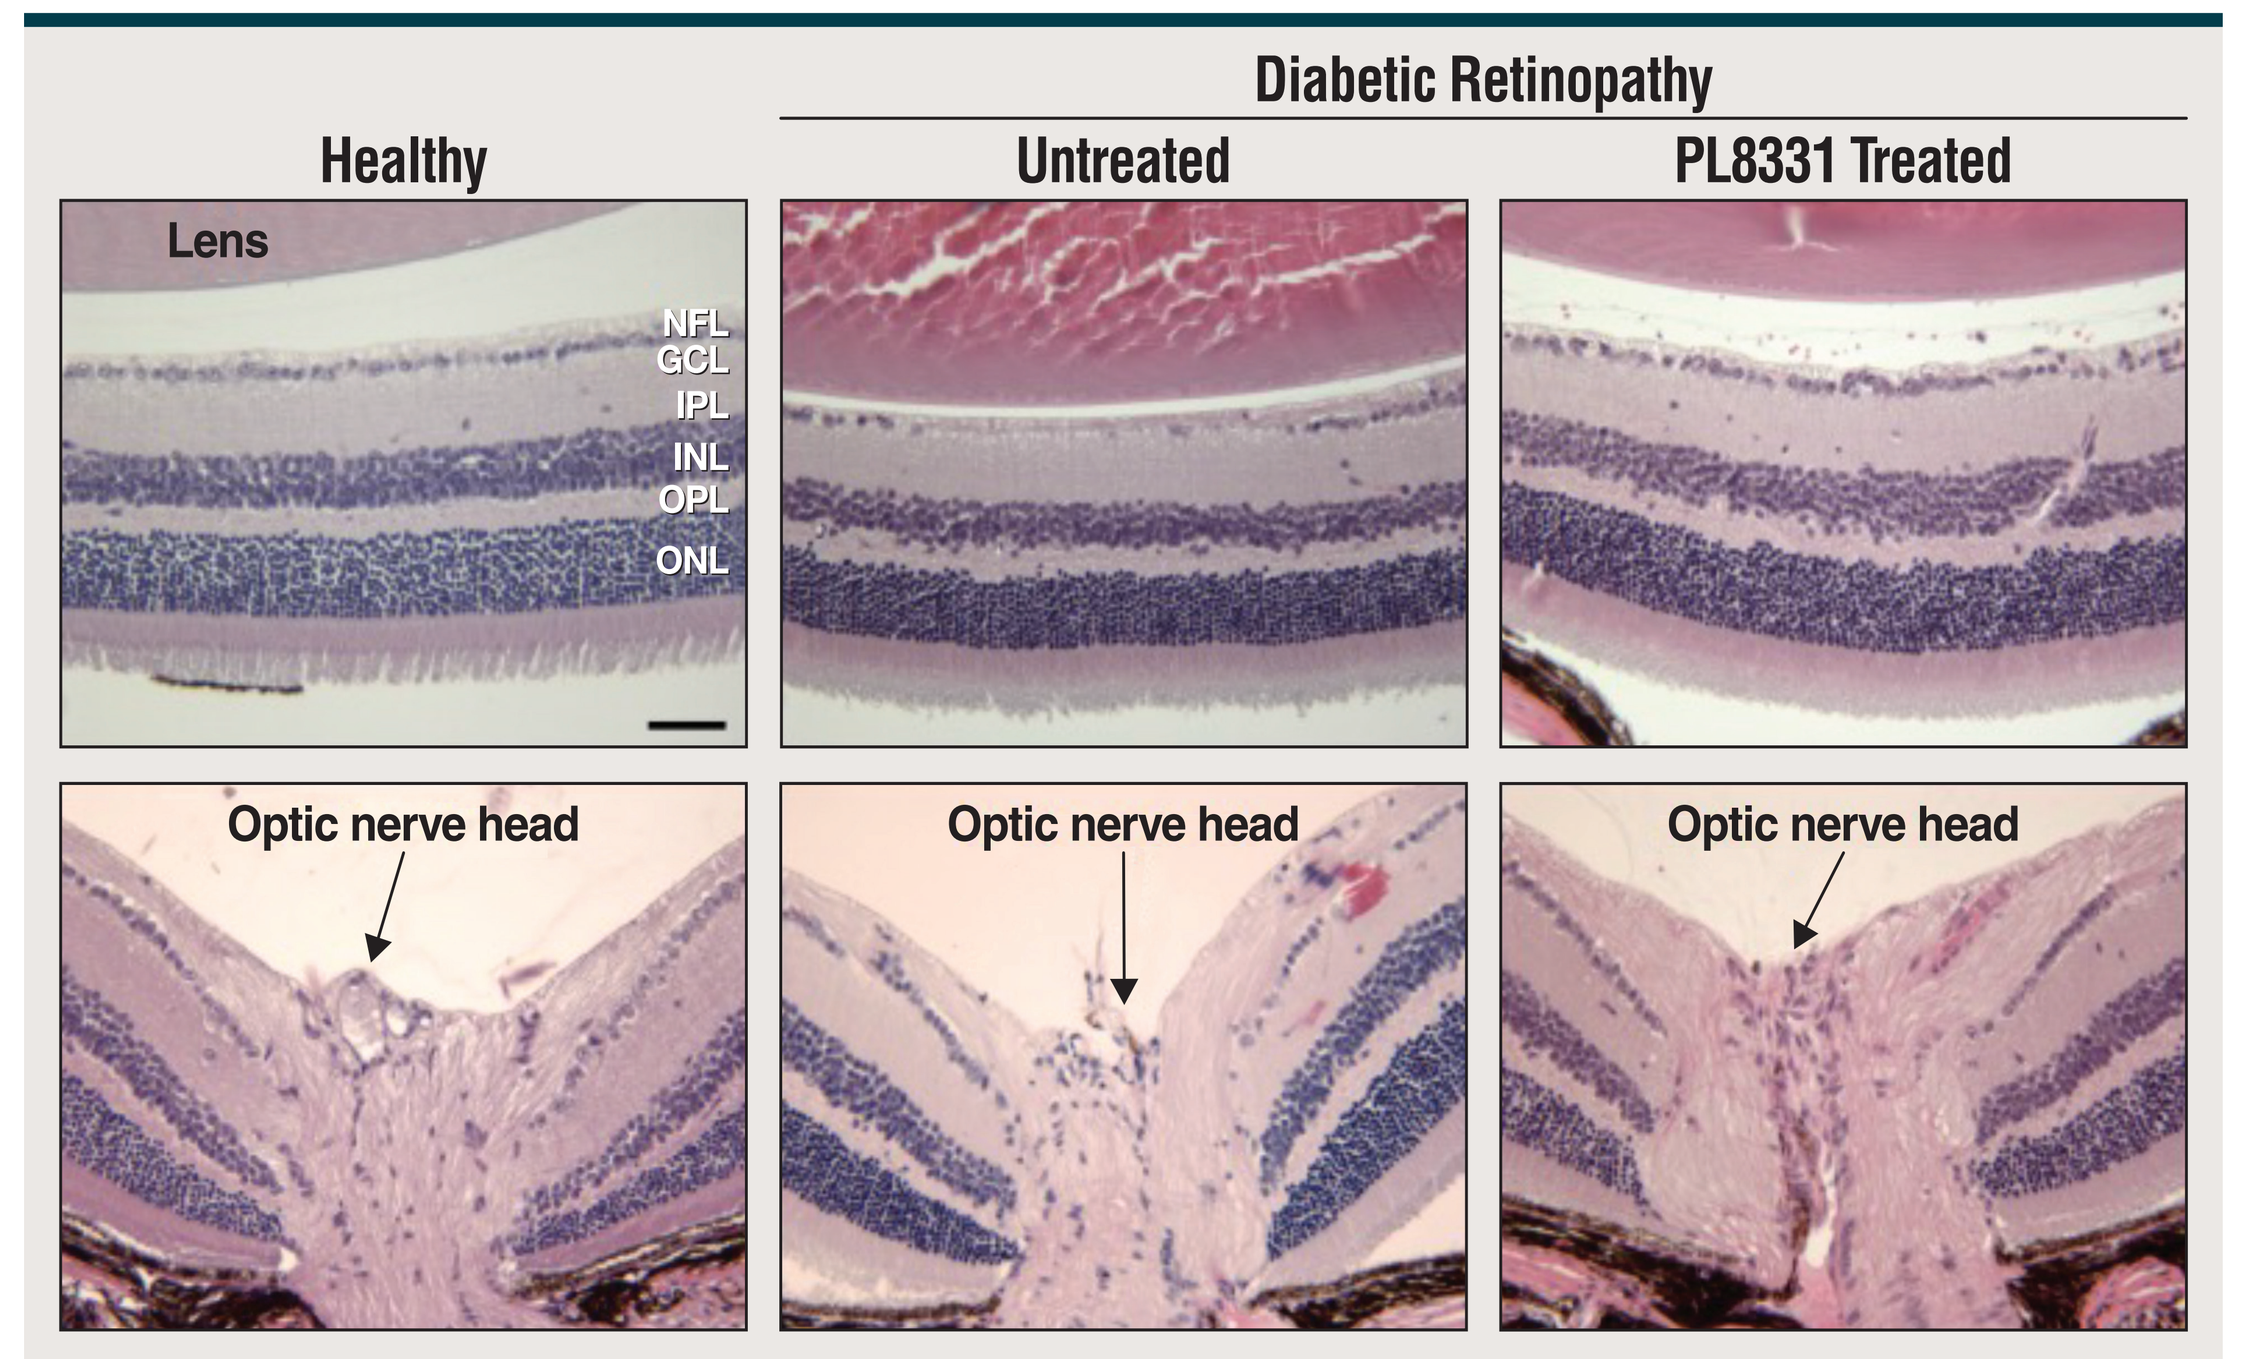

Diabetic Retinopathy
Healthy
Untreated
PL8331 Treated
Lens
NFL
GCL
IPL
INL
OPL
ONL
Optic nerve head

NFL, nerve fiber layer; GCL, ganglion cells layer; IPL, inner plexiform layer; INL, inner nuclear layer; OPL, outer plexiform layer; ONL, outer nuclear layer.

