Presented at:

Obesity Week 2025
November 4–7, 2025
Atlanta, GA

Co-Administration of Bremelanotide With Tirzepatide in Participants With Obesity

Robert Jordan[1]; Carl Spana, PhD[1]; Nazish Huq[1]; John Dodd, PhD[1]; Ryan Morgan, DO[2]; Ramses Vega-Casasnovas, MD[3]; Bruce Rankin, DO[4]; Shannon Conrad, DO[5]; Jesse Richards, DO[6]

[1]Palatin Technologies, Inc., Monmouth Junction, NJ; [2]Vitalis Metabolic Health, Oklahoma City, OK; [3]Lynn Health Science Institute, Chattanooga, TN; [4]Accel Research, Alcanza Clinical Research, DeLand, FL; [5]Coastal Carolina Research Center, Alcanza Clinical Research, North Charleston, SC; [6]Department of Internal Medicine, University of Oklahoma School of Community Medicine, Tulsa, OK

BACKGROUND

- The melanocortin 4 receptor (MC4R) in the hypothalamus plays a central role in the regulation of stored energy and food intake and is a well-validated target for obesity therapeutics[1-3]
 - In preclinical and clinical studies, the MC4R agonist bremelanotide has been shown to cause a decrease in food intake and corresponding weight loss[3-6]
- Tirzepatide, the dual GLP-1/GIP receptor agonist, decreases food intake and weight through activation of the endogenous incretin receptors GLP-1 and GIP[7]
 - Weight loss associated with tirzepatide may be limited due to treatment adverse events, such as nausea and vomiting,[6,8] which can limit long-term adherence and effectiveness in some individuals
 - Lowering treatment dose may mitigate adverse events; however, this strategy may come at the cost of reduced treatment effectiveness and may compromise long-term outcomes
- Previous data indicated that combining an incretin agonist with a MC4R agonist may result in increased weight loss at lower doses[2]
- This study assessed the safety and efficacy of co-administration of bremelanotide and tirzepatide in patients with obesity and evaluated whether low-dose MC4R agonism had an effect on weight-loss maintenance

METHODS

- This was a phase 2, prospective, randomized, double-blind, placebo (PBO)-controlled study (NCT06565611)

Participants

- Enrolled participants were aged 18–65 years and were clinically obese with a body mass index (BMI) of 30–45 kg/m^2
- Patients needed to be willing to self-inject treatment and should not have received the following treatments within the following timeframes:
 - Bremelanotide 6 months prior to study screening
 - Cyclosporine A, adrenocorticotropic hormones, long-term corticosteroids (>20 mg once daily or its equivalent), cytotoxic agents, or anti-obesity medications 3 months prior to study screening
- Patients were excluded if they had had clinically significant body weight change (≥5%) or dieting attempts in the previous 3 months or during Treatment Period 1

Treatment schedule

- All patients received subcutaneous tirzepatide 2.5 mg weekly for 4 weeks (Treatment Period 1), followed by randomization to 1 of 4 treatment arms shown in **Figure 1** for a further 4 weeks (Treatment Period 2), and were monitored for an additional 4 weeks without treatment

