087

Presented at:
14th Biennial International Podocyte Conference
Philadelphia, PA
May 24–26, 2023

Melanocortin 1 Receptor (MC1R) Agonist PL8177 Protects Against Podocyte Loss in a Streptozotocin-Induced Rat Model of Diabetic Nephropathy

Luipa Khandker, PhD; Priyanka Dhingra, PhD; John H. Dodd, PhD; Carl Spana, PhD; Paul S. Kayne, PhD

Palatin Technologies, Inc., Cranbury, NJ

Introduction

- Melanocortins are a family of neuropeptide hormone agonists that include several melanocyte-stimulating hormones (MSH) and adrenocorticotropin hormone (ACTH)[1-3]
- The melanocortin pathway plays an important role in resolving inflammation, promoting tissue healing processes, and maintaining immunological homeostasis[1,3]
- A growing body of evidence suggests that melanocortins have a protective role in many kidney diseases[4]
- Melanocortin-1 receptors (MC1Rs) are expressed in several kidney cell types, including podocytes, glomerular endothelial cells, mesangial cells, and tubular epithelial cells[4,5]
- PL8177 (Palatin Technologies, Inc) is a potent MC1R selective agonist that demonstrates binding characteristics similar to those of α-MSH, and it shows promise as a therapy that promotes the resolution of inflammation and tissue repair[6-8]
- PL8177 was investigated in a rat model of diabetic nephropathy to determine effects on kidney cell populations and on glomerular pathology

Acknowledgments Writing and editorial assistance was provided by Robin Smith, PhD, of The Curry Rockefeller Group, LLC (Tarrytown, NY, USA), which was funded by Palatin Technologies, Inc. We are grateful to Dr. Agnes Fogo for extensive discussions on immunohistochemistry data analysis of the kidney.

Support Palatin Technologies, Inc.

Disclosures Luipa Khandker, Priyanka Dhingra, John H. Dodd, Carl Spana, and Paul S. Kayne are employees of Palatin Technologies, Inc.

References **1.** Ahmed TJ, et al. *Int J Inflam*. 2013;2013: 985815. **2.** Perretti M, et al. *Trends Pharmacol Sci*. 2015;36(11): 737-755. **3.** Wang W, et al. *Front Endocrinol (Lausanne)*. 2019;10:683. **4.** Chang M, et al. *Front Physiol*. 2021;12:651236. **5.** Gong R. *Adv Chronic Kidney Dis*. 2014;21(2):134-151. **6.** Dodd J, et al. *Front Immunol*. 2023;14:1083333. **7.** Dodd J, et al. *Drugs R D*. 2021;21(4):431-443. **8.** Spana C, et al. *Front Pharmacol*. 2018;9:1535.

Methods

- Diabetes was induced in Norwegian brown rats by a single 50-mg/kg intraperitoneal injection of streptozotocin
- Diabetic rats were treated twice daily on days 4–113 with subcutaneously administered PL8177 (1.0 mg/kg) or vehicle (**Figure 1**)