Sections were hematoxylin and eosin (H&E) stained and labeled for the layers of the retina. Scale bar=50µm.

- Untreated diabetic mice had thinner retinas compared with healthy and PL8331-treated mice as well as optic nerve cupping (**Figure 3**)
- There was significant survival of retinal ganglion cells (RGC) in the diabetic eyes of mice treated with PL8331 compared with untreated in the diabetic mouse model

Figure 4. VEGF Expression in Healthy and Diabetic-Treated and -Untreated Mice

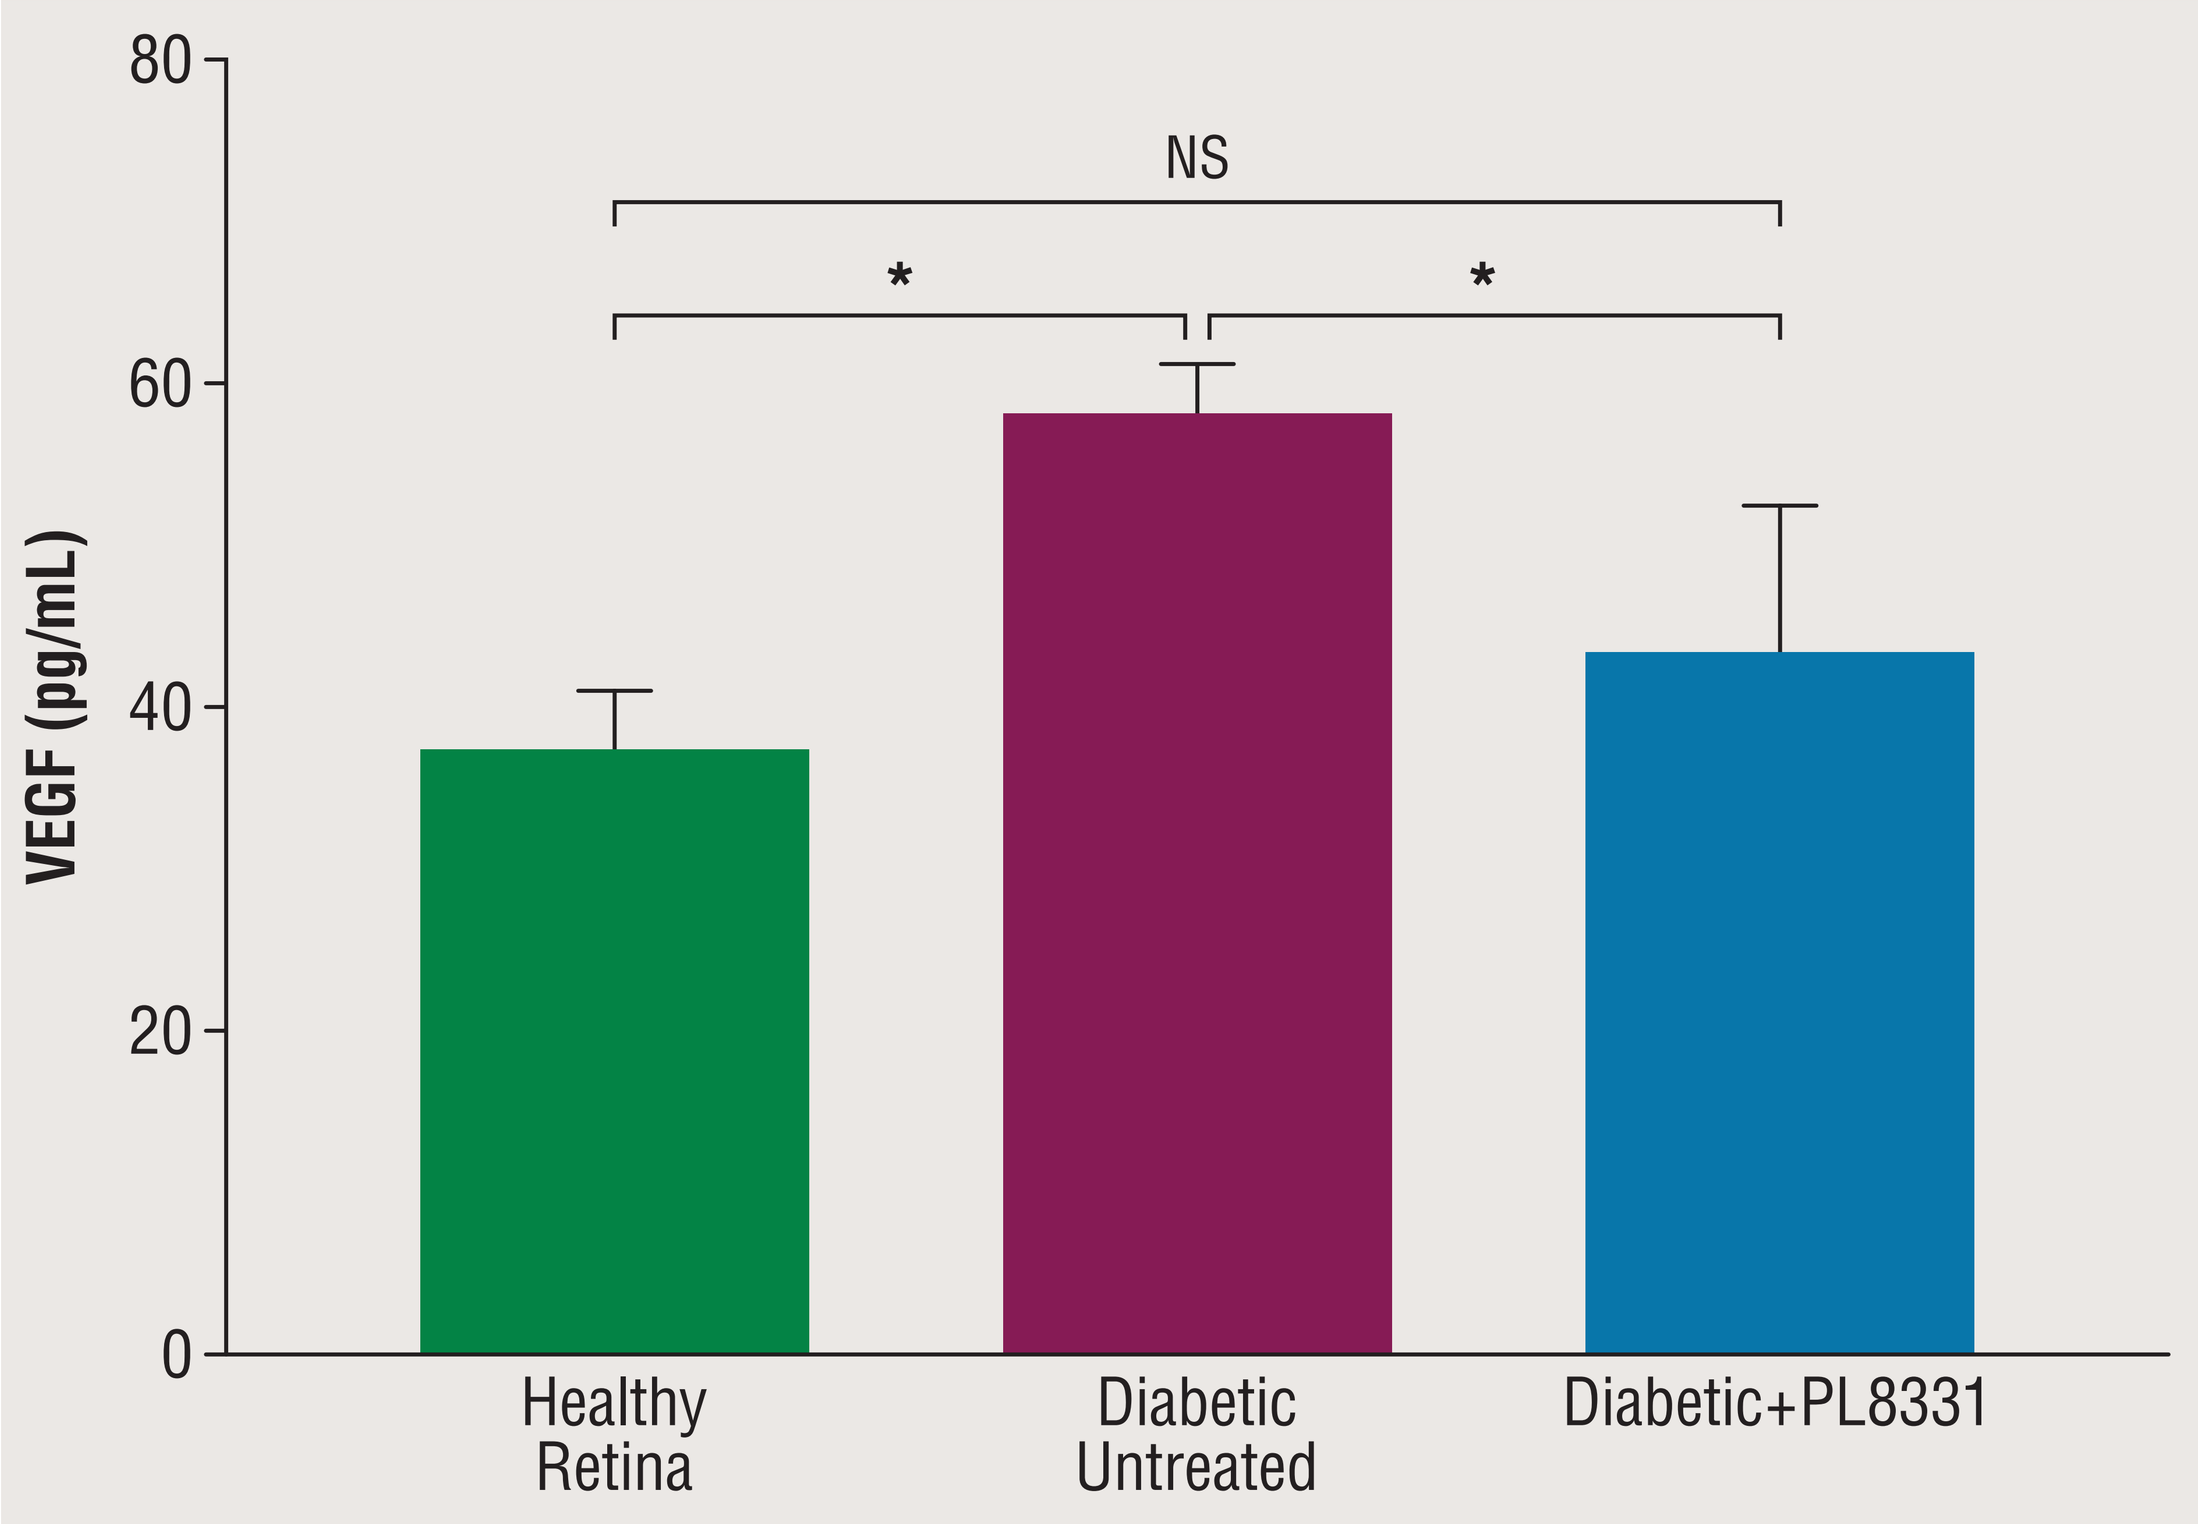

VEGF (pg/mL)
80
60
40
20
0
NS
*
*
Healthy Retina
Diabetic Untreated
Diabetic+PL8331

NS, no significant difference; VEGF, vascular endothelial growth factor.

*$P \leq 0.005$. VEGF was measured by enzyme-linked immunosorbent assay (ELISA).

- There was a significant increase in VEGF expression in the neuroretinas of diabetic mice compared with nondiabetic mice, but not in the retinas of diabetic mice treated with PL8331 (**Figure 4**)

Figure 5. Retinal Ganglion Cell Density in Healthy Control, Diabetic-Treated and -Untreated Mice

