Protective Effects of Two Melanocortin Agonists Delivered by Intravitreal Injection in Mouse Models of Retinopathy

John Dodd, PhD[1], Peter K. Kaiser, MD, FASRS[2], Wei H. Yang, PhD[1], Andrew W. Taylor, PhD[3], Tatfong Ng, PhD[3], Marie Makhlina, MS[1], Carl Spana, PhD
[1]Palatin Technologies, Inc., [2]Cleveland Clinic Lerner College of Medicine, [3]Boston University School of Medicine

OBJECTIVE: To investigate the efficacy of two melanocortin agonists in treating mouse models of diabetic retinopathy and CNV.

INTRODUCTION

The melanocortin system plays a key role in the inhibition of leukocyte activation and inflammation.[1-3] Targeting this pathway can be an alternative to current therapies for specific retinal diseases.

Melanocortin agonists have demonstrated anti-inflammatory effects in various experimental models of inflammatory disease.[4-6] PL8331 and PL9654 are melanocortin 1 receptor agonists being evaluated preclinically for the treatment of various ocular diseases.

METHODS

Study 1: Mice with streptozotocin-induced diabetic retinopathy were given IVT injections of PL8331 at weeks 1, 4, 8, 12, and 16. At week 17, retinas were evaluated for retinal ganglion cell (RGC) density, VEGF expression, and the effects of RPE on macrophage production of TNF, and IL-10.

Study 2: After induction of CNV by retinal laser burns, mice were given 1 of 6 treatments by IVT injection immediately and on day 15. Treatments: vehicle, anti-VEGF aflibercept, PL8331 (Concentrations X and X^2 μM), and PL9654 (Concentrations X and X^2 μM). FFA, SD-OCT, and immunohistochemistry were used to quantify neovascularization, extent of leakage, and fibrosis. Collagen deposition was also assessed.

RESULTS

Study 1: Mouse Model of Diabetic Retinopathy

Untreated Diabetic Retinas vs. healthy controls showed:
- Reduced RGC density ($P \leq 0.05$).
- Increased VEGF concentrations ($P < 0.005$).

Diabetic retinas treated with PL8331 vs. Untreated Diabetic Retinas:
- Maintained higher RGC density.
- Showed decreased levels of VEGF.
- RPE promoted macrophage production of the anti-inflammatory cytokine IL-10

Neuroretinal production of VEGF and RPE regulation of macrophage TNF and IL-10 production in mice with streptozotocin-induced diabetic retinopathy