Figure 1. Streptozotocin-Induced Diabetic Rat Model

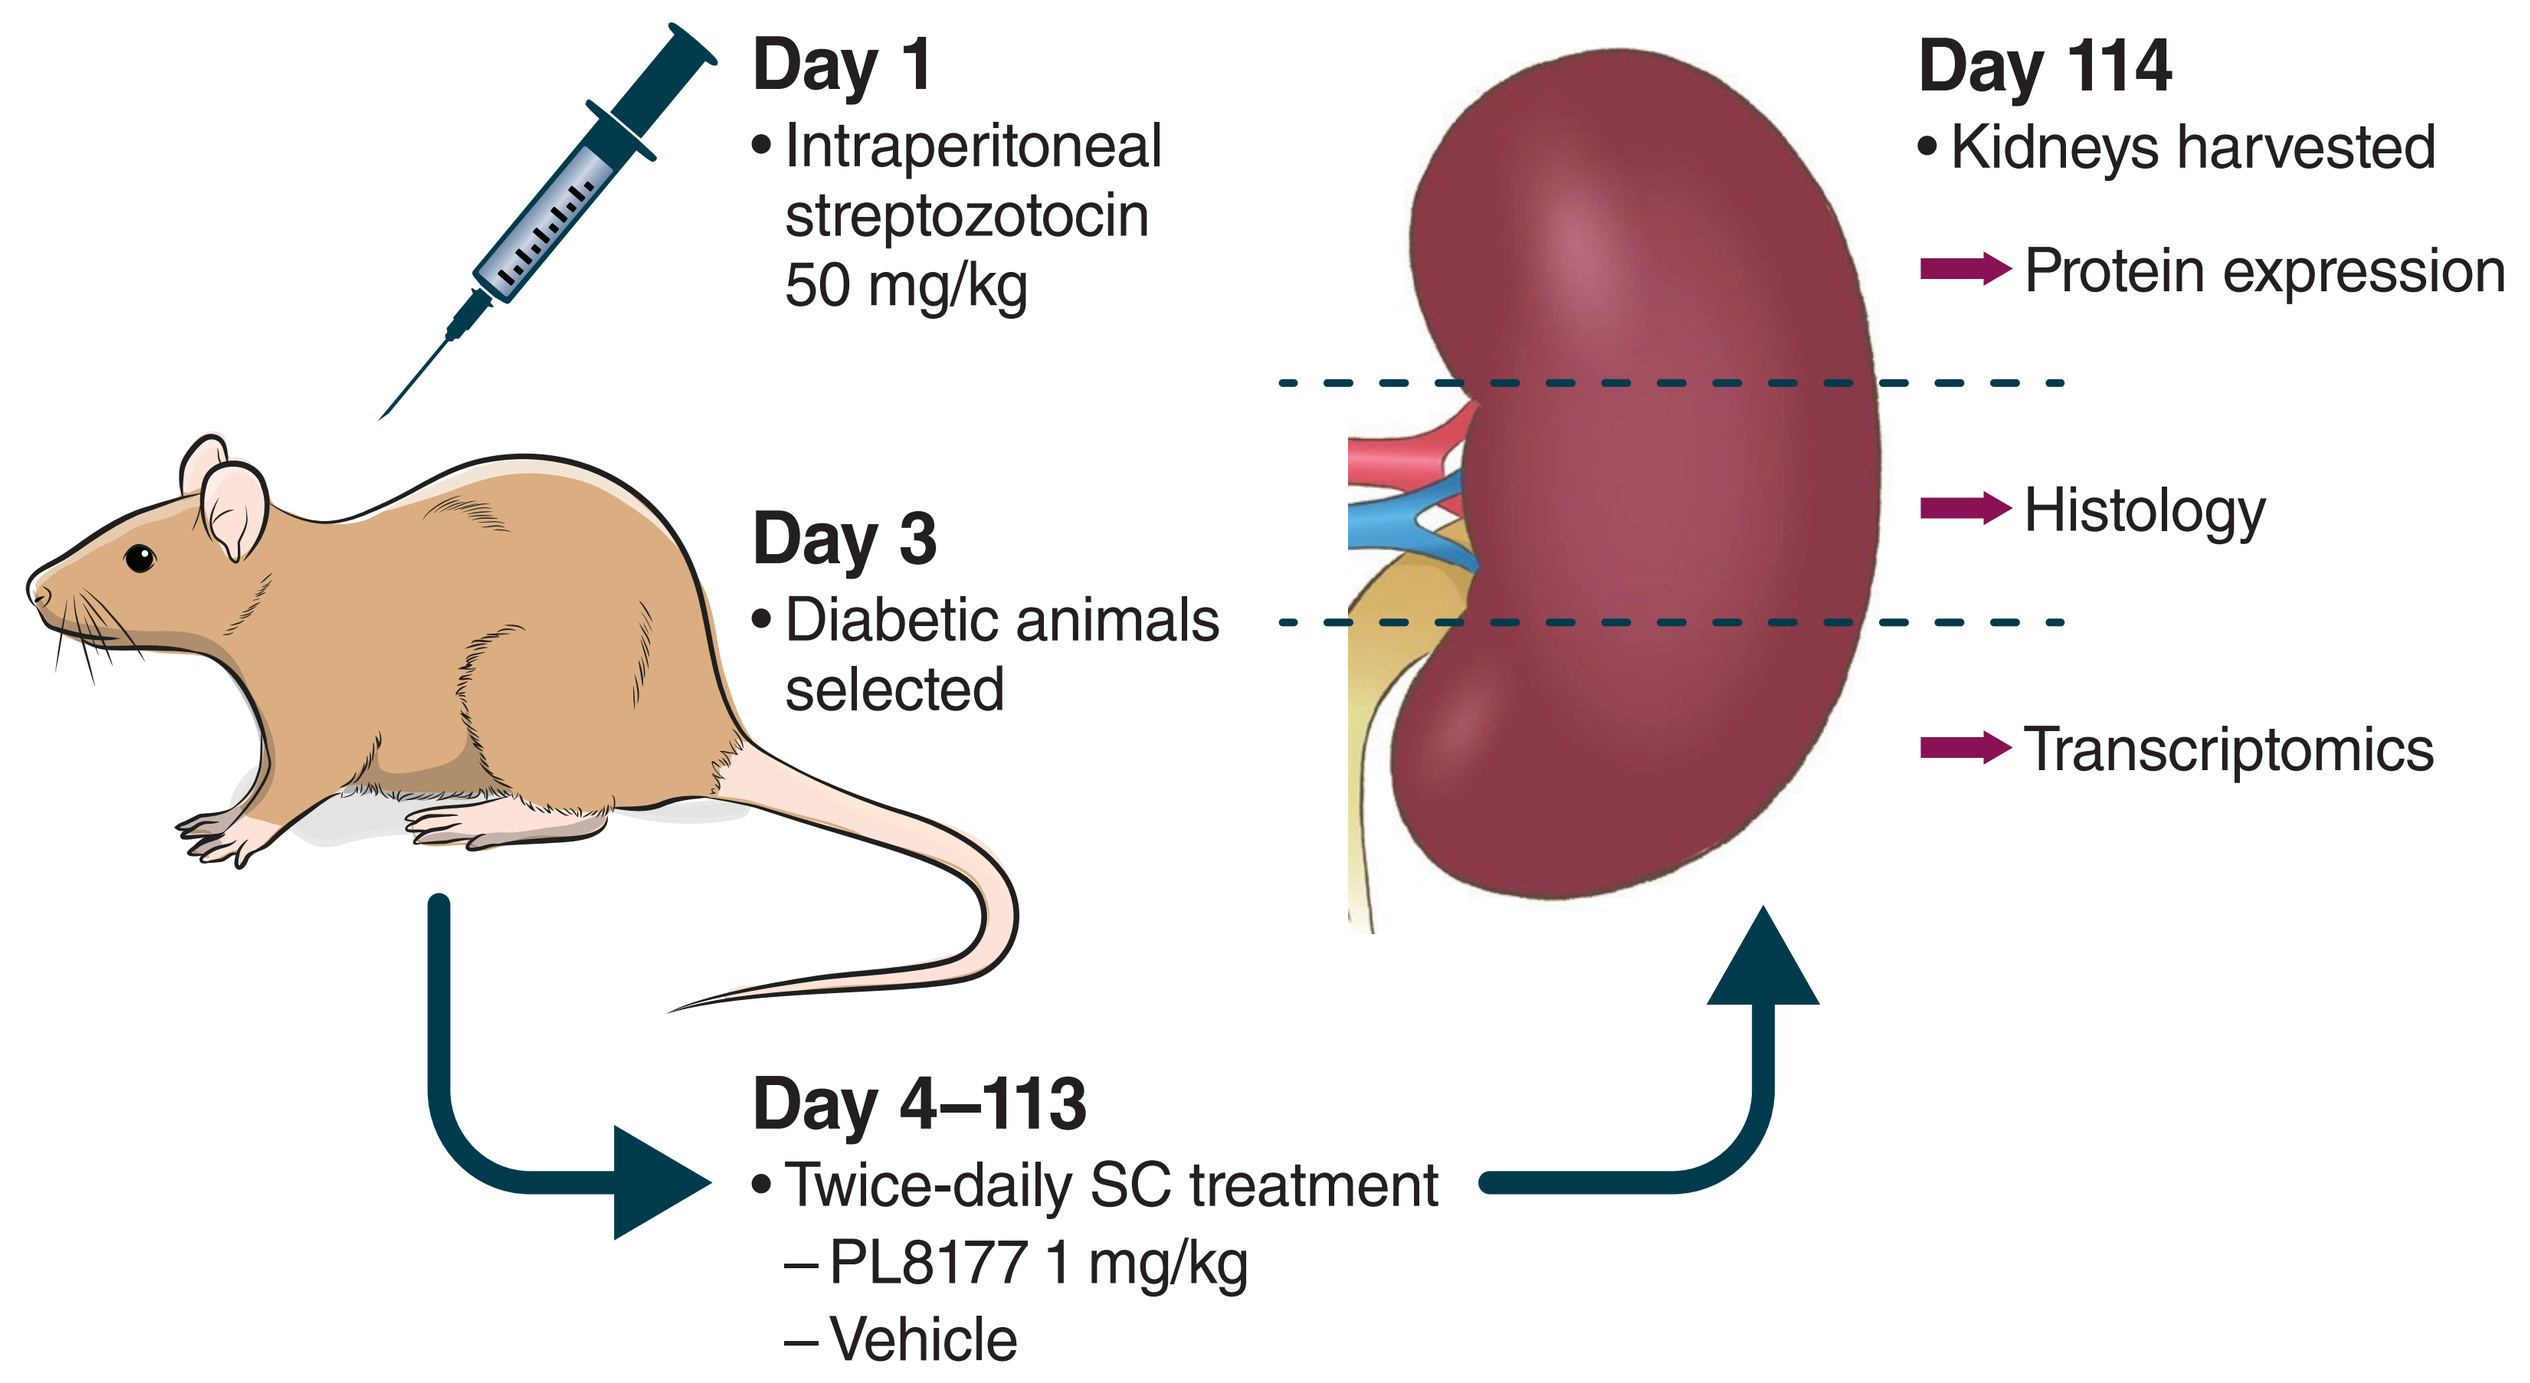

Day 1
• Intraperitoneal streptozotocin 50 mg/kg
Day 3
• Diabetic animals selected
Day 4–113
• Twice-daily SC treatment
– PL8177 1 mg/kg
– Vehicle
Day 114
• Kidneys harvested
Protein expression
Histology
Transcriptomics

SC, subcutaneous.

Histology

- Kidneys were snap-frozen at harvest. The middle blocks of the kidneys were formalin-fixed, paraffin-embedded, and sectioned into 4-μm horizontal/transverse plane sections. Sections were stained with Abcam WT1 antibody by HistoWiz (Long Island City, NY). 50 glomeruli per animal were annotated manually, and automated image analysis was performed
- Parameters measured
 –Total number of cells
 –WT1 positive/negative
 –Glomerular area (μm within annotations)

WT1 Protein Expression

- Measured by Western blotting of homogenized kidney tissue

Transcriptomics

- Single nucleus RNA sequencing (snRNA-seq) data for rat kidney samples (5 vehicle, 3 healthy control, and 5 PL8177 1.0-mg/kg treatment group) were generated using 10× Genomics Chromium technology
- Seurat toolkit v4.0.3 was used for downstream data analysis
- Seurat integration method was used to remove batch effects and enable identification of shared populations across samples within each treatment group
- Unbiased clustering on the gene expression profiles of 28,447 rat kidney nuclei identified 11 major kidney cell types
- Expression of cell type–specific marker genes was used to annotate each cluster into individual kidney cell types
- t-Distributed stochastic neighbor embedding (tSNE) was performed to visualize clustering of single nuclei from different kidney samples

Results

- WT1 protein expression (a marker for podocytes) was greater in both healthy control (non–streptozotocin treated) and PL8177-treated diabetic rat kidneys than in vehicle-treated diabetic kidneys (**Figure 2**)

Figure 2. WT1 Protein Expression in Healthy, Diabetic, and PL8177-Treated Rat Kidneys

