Sa1222

Presented at:
Digestive Disease Week
Chicago, IL
May 6–9, 2023

Efficacy of the Melanocortin Receptor Agonist PL8177 as a Potential Therapy for Gastrointestinal Inflammatory Diseases

John H. Dodd, PhD[1]; Robert Jordan, BS[1]; Barry Koplowitz, MS[2]; Luana Pesco Koplowitz, MD, PhD[2]; Carl Spana, PhD[1]

[1]Palatin Technologies, Inc., Cranbury, NJ; [2]DUCK FLATS Pharma, Flemington, NJ

Introduction

- The melanocortin 1 receptor (MC1R)–specific agonist PL8177 has demonstrated MC1R binding affinity and functional activity that mirrors that of α-melanocyte stimulating hormone (α-MSH)[1,2]
- α-MSH has been demonstrated to be effective in reducing inflammation in many experimental models[1,3,4]
- Studies in mice and humans have found that MC1R is expressed on the colon luminal surface, and mouse models have demonstrated an important role for MC1R in a dextran sulfate sodium (DSS)–induced model of colitis[5]
- PL8177 has shown significant protective effects vs vehicle (placebo) in rats with dinitrobenzene sulfonic acid (DNBS)–induced colitis after intracolonic administration via cannula of 1.5- and 5-μg doses[1] and after intracolonic treatment with 0.5- and 5-μg doses in the DSS model in rats
- Delayed-release microparticles of PL8177 were developed for oral delivery, allowing PL8177 to withstand the acidic environment of the stomach and release the active drug directly into the colon
- High potency and a lack of systemic absorption make the oral formulation of PL8177 a promising new candidate for clinical development as a treatment for inflammatory bowel disease

Objectives

- Oral PL8177 capsules were tested in DNBS- and DSS-treated rat in vivo models of ulcerative colitis (UC) and evaluated for efficacy and distribution
- Distribution of PL8177 in the gastrointestinal (GI) tracts of dogs and humans was investigated
- This data provided the basis for estimating a suitable dose for an ongoing human phase 2a safety and efficacy trial of oral PL8177 in patients with active UC

Methods

- Seven in vivo studies investigated PL8177 in rats, dogs, and humans, and a phase 2 study is currently recruiting patients (**Table 1**)

UC Induction in Rats (Studies A and B)

- UC was induced in rats by treatment with a single intracolonic delivery of DNBS (30 mg in 0.5 mL 30% ethanol) or DSS (5% in drinking water for 3 days)

Histopathology (Studies A and B)

- Colitis was assessed by diarrhea and rectal bleeding, changes in colon length and weight, and histopathological assessment on termination
- Histological criteria for the analysis of colitis included abnormalities of mucosal architecture, extent of inflammation, erosion or ulceration, epithelial regeneration, and the percentage involvement of the disease process
 - Scoring was based by independent observers by examining 3 colon sections from each animal. Scoring was from 0 = none to 4 = severe for each item, except for percentage involvement of disease process, which was scored from 1 (1%–25%) to 4 (76%–100%). Total scores for colitis resulted in the total colitis index

PL8177 Distribution in Rats and Dogs (Studies C and D)

- Cecum, small and large intestine, colon tissue contents, and feces were collected and homogenized
- PL8177 concentrations were analyzed with high-performance liquid chromatography, and analyte detection was performed using liquid chromatography tandem mass spectrometry

Human Phase 0 Study (Study E)

- The human phase 0 pharmacokinetic study used a single, oral, subclinical 70-μg dose of [14C]-PL8177 to investigate tissue distribution in healthy volunteers
- A laxative (MoviPrep®) was administered at 5, 8, 11, 14 and 17 h postdose. One cohort received no laxative
- Blood, urine, and feces were collected and plasma, urine, and feces pharmacokinetic samples were analyzed for [14C]-PL8177 and any metabolites, and total radioactivity using an ultra-performance liquid chromatography/accelerator mass spectrometry method

Table 1. Summary of In Vivo PL8177 Studies

	Type	Model/Population	N	Objective	Treatments	Assessments
A	Preclinical (3 studies)	DNBS rat	8–12 per treatment group	• Efficacy in UC model	• 10, 20, 50, 100, or 200 μg PL8177 BID for 7 days vs control (placebo) and sulfasalazine (positive control)	• Colon length/weight • Diarrhea and rectal bleeding • Stool consistency • Histopathology of GI tract at termination
B	Preclinical	DSS rat	8–12 per treatment group	• Efficacy in UC model	• 20, 50, 100 μg PL8177 BID for 7 days vs control (placebo) and mesalazine (positive control)	
C	Preclinical	Rat	12	• PL8177 distribution in GI tract	• Single dose of 550-μg PL8177	• PL8177 levels in cecum, small/large intestine, colon, and feces
D	Preclinical	Dog	5	• PL8177 distribution in GI tract	• Single dose of 20-mg PL8177	• PL8177 levels in cecum, stomach, small/large intestine, colon, and feces
E	Human (phase 0, open label)	Healthy human volunteers (aged 18–55 years; body mass index of 18.0–30.0 kg/m²)	24	• To demonstrate release of [14C]-PL8177 from the polymer-encapsulated form of [14C]-PL8177 in the colon after oral administration • Confirmation that oral [14C]-PL8177 does not result in systemic exposure to [14C]-PL8177	• Single, subclinical dose of 70-μg [14C]-PL8177 (35 kBq)	• PL8177 levels in blood, urine, and feces (with and without laxative use) • Safety: AEs, vital signs, lab assessments, and 12-lead ECG
F	Human (phase 2a, double-blind, placebo-controlled)	Adults with active UC	28	• Safety, tolerability, efficacy, pharmacokinetics/biomarker study	• Oral 20-mg PL8177 QD • Placebo	• Recruiting

AE, adverse event; BID, twice daily; DNBS, dinitrobenzene sulfonic acid; DSS, dextran sodium sulfate; ECG, electrocardiogram; GI, gastrointestinal; QD, once daily; UC, ulcerative colitis.

Phase 2a Study (Study F, Recruiting)

Title	A phase 2a, double-blind, randomized adaptive design, placebo-controlled, parallel group study to evaluate the safety, tolerability, efficacy, pharmacokinetics, and biomarkers with oral colon delivery PL8177 in adults with active UC
Trial number ClinicalTrials.gov identifier	PL8177-205 NCT05466890
Treatments	PL8177, 20 mg by mouth (PO) daily for 56 days; placebo, PO daily for 56 days
Duration	Screening: ~4 weeks. Dosing duration: 56 days (8 weeks). Post-treatment follow-up: 4 weeks. Total duration: up to ~16 weeks
Number of sites	~25 (US only)
Target enrollment	28 patients
Contact	rjordan@palatin.com

Results

DNBS Colitis Rat Model

- Colonic macroscopic damage scores and colon/body weight ratio were reduced, and the ulcer/inflammatory subscore and stool appearance were improved in rats treated with PL8177 compared to vehicle (placebo)
- Effects of the 50-μg dose of PL8177 on colonic damage (total score and ulcer/inflammatory subscore) were significant ($P<0.05$) (**Figure 1**)

Figure 1. Percentage Differences From Vehicle Across PL8177 Doses for the Macroscopic Damage Score (A) and Ulcer/Inflammatory Score (B) in DNBS-Treated Rats