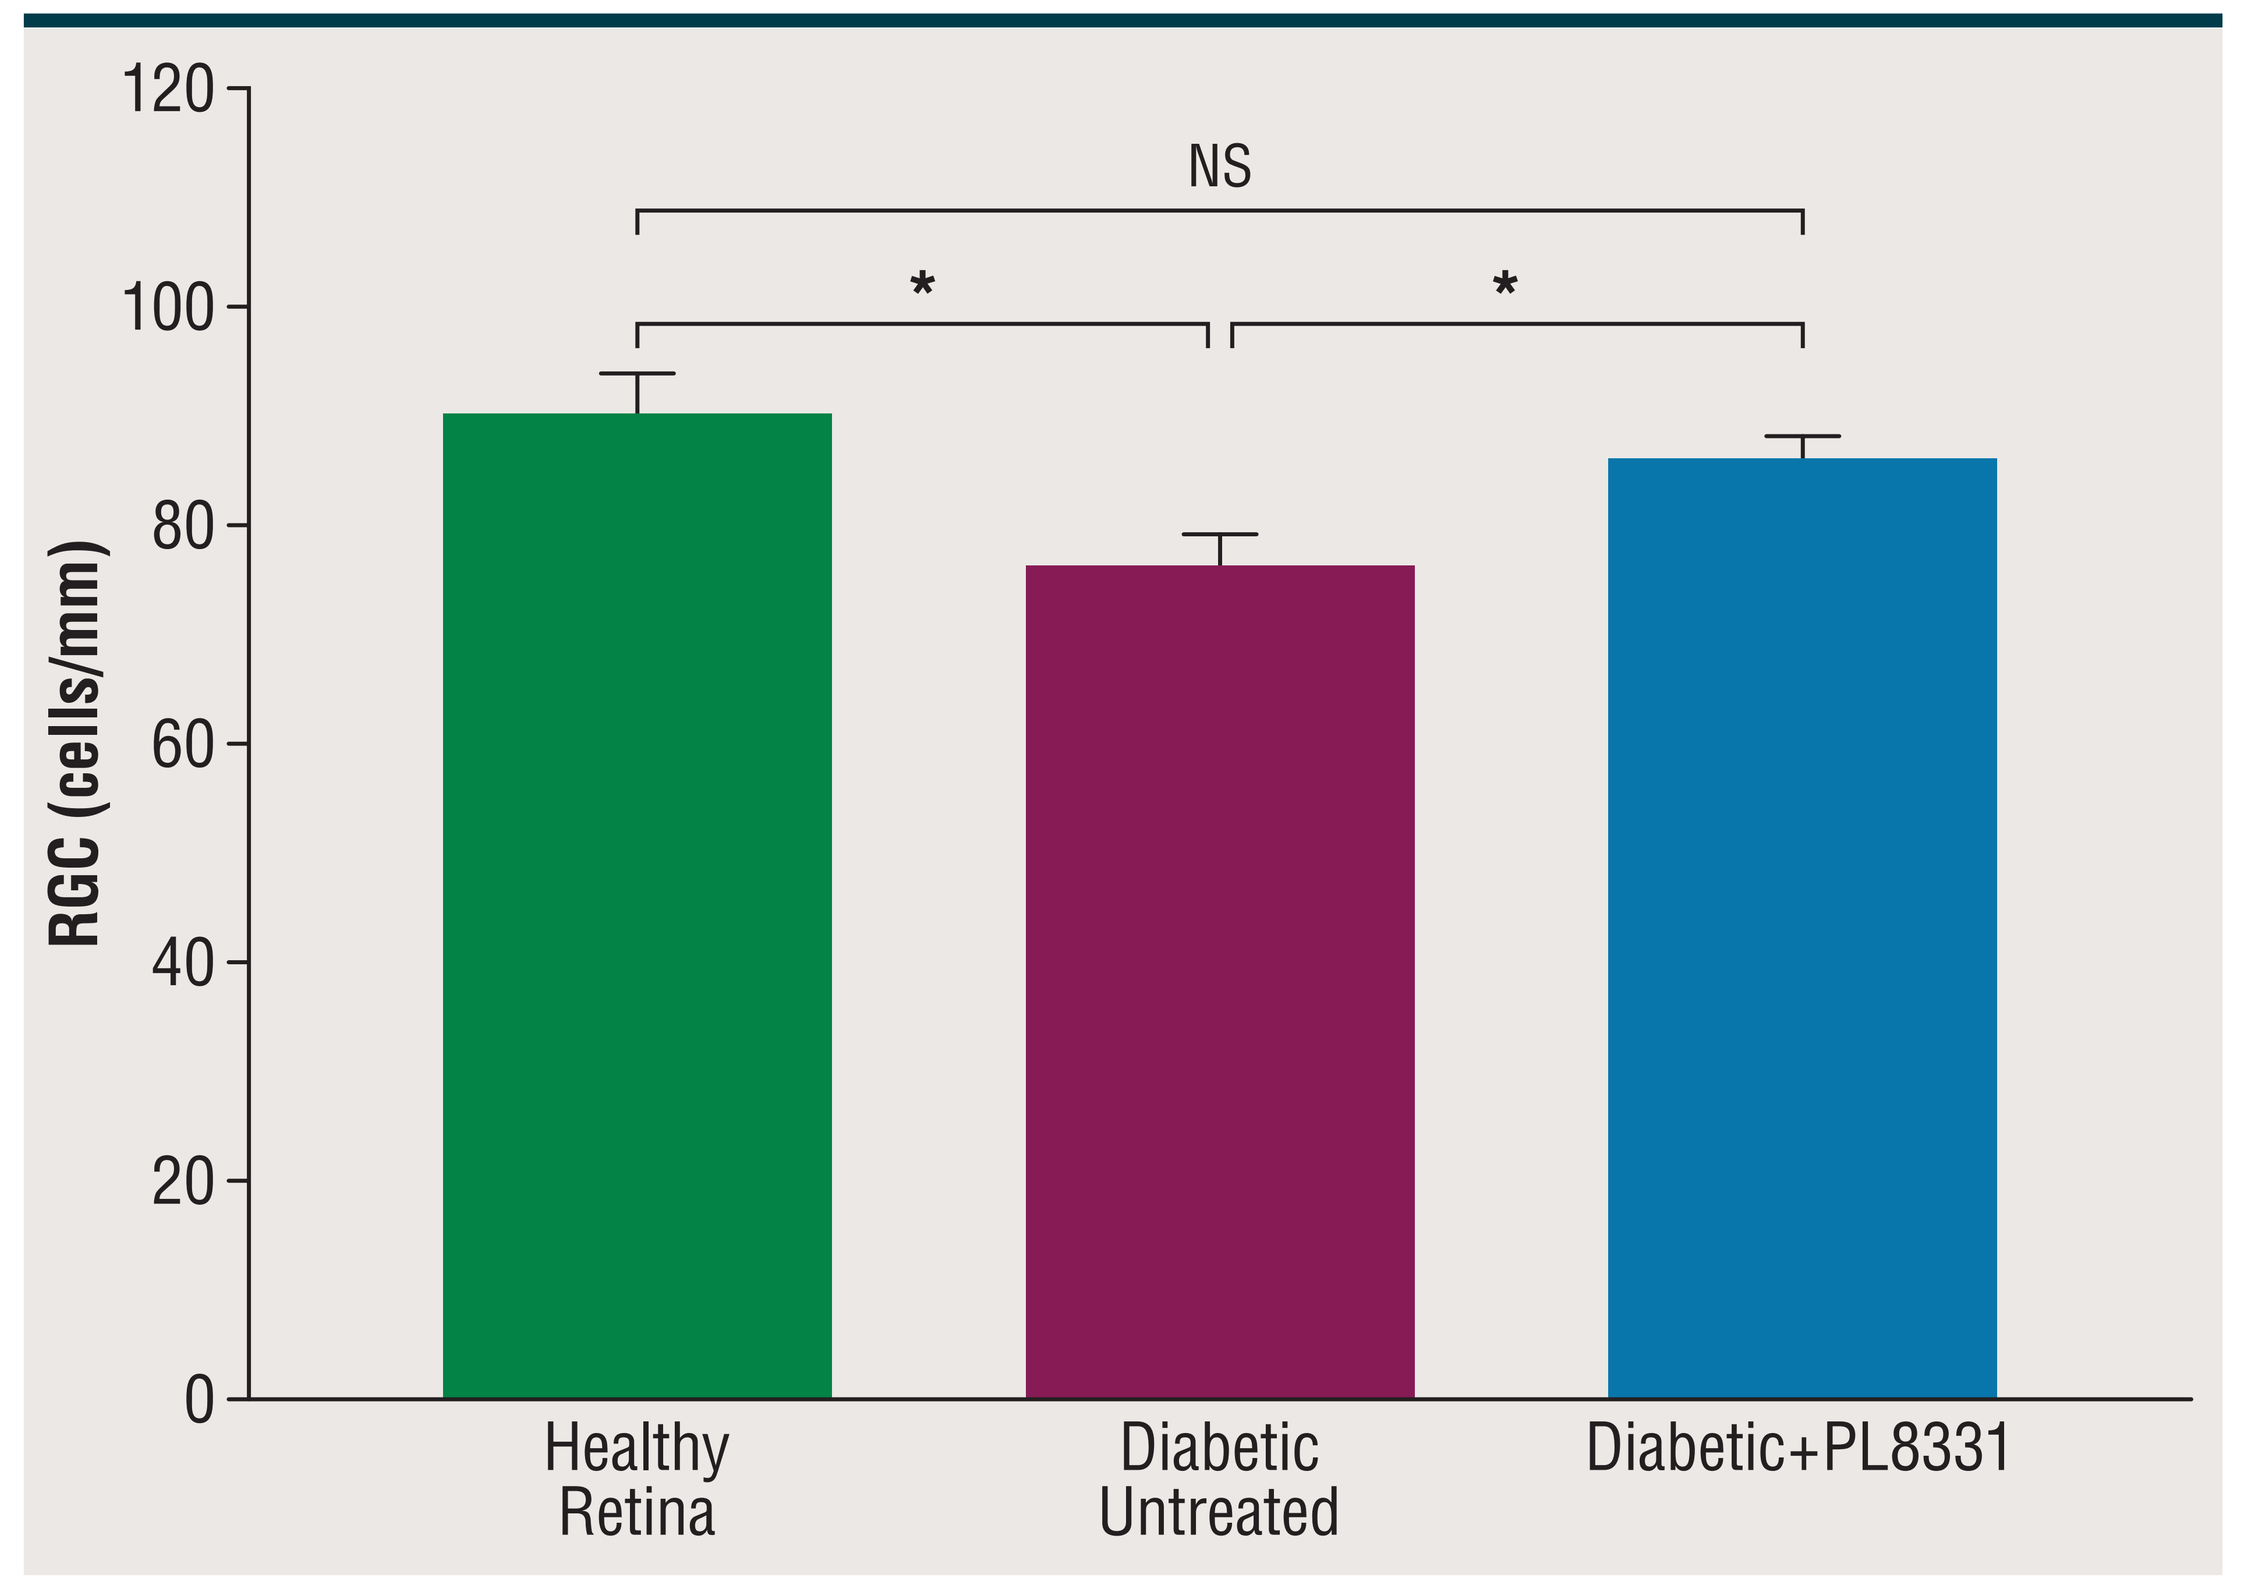

RGC (cells/mm)
120
100
80
60
40
20
0
NS
*
*
Healthy Retina
Diabetic Untreated
Diabetic+PL8331

NS, no significant difference; RGC, retinal ganglion cells.

*$P \leq 0.005$. RGCs were counted in one section of each eye per mm length. Data presented are mean±SEM (n=8 eyes) and statistically analyzed by one-way ANOVA with a Dunnett's multiple comparison test with $P \leq 0.05$ established as significant.

- RGC density was significantly lower in the retinas of diabetic untreated mice compared with healthy retinas or diabetic mice treated with PL8331 (**Figure 5**)
- RGC density was not significantly different between healthy control and PL8331-treated diabetic mice