Figure 1. Study treatment and follow-up schedule

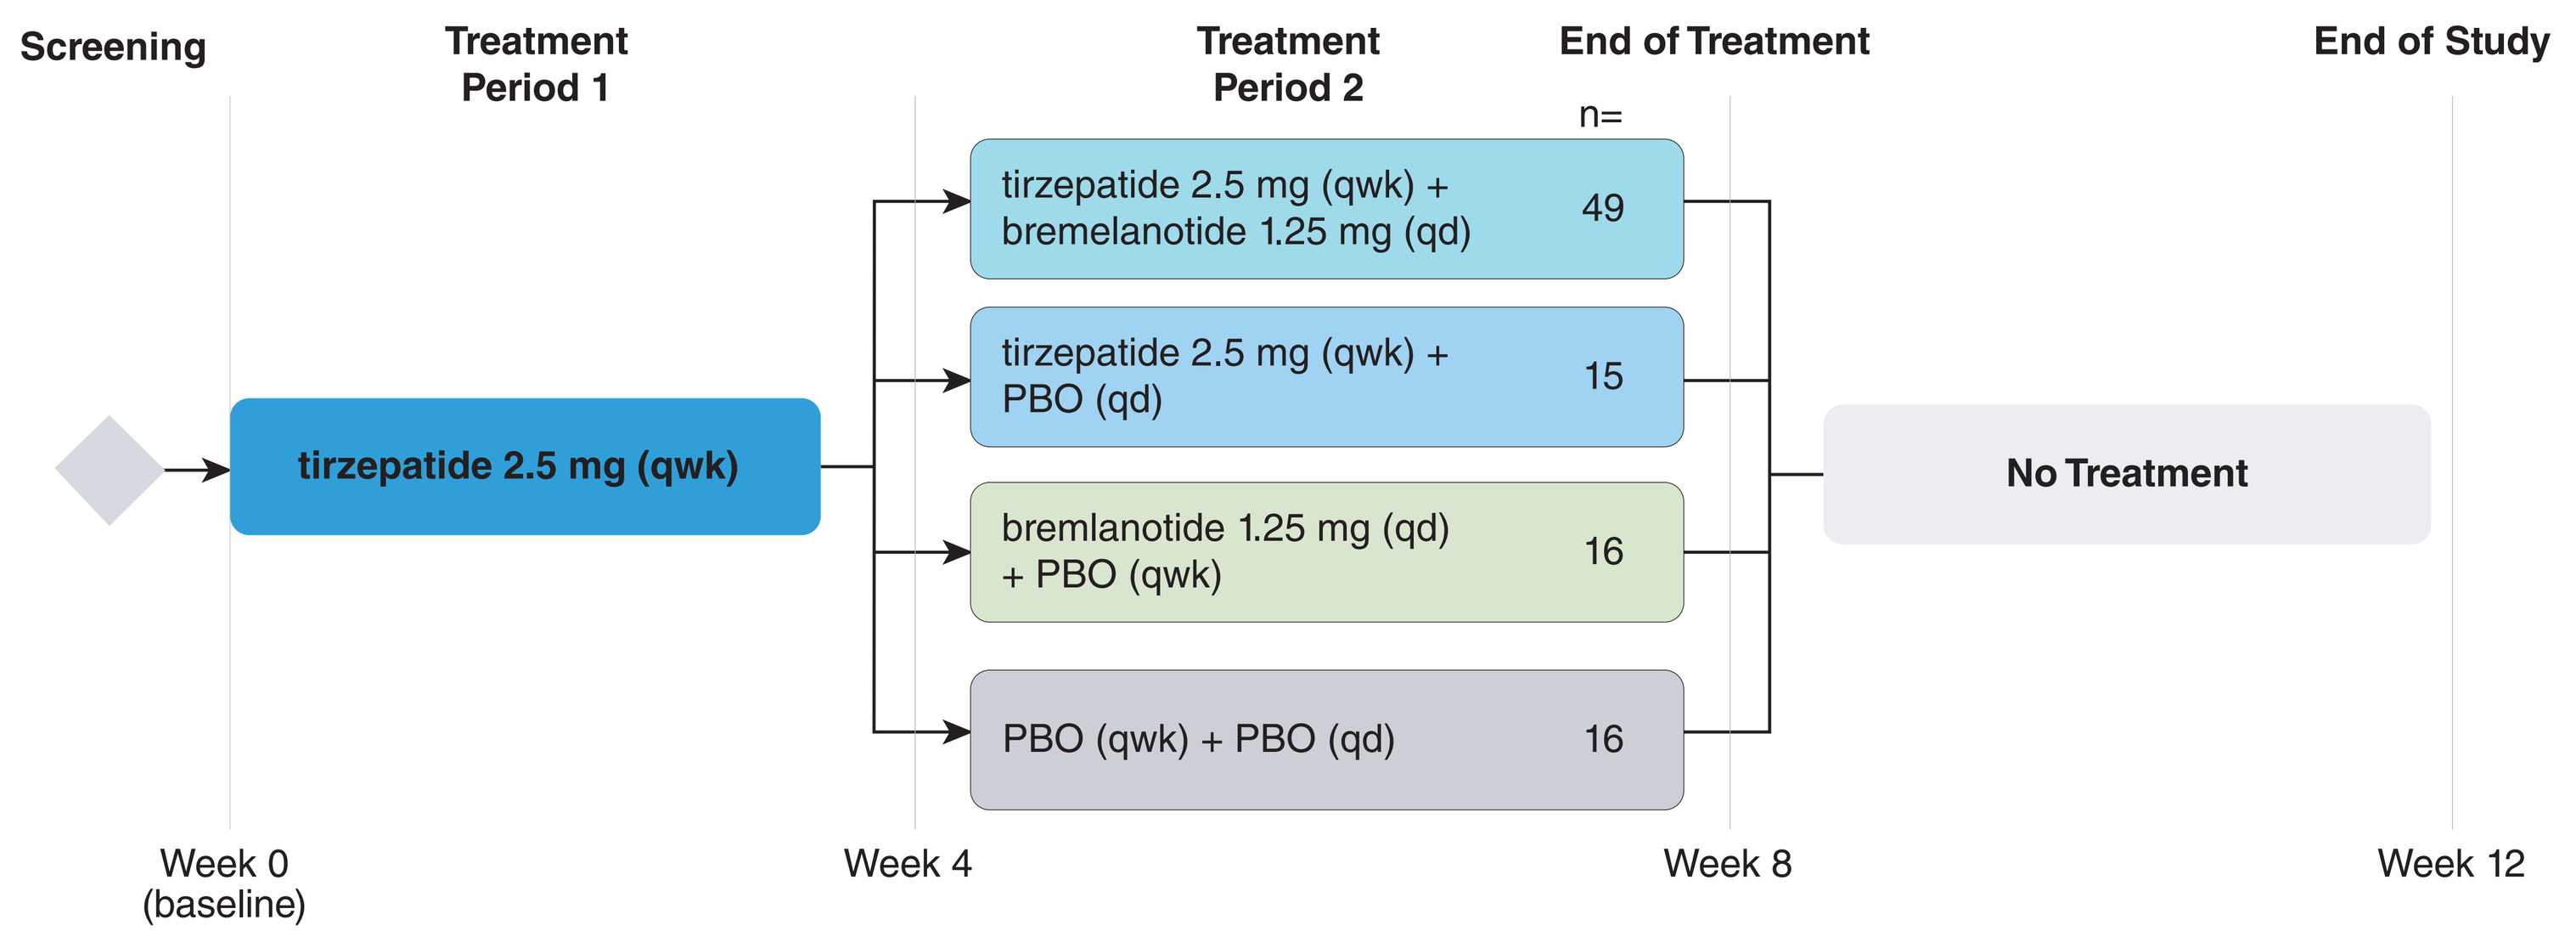
Screening
Treatment Period 1
Treatment Period 2
End of Treatment
End of Study
n=
tirzepatide 2.5 mg (qwk)
tirzepatide 2.5 mg (qwk) + bremelanotide 1.25 mg (qd)
49
tirzepatide 2.5 mg (qwk) + PBO (qd)
15
bremlanotide 1.25 mg (qd) + PBO (qwk)
16
PBO (qwk) + PBO (qd)
16
No Treatment
Week 0 (baseline)
Week 4
Week 8
Week 12

PBO, placebo; qd, once daily; qwk, once weekly.