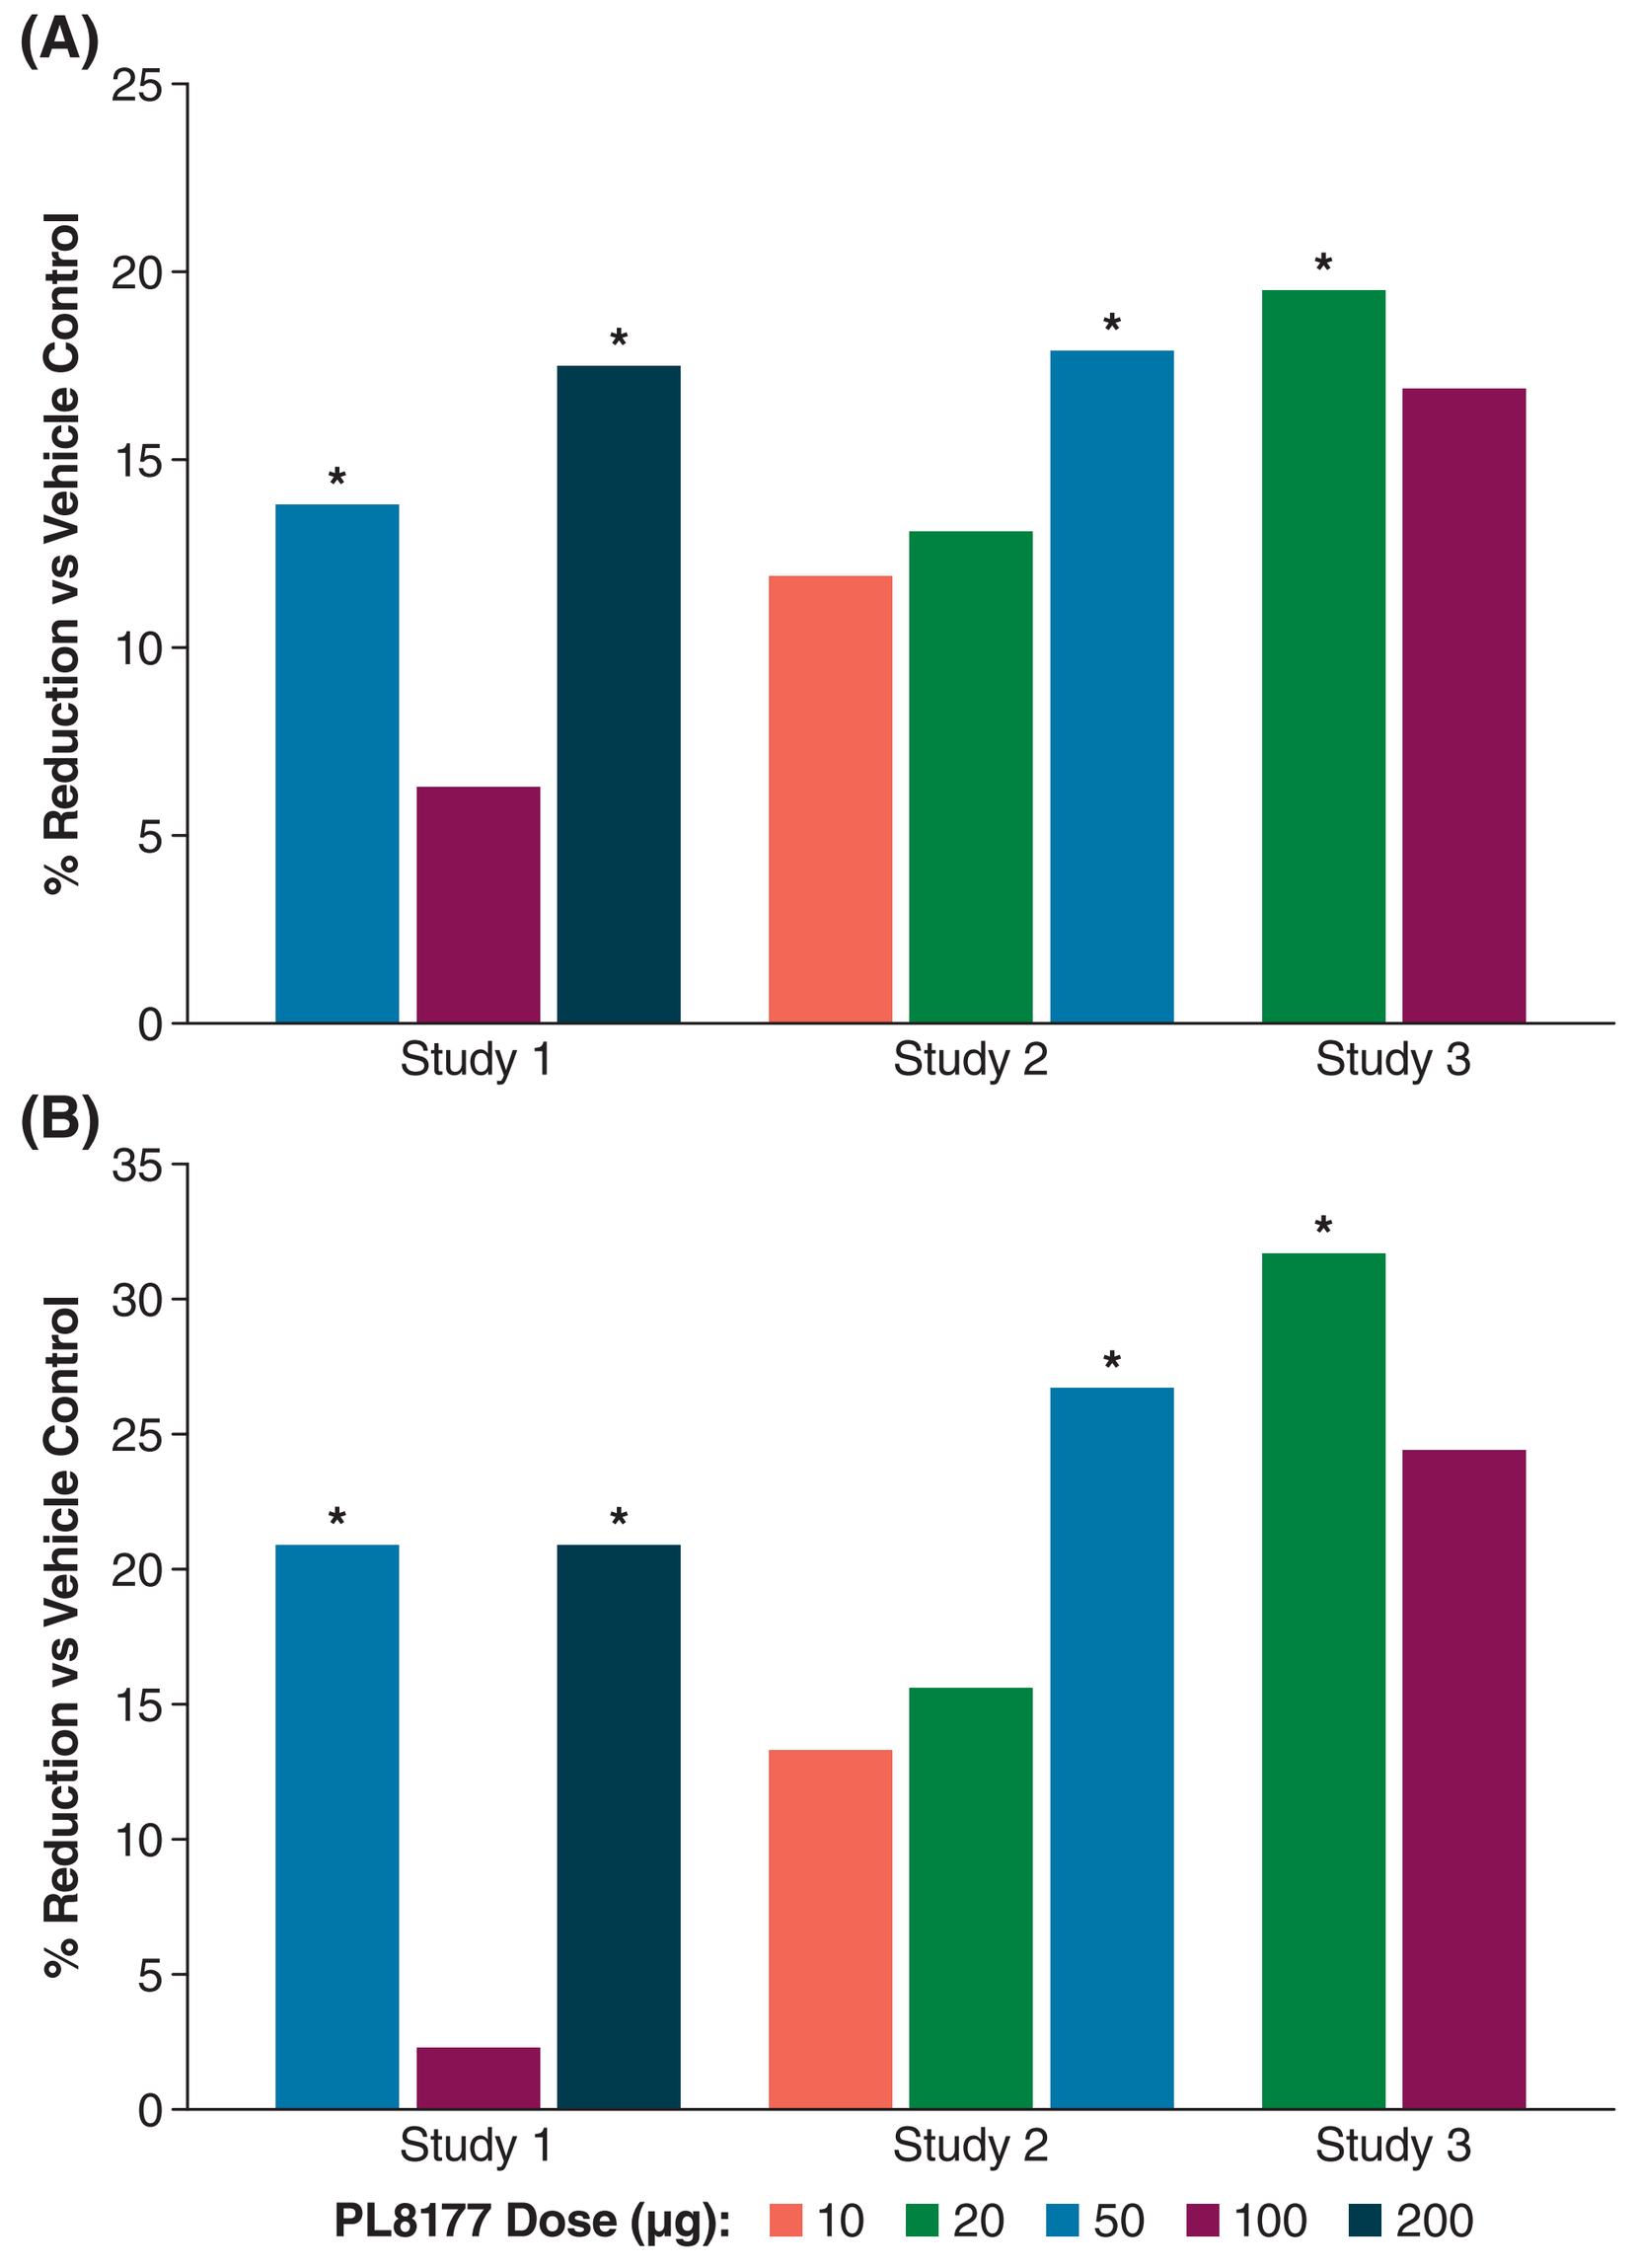

(A)
% Reduction vs Vehicle Control
Study 1
Study 2
Study 3
(B)
PL8177 Dose (μg): 10 20 50 100 200

*$P<0.05$, treated vs vehicle control; 1-way analysis of variance followed by Dunnett's test.
DNBS, dinitrobenzene sulfonic acid.

- Overall, activity was observed from 20 μg to 200 μg. The 20- and 50-μg doses were consistently efficacious in this rat model of colitis, informing dosing in human studies

DSS Colitis Rat Model

- Oral PL8177 at 50 μg/animal showed significant ($P<0.05$) improvement in stool consistency score from day 5 to day 8 and significant ($P<0.05$) improvement in fecal occult blood score on day 8 when compared to the vehicle group (**Figure 2**)
- There was a significant ($P<0.05$) improvement observed in the total colitis index for the PL8177 100-μg group compared to the vehicle control group, and all PL8177 cohorts showed improvement compared to vehicle (**Figure 3**)
- The decline in the total colitis index for mesalazine-treated rats was less than that observed in any of the PL8177-treated rats
- Oral PL8177 (50 μg/animal) treatment showed a significant improvement in colon weight (53% reduction) vs vehicle ($P<0.05$)
- Oral mesalazine 300 mg/kg (positive control) was associated with significant reduction in colon length but only moderate improvement (35%) in colon weight gain