Study 2

Figure 6. Areas of Leakage Measured Using Fundus Fluorescein Angiography

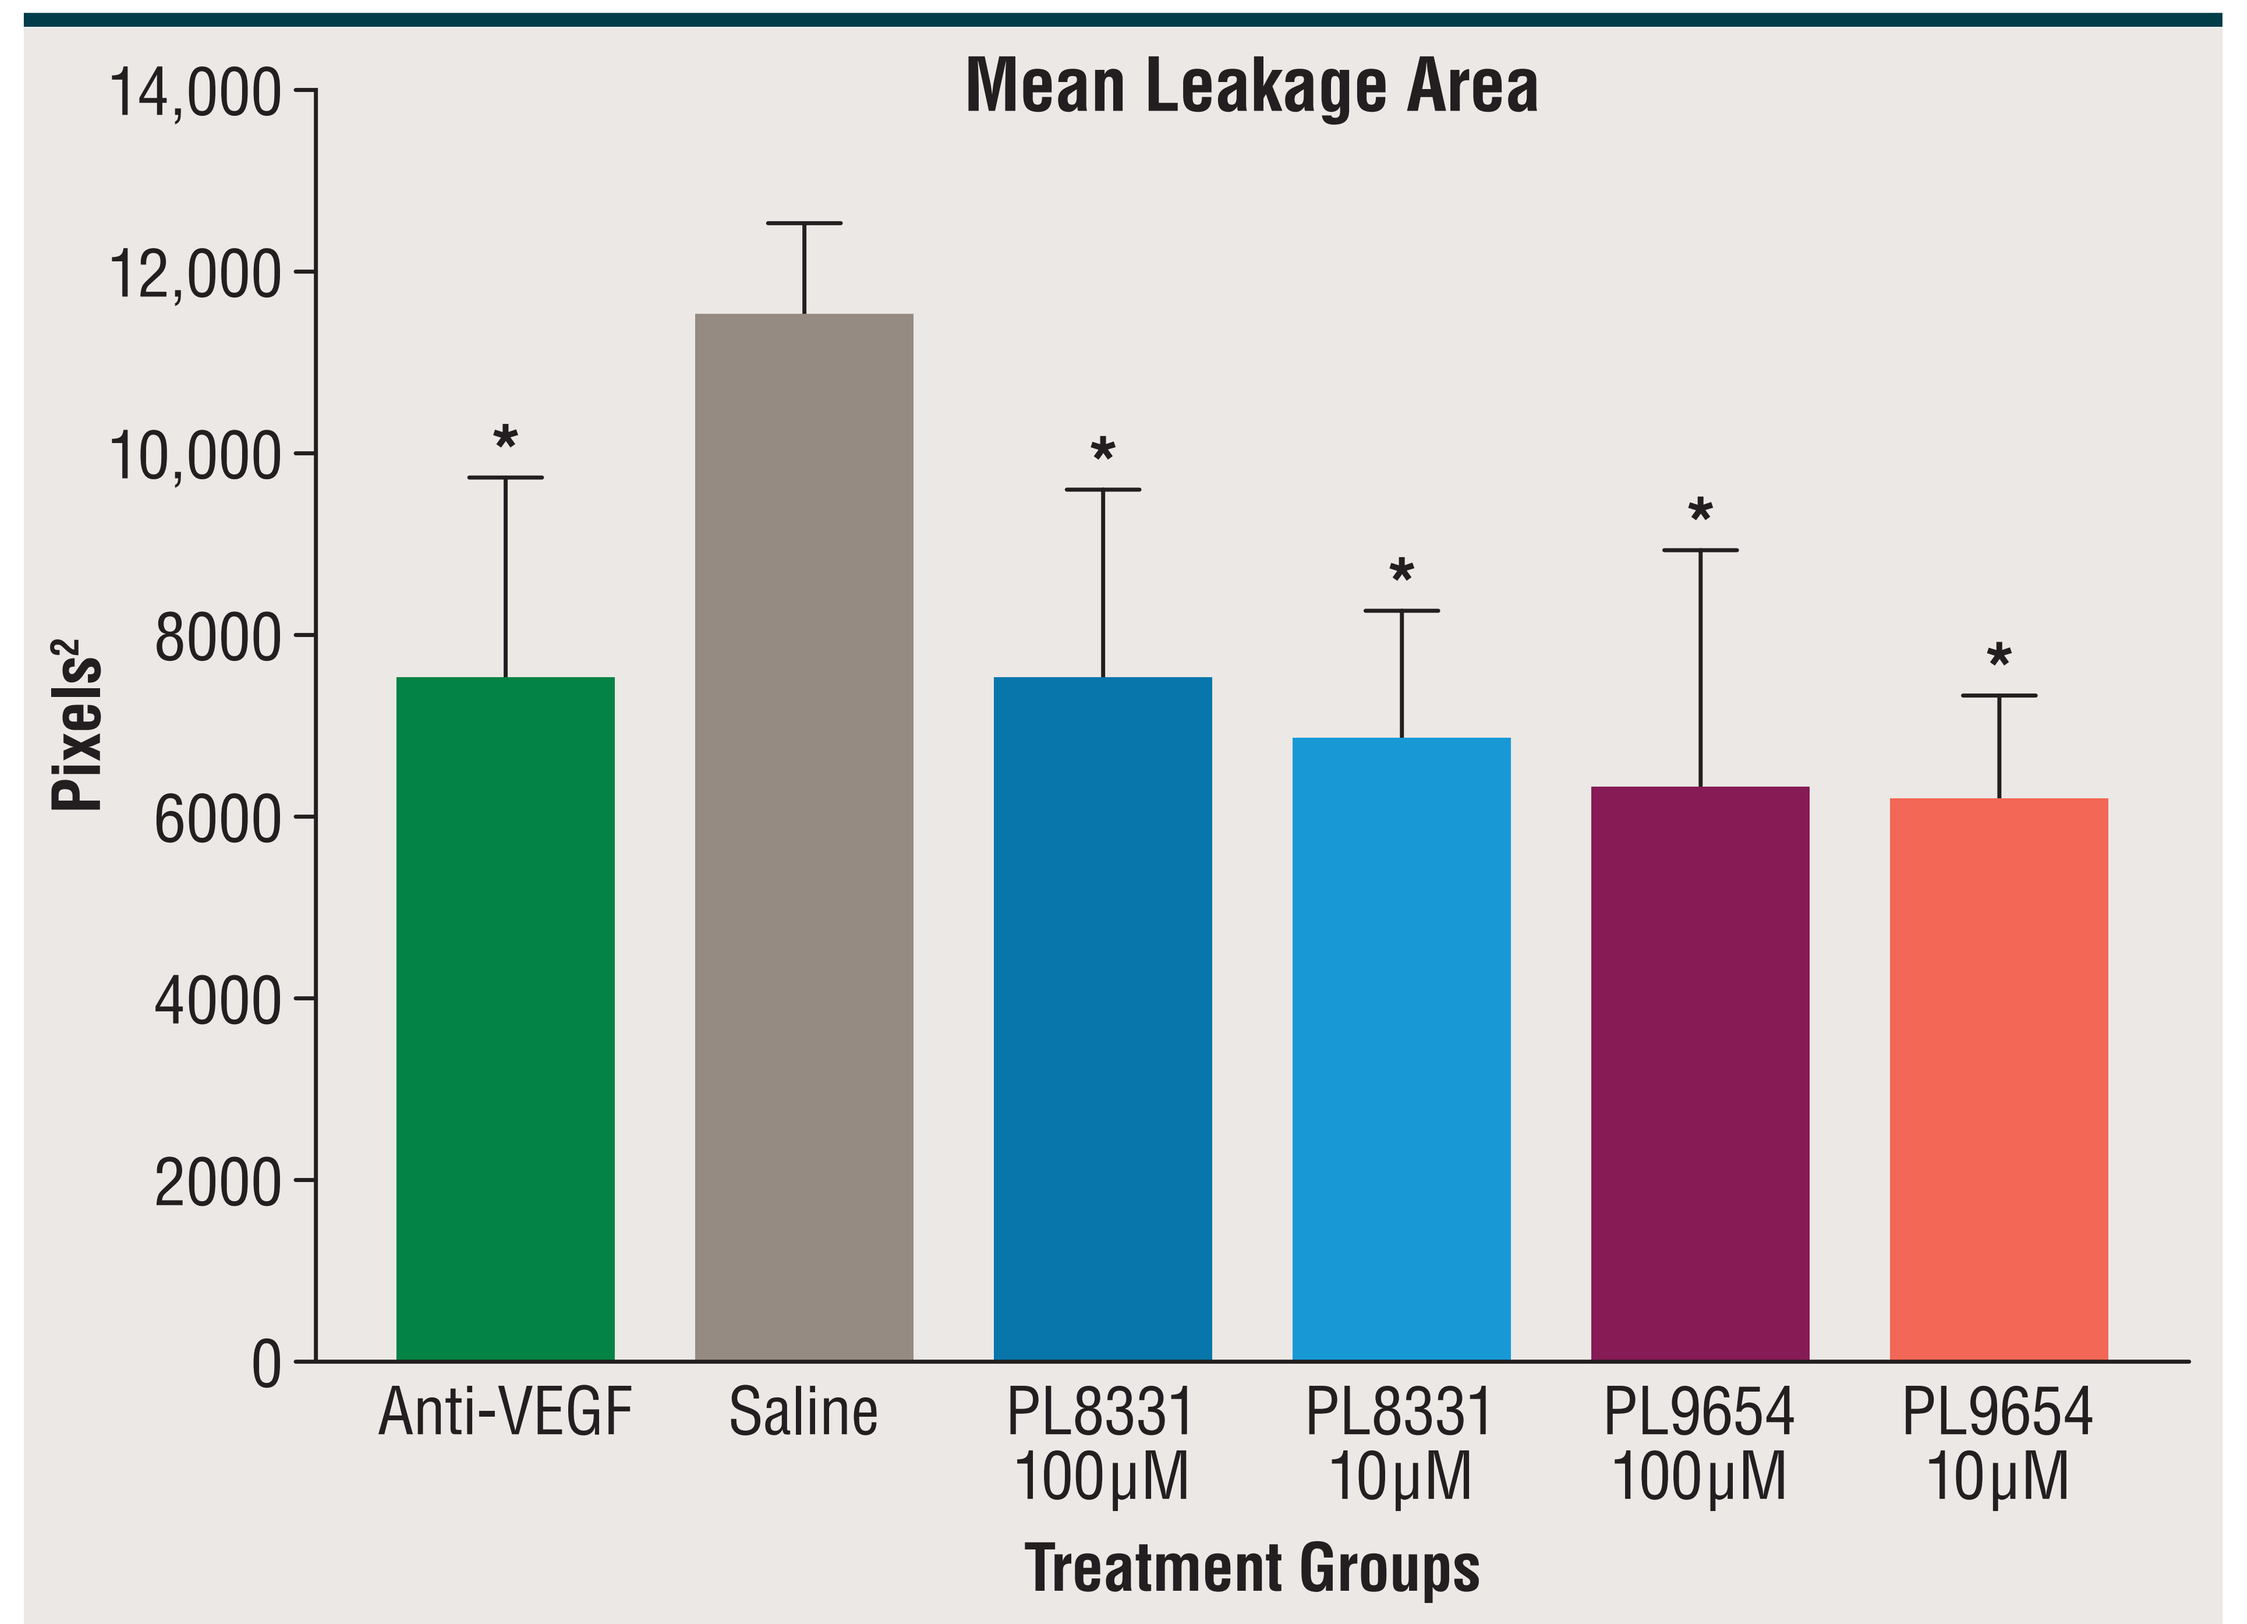

Mean Leakage Area
Pixels²
14,000
12,000
10,000
8000
6000
4000
2000
0
Anti-VEGF
Saline
PL8331 100µM
PL8331 10µM
PL9654 100µM
PL9654 10µM
Treatment Groups

VEGF, vascular endothelial growth factor.