Endpoints

- **Additive effect of bremelanotide:**
 - Percentage of patients with ≥5% weight loss at week 8 with tirzepatide + bremelanotide vs PBO + PBO
 - Percentage of patients with greater weight loss in Treatment Period 2 (week 8) vs Treatment Period 1 (week 4), tirzepatide + bremelanotide compared to PBO + PBO
 - Percentage change in weight loss with tirzepatide + bremelanotide vs PBO + PBO during Treatment Period 2 (ie, study weeks 4–8)
- **Co-administration vs PBO:** Percentage change in weight loss tirzepatide + bremelanotide compared to PBO + PBO across both treatment periods (week 8)
- **Weight loss maintenance:** Percentage change in weight loss with bremelanotide + PBO vs PBO + PBO in Treatment Period 2 (study weeks 4–8)

RESULTS

Baseline demographics

- Ninety-six randomized participants were ≈46 years old (75% female), with an average BMI of ≈38 and an average weight of ≈239 lb at baseline
- Baseline demographics were generally well balanced between the 4 groups in Treatment Period 2 (**Table 1**)
 - In the bremelanotide-only group, baseline weight was somewhat higher, and there were fewer females

Table 1. Patient baseline demographics by treatment group

Mean	Group 1 bremelanotide + tirzepatide (n=49)	Group 2 tirzepatide + PBO (n=15)	Group 3 bremelanotide + PBO (n=16)	Group 4 PBO + PBO (n=16)
Female, n (%)	37 (76)	13 (87)	10 (63)	12 (75)
Age, mean, y	46	47	43	47
Weight, mean, lb	230	232	272	241
BMI, kg/m^2	37	37	41	38

BMI, body mass index; PBO, placebo.

Safety

- Addition of bremelanotide did not appreciably affect the safety profile of tirzepatide (**Table 2**)
- The most common adverse events (AEs) for tirzepatide alone were gastrointestinal (GI)-related at 21% in Treatment Period 1 and 27% for group 2 in Treatment Period 2
- Hyperpigmentation was the most common AE for bremelanotide alone at 50% in group 3, Treatment Period 2

Efficacy

Treatment groups with ≥5% weight loss

- A ≥5% reduction in percent body weight from baseline to week 8 was achieved in group 1 (tirzepatide + bremelanotide) and group 2 (tirzepatide + PBO) and was statistically significant (P<0.05; **Figure 2**)
 - 40% of patients treated with tirzepatide + bremelanotide lost ≥5% of body weight compared with 27% treated with tirzepatide + PBO
- Only group 1 (tirzepatide + bremelanotide) achieved a ≥7% reduction in percent body weight by week 8 at 19% (P≤0.1)

Table 2. Adverse events by treatment periods and treatment groups

	Treatment Period 1 tirzepatide (N=113)	Treatment Period 2			
		Group 1 bremelanotide + tirzepatide (n=49)	Group 2 tirzepatide + PBO (n=15)	Group 3 bremelanotide + PBO (n=16)	Group 4 PBO + PBO (n=16)
All AEs, n (%)	47 (42)	32 (65)	8 (53)	13 (81)	4 (25)
AEs excluding hyperpigmentation	47 (42)	24 (49)	8 (53)	11 (69)	4 (25)
All GI AEs, n (%)	24 (21)	14 (29)	4 (27)	4 (25)	2 (13)
Nausea	11 (10)	6 (12)	1 (7)	4 (25)	2 (13)
Diarrhea	6 (5)	1 (2)	1 (7)	0 (0)	0 (0)
Emesis	2 (2)	0 (0)	0 (0)	1 (6)	0 (0)
Other	12 (11)	8 (16)	3 (20)	0 (0)	0 (0)
Headache, n (%)	5 (4)	6 (12)	1 (7)	1 (6)	0 (0)
Hyperpigmentation, n (%)	0 (0)	23 (47)	0 (0)	8 (50)	0 (0)
Flushing, n (%)	0 (0)	3 (6)	0 (0)	2 (13)	0 (0)
Elevated BP, n (%)	1 (1)	0 (0)	0 (0)	1 (6)	3 (19)
Severe AE, n (%)	4 (4)	0 (0)	0 (0)	1 (6)	0 (0)

AE, adverse event; BP, blood pressure; GI, gastrointestinal; PBO, placebo.

Figure 2. Percentage of patients with reductions in percent body weight by week 8[a]