A. Western Blots From Kidney Homogenates

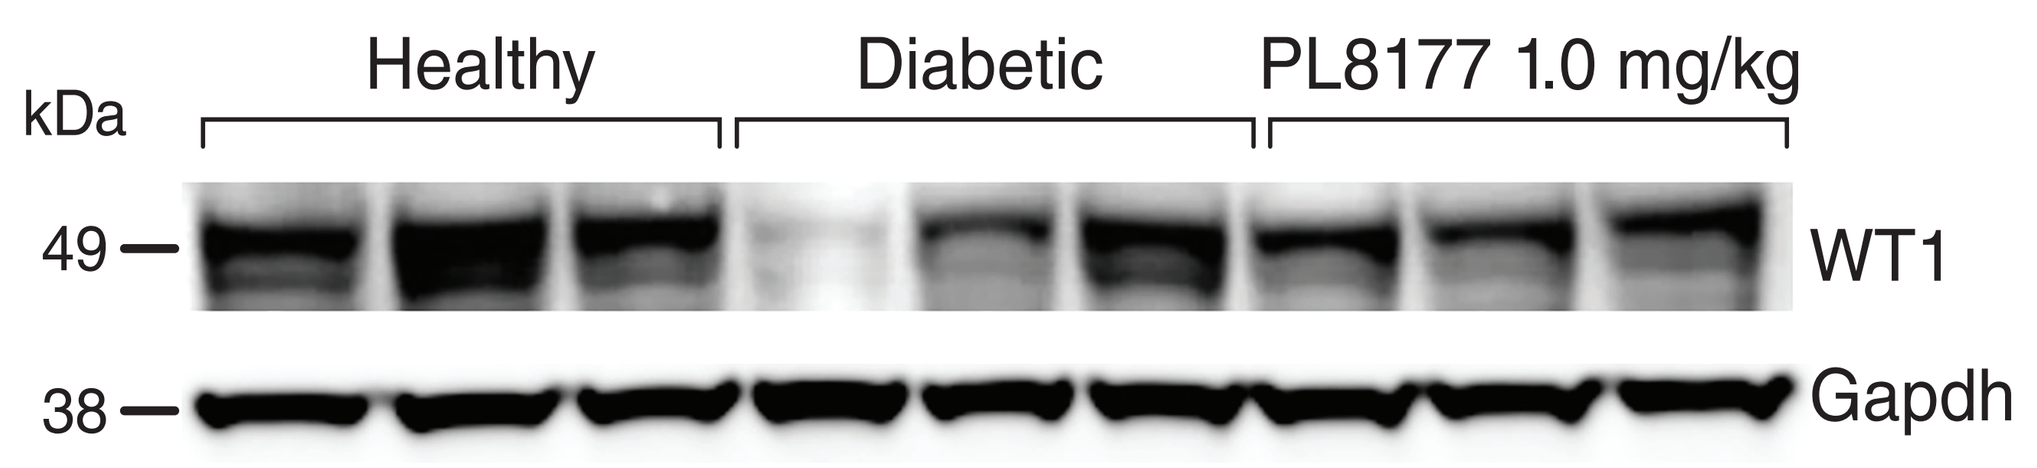

Healthy
Diabetic
PL8177 1.0 mg/kg
kDa
49
38
WT1
Gapdh

B. Relative WT1 Expression

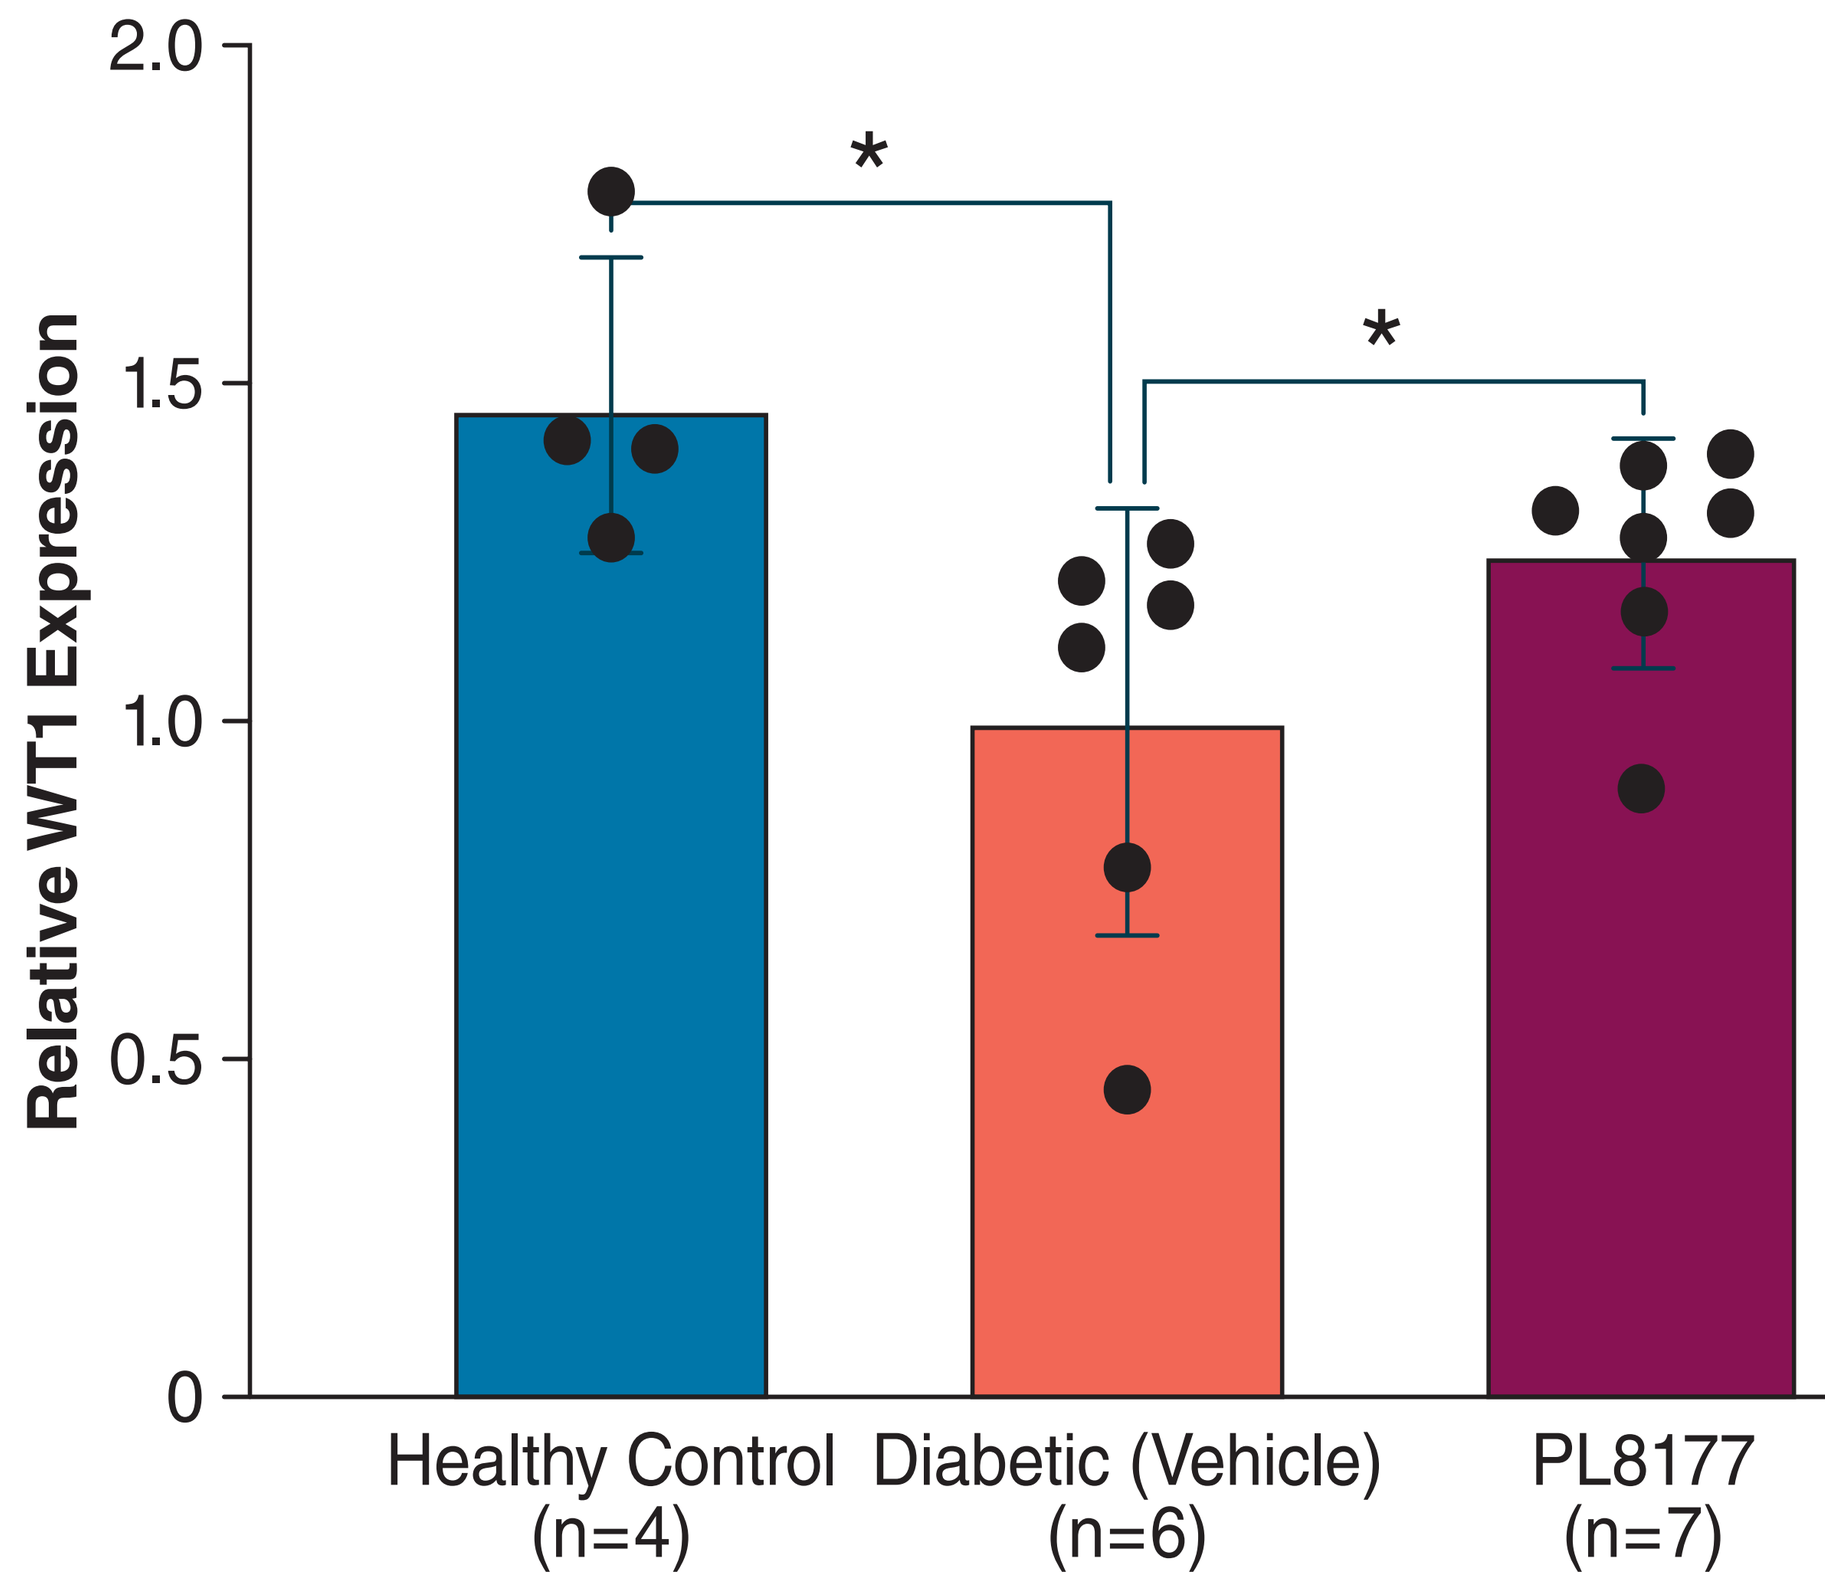

Relative WT1 Expression
2.0
1.5
1.0
0.5
0
*
*
Healthy Control (n=4)
Diabetic (Vehicle) (n=6)
PL8177 (n=7)

Gapdh, glyceraldehyde-3-phosphate dehydrogenase.
Data are shown as mean ± SD, plus data points. *$P<0.05$ vs vehicle.

- Podocyte density, as shown by immunostaining for WT1 protein, was reduced in diabetic kidneys compared to healthy controls and PL8177-treated diabetic kidneys (**Figure 3**)

