Presented at:
TIDES USA Oligonucleotide and Peptide Therapeutics 2021
Boston, MA
20–30 September 2021

Effect of the Melanocortin Receptor Agonist PL8177 on DSS-Induced Colitis in Rats and a Toxicologic Assessment of PL8177 in Beagle Dogs

Carl Spana, PhD; John Dodd, PhD; Robert Jordan, BS; Wei H. Yang, PhD; Marie Makhlina, MSc; Alison Obr, PhD; Paul S. Kayne, PhD
Palatin Technologies, Inc., Cranbury, NJ

Introduction

- Melanocortin-1 receptor (MC1r) agonists have been shown to induce the resolution of inflammation and its natural ligand, α-melanocyte stimulating hormone, (α-MSH) has been demonstrated to be effective in reducing inflammation in numerous experimental models[1,2]
- The MC1r-specific agonist PL8177 and its main metabolite PL8435 have demonstrated MC1r binding affinity and functional activity that mirrors that of α-MSH[2]
- Murine and human studies have found MC1r expression in the colon luminal surface, and mouse models have demonstrated an important role for MC1r in a dextran sodium sulfate (DSS)–induced model of colitis[3]
- PL8177 has shown significant protective effects against dinitrobenzene sulfonic acid (DNBS)-induced colitis in rats when doses of 1.5 and 5 µg were given by intracolonic administration via cannula[2]
- High potency and a lack of systemic absorption make PL8177 a promising new candidate for clinical development of an oral formulation for the treatment of inflammatory bowel disease
- Delayed-release microparticles of PL8177 were developed for oral delivery, allowing the agent to withstand the acidic environment of the stomach and release the active drug directly into the colon
- Here, we report on a study investigating the effects of PL8177 in a DSS–induced rat model of colitis and a toxicity study investigating its safety in beagle dogs

Methods

DSS-Induced Rat Model of Colitis

Figure 1. Study Design

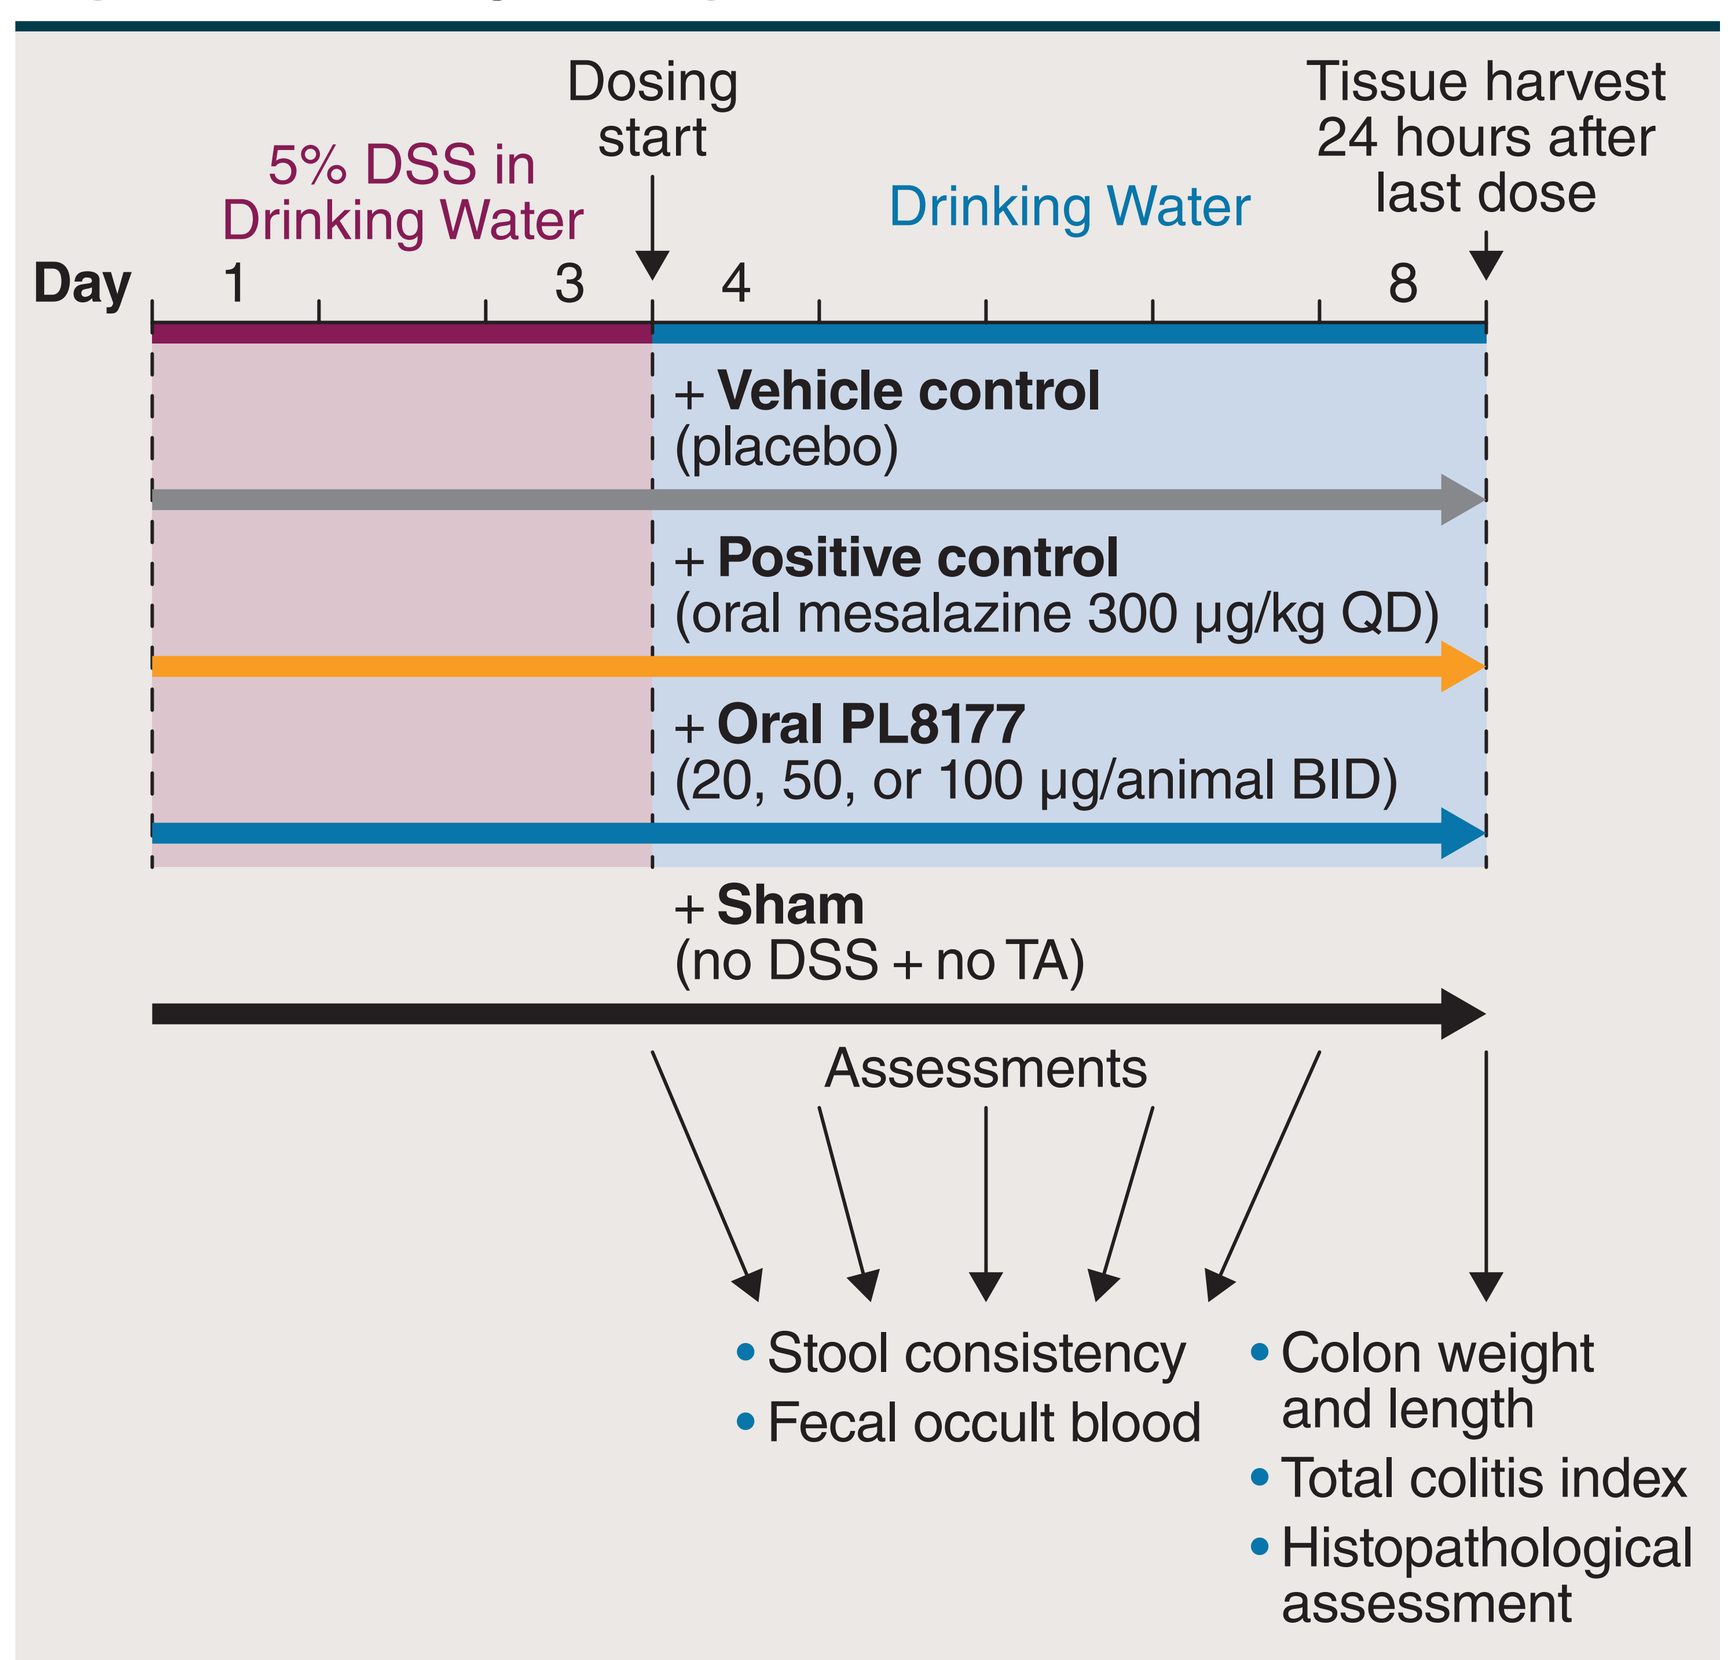
Dosing start
Tissue harvest 24 hours after last dose
5% DSS in Drinking Water
Drinking Water
Day 1 3 4 8
+ Vehicle control (placebo)
+ Positive control (oral mesalazine 300 µg/kg QD)
+ Oral PL8177 (20, 50, or 100 µg/animal BID)
+ Sham (no DSS + no TA)
Assessments
• Stool consistency
• Fecal occult blood
• Colon weight and length
• Total colitis index
• Histopathological assessment