- Histopathology analysis showed PL8177 treatment resulted in the maintenance of intact colon structure and barrier and reduced immune cell infiltration

PL8177 Distribution in Rats and Dogs

- In both the rat and dog studies, PL8177 was detected in higher amounts in the colon vs the upper GI tract
- In rats, the mean amount of PL8177 was highest in the jejunum, ileum, and cecum at 3 h postdose (25.7 μg–33.8 μg) and was highest in the colon at 6 h (33.2 μg) and 10 h (19.8 μg)
- In dogs, mean total PL8177 concentrations at 6 h after a single oral dose were very low (≤0.8 μg/g) in the upper GI tract tissues (stomach, duodenum, jejunum, ileum); the highest concentrations were observed in the transverse colon, descending colon, rectum, ascending colon, and cecum (mean ± SEM, 24.2±1.7, 16.9±3.2, 15.0±6.3, 10.4±2.9, and 6.4±1.9 μg/g, respectively)
 - Percentage of the total oral PL8177 dose in the colon at 6 h (4.2%) was ≥22-fold greater than the percentage observed across upper GI tract organs
 - PL8177 and its main metabolite PL8435 were below the lower limits of quantification in plasma and urine.

Figure 2. Changes in Stool Consistency (A) and Fecal Occult Blood (B) in DSS-Treated Rats