Figure 3. Podocyte Density in Healthy, Diabetic, and PL8177-Treated Rat Kidneys

A. Podocyte Density

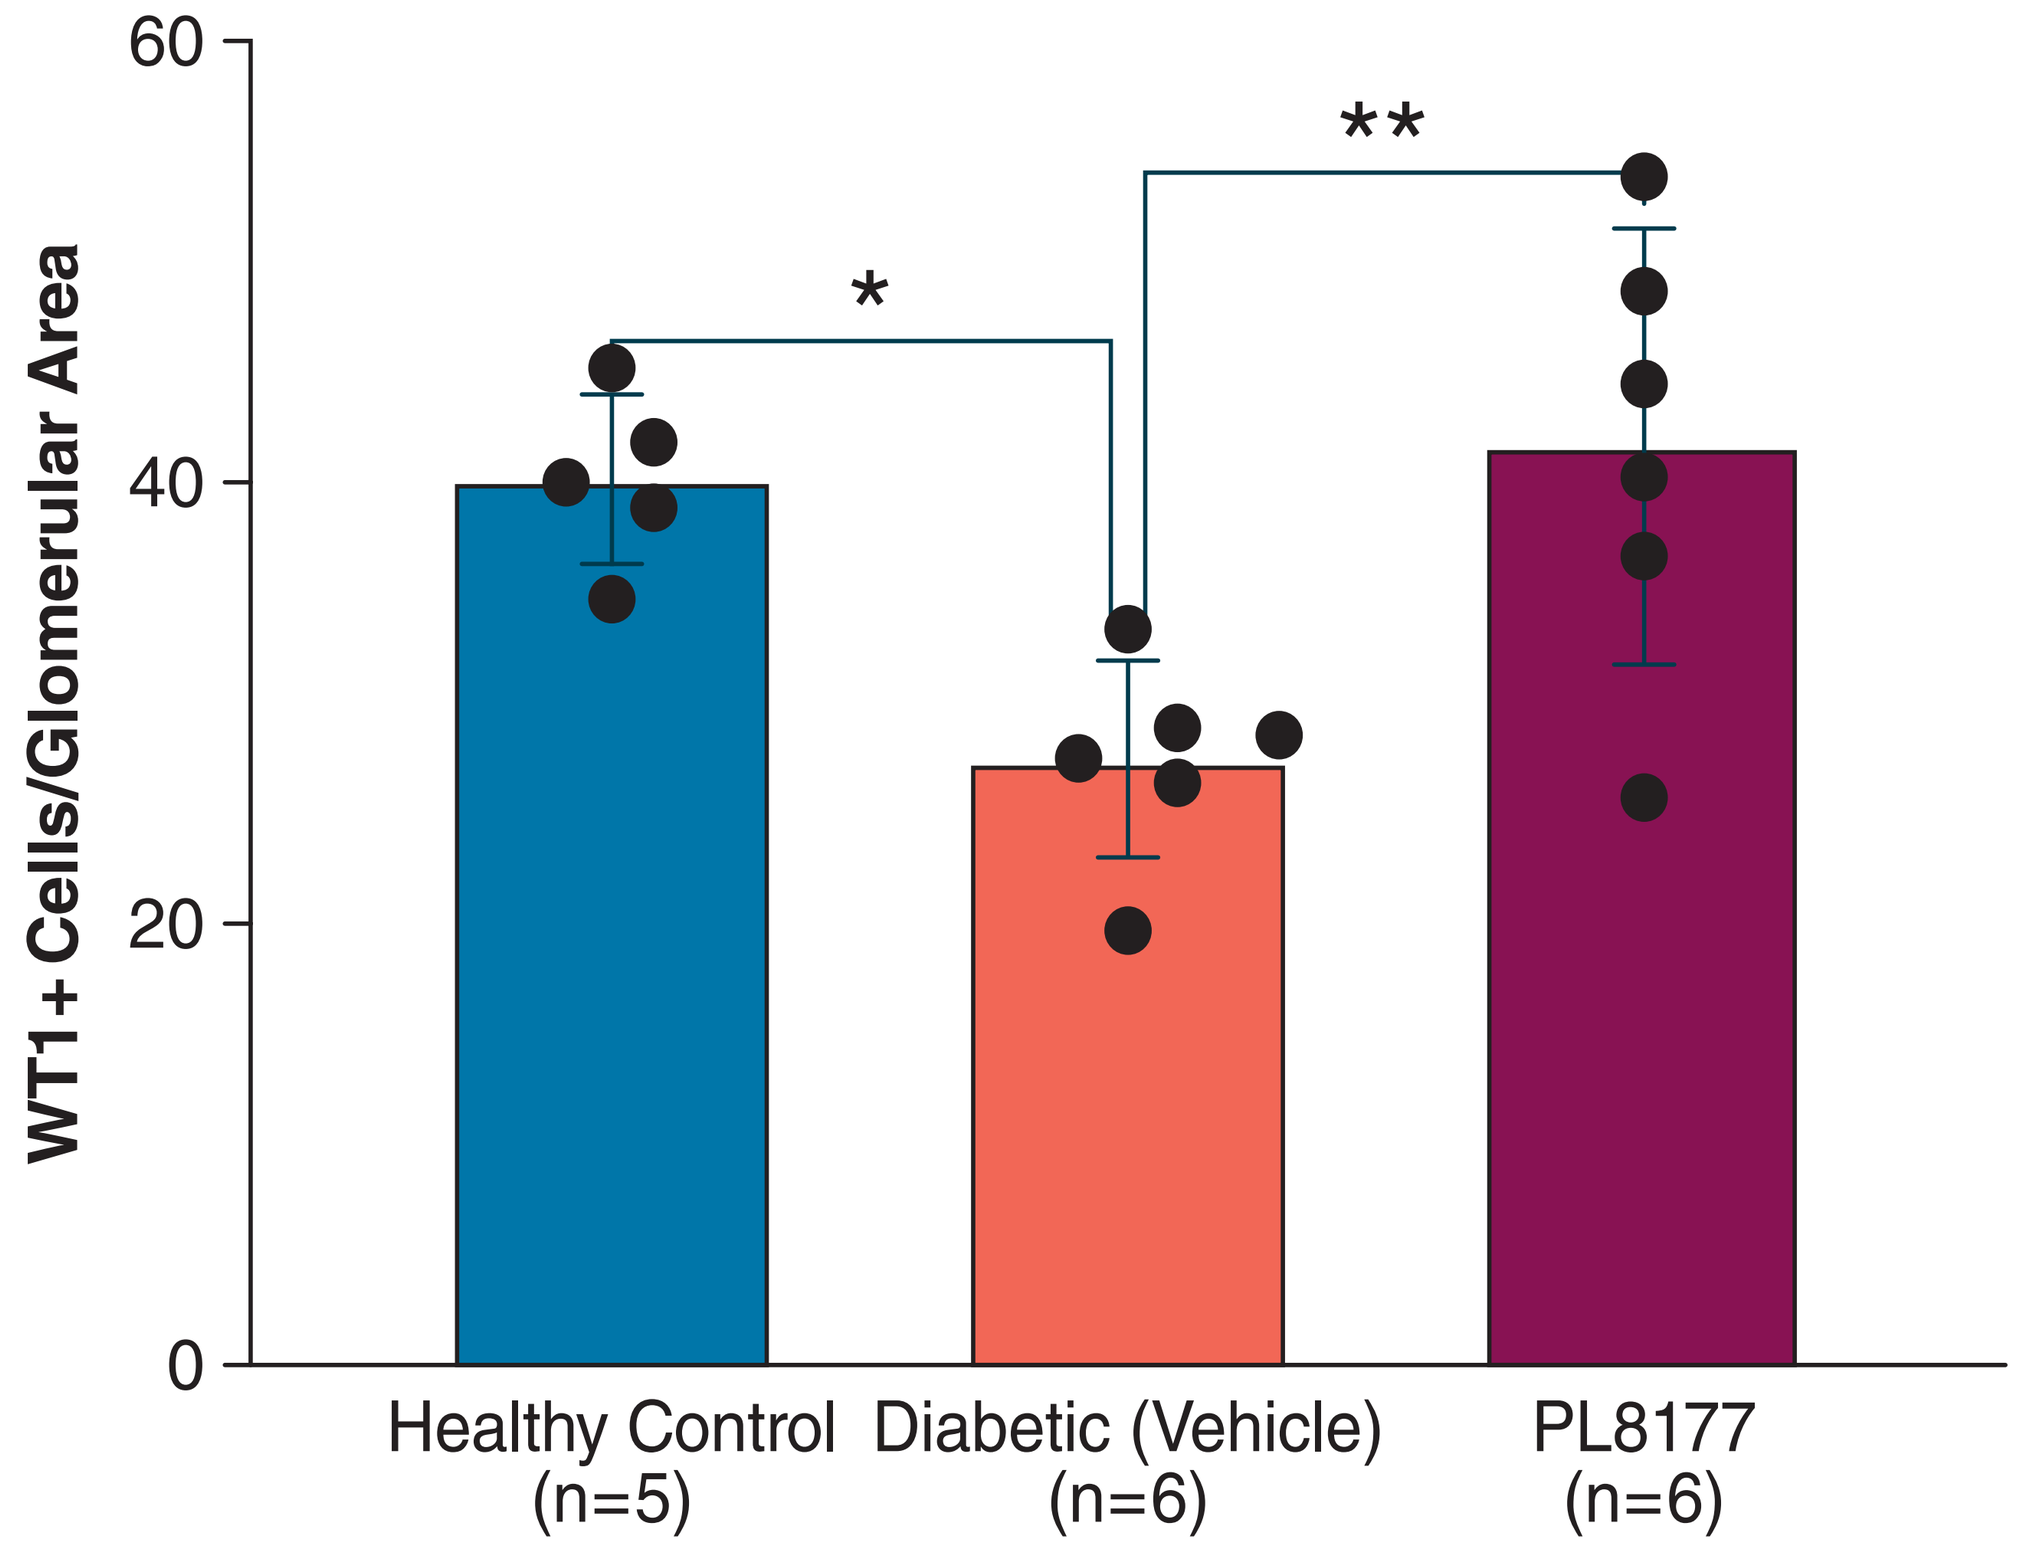

WT1+ Cells/Glomerular Area
60
40
20
0
*
**
Healthy Control (n=5)
Diabetic (Vehicle) (n=6)
PL8177 (n=6)

B. Representative WT1 Immunohistochemistry

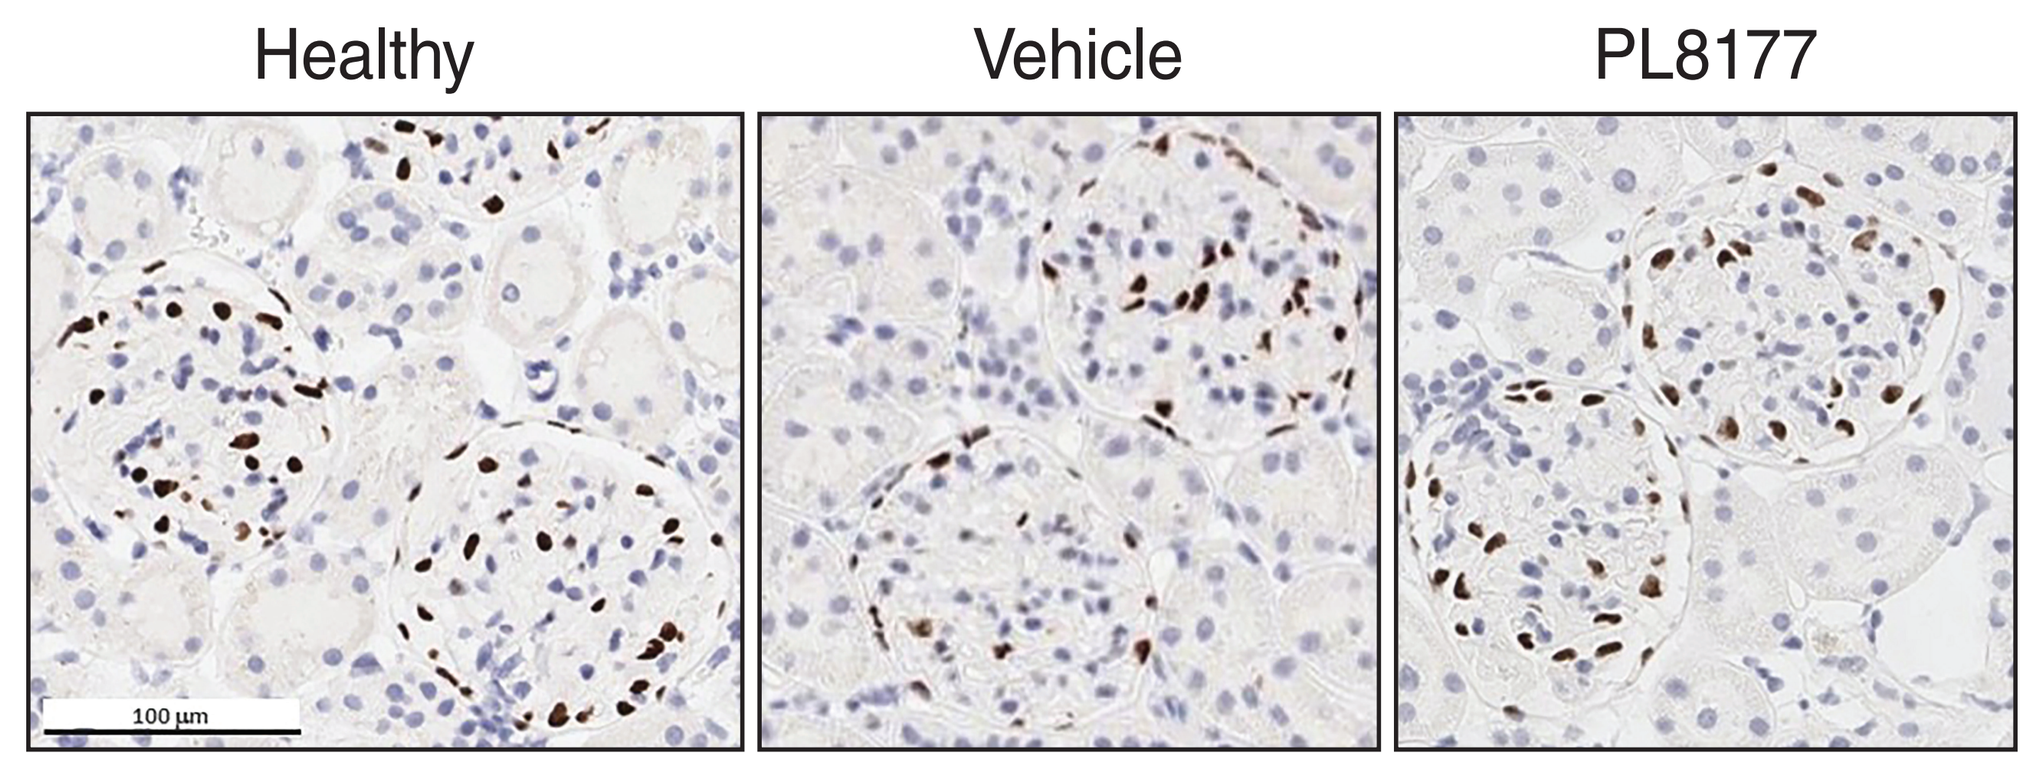

Healthy
Vehicle
PL8177

Mean podocyte density (WT1+ cells by immunostaining) in PL8177-treated rats was 42 cells per glomerular area vs 27 cells per glomerular area for vehicle and was similar to that seen in healthy rats (40 cells per glomerular area). Data are shown as mean ± SD, plus data points. *$P<0.05$ vs vehicle. **$P<0.01$ vs vehicle.