BID, twice daily; DSS, dextran sodium sulfate; QD, once daily; TA, test agent.

- All rats, except those in the sham group (each group n=6), received 5% DSS in the drinking water for 3 days to induce colitis. Rats in the sham group had drinking water only
- Other groups received vehicle control (placebo)–filled capsules; PL8177-filled capsules at 20, 50, and 100 µg per animal (by oral gavage); or oral mesalazine 300 mg/kg (as positive control)
- Colitis was assessed by disease activity index (diarrhea and rectal bleeding), colon length shortening, colon weight gain, and histopathological assessment after organ harvesting at Day 8
- The total colitis index, an assessment that included separate items of inflammatory damage, was assessed
 - Total colitis index scoring was based on independent observers examining and summing the scores from 3 sections from each colon per animal. Separate scored items consisted of abnormalities of mucosal architecture (0–4), extent of inflammation (0–4), erosion or ulceration (0–4), epithelial regeneration (0–4), percentage involvement (1 [1%–25%] to 4 [76%–100%]), and severity of colitis (graded semi-quantitatively from 0–20). Total colitis index score range is 0–60

Toxicology Study in Beagle Dogs

Figure 2. Study Design

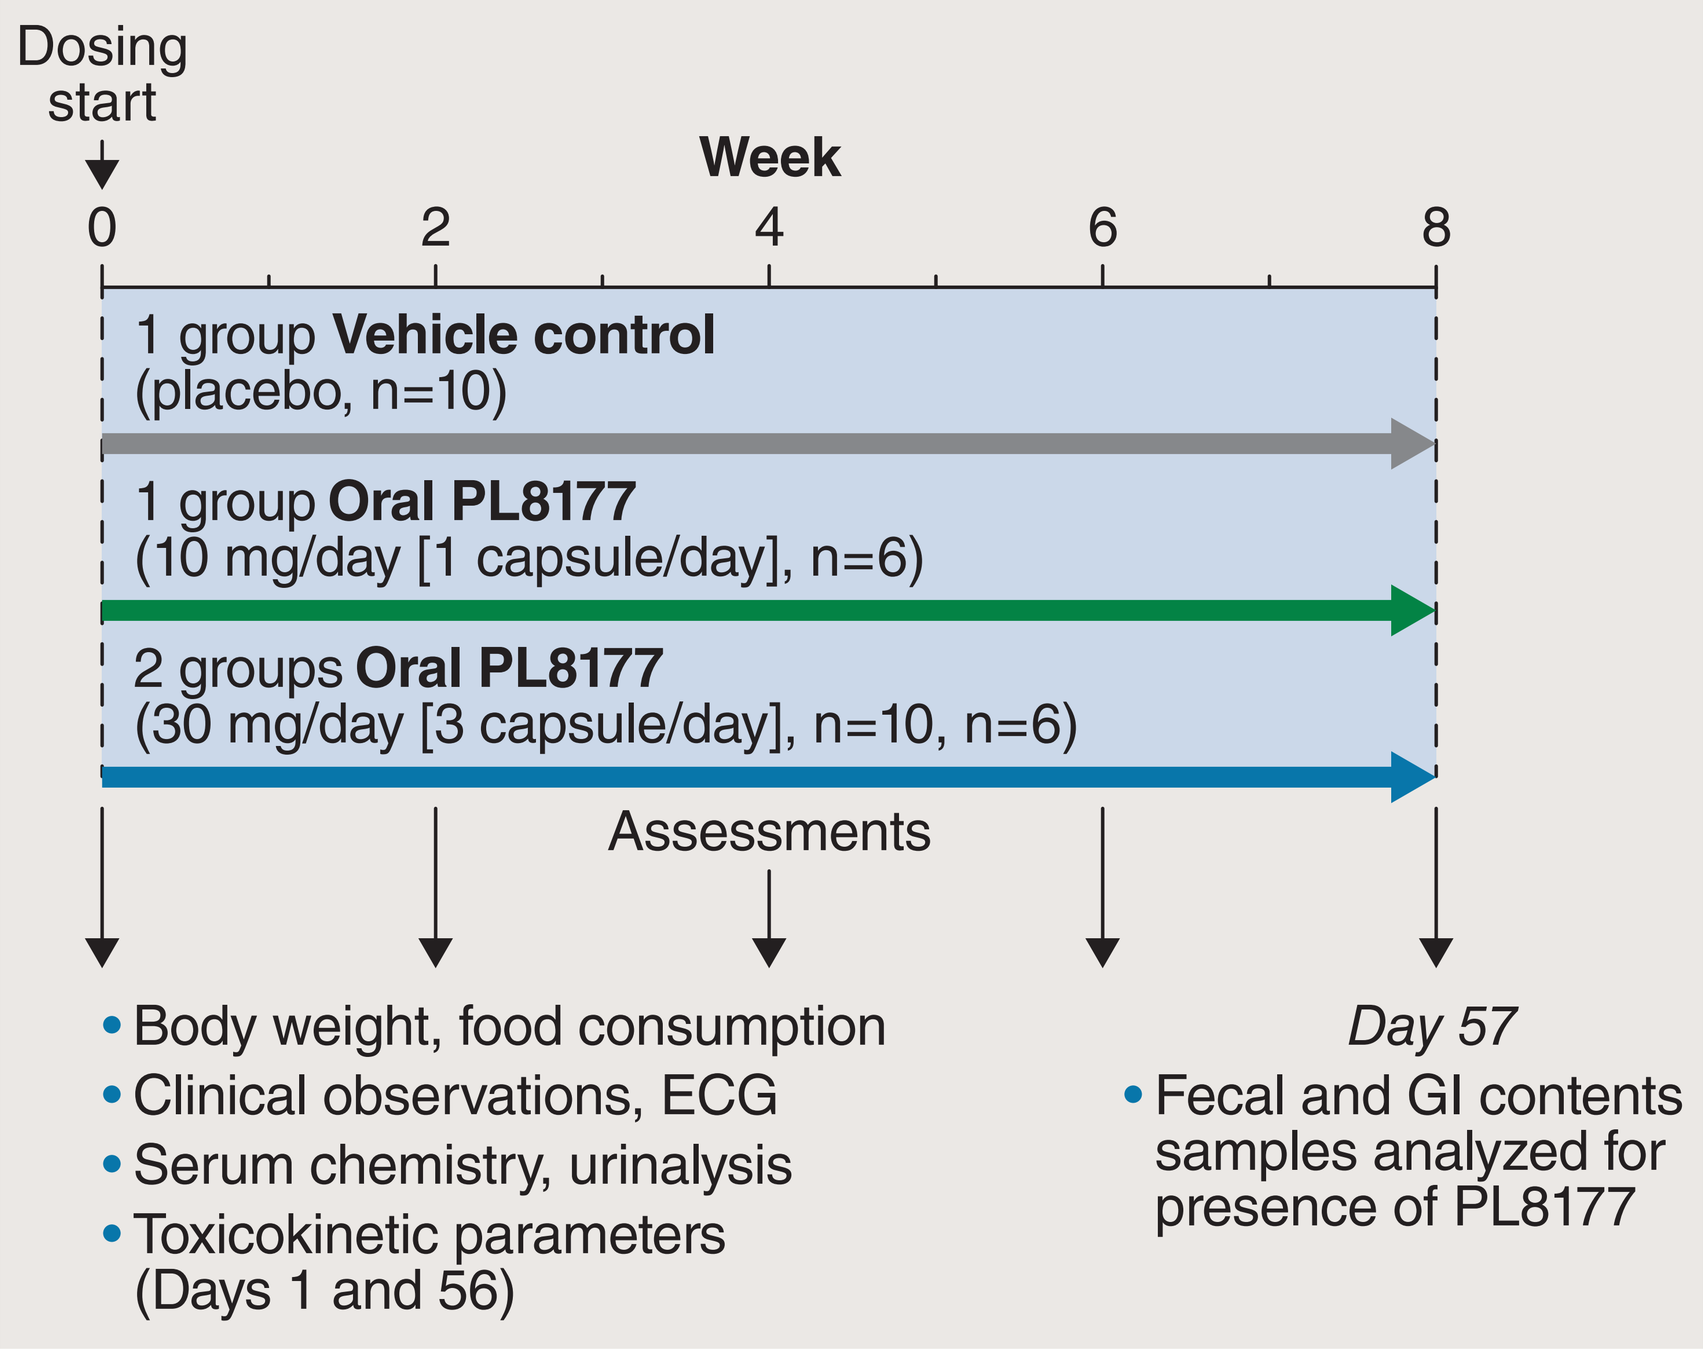
Dosing start
Week
0 2 4 6 8
1 group Vehicle control (placebo, n=10)
1 group Oral PL8177 (10 mg/day [1 capsule/day], n=6)
2 groups Oral PL8177 (30 mg/day [3 capsule/day], n=10, n=6)
Assessments
• Body weight, food consumption
• Clinical observations, ECG
• Serum chemistry, urinalysis
• Toxicokinetic parameters (Days 1 and 56)
Day 57
• Fecal and GI contents samples analyzed for presence of PL8177