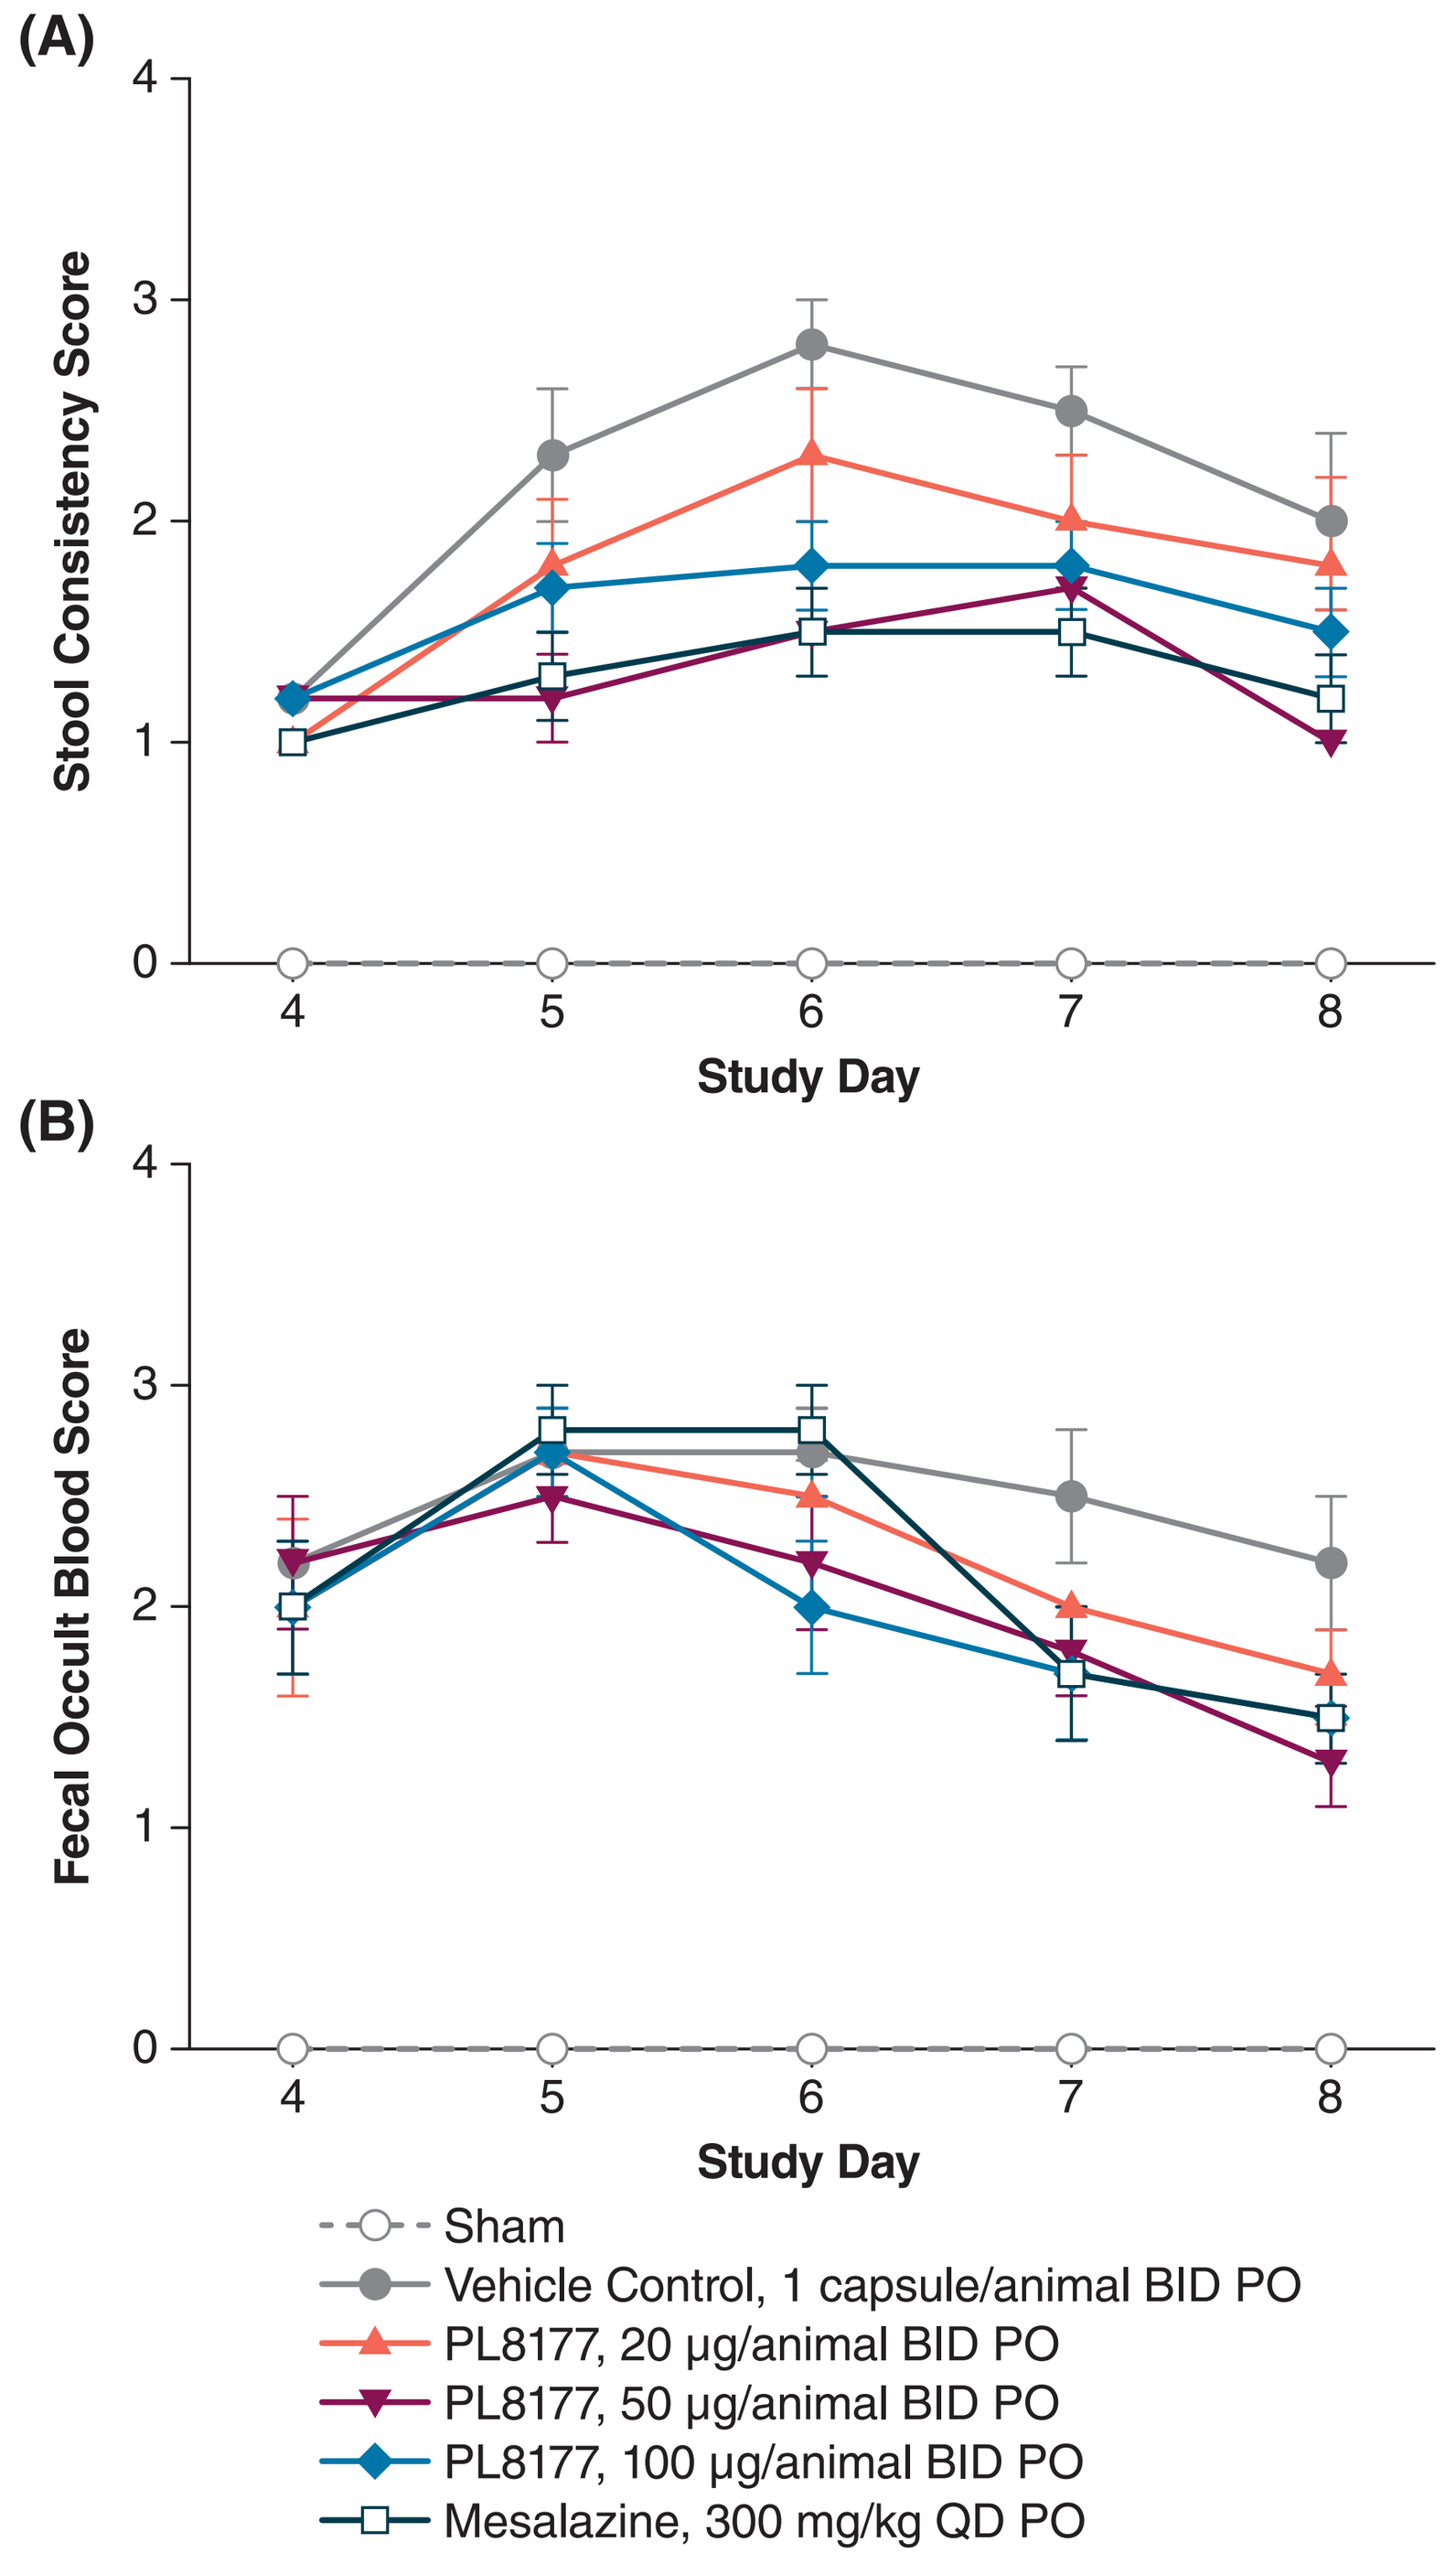

(A)
Stool Consistency Score
Study Day
(B)
Fecal Occult Blood Score
Study Day
Sham
Vehicle Control, 1 capsule/animal BID PO
PL8177, 20 μg/animal BID PO
PL8177, 50 μg/animal BID PO
PL8177, 100 μg/animal BID PO
Mesalazine, 300 mg/kg QD PO

BID, twice daily; DSS, dextran sulfate sodium; PO, by mouth; QD, once daily. Note: all animals, except those in sham group, received 5% DSS in the drinking water for 3 days, from day 1 to day 3, and then changed to normal drinking water for the following 5 days. Tissue harvest occurred on day 8. Mesalazine is a positive control. Data are mean (SEM).