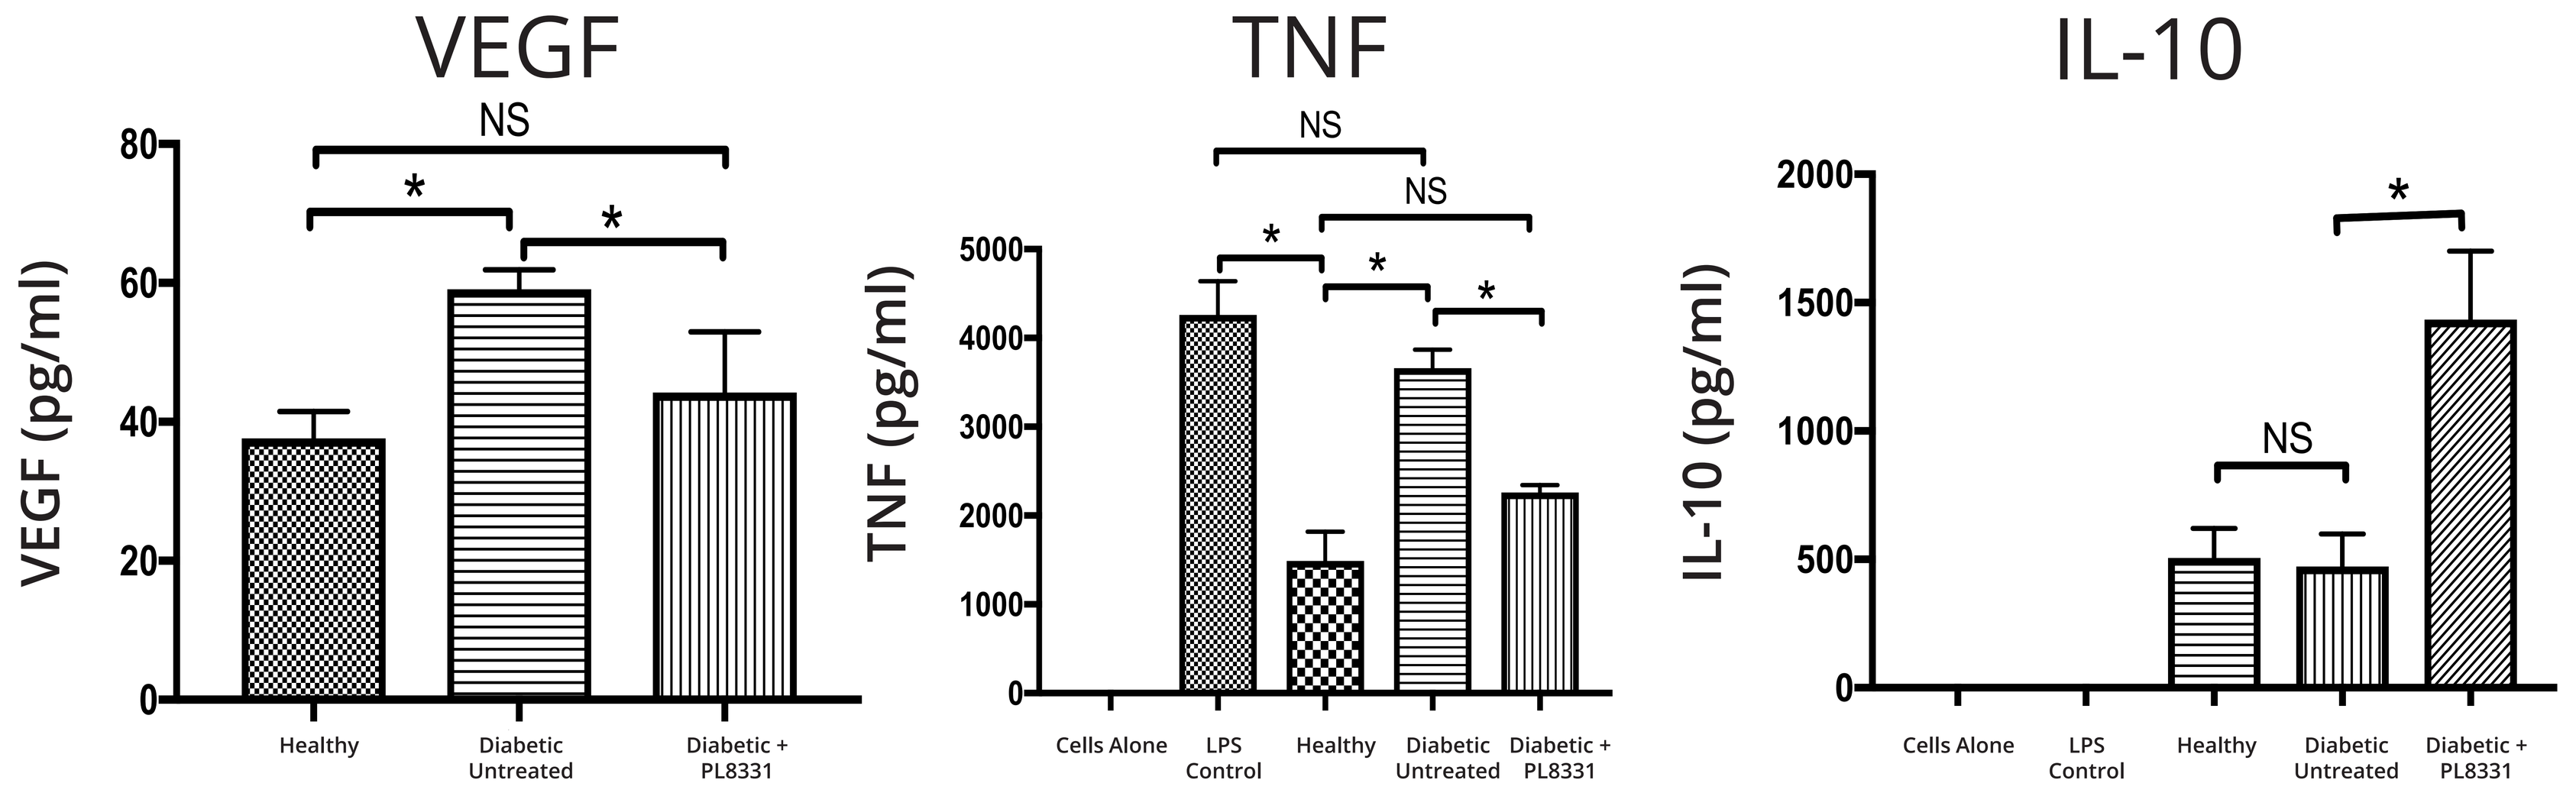
VEGF
VEGF (pg/ml)
80
60
40
20
0
NS
*
*
Healthy
Diabetic Untreated
Diabetic + PL8331
TNF
TNF (pg/ml)
5000
4000
3000
2000
1000
0
NS
NS
*
*
*
Cells Alone
LPS Control
Healthy
Diabetic Untreated
Diabetic + PL8331
IL-10
IL-10 (pg/ml)
2000
1500
1000
500
0
*
NS
Cells Alone
LPS Control
Healthy
Diabetic Untreated
Diabetic + PL8331

Histologic images of retinas from mice with streptozotocin-induced diabetic retinopathy