ECG, electrocardiogram; GI, gastrointestinal.

- PL8177 (10 and 30 mg/kg) or placebo control was administered orally to 4 groups of beagle dogs once daily for 8 weeks
- Samples were collected for toxicokinetic assessment on Days 1 and 56
- Feces and gastrointestinal (GI) content samples were collected and analyzed on Day 57
- Necropsy was performed on 3 dogs/sex/group on Day 57, and on 2 dogs/sex/group after a 4-week recovery period

Results

DSS-Induced Rat Model of Colitis

- Colitis was successfully induced in rats treated with DSS, inducing significant ($P<0.05$) body weight loss, diarrhea, rectal bleeding, colon weight gain, and colon length shortening vs sham group (no DSS)

Body Weight, Stool Consistency, and Fecal Occult Blood

- Body weight gain between the vehicle control (placebo) and treated groups were similar (**Figure 3A**)

Figure 3. Changes in (A) Body Weight, (B) Stool Consistency, and (C) Fecal Occult Blood in DSS-Colitis-Induced Rats

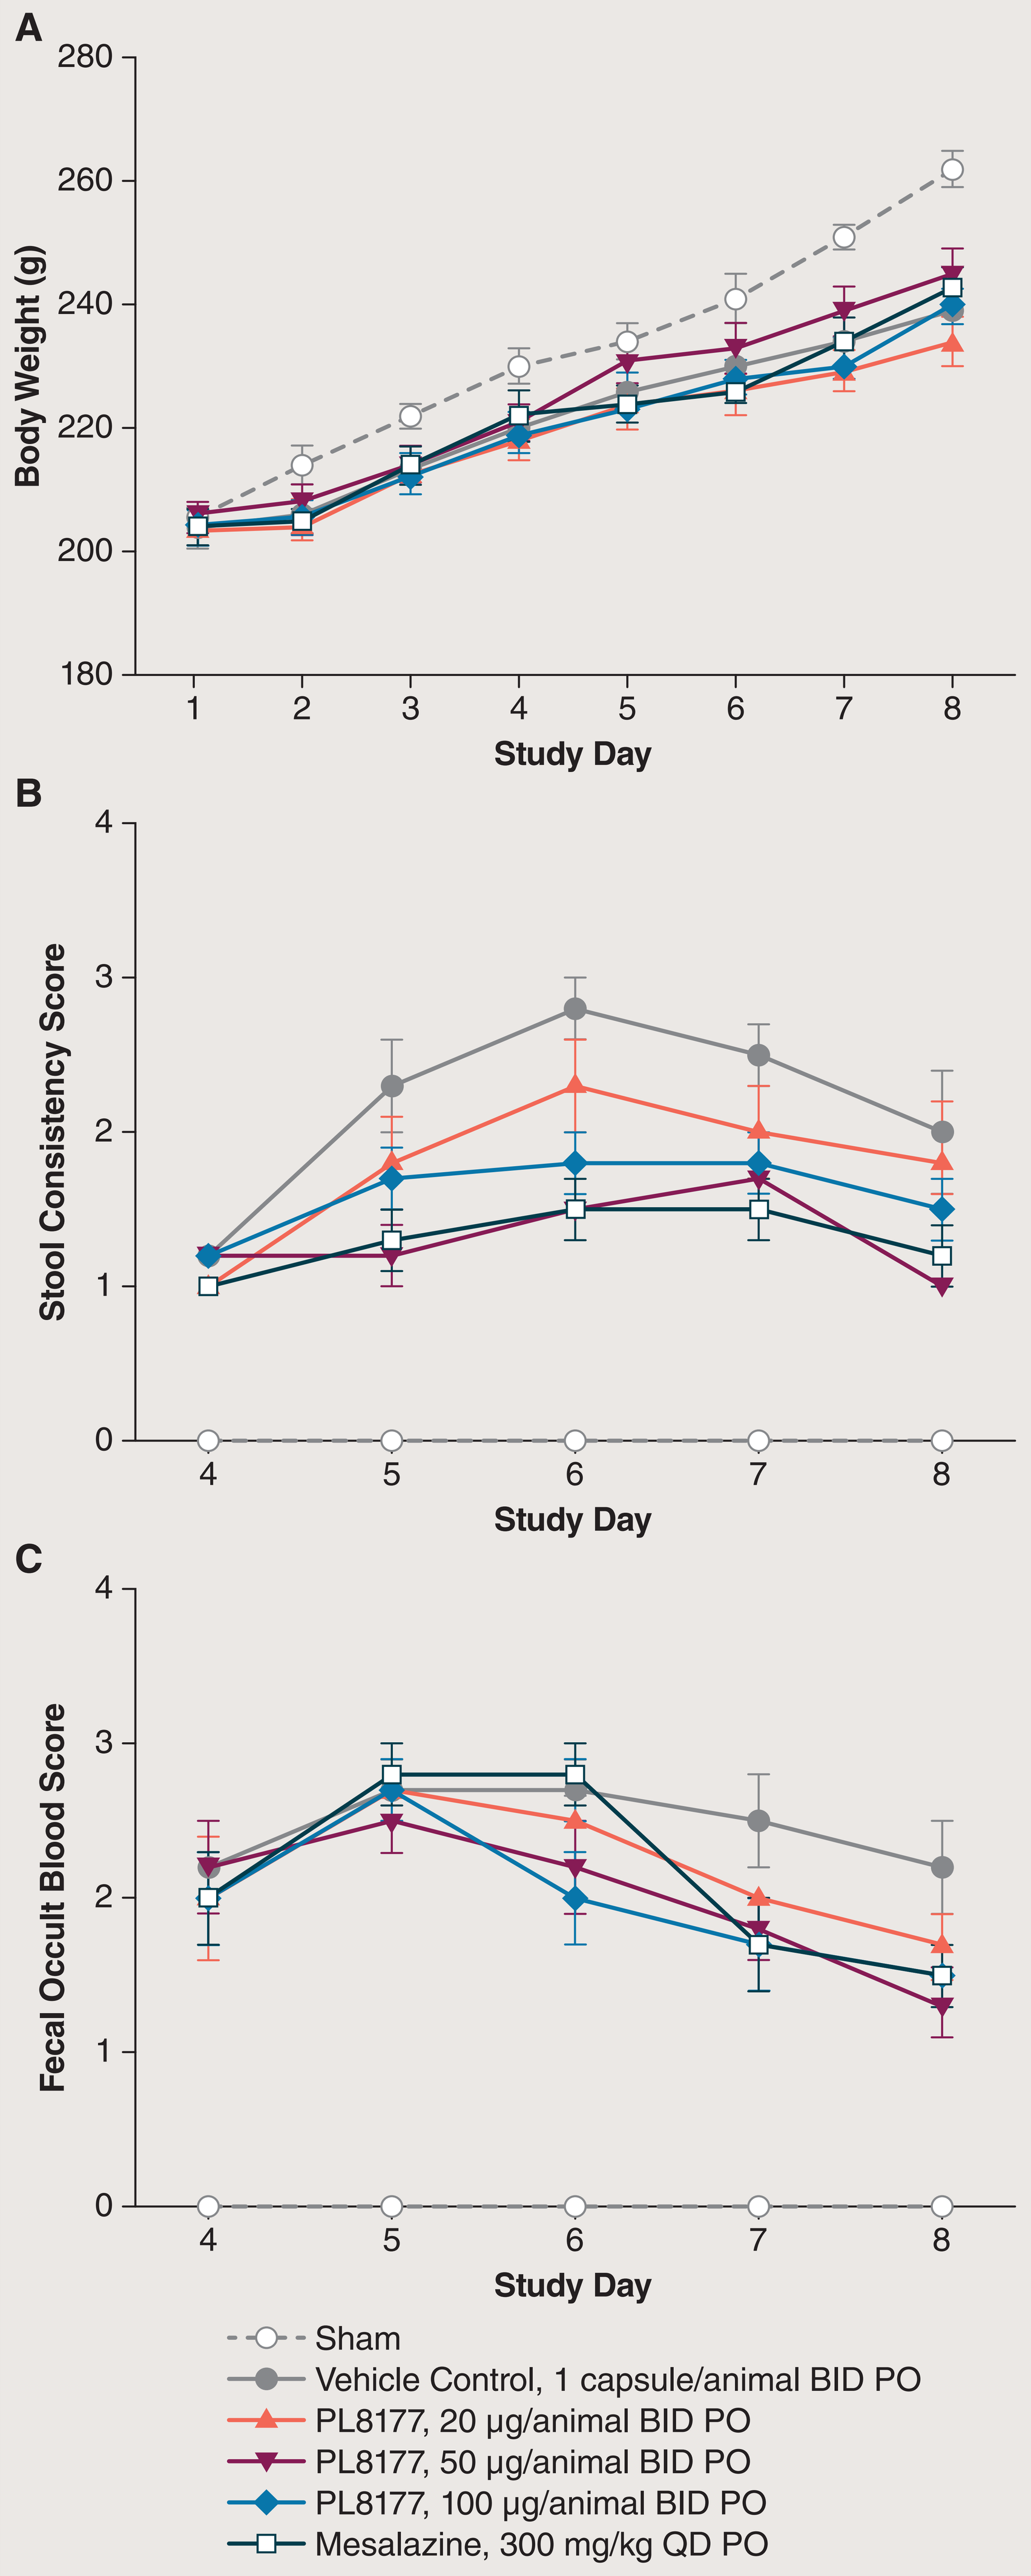
A
Body Weight (g)
280
260
240
220
200
180
1 2 3 4 5 6 7 8
Study Day
B
Stool Consistency Score
4 3 2 1 0
4 5 6 7 8
Study Day
C
Fecal Occult Blood Score
4 3 2 1 0
4 5 6 7 8
Study Day
Sham
Vehicle Control, 1 capsule/animal BID PO
PL8177, 20 µg/animal BID PO
PL8177, 50 µg/animal BID PO
PL8177, 100 µg/animal BID PO
Mesalazine, 300 mg/kg QD PO

Note: All animals, except those in sham group, received 5% DSS in the drinking water from Day 1 to Day 3 for 3 days, and then changed to normal drinking water for the following 5 days. Tissue harvest occurred on Day 8.
BID, twice daily; DSS, dextran sodium sulfate; PO, by mouth; QD, once daily.

- Oral PL8177 at 50 µg/animal showed significant ($P<0.05$) improvement in stool consistency score from Day 5 to Day 8 and significant ($P<0.05$) improvement in fecal occult blood score on Day 8, when compared to the vehicle group (**Figure 3B** and **C**)
- Oral PL8177 at 100 µg/animal had a significant ($P<0.05$) effect on stool consistency score on Day 6
- Mesalazine also showed significant improvement vs vehicle control for Days 5–8

Total Colitis Index and Histological Assessment

- There was a significant ($P<0.05$) improvement observed in the total colitis index for the PL8177 100-µg group compared to the vehicle control group (**Figure 4**)