Figure 3. Total Colitis Index on Day 8 in DSS-Treated Rats

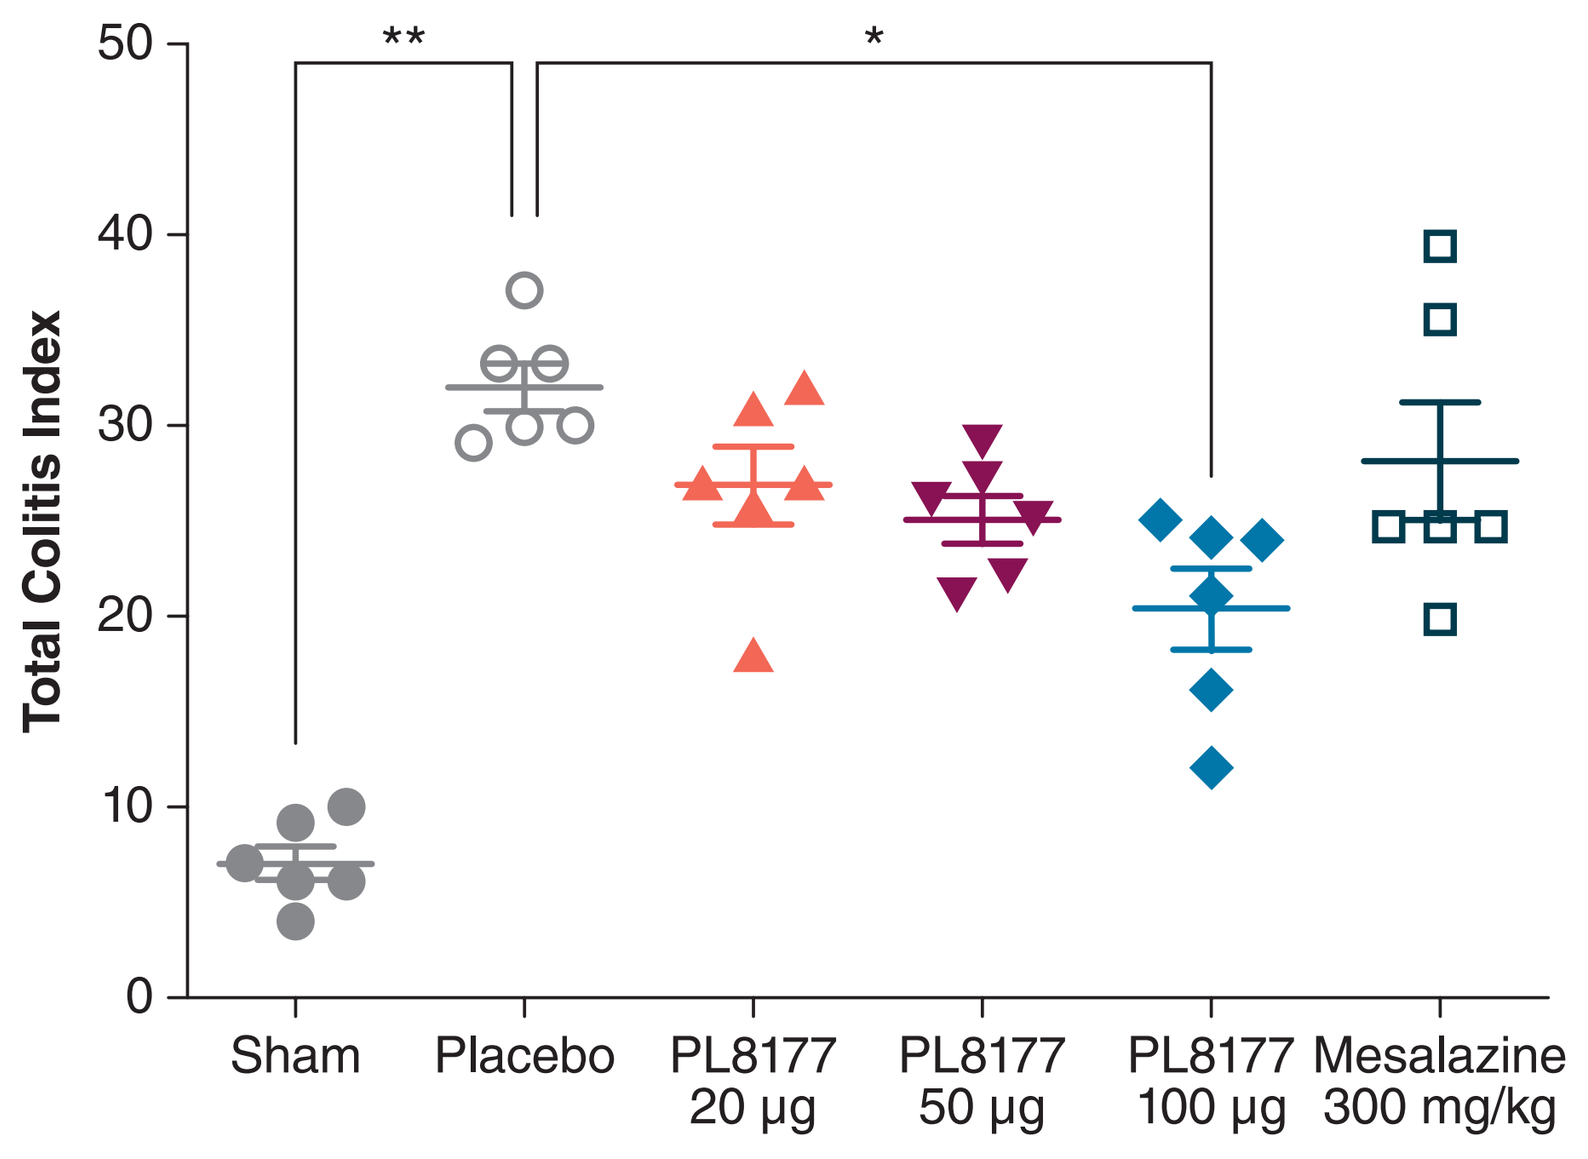

Total Colitis Index
**
*
Sham
Placebo
PL8177 20 μg
PL8177 50 μg
PL8177 100 μg
Mesalazine 300 mg/kg

*$P<0.01$, **$P<0.05$. DSS, dextran sulfate sodium. Note: all animals, except those in sham group, received 5% DSS in the drinking water for 3 days, from day 1 to day 3, and then changed to normal drinking water for the following 5 days. Tissue harvest occurred on day 8. Mesalazine is a positive control.

Human Phase 0 Study

- The human phase 0 pharmacokinetic study used a single, subclinical 70-μg dose of [14C]-PL8177 to investigate tissue distribution in healthy volunteers
- Neither PL8177 nor its main metabolite (PL8435) were detected in plasma or urine following oral administration of [14C]-PL8177 in a capsule delayed-release formulation, suggesting that PL8177 does not reach the systemic circulation
- In plasma and urine, the majority of radio-activity was identified as [14C]-phenylalanine, the radiolabeled amino acid component of [14C]-PL8177
- Radioactivity was seen in plasma approximately 1 h postdose, consistent with release of PL8177 from the polymer in the small intestine and degradation into absorbable peptide fragments
- Approximately 20%–30% of the dose was recovered in urine at 24 h postdose, regardless of laxative administration
- PL8177's main active metabolite (PL8435) was detected in feces, indicating release of PL8177 from the polymer. Approximately 30%–50% of the radioactivity was detected in feces
- This is consistent with rapid degradation into peptide fragments, which were then either absorbed or eliminated in feces. Fecal elimination was higher when PL8177 was co-administered with a laxative

Safety in Phase 0 Study

- Treatment-emergent adverse events (AEs) (>10%) in the human study were consistent with laxative use (abdominal pain, diarrhea, flatulence, nausea, vomiting, fatigue, and dizziness). No AE was considered related to [14C]-PL8177 and there were no changes observed in clinical laboratory parameters, vital signs, 12-lead electrocardiogram, or physical examinations

Phase 2 Study (Recruiting)

A phase 2, double-blind, placebo-controlled study will evaluate the safety, tolerability, and efficacy of oral PL8177 in adult males and nonpregnant, nonlactating females with active UC (**Figure 4**)

- The most efficacious doses ($P<0.05$ vs vehicle) in the rat studies were 20 μg and 50 μg BID, which provide the basis for estimating suitable doses for the phase 2 trial. Colonic surface area for humans is ~3,000 times greater than rat. Considering species-specific MC1R receptor sensitivity differences and other variables, a dose of 20 mg/day was estimated to be the best choice for observation of efficacy in the phase 2 trial
- The rationale for selecting the PL8177 20-mg daily dose is shown in **Figure 5**

Figure 4. Study Design for Phase 2a Study (PL8177-205)