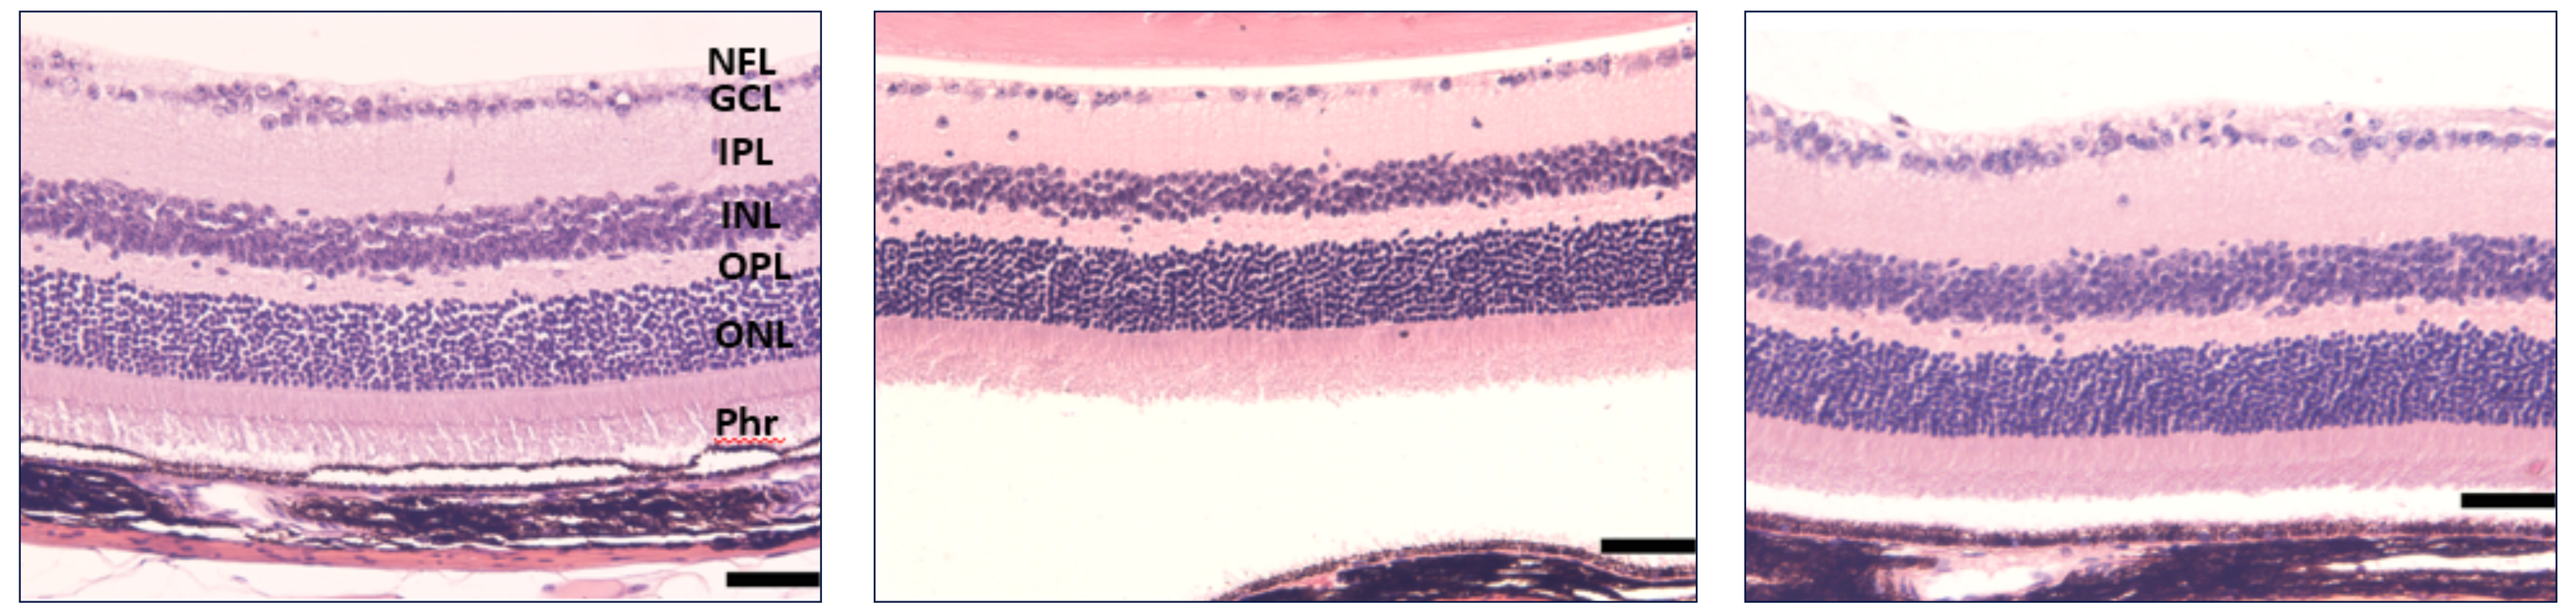
NFL
GCL
IPL
INL
OPL
ONL
Phr

Healthy mouse eye | Diabetic mouse eye | Diabetic +PL8331

Study 2: Mouse Model of CNV

Compared to mice treated with saline, treatment with positive control (aflibercept) and both dose levels of PL8331 and PL9654 significantly reduced ($P < 0.05$) the mean area of CNV leakage, CNV area, and area of fibrosis.