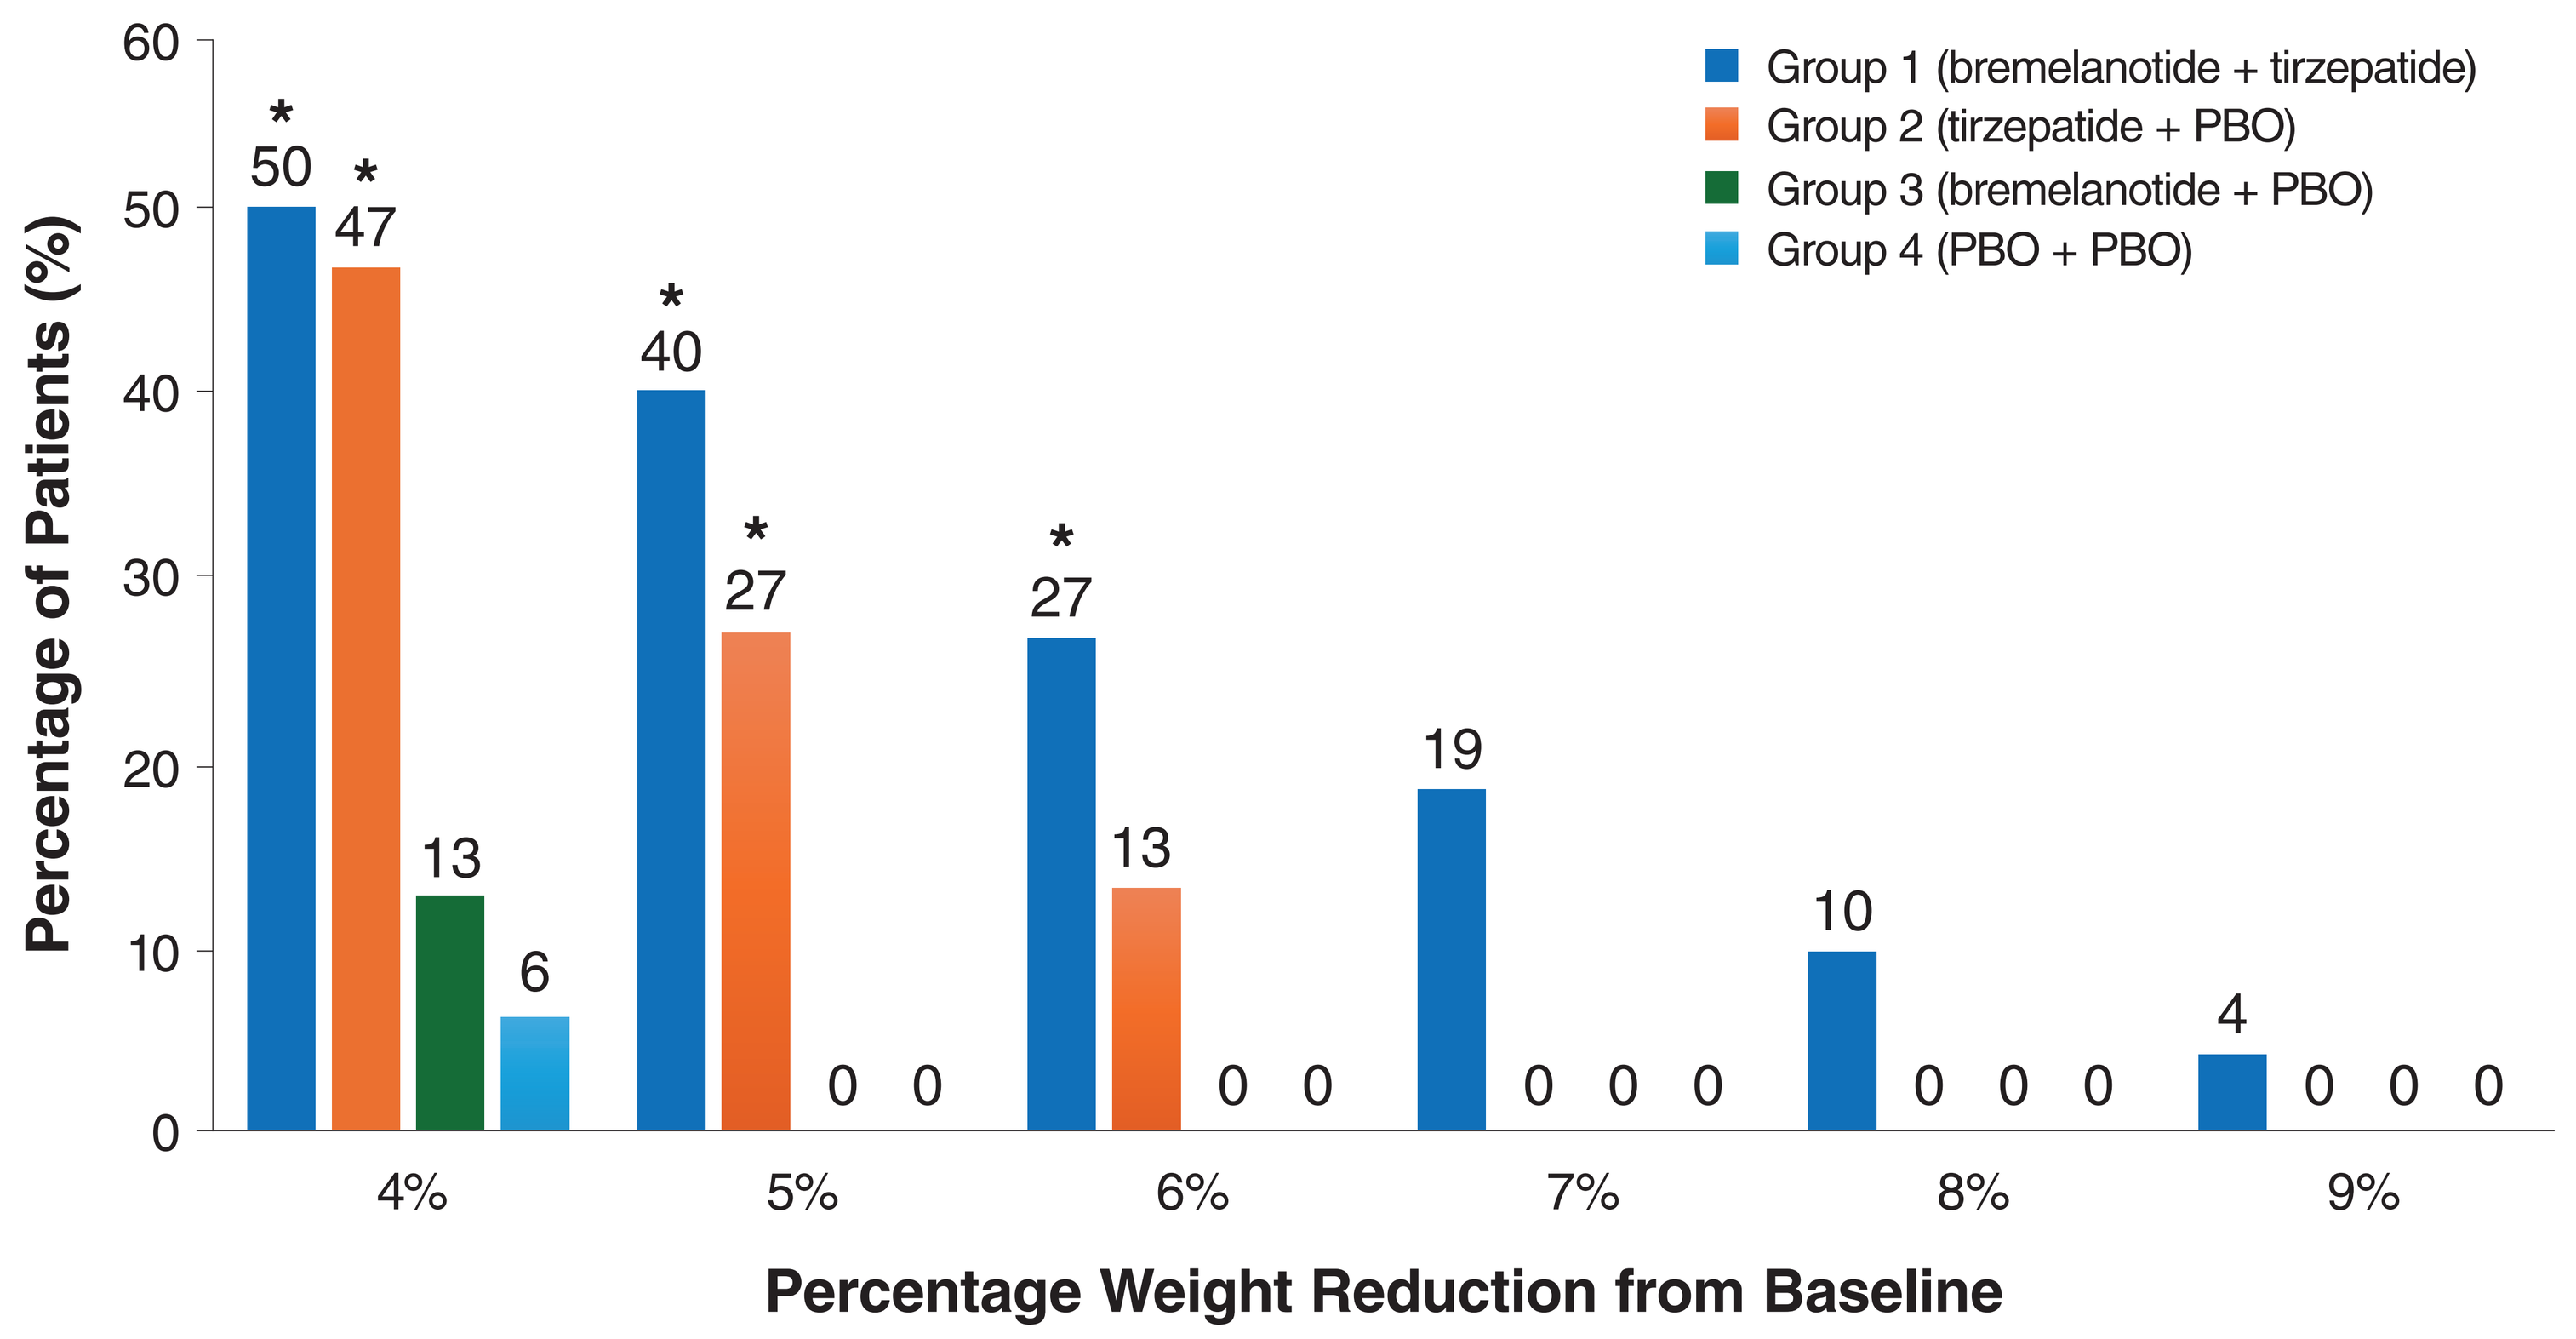
Group 1 (bremelanotide + tirzepatide)
Group 2 (tirzepatide + PBO)
Group 3 (bremelanotide + PBO)
Group 4 (PBO + PBO)
Percentage of Patients (%)
60
50
40
30
20
10
0
*
50
*
47
13
6
*
40
*
27
0 0
*
27
13
0 0
19
0 0 0
10
0 0 0
4
0 0 0
4%
5%
6%
7%
8%
9%
Percentage Weight Reduction from Baseline