*$P \leq 0.05$. Areas were outlined manually and measured using Image J (University of Wisconsin, Madison, WI, USA). The average area of leakage for each animal was used for group comparison using one-way ANOVA.

- Laser retina burns resulted in retinal leakage in saline-injected mice (**Figure 6**)
- Mice treated with anti-VEGF antibody, PL8331, or PL9654 had significantly reduced leakage compared with saline-treated animals ($P \leq 0.05$)

Figure 7. Mean Angiogenesis in Retinas of Mice Who Received Ocular Laser Burns

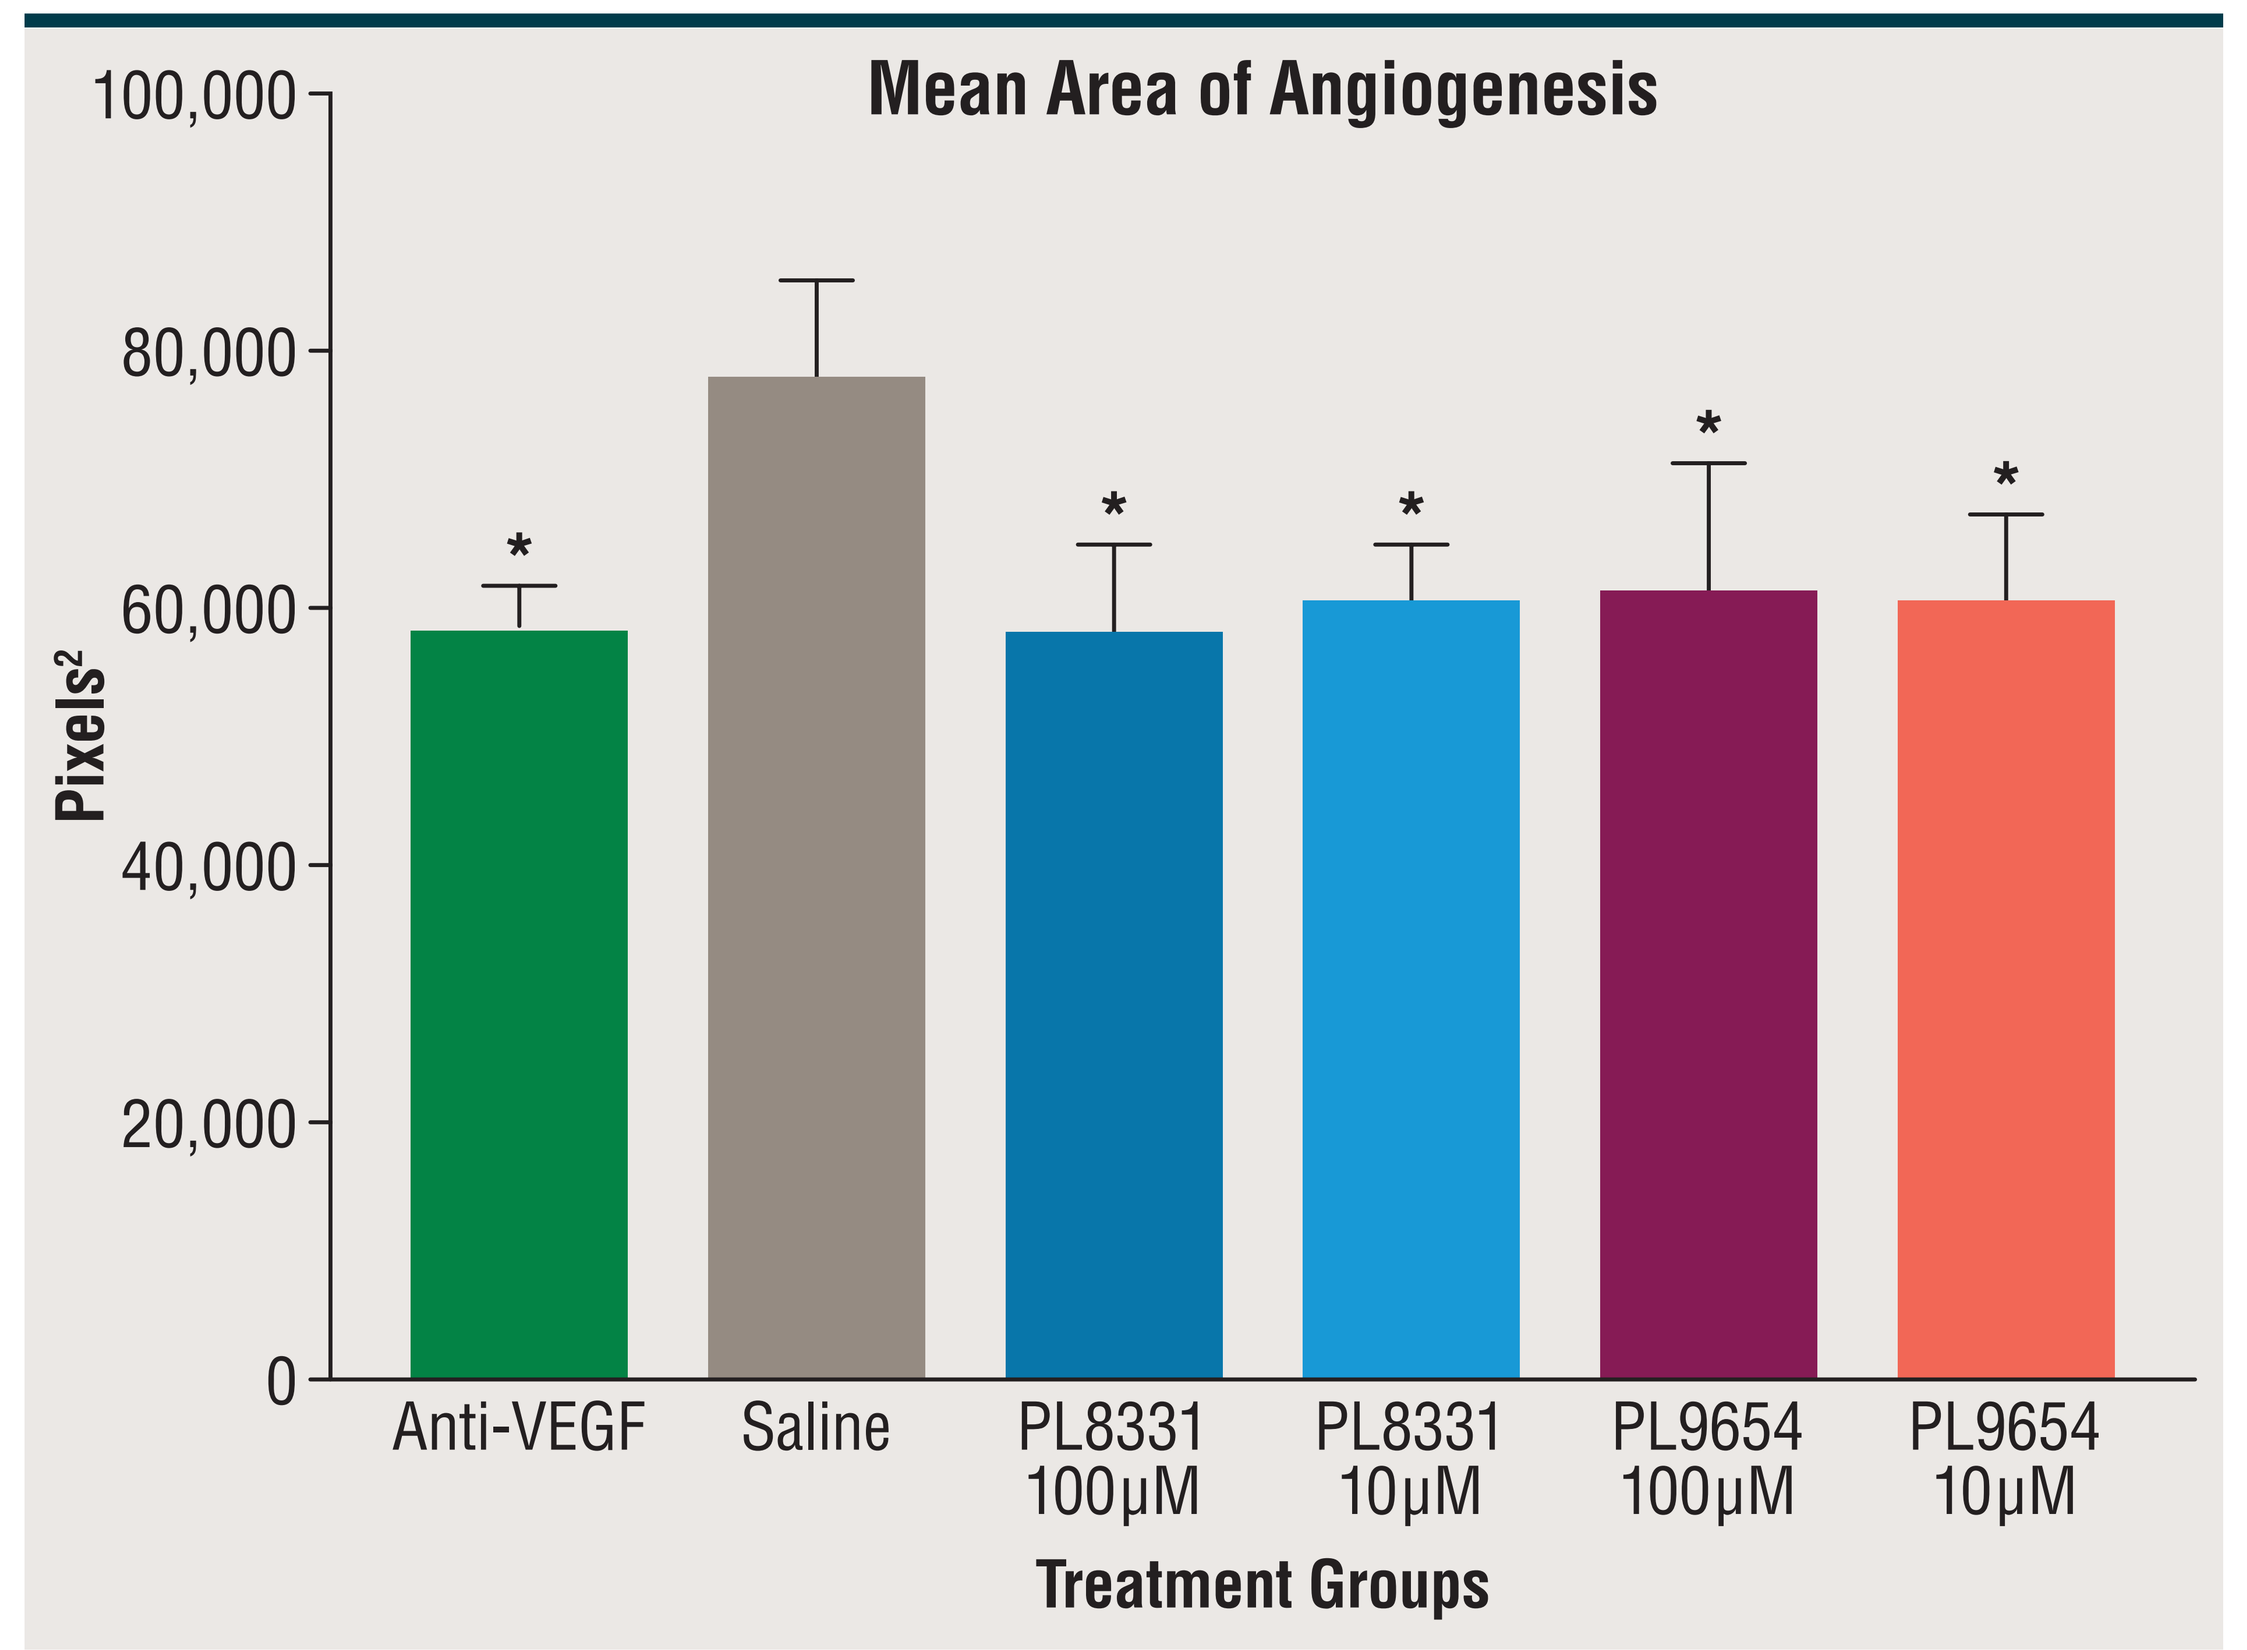

Mean Area of Angiogenesis
Pixels²
100,000
80,000
60,000
40,000
20,000
0
Anti-VEGF
Saline
PL8331 100µM
PL8331 10µM
PL9654 100µM
PL9654 10µM
Treatment Groups

VEGF, vascular endothelial growth factor.

*$P \leq 0.05$. Angiogenesis was determined by immunohistochemistry using isolectin B4 staining. Mean areas of staining were using for group comparison using one-way ANOVA.

- Laser retina burns resulted in increased angiogenesis in vehicle-injected mice (**Figure 7**)
- Mice treated with anti-VEGF antibody, PL8331, or PL9654 showed statistically significant reduction in angiogenesis

Figure 8. Mean Area of Fibrosis in Retinas of Mice Who Received Ocular Laser Burns