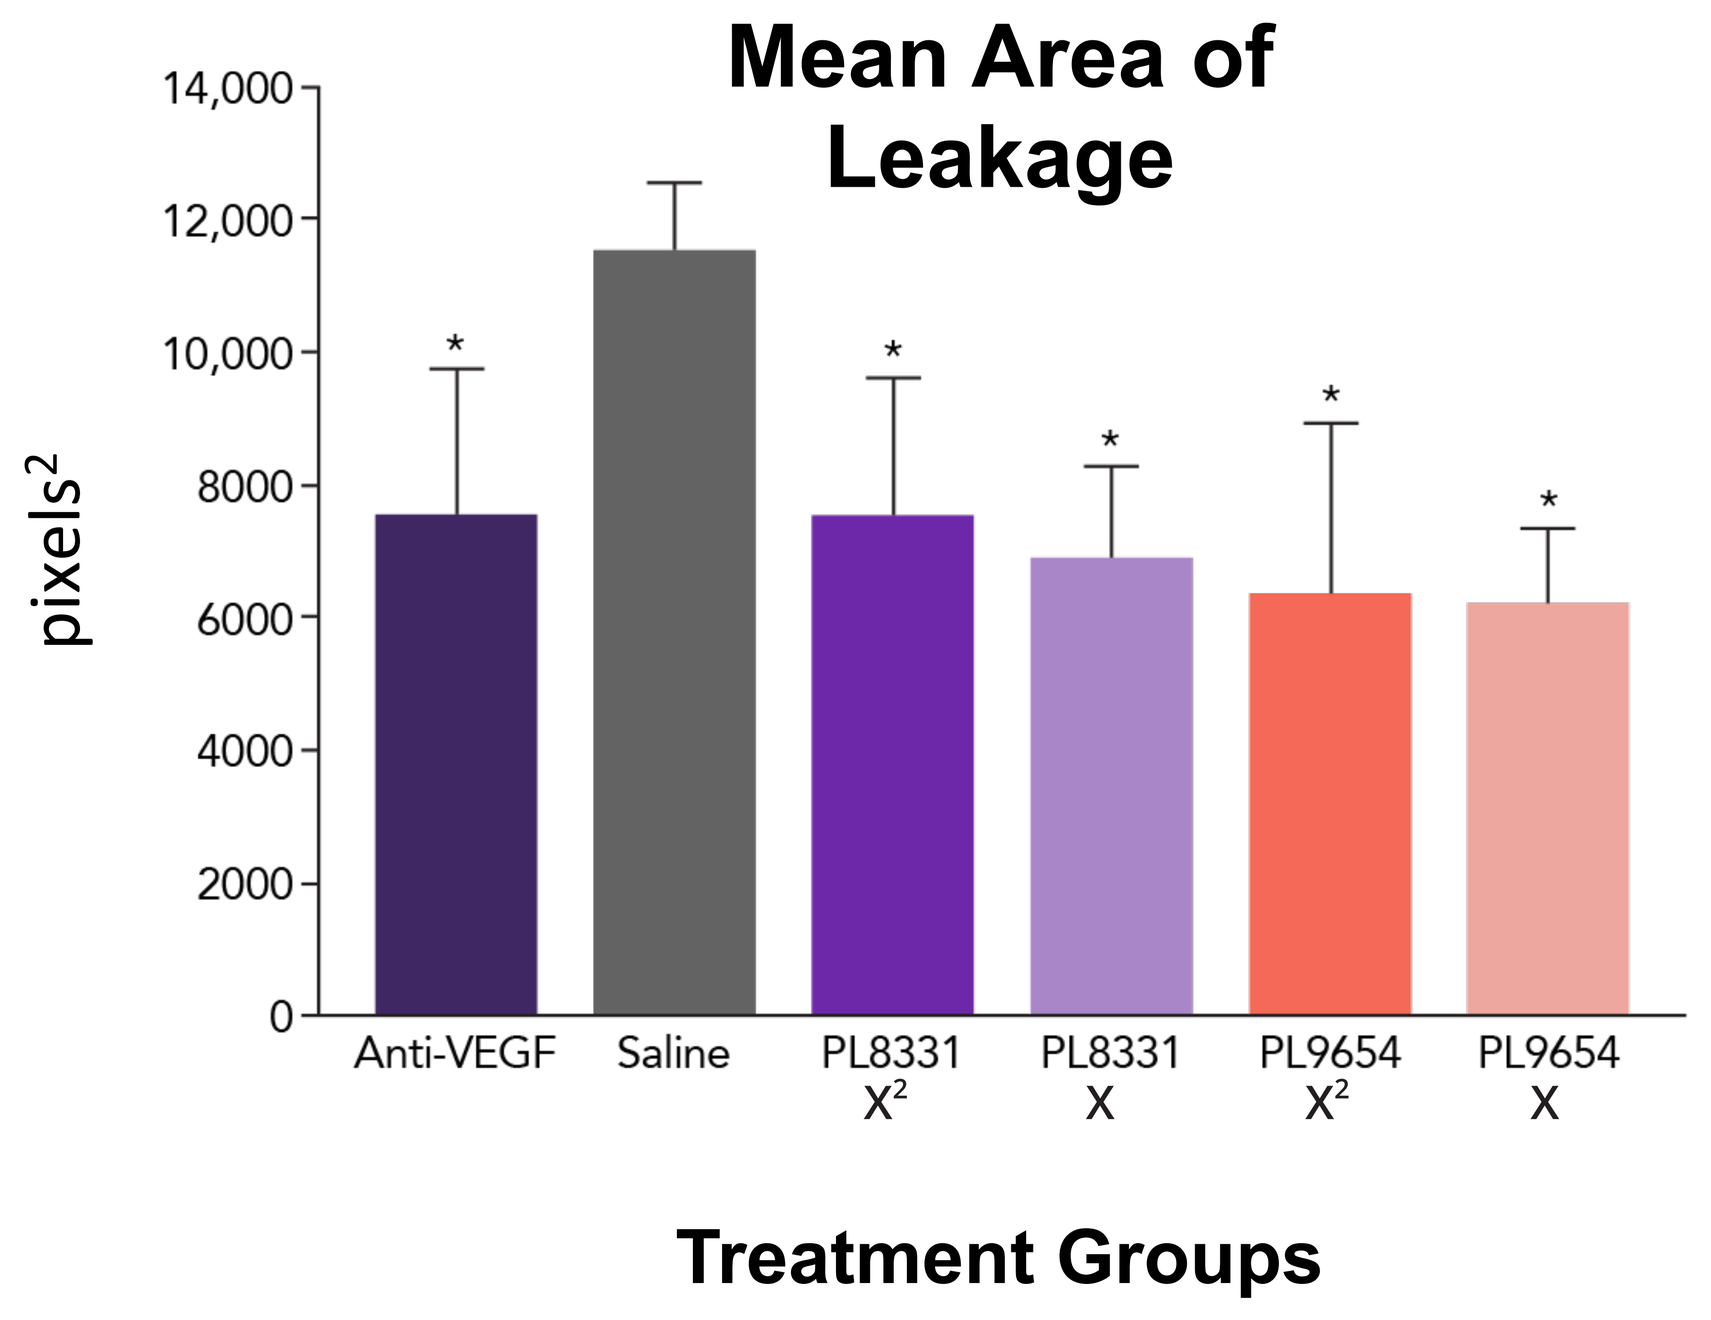
Mean Area of Leakage
pixels²
14,000
12,000
10,000
8000
6000
4000
2000
0
Anti-VEGF
Saline
PL8331 X²
PL8331 X
PL9654 X²
PL9654 X
Treatment Groups