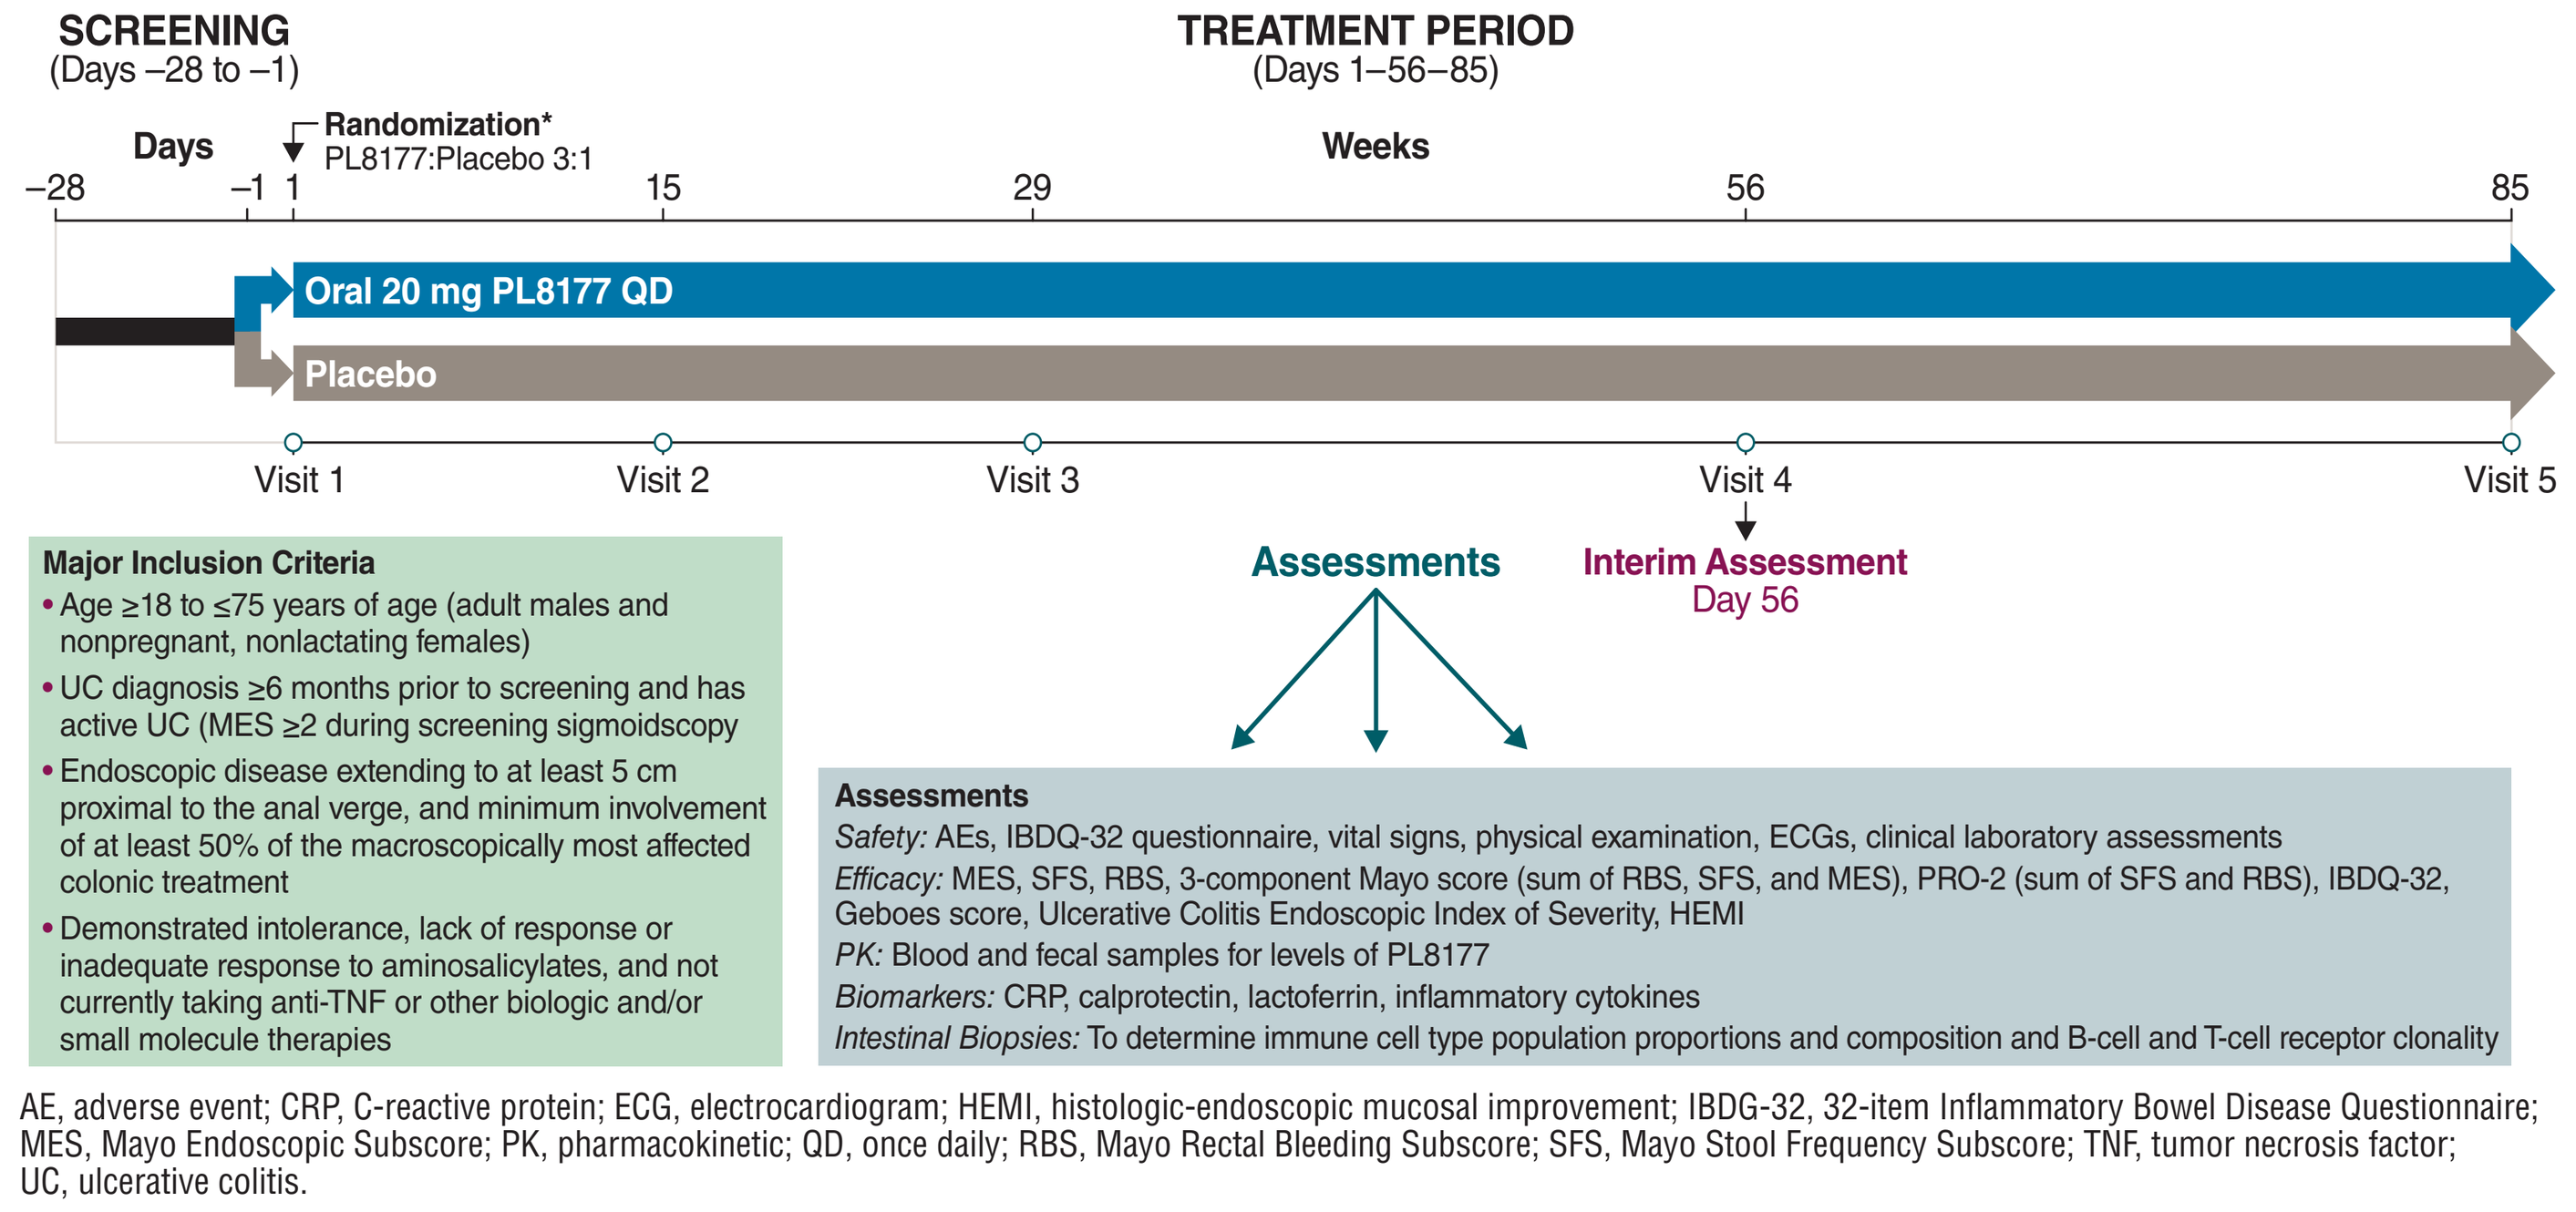

SCREENING
(Days −28 to −1)
TREATMENT PERIOD
(Days 1–56–85)
Days
Randomization*
PL8177:Placebo 3:1
Weeks
−28
−1
1
15
29
56
85
Oral 20 mg PL8177 QD
Placebo
Visit 1
Visit 2
Visit 3
Visit 4
Visit 5
Major Inclusion Criteria
• Age ≥18 to ≤75 years of age (adult males and nonpregnant, nonlactating females)
• UC diagnosis ≥6 months prior to screening and has active UC (MES ≥2 during screening sigmoidscopy
• Endoscopic disease extending to at least 5 cm proximal to the anal verge, and minimum involvement of at least 50% of the macroscopically most affected colonic treatment
• Demonstrated intolerance, lack of response or inadequate response to aminosalicylates, and not currently taking anti-TNF or other biologic and/or small molecule therapies
Assessments
Interim Assessment
Day 56
Assessments
Safety: AEs, IBDQ-32 questionnaire, vital signs, physical examination, ECGs, clinical laboratory assessments
Efficacy: MES, SFS, RBS, 3-component Mayo score (sum of RBS, SFS, and MES), PRO-2 (sum of SFS and RBS), IBDQ-32, Geboes score, Ulcerative Colitis Endoscopic Index of Severity, HEMI
PK: Blood and fecal samples for levels of PL8177
Biomarkers: CRP, calprotectin, lactoferrin, inflammatory cytokines
Intestinal Biopsies: To determine immune cell type population proportions and composition and B-cell and T-cell receptor clonality

AE, adverse event; CRP, C-reactive protein; ECG, electrocardiogram; HEMI, histologic-endoscopic mucosal improvement; IBDG-32, 32-item Inflammatory Bowel Disease Questionnaire; MES, Mayo Endoscopic Subscore; PK, pharmacokinetic; QD, once daily; RBS, Mayo Rectal Bleeding Subscore; SFS, Mayo Stool Frequency Subscore; TNF, tumor necrosis factor; UC, ulcerative colitis.

*16 patients (12 PL8177, 4 placebo) will be enrolled for treatment until visit 4 (day 56) when data will be assessed in an interim assessment, after which an additional 12 patients could be enrolled or, depending on the conclusions of the interim assessment, additional patients beyond the target sample of 28 patients could be enrolled to a maximum of 84 patients.