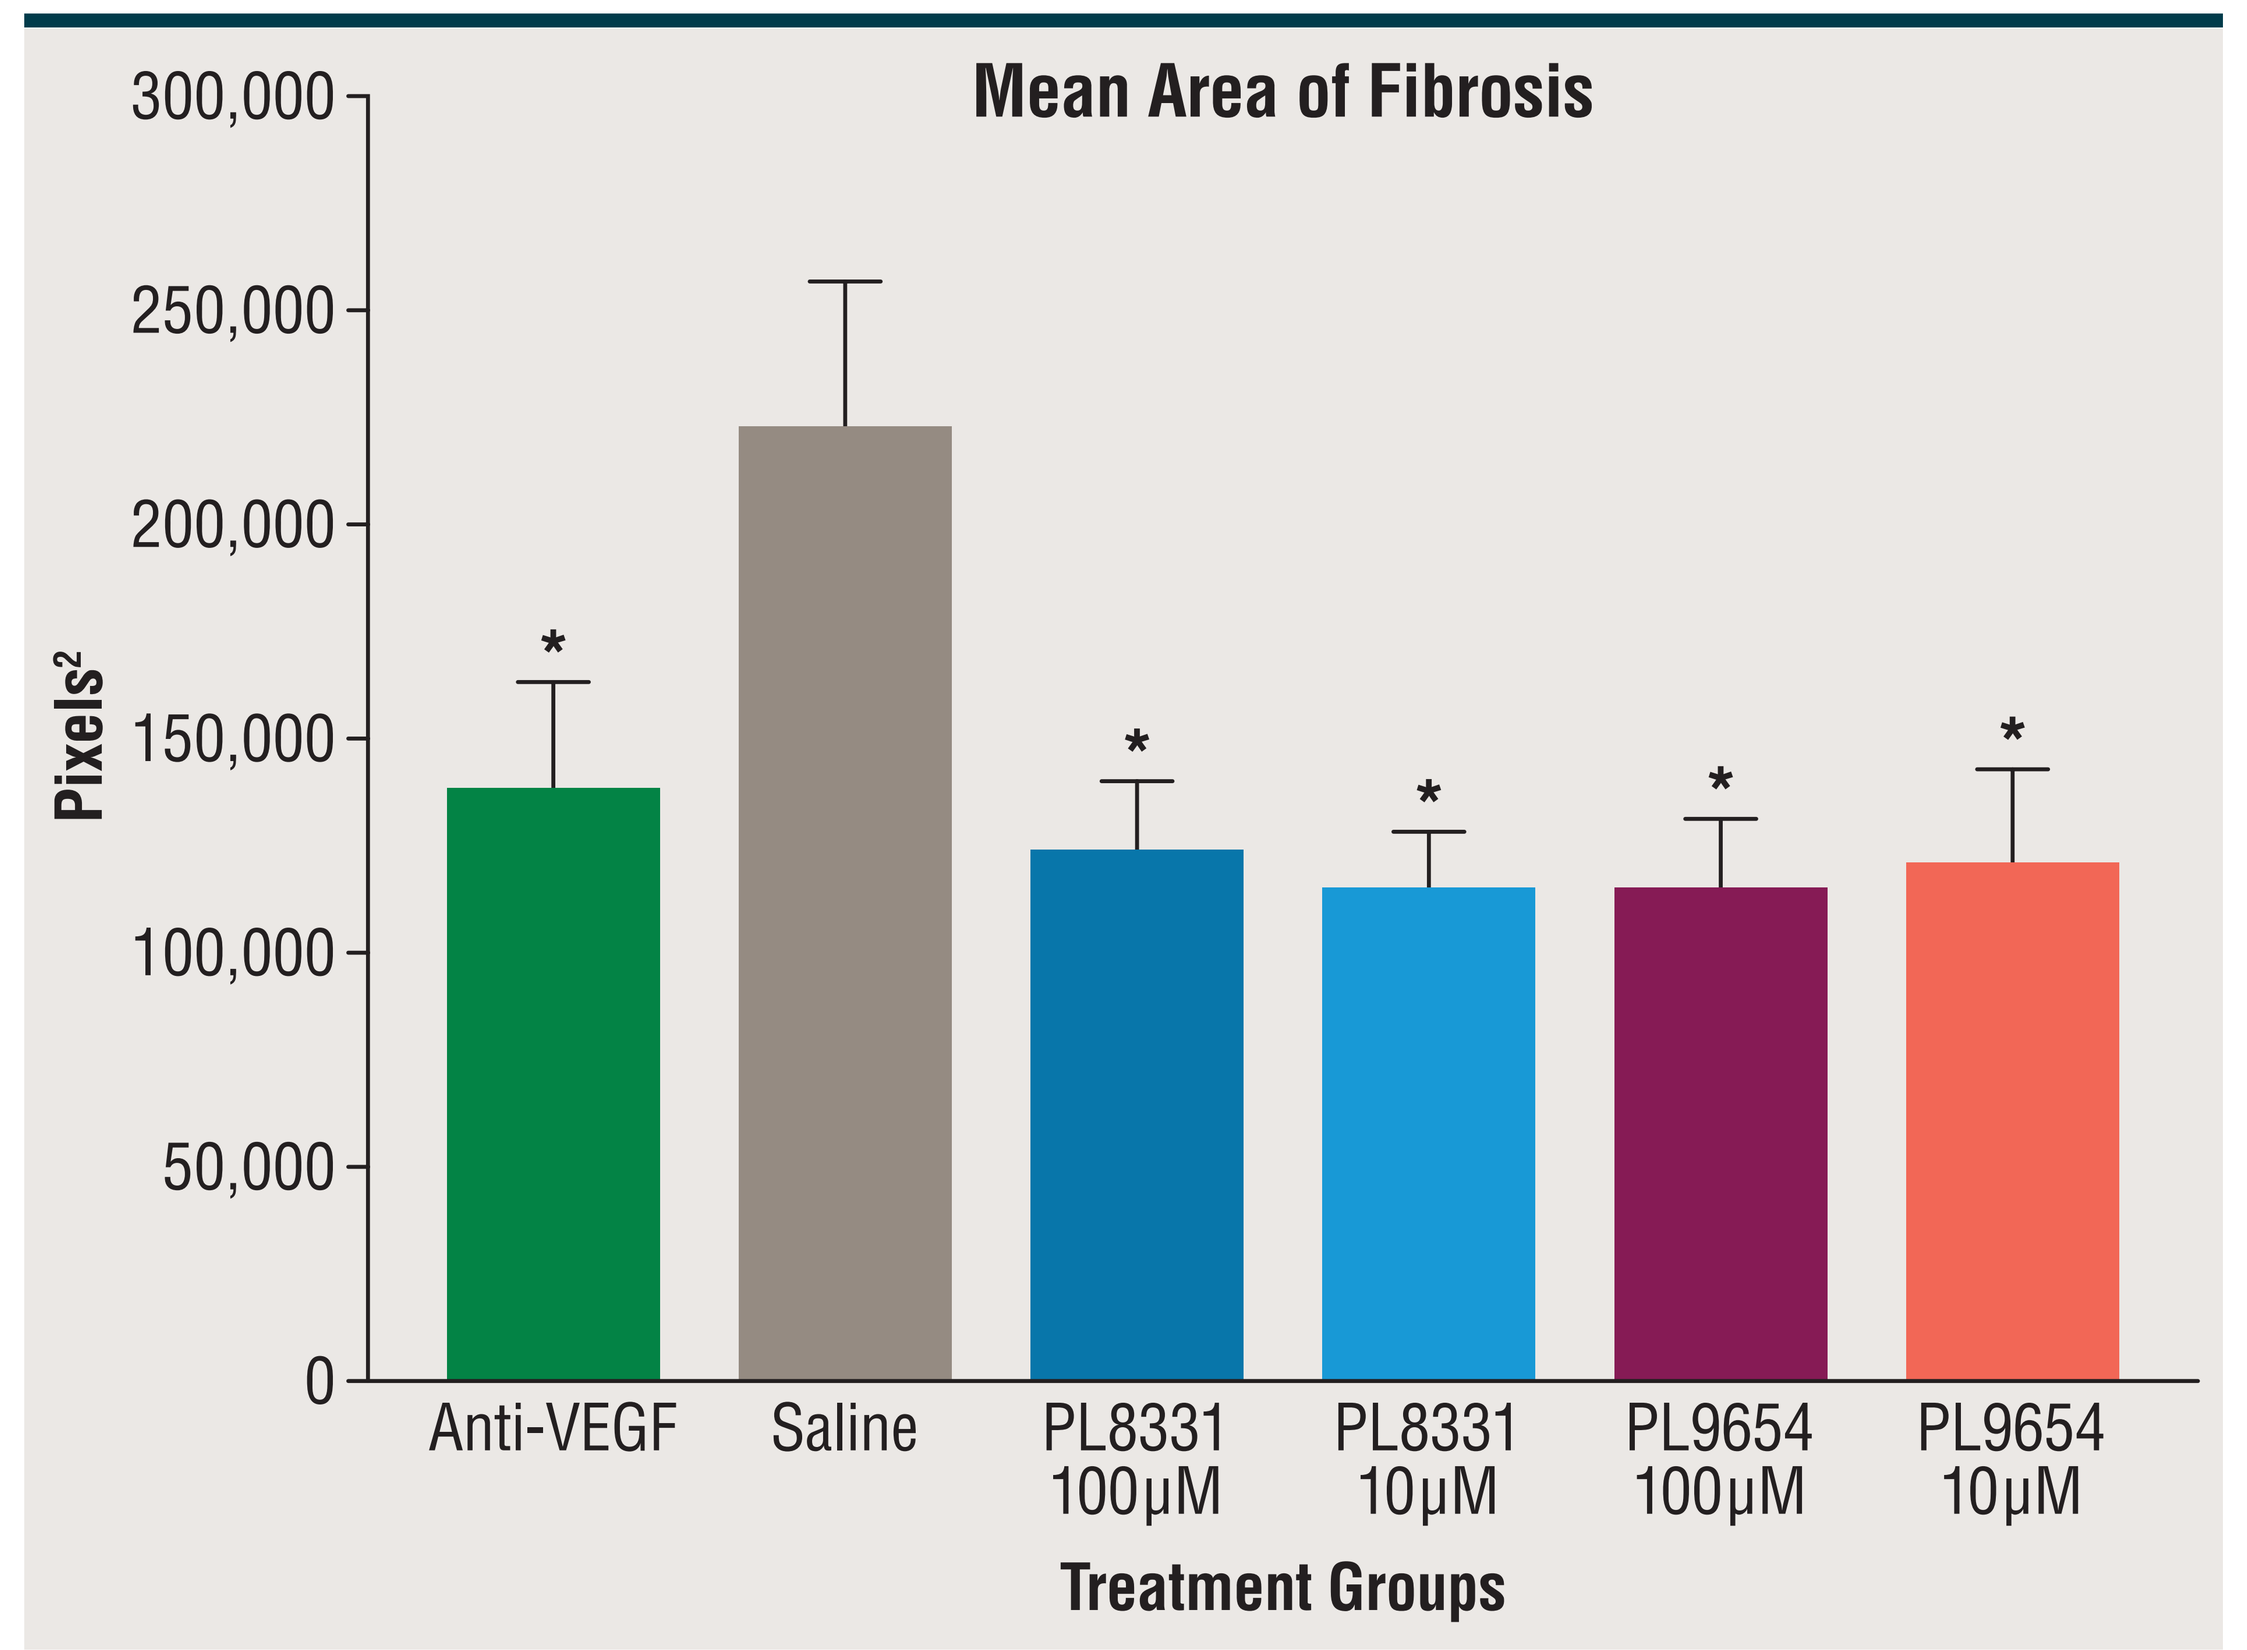

Mean Area of Fibrosis
Pixels²
300,000
250,000
200,000
150,000
100,000
50,000
0
Anti-VEGF
Saline
PL8331 100µM
PL8331 10µM
PL9654 100µM
PL9654 10µM
Treatment Groups

VEGF, vascular endothelial growth factor.

*$P \leq 0.05$. Fibrosis was determined by immunohistochemistry using positive collagen I staining. Mean areas of staining were compared using one-way ANOVA.

- Laser burns resulted in increased fibrosis in vehicle-injected mice (**Figure 8**)
- Mice treated with anti-VEGF antibody, PL8331, or PL9654 had significantly reduced fibrosis compared with vehicle control

Summary and Conclusions

- PL8331 treatment preserved retinal thickness and mitigated optic nerve cupping in a mouse model of diabetic retinopathy
- PL8331 therapy promoted preservation of retinal structure observed by RGC survival and histology of the retinas
- PL8331 suppressed VEGF production in the diabetic retina, suggesting that PL8331 treatment may block vascular leakage and neovascularization
- In a mouse model of age-related macular degeneration, PL8331 and PL9654 reduced retinal leakage, angiogenesis, and fibrosis compared with control to an extent equivalent to anti-VEGF treatment