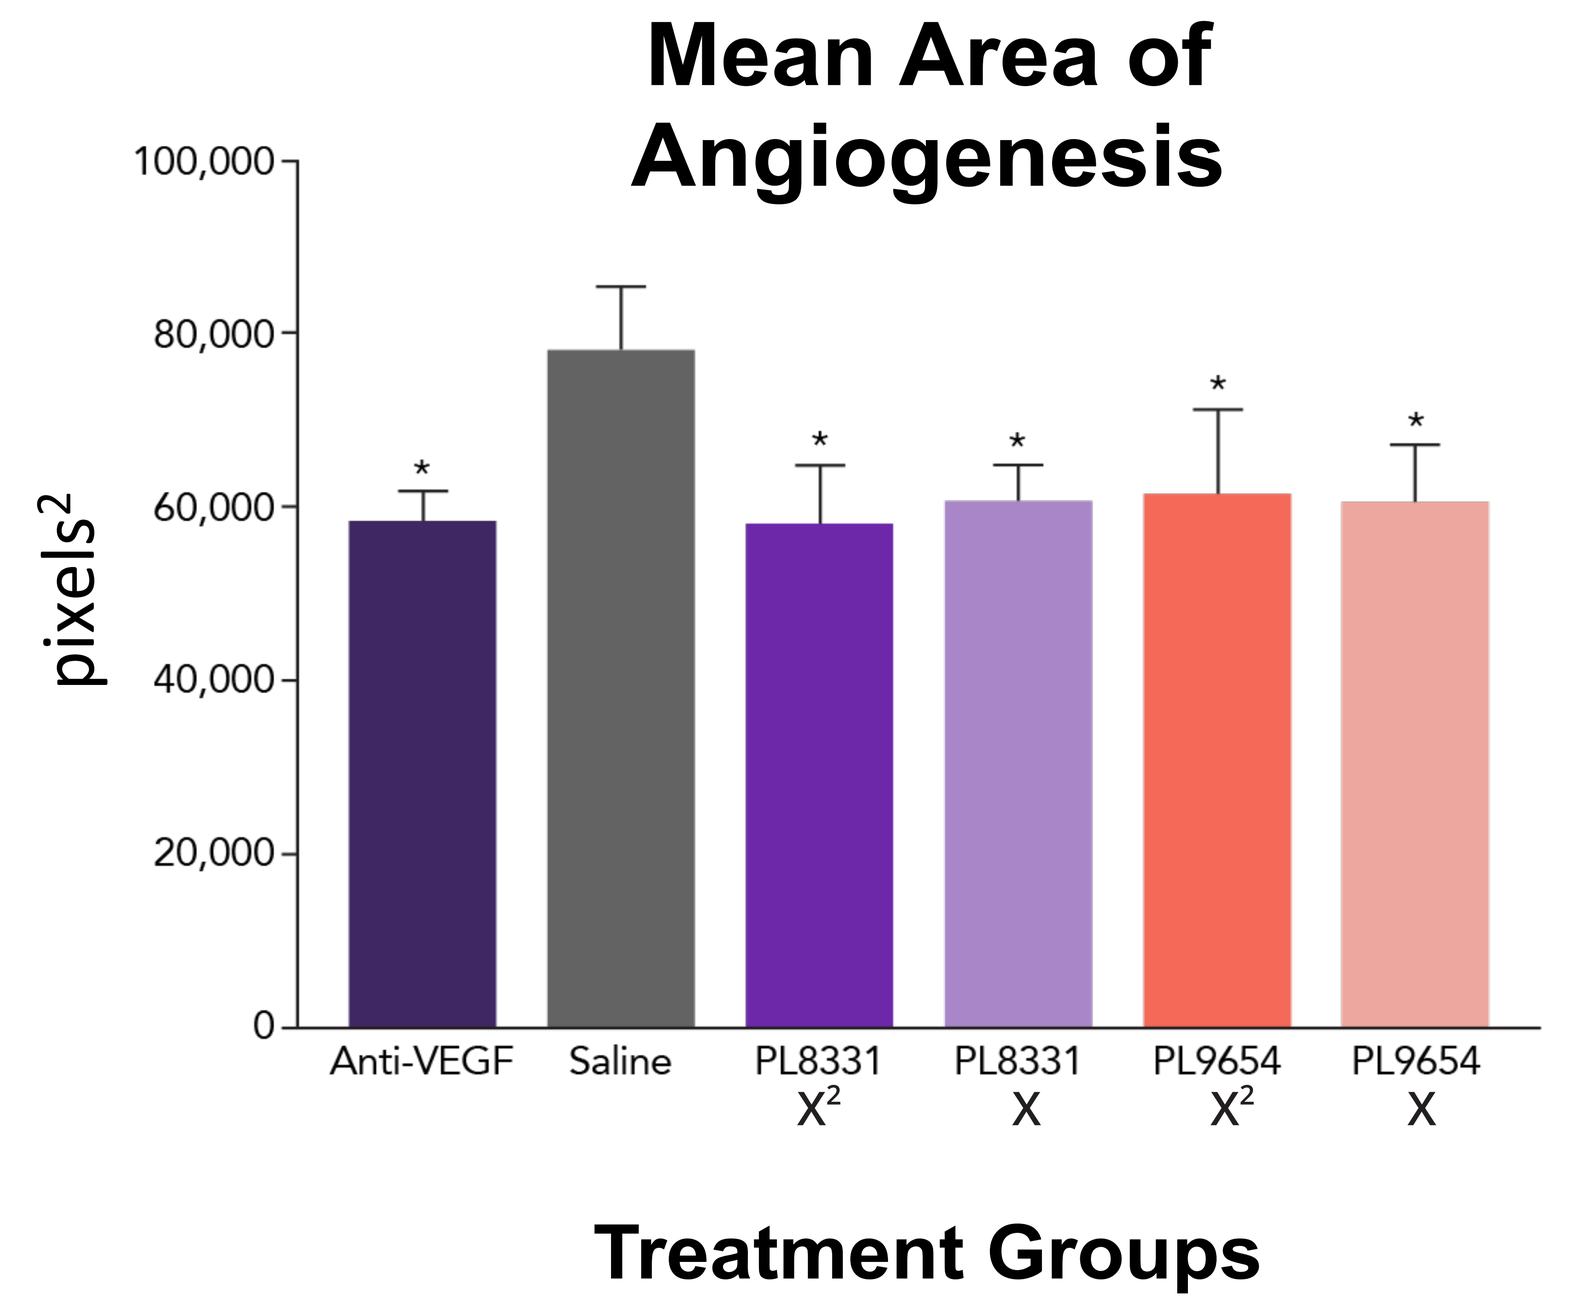
Mean Area of Angiogenesis
pixels²
100,000
80,000
60,000
40,000
20,000
0
Anti-VEGF
Saline
PL8331 X²
PL8331 X
PL9654 X²
PL9654 X
Treatment Groups