- Glomerular area and total number of cells (nuclear staining within glomeruli) were both trending lower in PL8177-treated rats compared to vehicle, suggesting less glomerular hypertrophy with PL8177 treatment (**Figure 4**)
- Representative images of kidney sections from healthy, diabetic (vehicle), and PL8177-treated rats harvested at day 114 are shown in **Figure 3B**
- snRNA-seq revealed cellular composition of kidney tissues in control, diabetic (vehicle), and PL8177-treated animals (**Figure 5**)

Figure 4. Glomerular Area and Total Number of Cells in Healthy, Diabetic, and PL8177-Treated Rat Kidneys

A. Glomerular Area

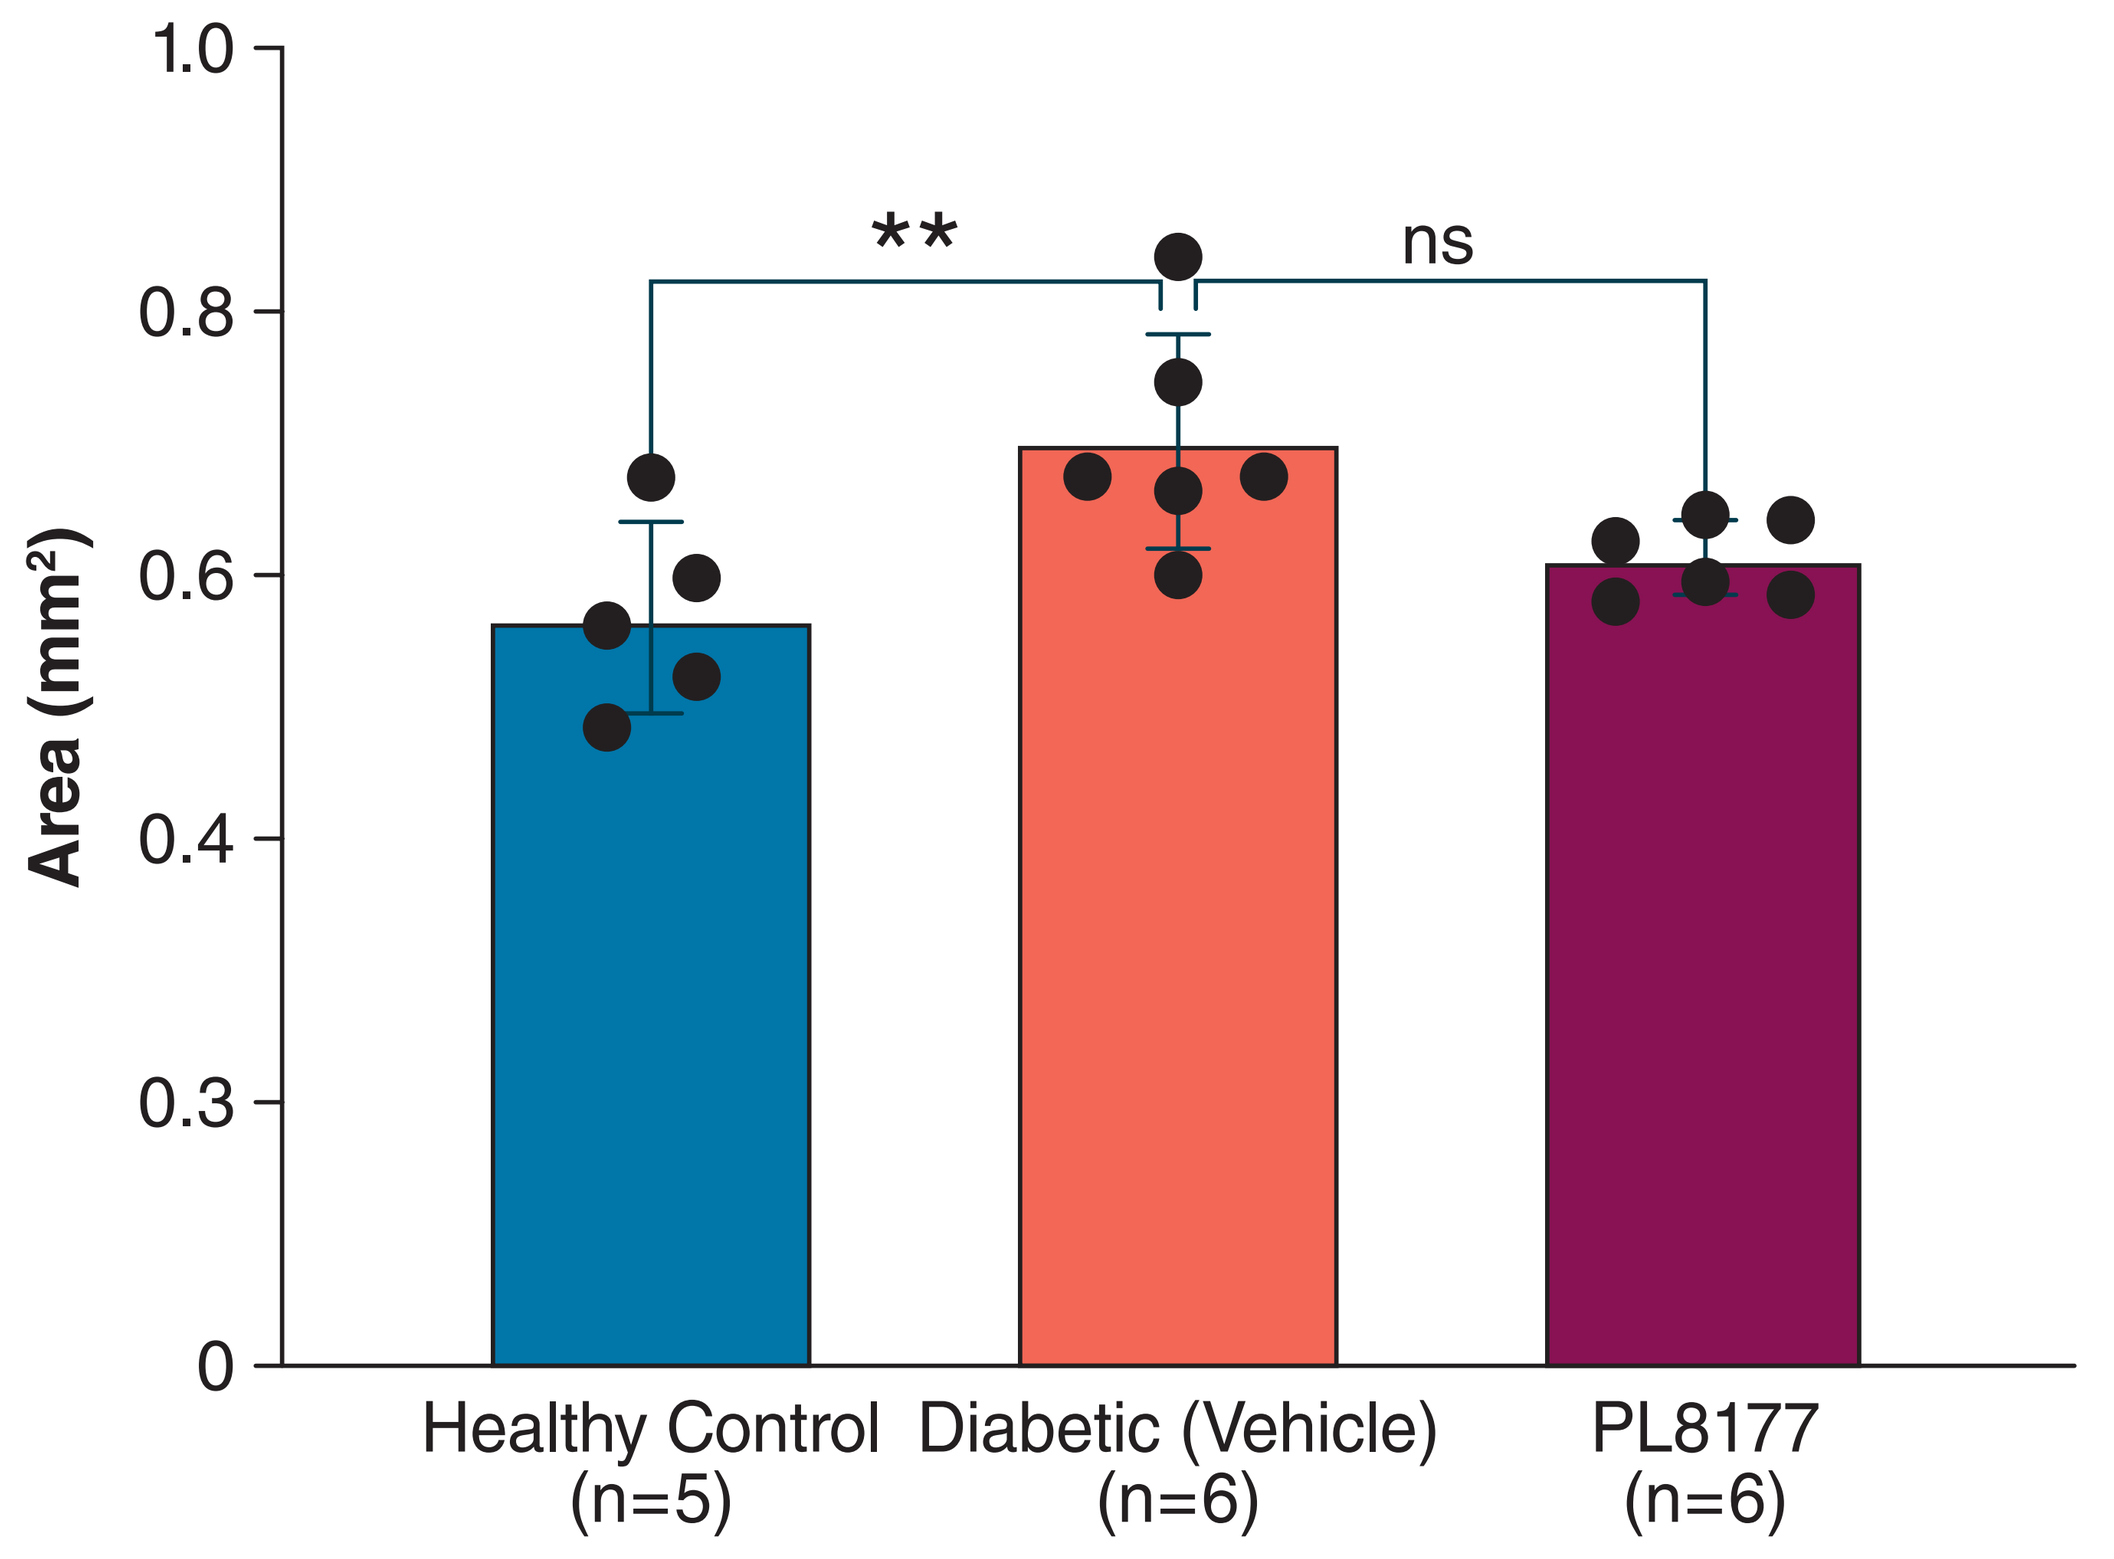

Area (mm²)
1.0
0.8
0.6
0.4
0.3
0
**
ns
Healthy Control (n=5)
Diabetic (Vehicle) (n=6)
PL8177 (n=6)