- In conclusion, intravitreal delivery of the melanocortin agonists PL8331 and PL9654 significantly reduced several markers of retinal damage in mouse models of eye injury, supporting the continued development of PL8331 and PL9654 for the treatment of ocular disease

Support Palatin Technologies Inc.

Acknowledgements Writing and editorial assistance was provided by Miranda Tradewell for The Curry Rockefeller Group, LLC, Tarrytown, NY, USA, which was funded by Palatin Technologies Inc.

Disclosures John Dodd, Marie Makhlina, Wei H. Yang, and Carl Spana are employees of Palatin Technologies Inc. Andrew W. Taylor received consulting fees from Palatin Technologies Inc. Tat Fong Ng received consulting fees from Palatin Technologies Inc.

References **1.** Manna SK, et al. *J Immunol*. 1998;161:2873-2880. **2.** Catania A, et al. *Pharmacol Rev*. 2004;56(1):1-29. **3.** Perretti M, et al. *Trends Pharmacol Sci*. 2015;36(11):737-755. **4.** Raap U, et al. *J Immunol*. 2003;171:353-359. **5.** Lipton JM, et al. *Ann N Y Acad Sci.* 1994;741:137-148. **6.** Rajora N, et al. *J Neurosci*. 1997;17(6):2181-2186.