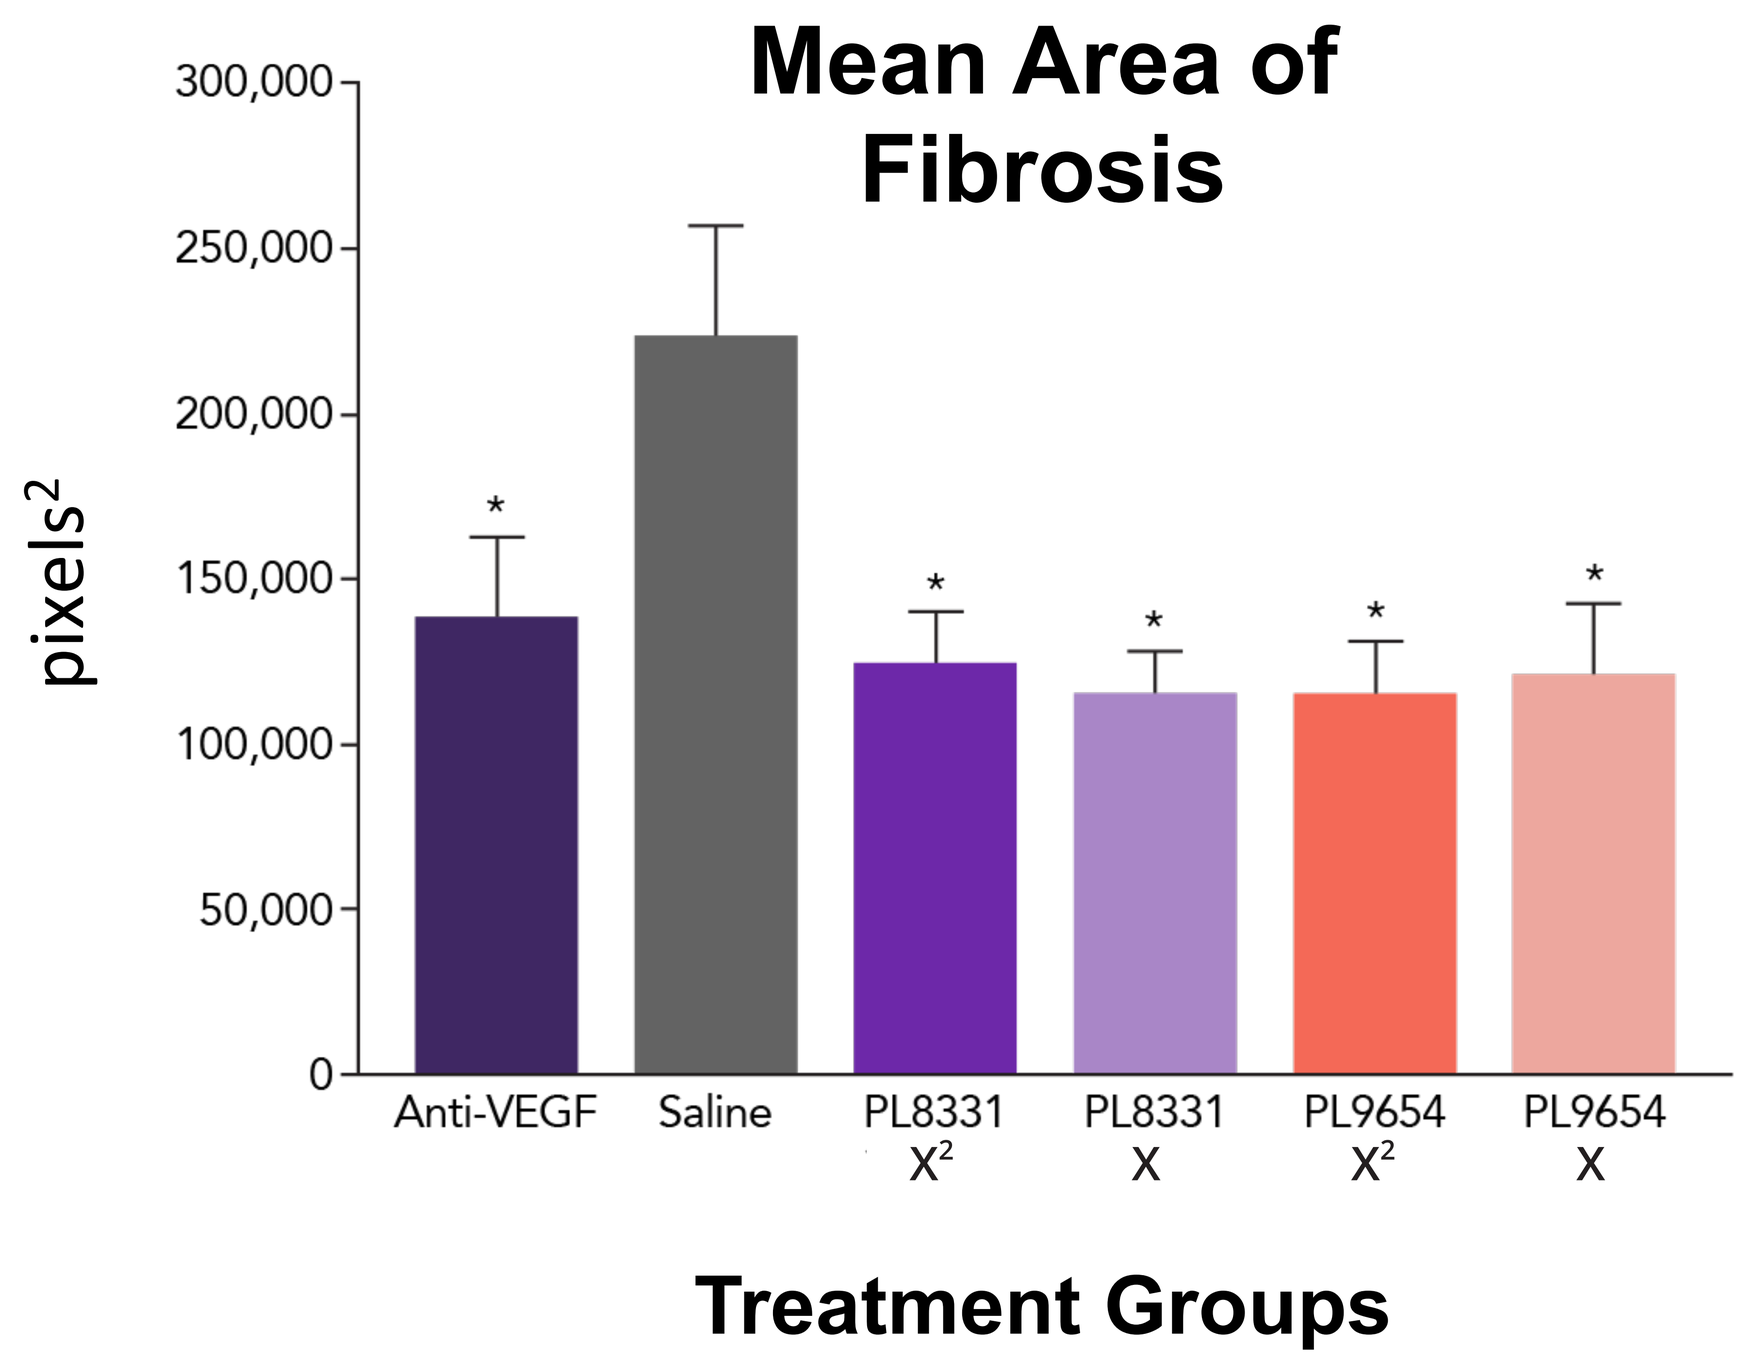
Mean Area of Fibrosis
pixels²
300,000
250,000
200,000
150,000
100,000
50,000
0
Anti-VEGF
Saline
PL8331 X²
PL8331 X
PL9654 X²
PL9654 X
Treatment Groups

CONCLUSIONS

In a mouse model of diabetic retinopathy, monthly IVT treatment with PL8331 preserved retinal structure, suppressed VEGF production, and promoted anti-inflammatory activity.

IVT treatment with the melanocortin agonists PL8331 and PL9654 decreased mean area of leakage, angiogenesis, and fibrosis.

These findings support the continued development of both agents for the treatment of retinal diseases.

Support: Palatin Technologies Inc.

Acknowledgements: Writing and editorial assistance was provided by i2Vision, which was funded by Palatin Technologies Inc.

Disclosures: JD, MM, WHY, and CS are employees of Palatin Technologies Inc. AWT and TFN received consulting fees from Palatin Technologies Inc.

References: 1. Manna SK, et al. J Immunol. 1998;161:2873-2880. 2. Catania A, et al. Pharmacol Rev. 2004;56(1):1-29. 3. Perretti M, et al. Trends Pharmacol Sci. 2015;36(11):737-755. 4. Raap U, et al. J Immunol. 2003;171:353-359. 5. Lipton JM, et al. Ann N Y Acad Sci. 1994;741:137-148. 6. Rajora N, et al. J Neurosci. 1997;17(6):2181-2186.