Figure 4. Total Colitis Index on Day 8

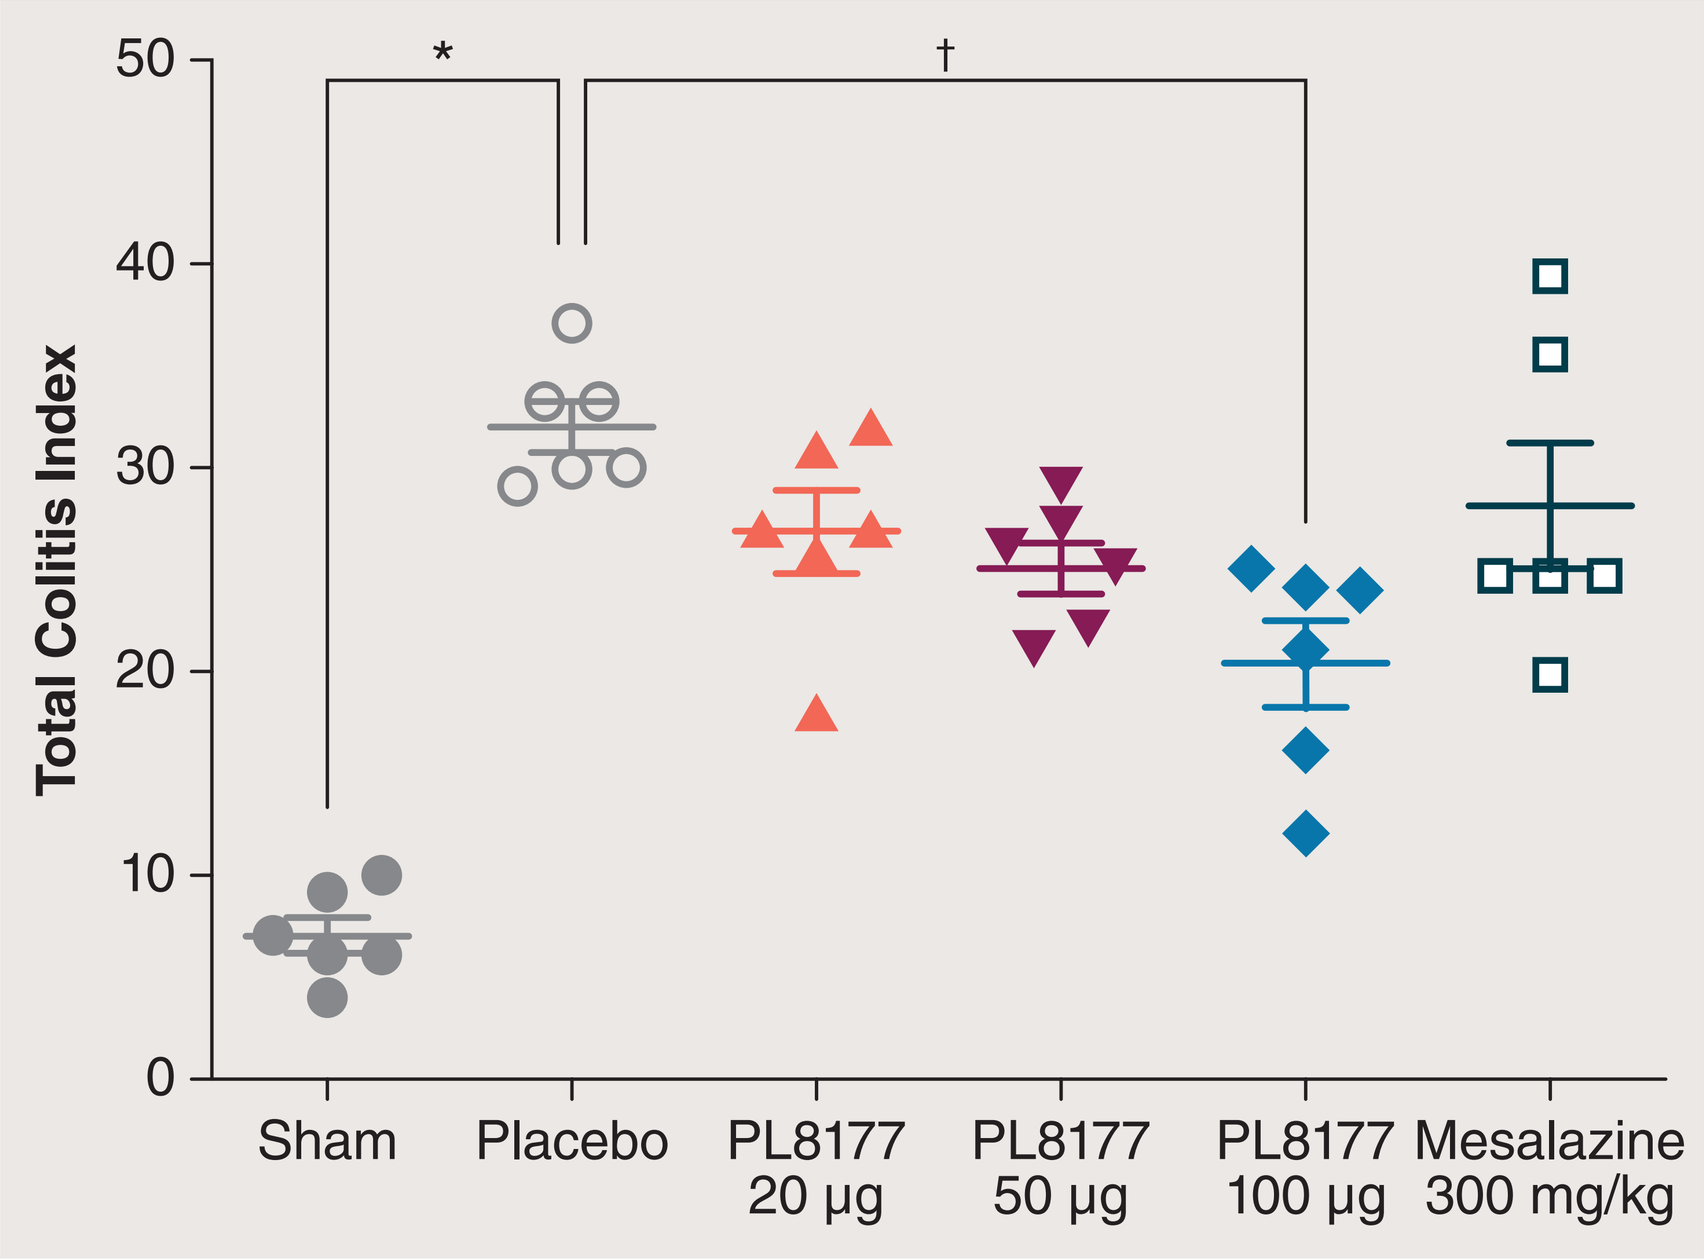
Total Colitis Index
50
40
30
20
10
0
*
†
Sham
Placebo
PL8177 20 µg
PL8177 50 µg
PL8177 100 µg
Mesalazine 300 mg/kg

Mean (SEM).
*$P<0.05$ vs sham. †$P<0.05$ vs vehicle control (placebo).

- The decline in the total colitis index for mesalazine-treated rats was less than that observed in any of the PL8177-treated rats
- Colon histopathological examination showed injury, and prominent ulcerations to the mucosa of the distal colon that extended for 2.5 to 7 cm in treated inflamed rats. Submucosal focal edema and pronounced transmural thickening of the colonic wall were also observed (**Figure 5**)