Figure 5. Flow Chart for Determination of PL8177 Dose in the Phase 2a Study

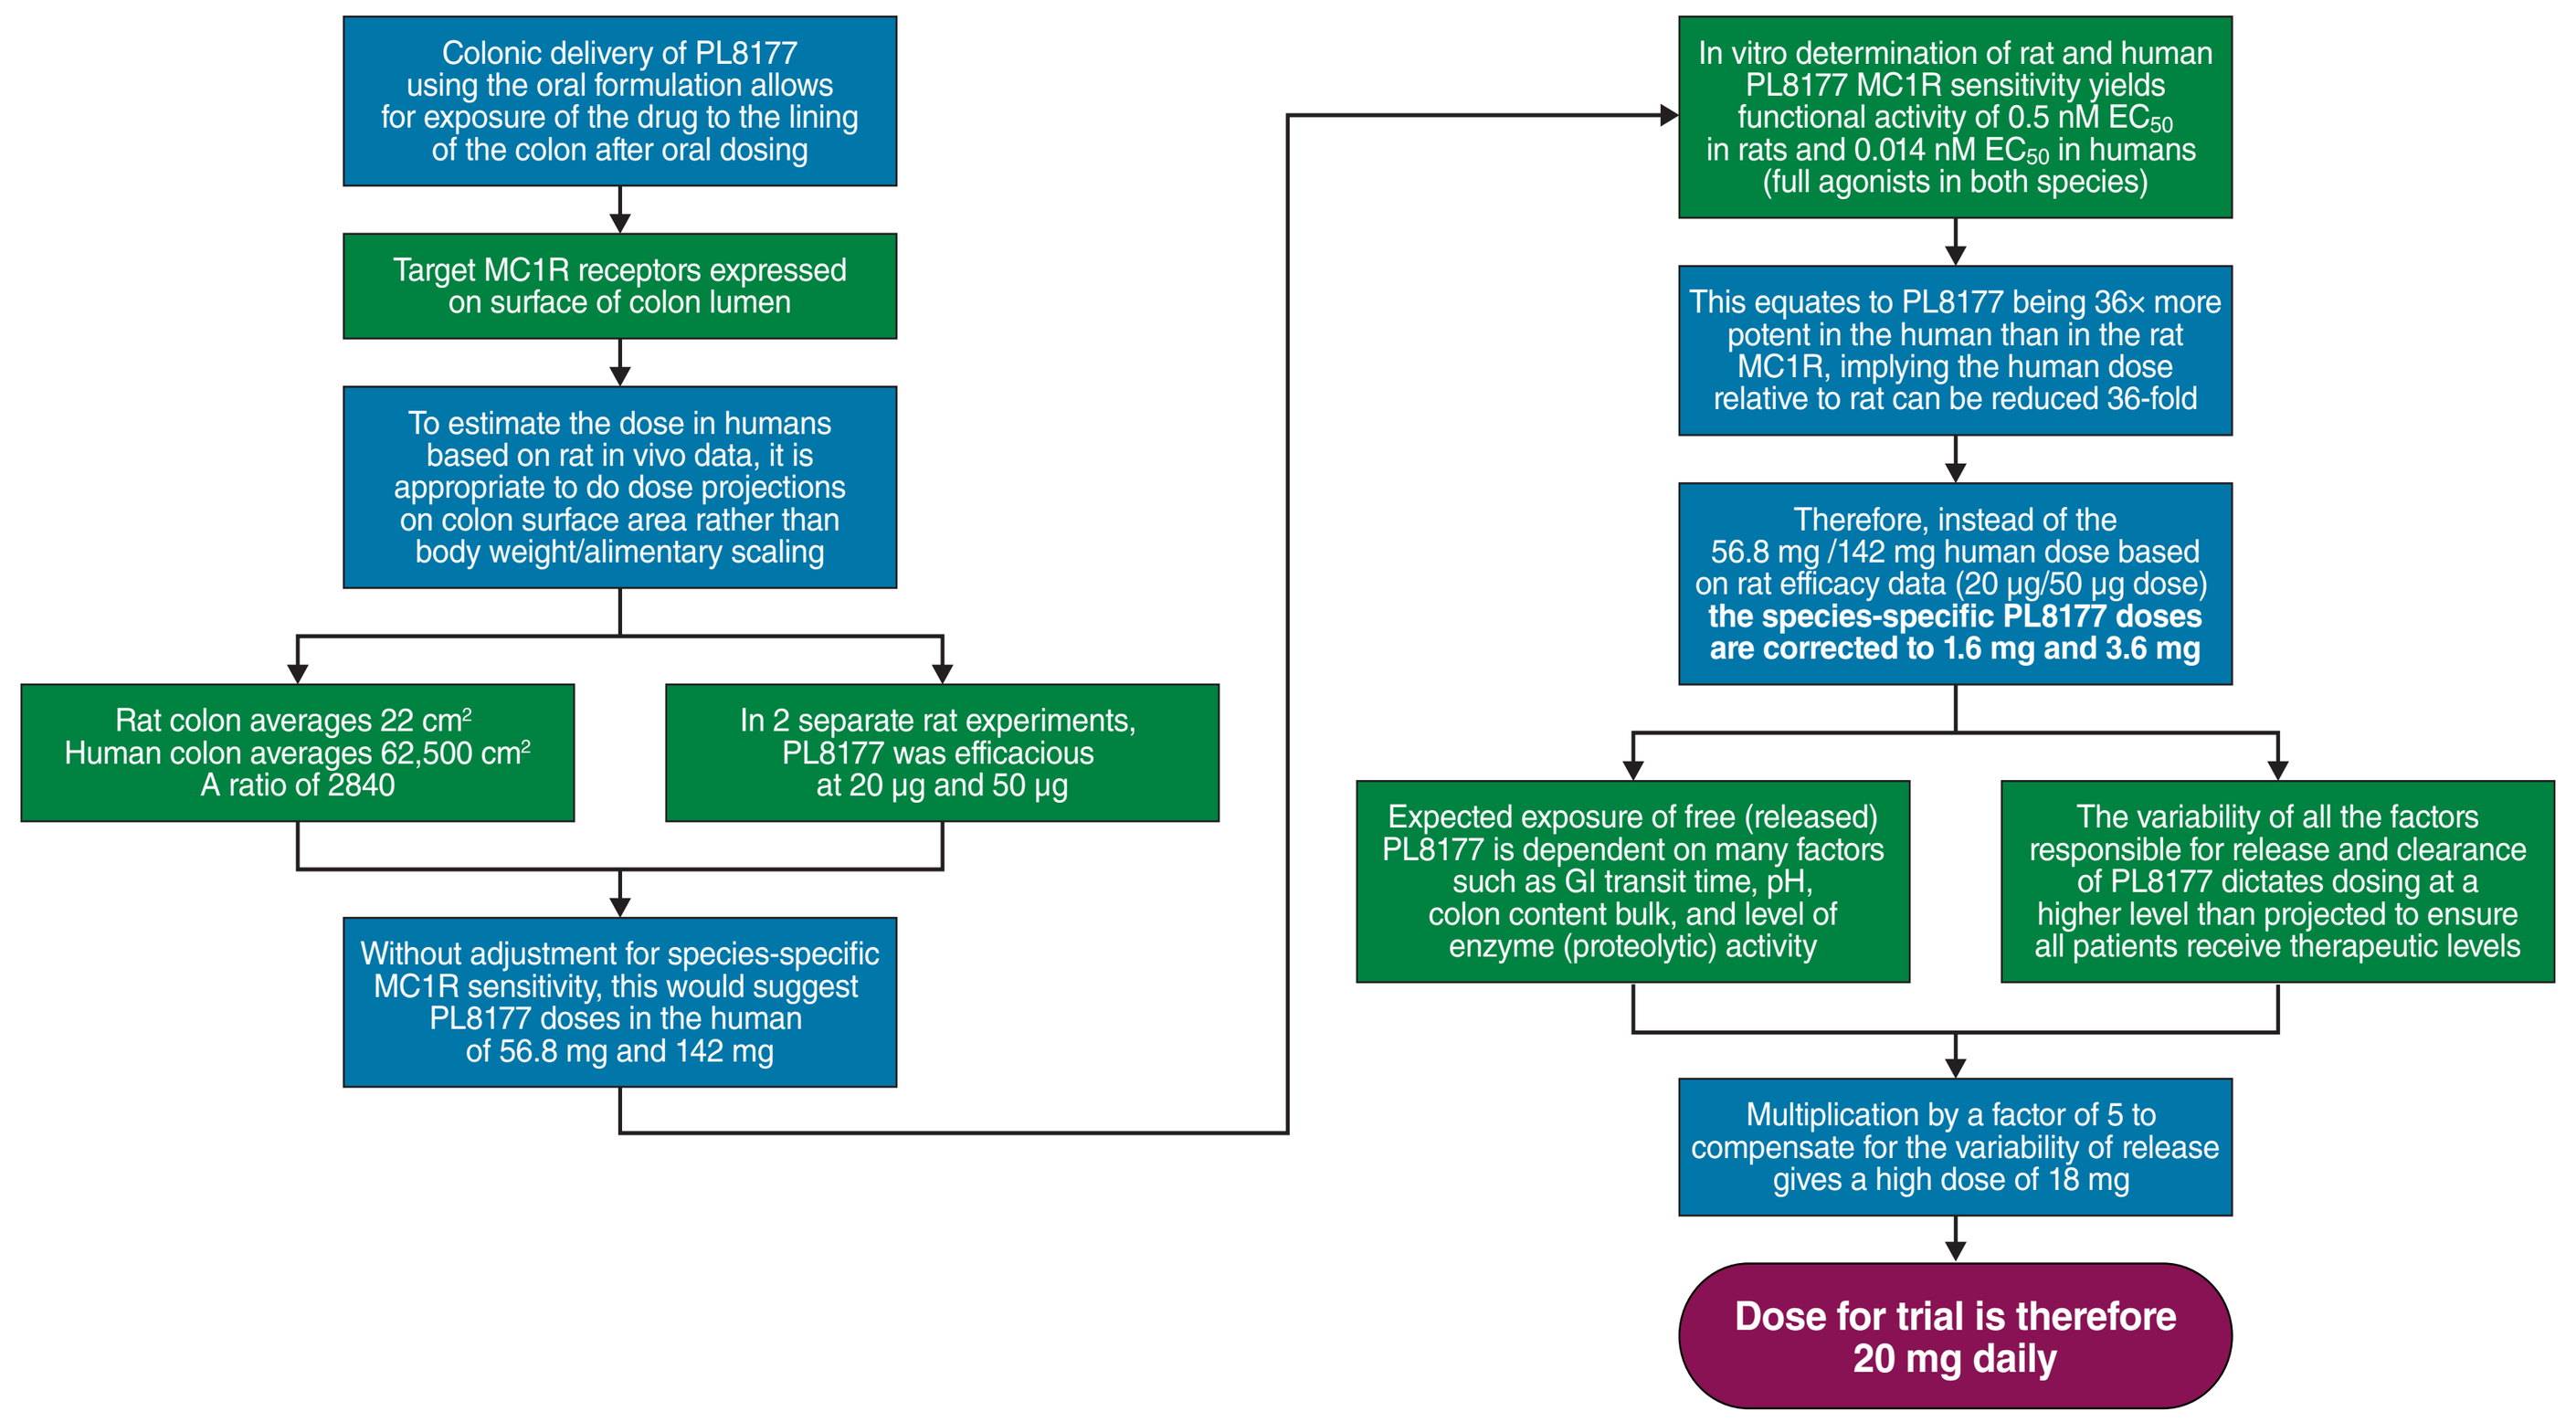

Colonic delivery of PL8177 using the oral formulation allows for exposure of the drug to the lining of the colon after oral dosing
Target MC1R receptors expressed on surface of colon lumen
To estimate the dose in humans based on rat in vivo data, it is appropriate to do dose projections on colon surface area rather than body weight/alimentary scaling
Rat colon averages 22 cm²
Human colon averages 62,500 cm²
A ratio of 2840
In 2 separate rat experiments, PL8177 was efficacious at 20 μg and 50 μg
Without adjustment for species-specific MC1R sensitivity, this would suggest PL8177 doses in the human of 56.8 mg and 142 mg
In vitro determination of rat and human PL8177 MC1R sensitivity yields functional activity of 0.5 nM EC50 in rats and 0.014 nM EC50 in humans (full agonists in both species)
This equates to PL8177 being 36× more potent in the human than in the rat MC1R, implying the human dose relative to rat can be reduced 36-fold
Therefore, instead of the 56.8 mg /142 mg human dose based on rat efficacy data (20 μg/50 μg dose) the species-specific PL8177 doses are corrected to 1.6 mg and 3.6 mg
Expected exposure of free (released) PL8177 is dependent on many factors such as GI transit time, pH, colon content bulk, and level of enzyme (proteolytic) activity
The variability of all the factors responsible for release and clearance of PL8177 dictates dosing at a higher level than projected to ensure all patients receive therapeutic levels
Multiplication by a factor of 5 to compensate for the variability of release gives a high dose of 18 mg
Dose for trial is therefore 20 mg daily

EC_{50}, half maximal effective concentration; GI, gastrointestinal; MC1R, melanocortin 1 receptor.