[a]Statistics are based on comparison to group 4 (PBO + PBO).
*P<0.05
PBO, placebo.

Patient weight change from baseline at week 0

- The mean percent change in weight from baseline to week 8 (PBO subtracted) was significant for the bremelanotide + tirzepatide (–2.8%, P=0.0001) and tirzepatide + PBO (–1.9% with P<0.05) treatment groups vs PBO alone (**Figure 3**)

Patient weight change during treatment periods

- All patients lost weight during Treatment Period 1 (2.5 mg tirzepatide once weekly for 4 weeks; **Figure 4**)
- During Treatment Period 2, participants in the PBO + PBO group regained 0.4% of their weight compared with a loss of 0.3% for those in the bremelanotide + PBO group

Figure 3. Patient weight change (%) week 0 to week 8 compared to PBO

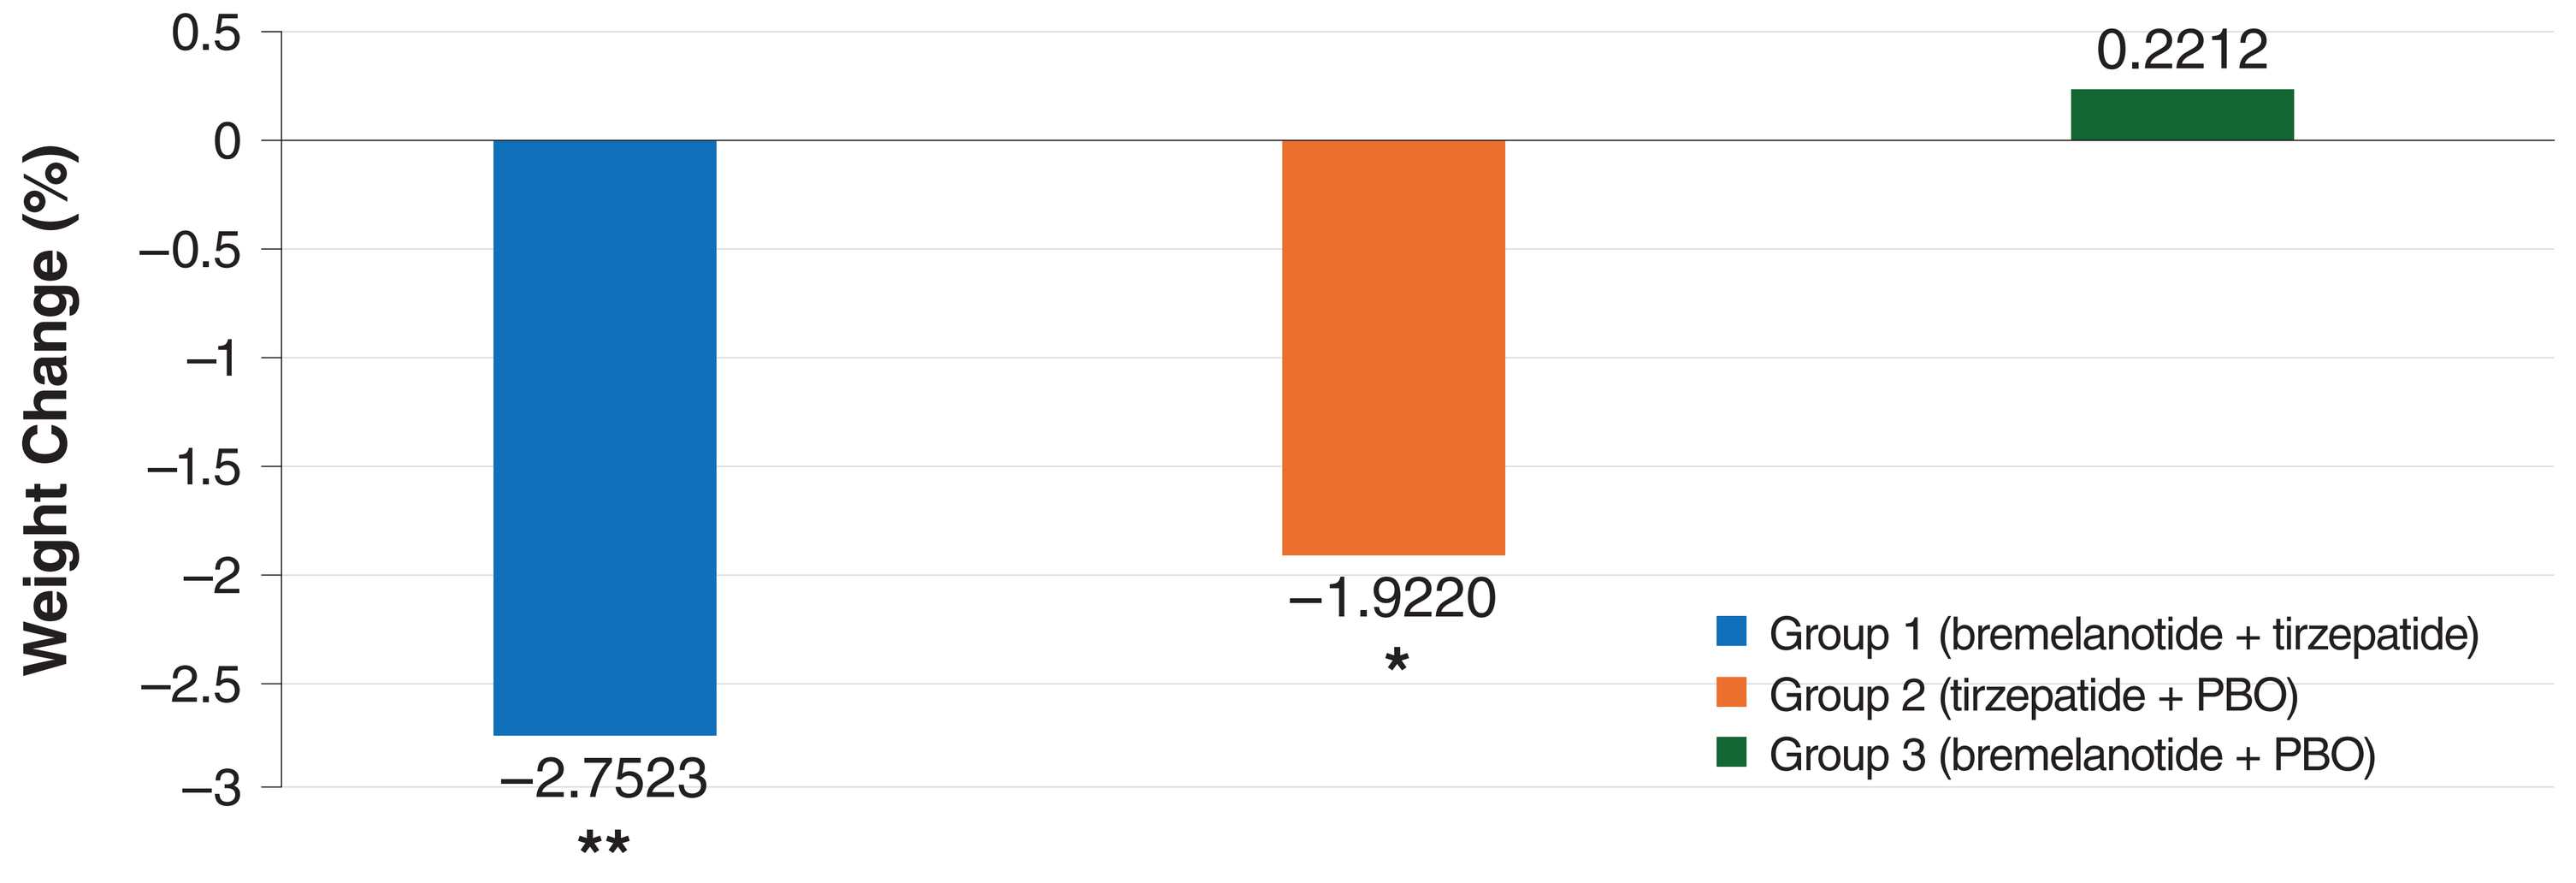
Weight Change (%)
0.5
0
−0.5
−1
−1.5
−2
−2.5
−3
−2.7523
**
−1.9220
*
0.2212
Group 1 (bremelanotide + tirzepatide)
Group 2 (tirzepatide + PBO)
Group 3 (bremelanotide + PBO)