Figure 5. Representative Colon Histological Sections of DSS-Colitis-Induced Rats

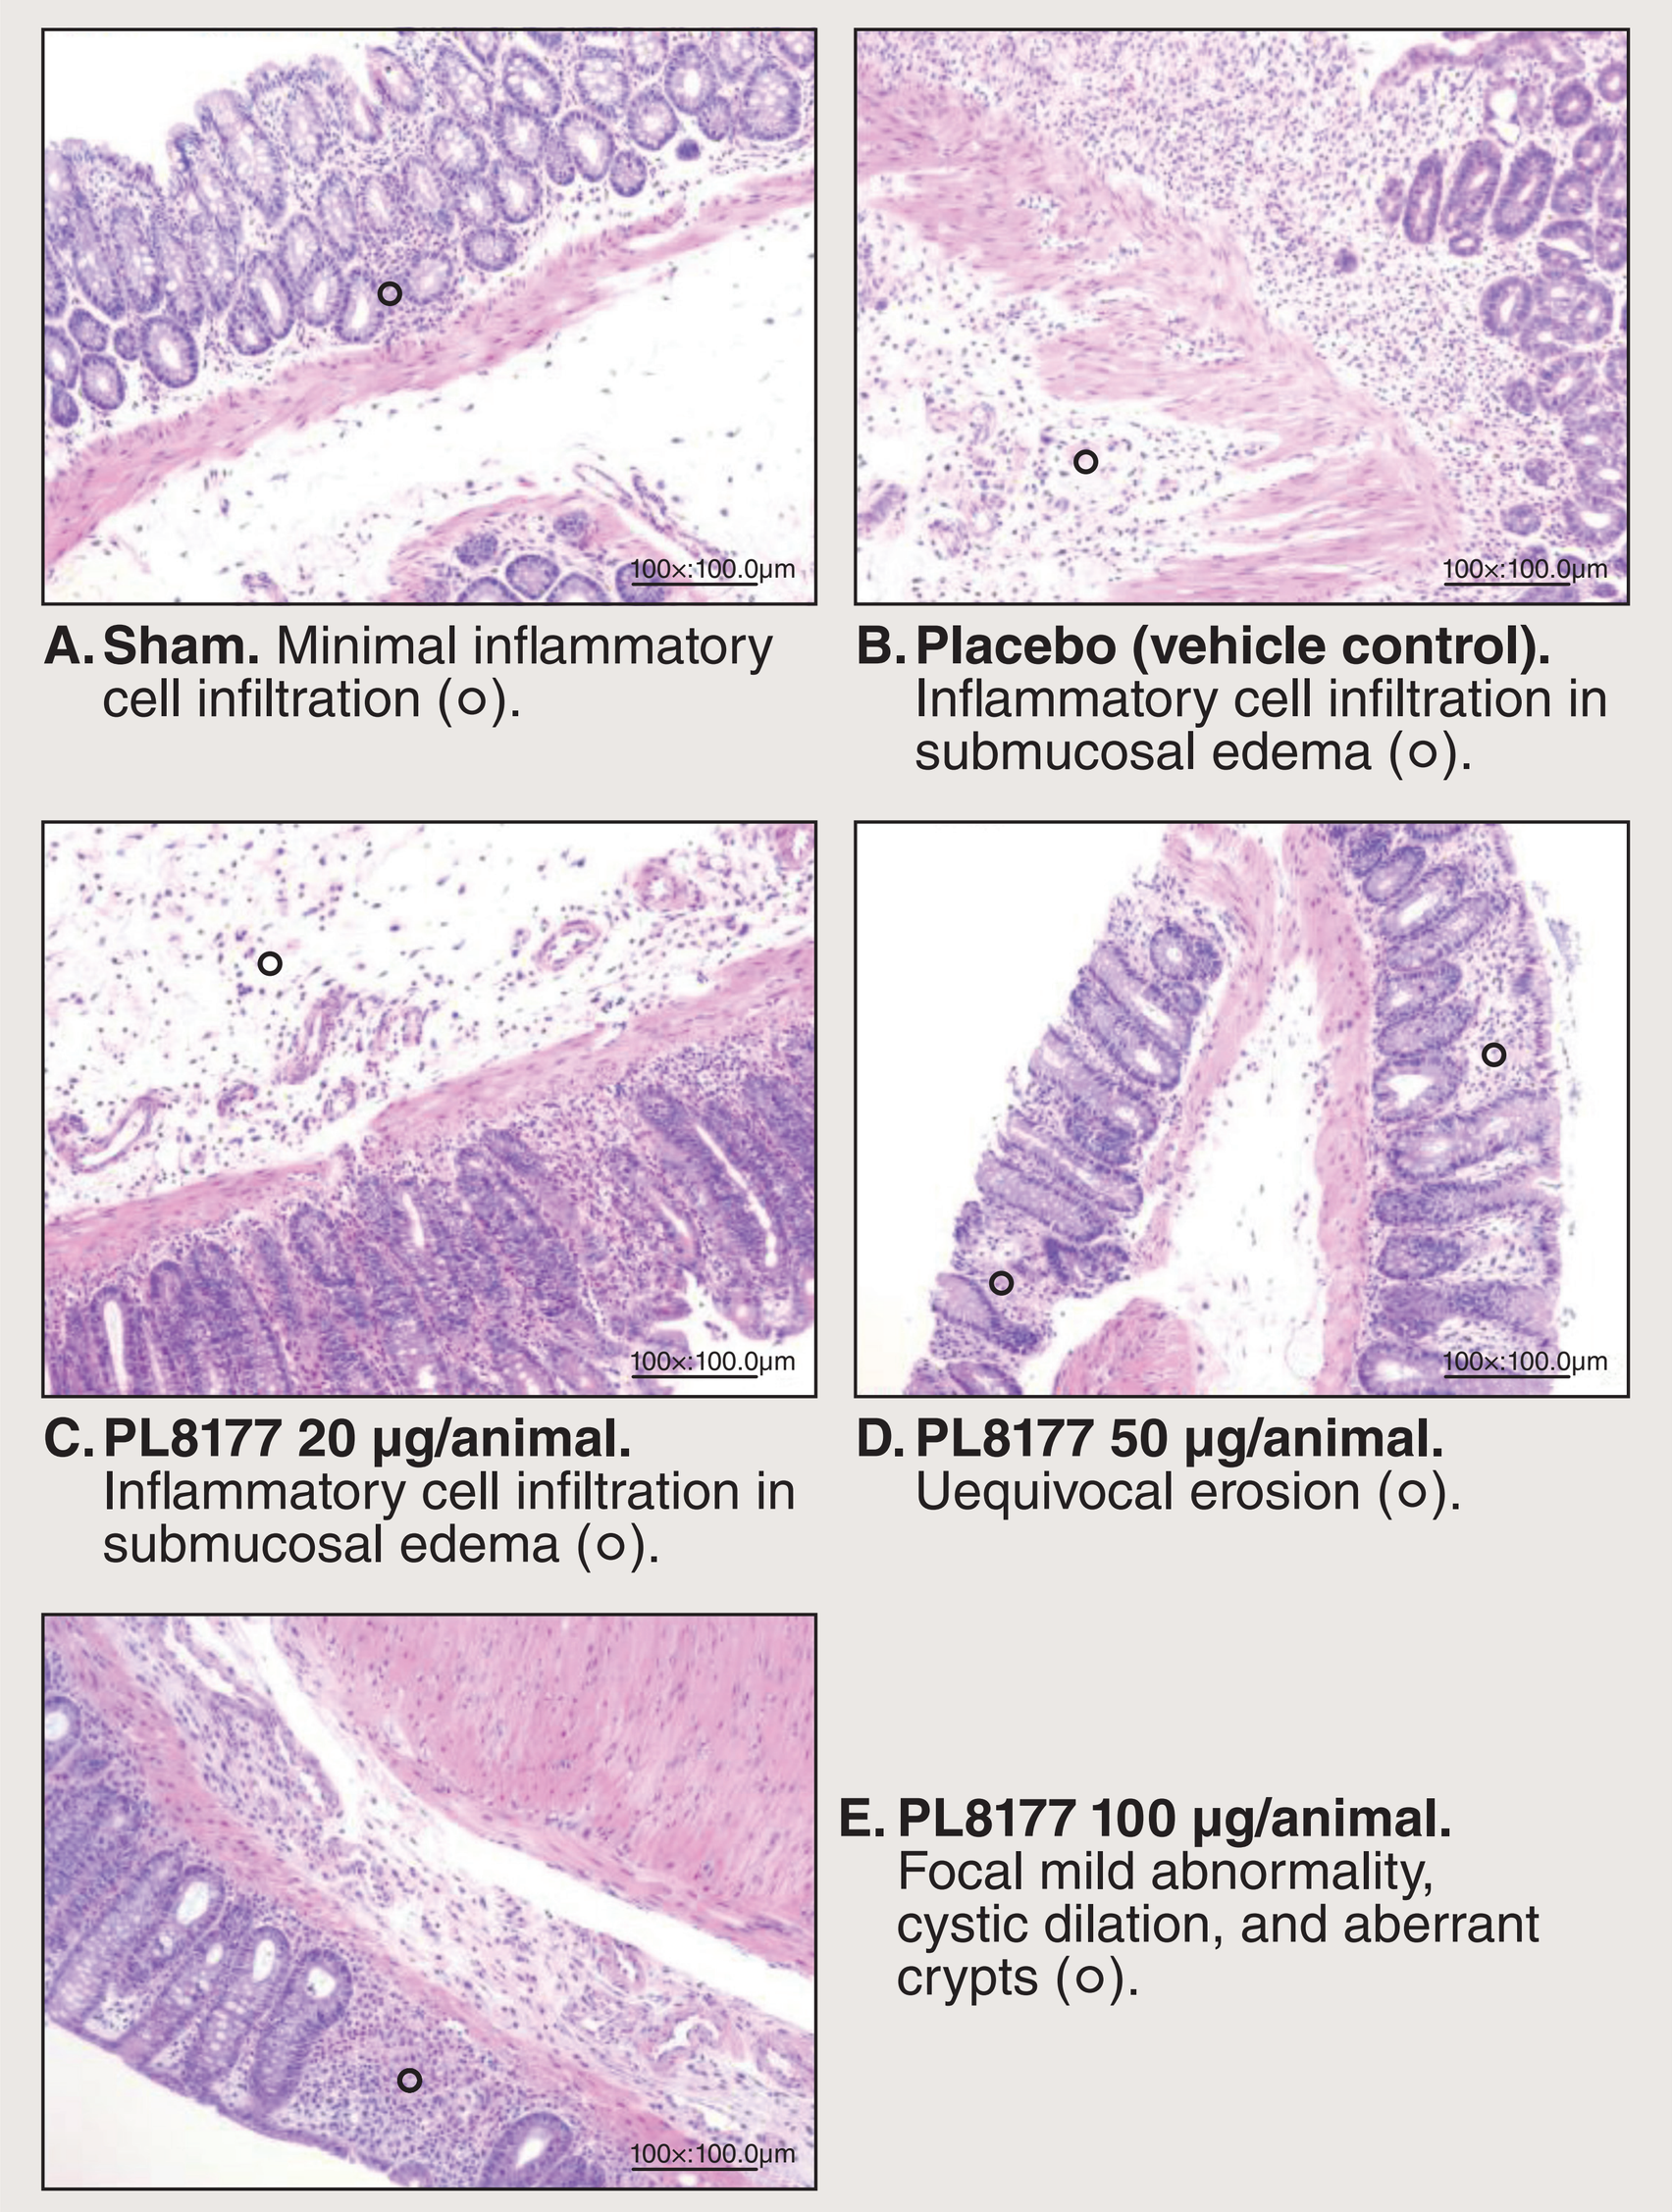
100×:100.0µm

A. Sham. Minimal inflammatory cell infiltration (○).
B. Placebo (vehicle control). Inflammatory cell infiltration in submucosal edema (○).
C. PL8177 20 µg/animal. Inflammatory cell infiltration in submucosal edema (○).
D. PL8177 50 µg/animal. Uequivocal erosion (○).