B. Total Number of Cells

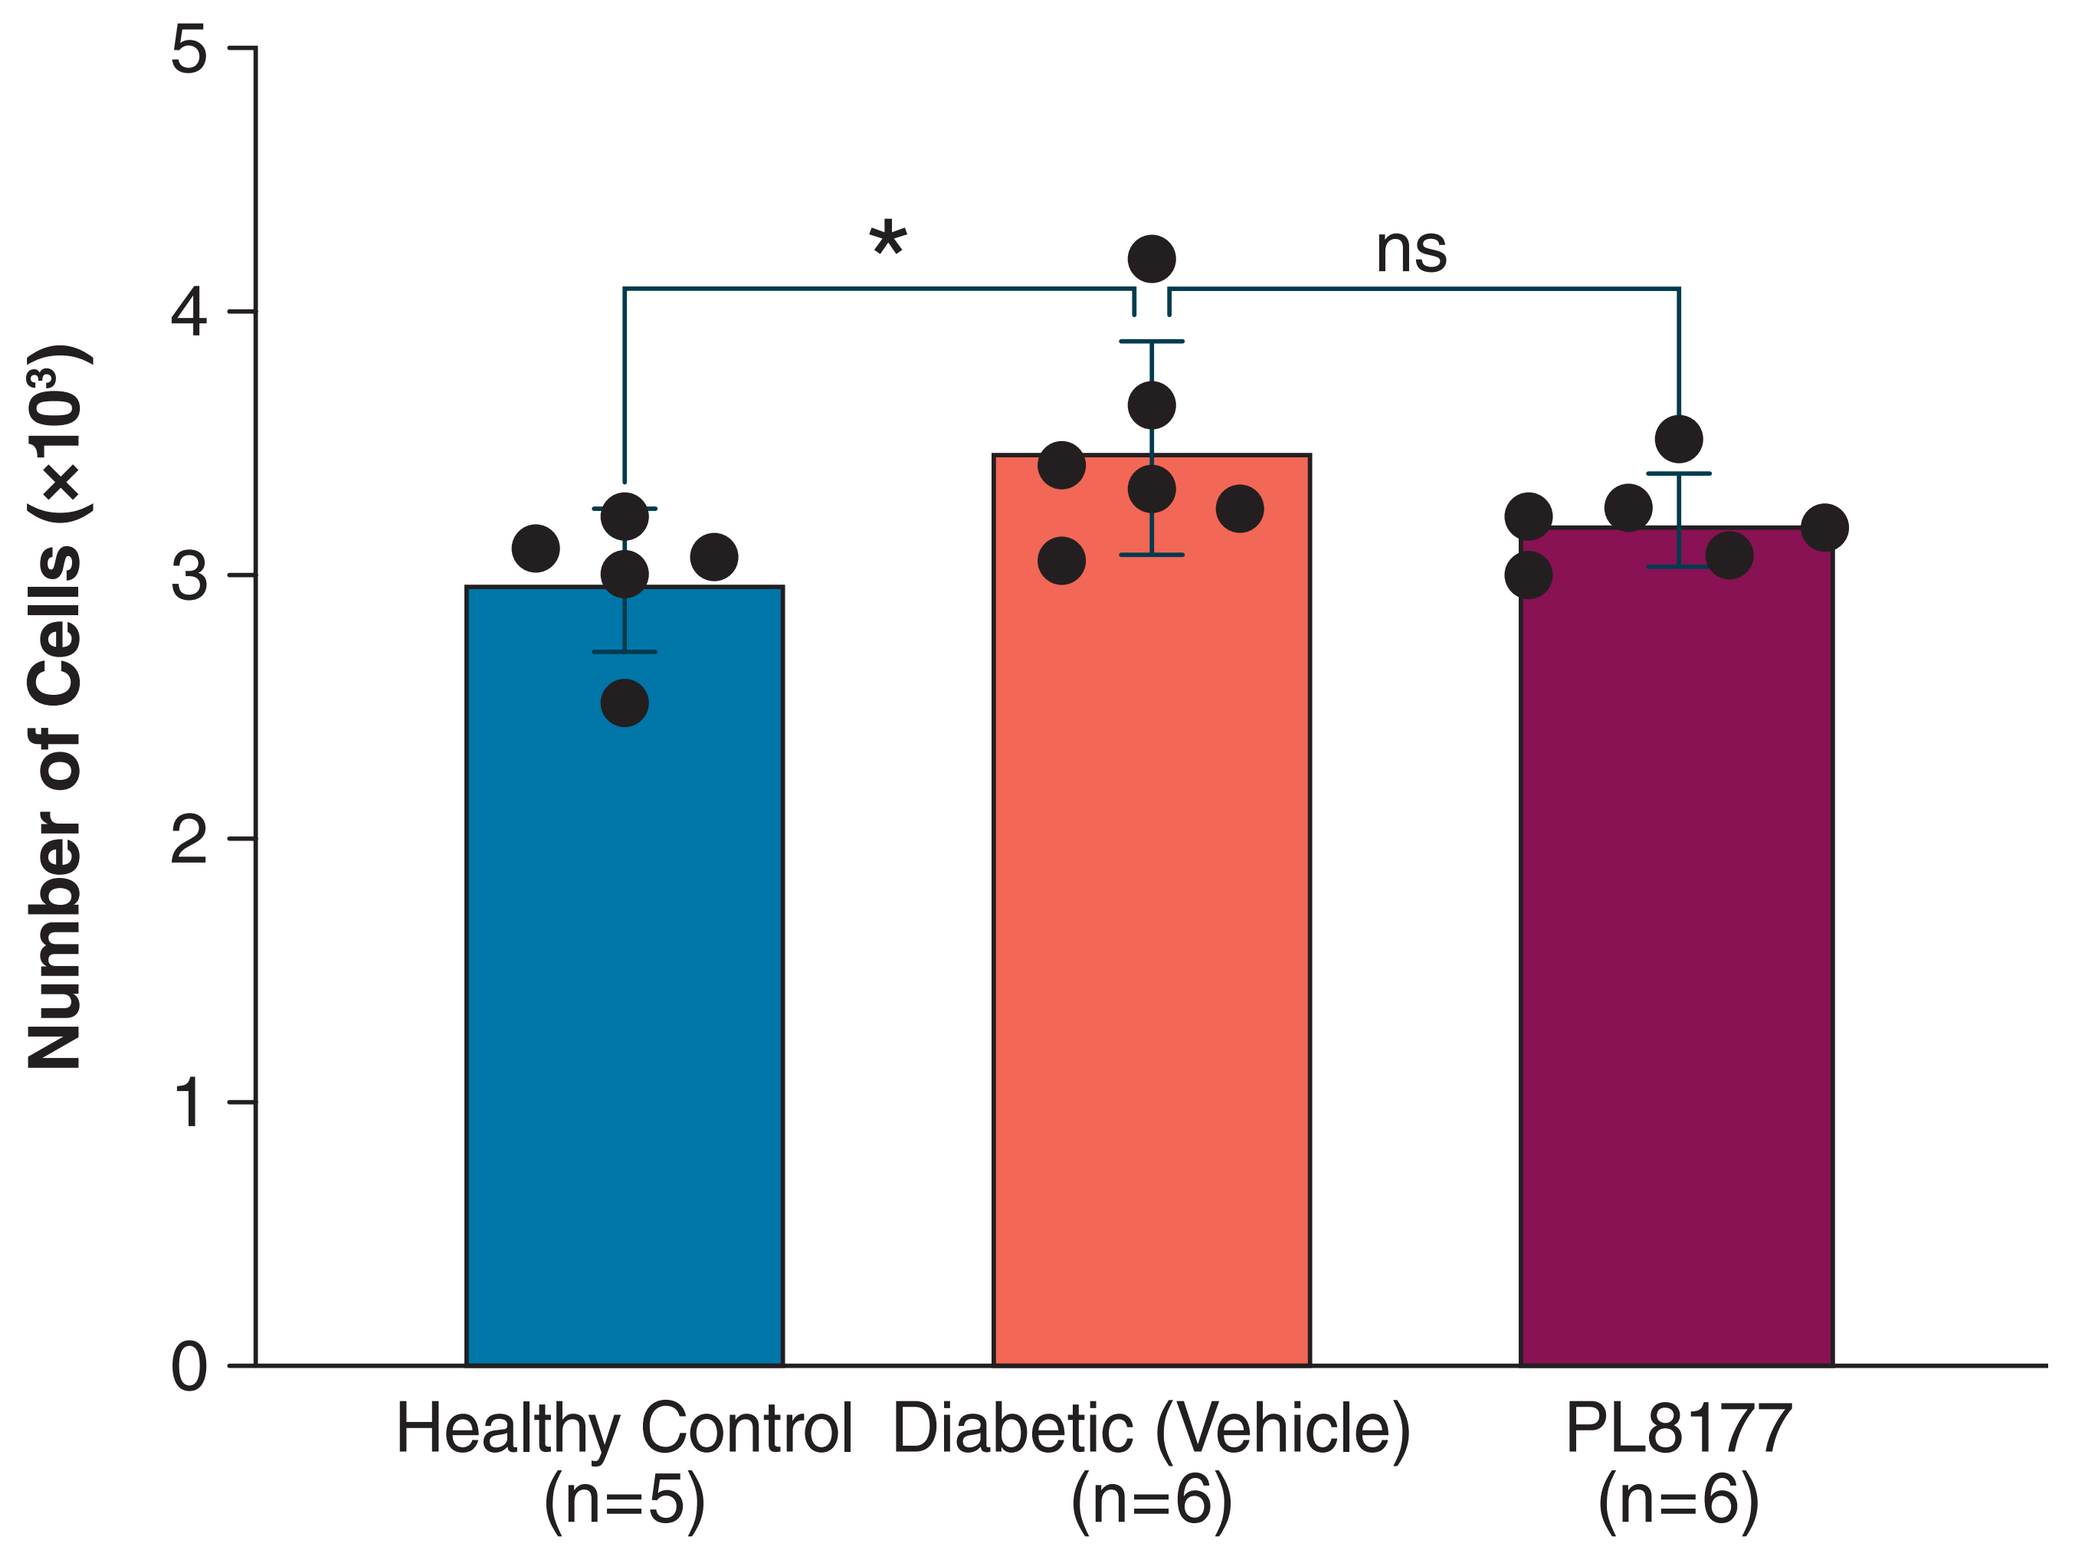

Number of Cells (×10³)
5
4
3
2
1
0
*
ns
Healthy Control (n=5)
Diabetic (Vehicle) (n=6)
PL8177 (n=6)

ns, not significant. Data are shown as mean ± SD, plus data points. *$P<0.05$ vs vehicle.**$P<0.01$ vs vehicle.