**P<0.001; *P<0.05.
PBO, placebo.

Figure 4. Percent change in weight over time across treatment groups

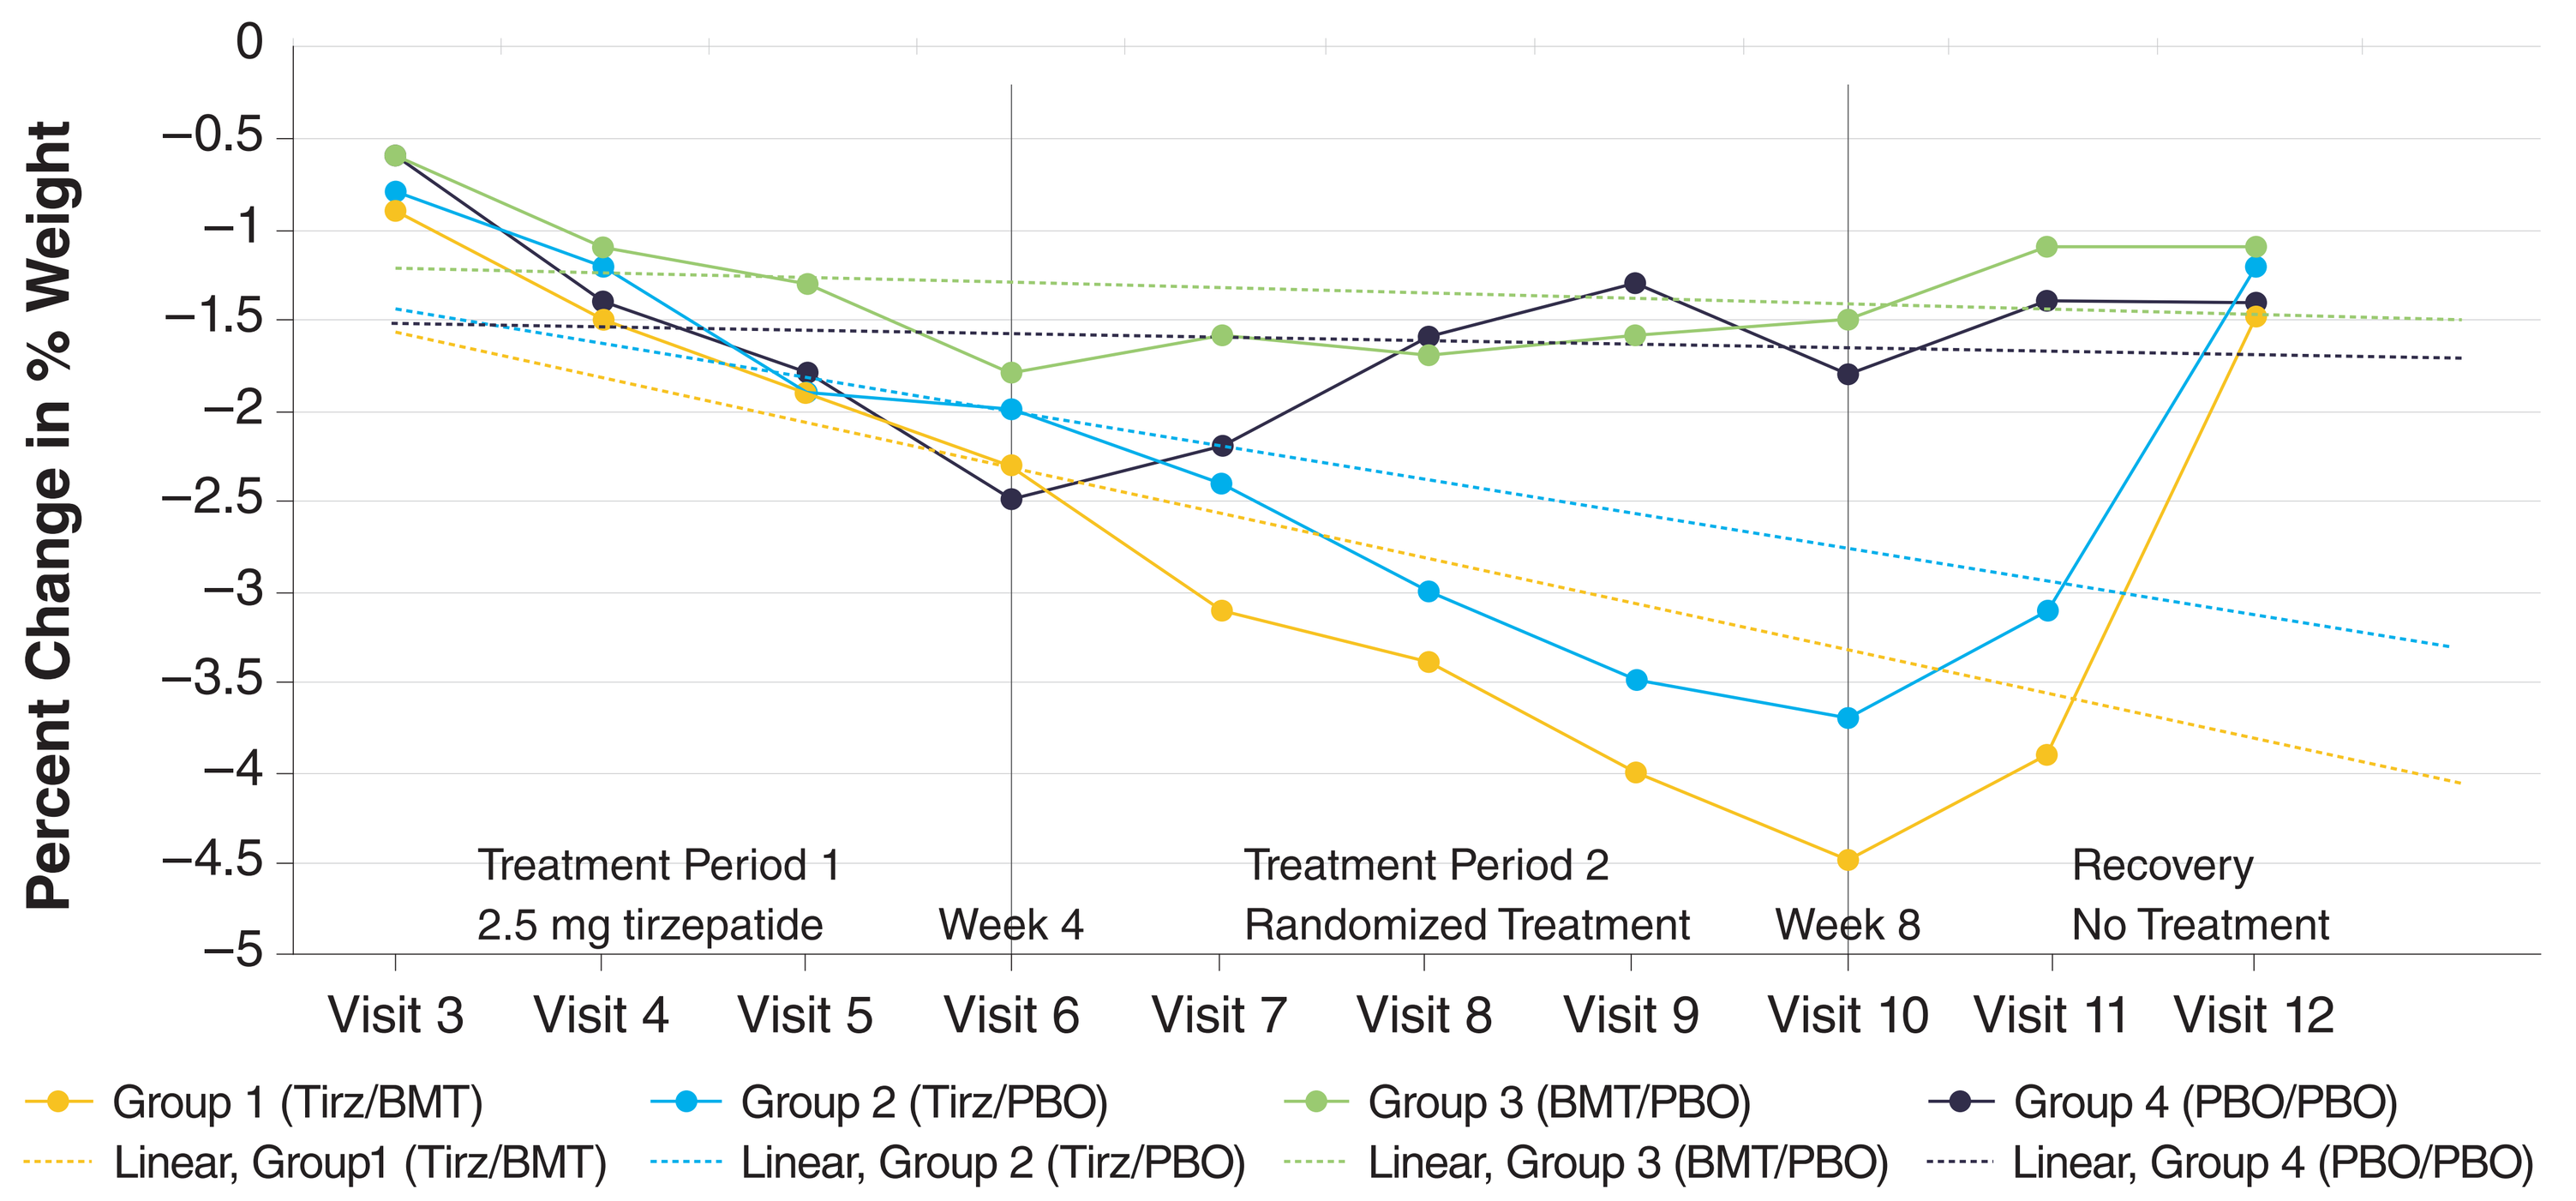
Percent Change in % Weight
0
−0.5
−1
−1.5
−2
−2.5
−3
−3.5
−4
−4.5
−5
Treatment Period 1
2.5 mg tirzepatide
Week 4
Treatment Period 2
Randomized Treatment
Week 8
Recovery
No Treatment
Visit 3
Visit 4
Visit 5
Visit 6
Visit 7
Visit 8
Visit 9
Visit 10
Visit 11
Visit 12
Group 1 (Tirz/BMT)
Group 2 (Tirz/PBO)
Group 3 (BMT/PBO)
Group 4 (PBO/PBO)
Linear, Group1 (Tirz/BMT)
Linear, Group 2 (Tirz/PBO)
Linear, Group 3 (BMT/PBO)
Linear, Group 4 (PBO/PBO)