E. PL8177 100 µg/animal. Focal mild abnormality, cystic dilation, and aberrant crypts (○).

Note: Sham (A) is no challenge and no treatment. Vehicle control (placebo) (B) is no treatment but DSS challenge. C, D, and E are DSS challenge and treatment with PL8177. DSS, dextran sodium sulfate.

Colon Length and Weight

- Oral PL8177 (50 µg/animal) treatment showed a significant improvement in colon weight (53% reduction) (**Figure 6**)

Figure 6. Colon Length and Weight on Day 8

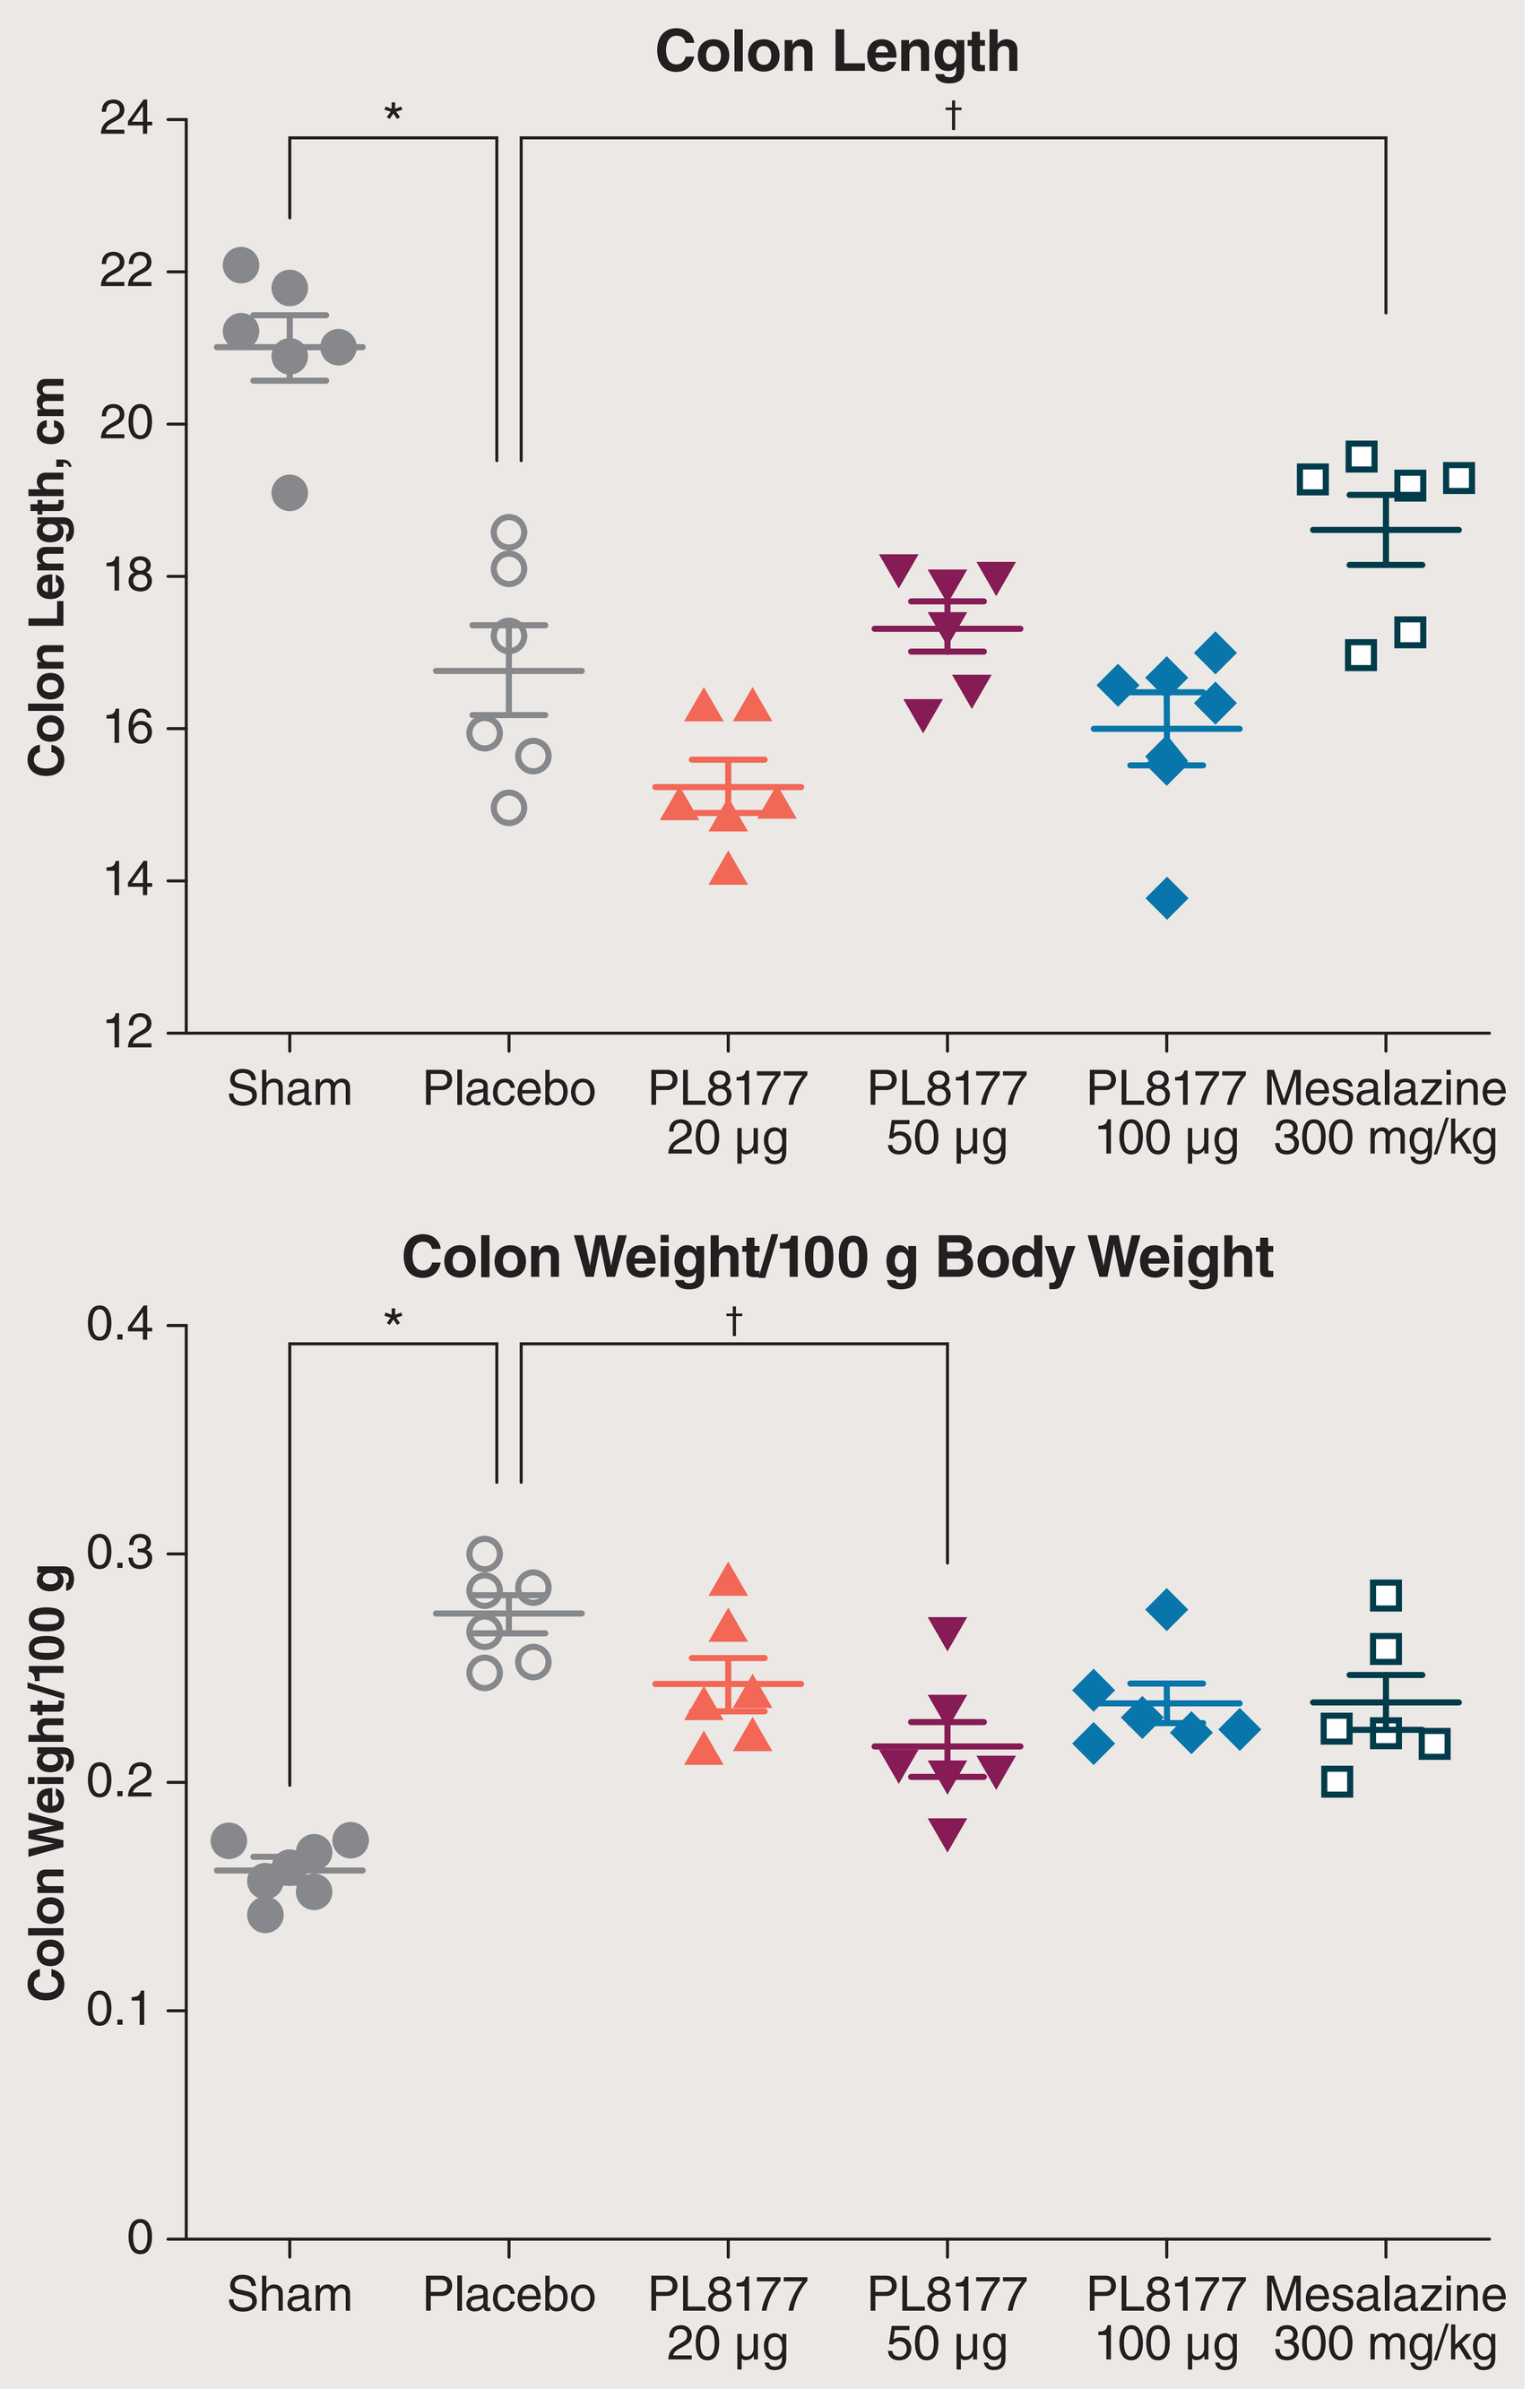
Colon Length
Colon Length, cm
24
22
20
18
16
14
12
*
†
Sham
Placebo
PL8177 20 µg
PL8177 50 µg
PL8177 100 µg
Mesalazine 300 mg/kg
Colon Weight/100 g Body Weight
Colon Weight/100 g
0.4
0.3
0.2
0.1
0
*
†
Sham
Placebo
PL8177 20 µg
PL8177 50 µg
PL8177 100 µg
Mesalazine 300 mg/kg