Figure 5. Cell Populations in Healthy, Diabetic, and PL8177-Treated Rat Kidneys

A. tSNE Plot

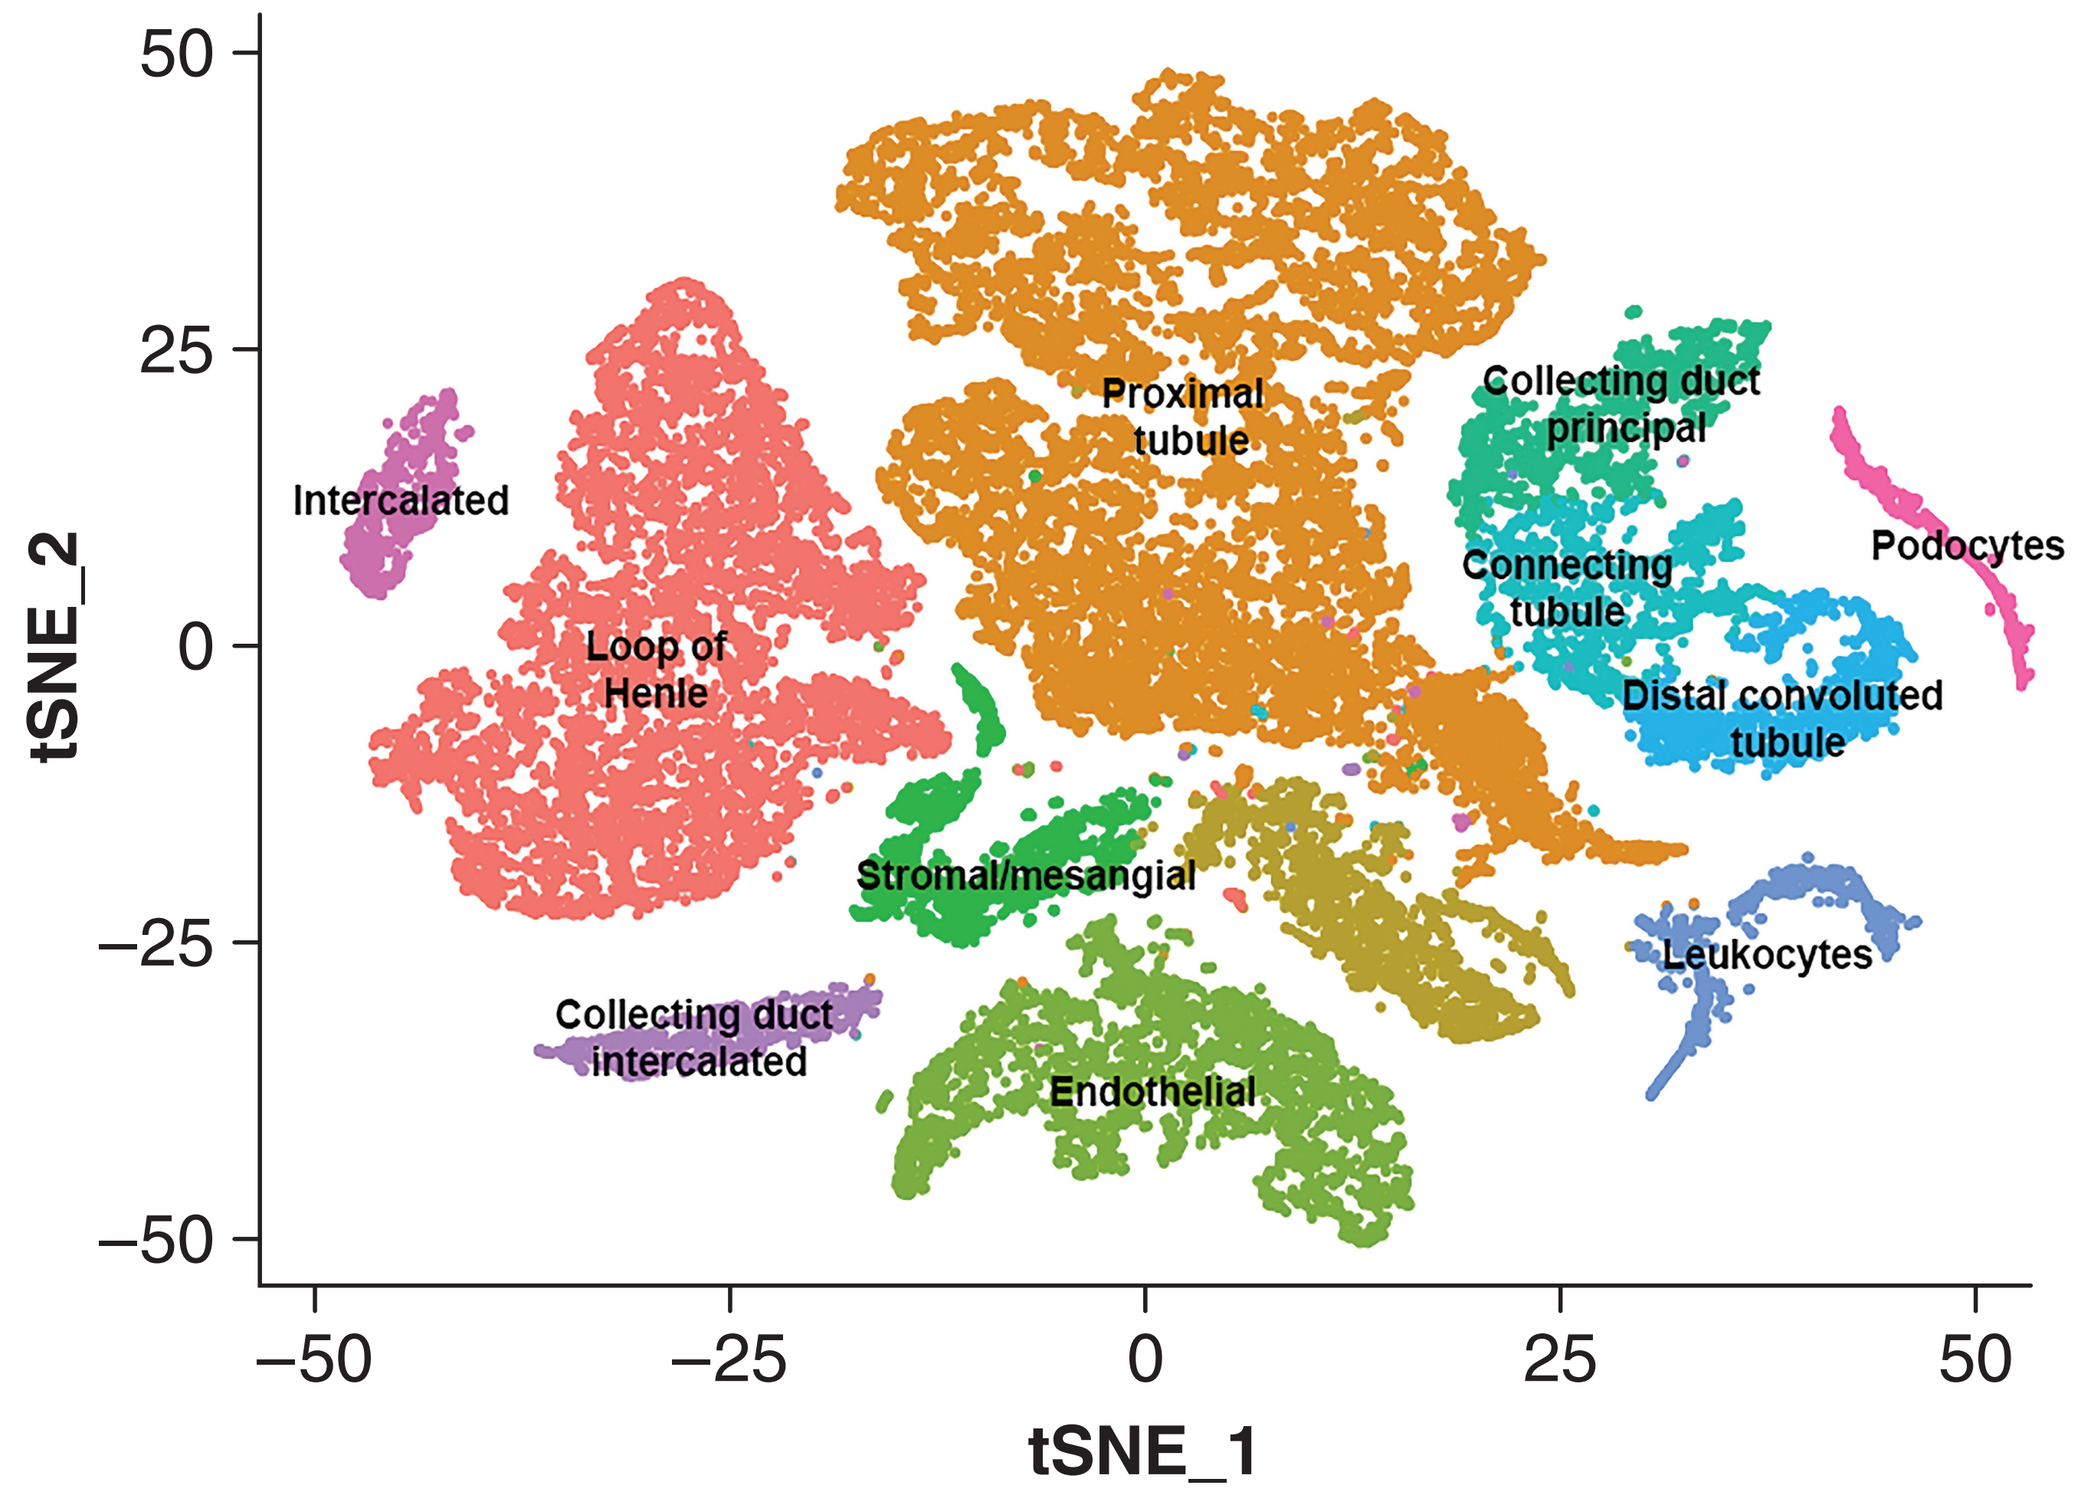

tSNE_2
tSNE_1
50
25
0
−25
−50
−50
−25
0
25
50
Intercalated
Loop of Henle
Proximal tubule
Collecting duct principal
Connecting tubule
Podocytes
Distal convoluted tubule
Stromal/mesangial
Leukocytes
Collecting duct intercalated
Endothelial