Conclusions

- High potency and a lack of systemic absorption make a delayed-release microparticle oral formulation of the MC1R agonist PL8177 a promising new candidate for clinical development for the treatment of inflammatory bowel disease
- In rat animal models of UC, oral PL8177 was found to be efficacious in reducing colonic damage and inflammation and improving stool consistency and fecal occult blood; results were supported by histopathological analysis
- In rats and dogs, PL8177 was detected in higher amounts in the colon vs the upper GI tract, showing that PL8177 is protected from metabolic breakdown and released in the lower GI tract where it can exert its effect
- A human study using [14C]-PL8177 showed that PL8177 did not reach the systemic circulation; it was released in the GI tract and detected in the feces
- As the next step in the clinical development of an oral formulation of PL8177 for the treatment of UC, a phase 2, double-blind safety and efficacy trial is currently recruiting patients with active UC. The oral PL8177 dose in the trial will be 20 mg per day

Support Palatin Technologies, Inc.

Acknowledgments Writing and editorial assistance was provided by Robin Smith, PhD, of The Curry Rockefeller Group, LLC (Tarrytown, NY, USA), which was funded by Palatin Technologies, Inc.

Disclosures John H. Dodd, Robert Jordan, and Carl Spana are employees of Palatin Technologies, Inc. Barry Koplowitz and Luana Pesco Koplowitz are paid consultants for Palatin Technologies, Inc.

References **1.** Spana C, et al. *Front Pharmacol.* 2018;9:1535; **2.** Dodd J, et al. *Drugs R D.* 2021;21(4):431-443; **3.** Ahmed TJ, et al. *Int J Inflam.* 2013;2013:985815; **4.** Wang W, et al. *Front Endocrinol (Lausanne).* 2019;10:683; **5.** Maaser C, et al. *Gut.* 2006;55(10):1415-1422.