Figures show mean (SEM).
*$P<0.05$ vs sham. †$P<0.05$ vs vehicle control (placebo).

- Oral mesalazine 300 mg/kg (positive control) was associated with significant reduction in colon length, but only moderate improvement (35%) in colon weight gain

Toxicology Study in Beagle Dogs

- A total of 276 plasma samples, 9 feces samples, and 42 GI tract content samples were analyzed for the presence of PL8177
- All systemic samples were below the lower limit of quantition for PL8177
- PL8177 was not systemically absorbed, and plasma exposure was below the lower limit of quantitation
- Dogs tolerated 10- and 30-mg/kg/day oral gavage doses of PL8177 for up to 57 consecutive days without adverse effects
- PL8177 and PL8435 sample concentration and amounts were generally higher in the cecum, colon, and rectum compared to the upper sections of the GI tract, consistent with movement of the dose through the GI tract
- The majority of the administered dose in GI content and feces samples was accounted for as PL8177, with lesser amounts of the main metabolite (PL8435)
 - The mean amount of PL8177 as a percentage of administered dose in GI sections and feces was 81% for males and 102% for females. The mean amount of PL8177 in feces was 25% for males and 23% for females

Conclusions

- Direct application of oral PL8177 to inflamed colon showed significant improvement in markers of colitis in the DSS rat model compared to the vehicle group
 - Improvement in stool consistency score and blood score compared to vehicle
 - Improvement in total colitis index
 - Improvement in colon weight
- In the dog toxicity study, after oral administration, no adverse events were noted at any dose, and PL8177 was concentrated in the lower GI tract
- Reported results are consistent with the aim of ultimately treating inflammatory bowel disease in humans

Funding These studies were funded by Palatin Technologies Inc. (Cranbury, NJ).

Acknowledgments Editorial support was provided by Robin Smith, PhD, of The Curry Rockefeller Group, LLC (Tarrytown, NY), and was funded by Palatin Technologies, Inc. (Cranbury, NJ).

Disclosures Carl Spana, John Dodd, Robert Jordan, Alison Obr, and Paul S. Kayne are employees of Palatin Technologies, Inc. Marie Makhlina and Wei H. Yang are former employees of Palatin Technologies.

References **1.** Ahmed TJ, Montero-Melendez T, Perretti M, Pitzalis C. Curbing inflammation through endogenous pathways: focus on melanocortin peptides. *Int J Inflam*. 2013;2013:985815. **2.** Spana C, Taylor AW, Yee DG, Makhlina M, Yang W, Dodd J. Probing the role of melanocortin type 1 receptor agonists in diverse immunological diseases. *Front Pharmacol*. 2018;9:1535. **3.** Maaser C, Kannengiesser K, Specht C, et al. Crucial role of the melanocortin receptor MC1R in experimental colitis. *Gut*. 2006;55(10):1415-1422.