BMT, bremelanotide; PBO, placebo; Tirz, tirzepatide.

CONCLUSIONS

- In this controlled study, adding a low-dose MC4R agonist to low-dose tirzepatide resulted in statistically greater weight loss over low-dose tirzepatide alone
 - There were no safety signals with co-administration; expected tolerability was observed in all treatment arms
 - These results indicate a potential positive, synergistic effect between the 2 treatments' mechanisms of action
- Participants who were randomized to PBO + PBO after 4 weeks of tirzepatide treatment experienced rapid weight regain
 - The use of low-dose MC4R agonism alone was able to blunt this weight regain
- Combined treatment was well tolerated, and there was no increase in GI AEs
- Findings support the addition of a low-dose MC4R agonist to incretin therapy as a safe and potentially effective strategy for achieving and maintaining weight loss, and these results will inform planned clinical and development programs in obesity and weight loss

Acknowledgments: Medical writing support was provided by Emily Doogan, PhD, for Citrus Health Group, Inc. (Chicago, Illinois) and was funded by Neurelis, Inc. (San Diego, California)

Disclosures: Robert Jordan, Carl Spana, Nazish Huq, and John Dodd are employees of Palatin Technologies, Inc. Ryan Morgan serves on the Speakers Bureau for Rhythm Pharmaceuticals and Eli Lilly, and is a consultant to Novo Nordisk and Madrigal Pharmaceuticals. Jesse Richards serves on the Speakers Bureau for Eli Lilly; Novo Nordisk; and Rhythm pharmaceuticals, is a member of the medical advisory board for Novo Nordisk and Eli Lilly, and has received research funding from Palatin Technologies, Inc. Ramses Vega-Casasnovas, Bruce Rankin, and Shannon Conrad have nothing to disclose.

References: 1. Singh M, Mukhopadhyay K. *Biomed Res Int*. 2014;2014:874610. 2. Dahir NS, et al. *J Clin Invest*. 2024;134(14):e178250. 3. Spana C, et al. *Diabetes Obes Metab*. 2022;24(6):1084-1093. 4. Yang Y, Xu Y. *J Mol Cell Biol*. 2020;12(10):785-797. 5. Tao YX. Endocr Rev. 2010;31(4):506-543. 6. Vyleesi (bremelanotide injection). Full Prescribing Information. 2024. 7. Ciardullo S, et al. *Acta Diabetol*. 2024;61(8):941-950. 8. ZEPBOUND (tirzepatide). Full Prescribing Information. Lilly USA; 2022.