B. Transcriptomic Analysis

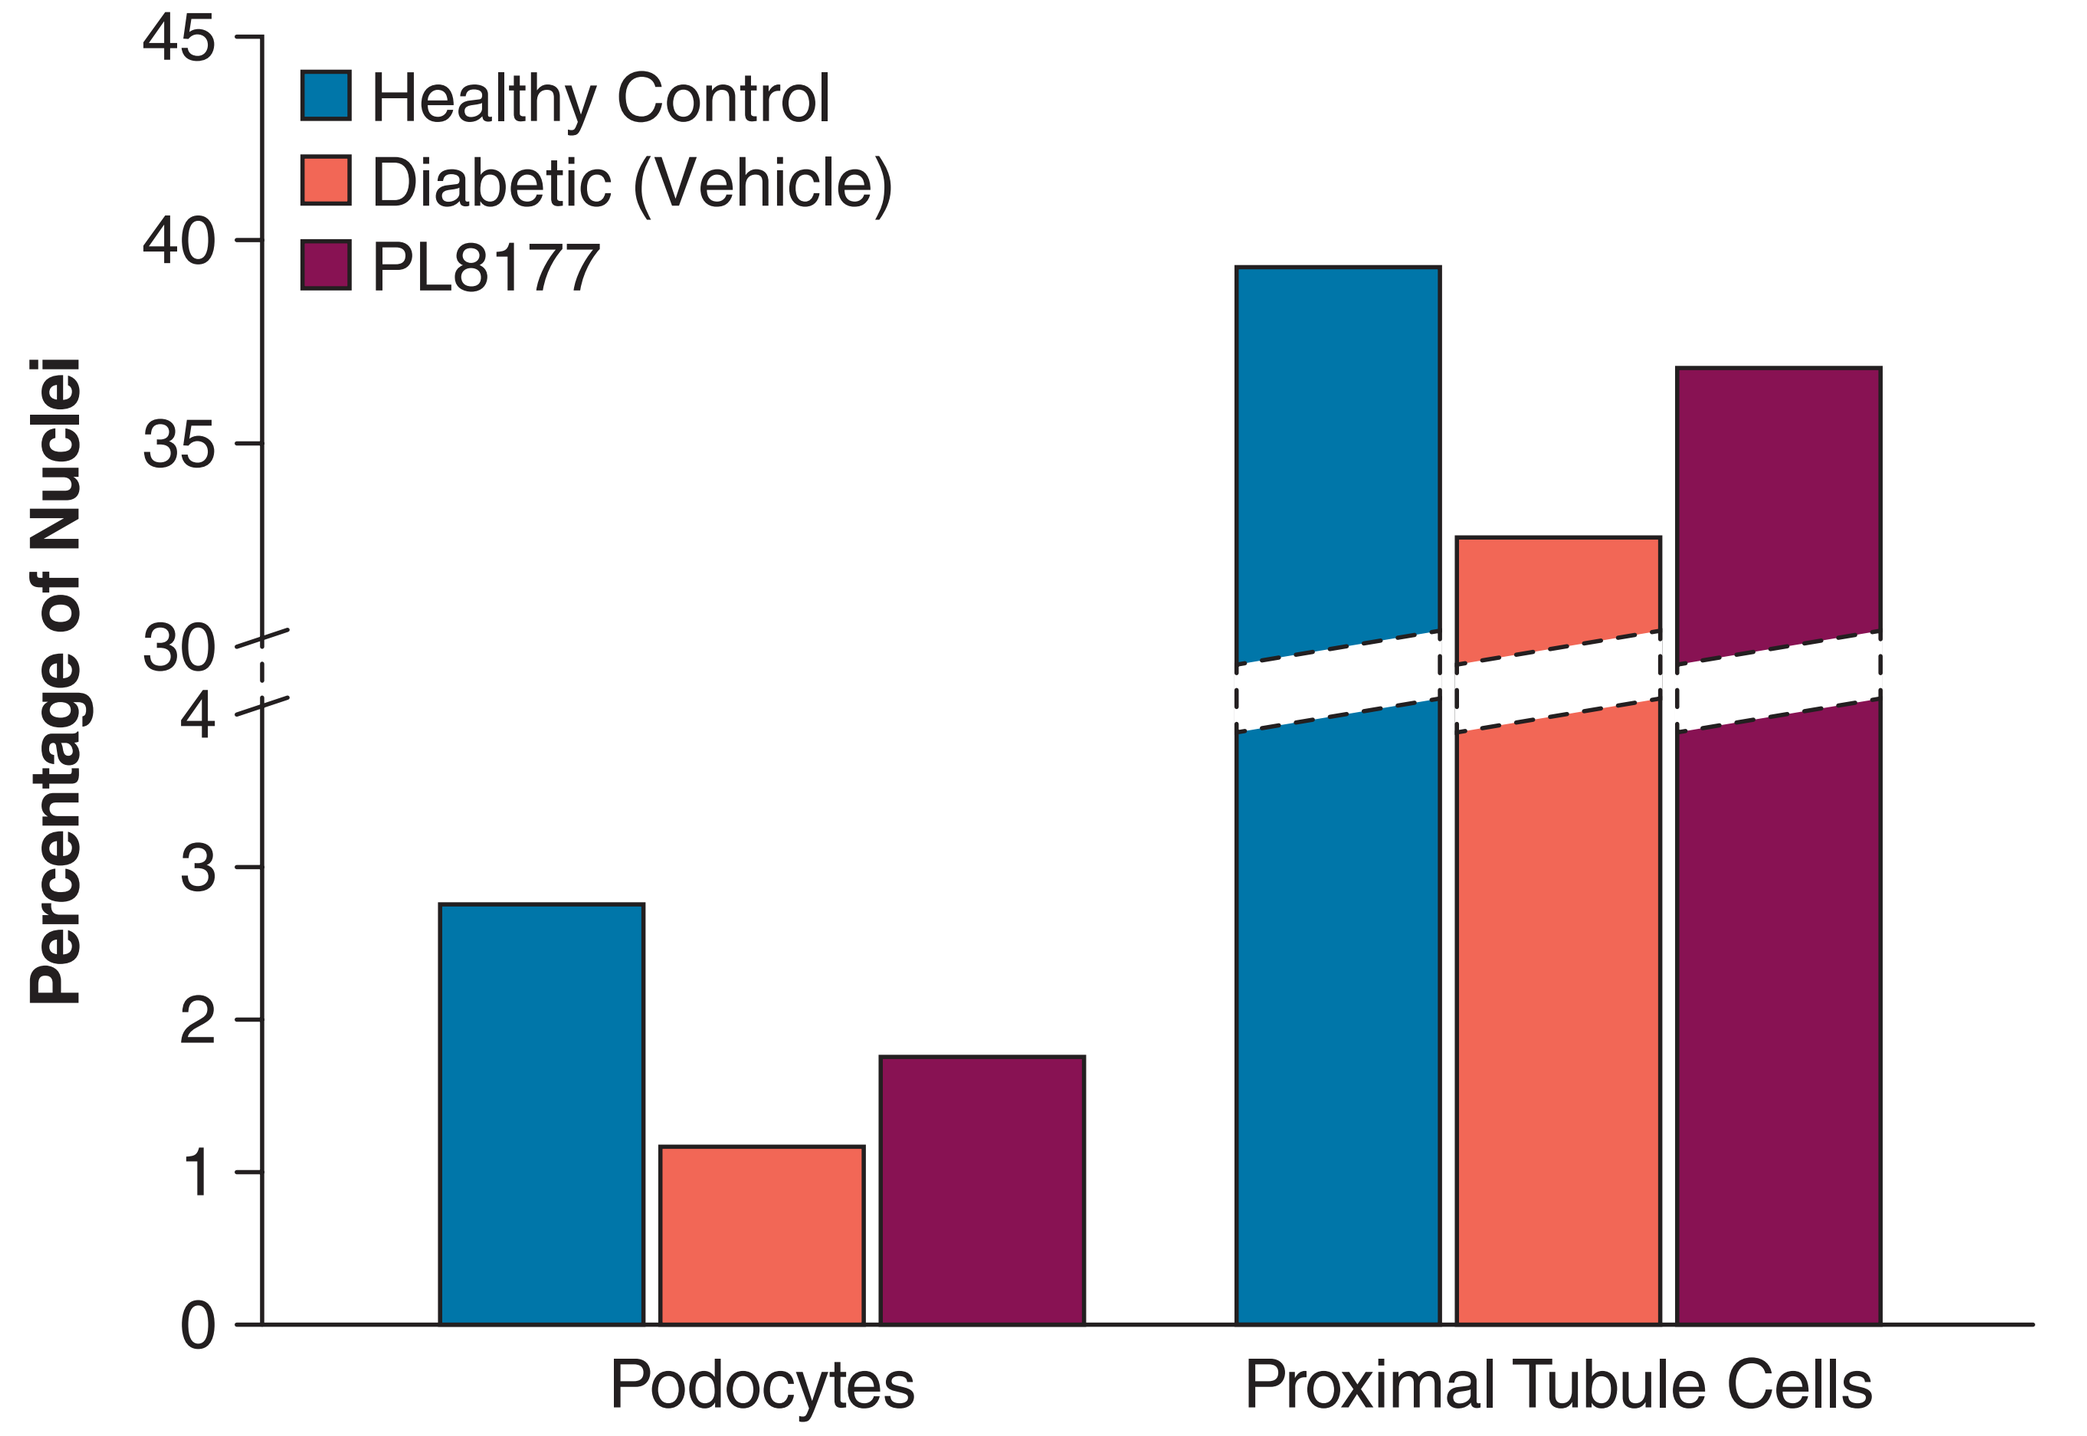

Healthy Control
Diabetic (Vehicle)
PL8177
Percentage of Nuclei
45
40
35
30
4
3
2
1
0
Podocytes
Proximal Tubule Cells

A. tSNE plot displays cell-type clusters derived from all the treatment groups. Each dot represents a nucleus. Nuclei are derived from 5 vehicle, 3 healthy control, and 5 PL8177 (1.0 mg/kg) samples. B. Transcriptomic analysis shows that PL8177-treated rats had increased relative proportions of podocytes and proximal tubule cells compared to vehicle.
tSNE, t-distributed stochastic neighbor embedding.

Conclusions

- PL8177 treatment of streptozotocin-induced diabetic rats increased the expression of WT1 protein and increased the number of podocytes and proximal tubule cells compared to vehicle-treated controls
- Glomerular hypertrophy, a characteristic of kidney injury, trended toward reduction in PL8177-treated kidneys
- These results suggest that melanocortin agonists may represent a new therapeutic avenue to treat diabetic nephropathy