Presented at:

Obesity Week 2025
November 4–7, 2025
Atlanta, GA

Characterization of the Melanocortin Receptor 4 Oral Small Molecule Agonist PL7737

John Dodd, PhD[1]; Carl Spana, PhD[1]; Marie Makhlina, MSc[1]; Lakmal W. Boteju, PhD[1]

[1]Palatin Technologies, Inc., Monmouth Junction, NJ

INTRODUCTION

- The melanocortin 4 receptor (MC4R) in the hypothalamus plays a central role in the regulation of stored energy and food intake and is a well validated target for obesity therapeutics[1]
 - To date there has been limited success in the identification of orally available, small molecule MC4R agonists with acceptable drug characteristics for clinical development
- Here, we describe the characterization of PL7737, an oral, small molecule MC4R agonist in development for various obesity etiologies
 - Oral PL7737 is a potent MC4R agonist (2 nM EC_{50}, 95% E_{max}, data on file)
 - PL7737 has reduced potency against MC1R
 - PL7737 is inactive against MC3R
 - PL7737 has received FDA orphan drug designation for obesity due to leptin receptor deficiency[2]

METHODS

- Oral PL7737 was evaluated for effects on weight loss and blood pressure in diet-induced obese (DIO) mice
- In addition, PL7737 was evaluated in hERG, Ames, and a 28-day non-GLP toxicology study in rats

Mouse Study 1: Two-Week Body Weight and Systolic Blood Pressure Study

- Male C57BL/6 DIO mice (n=6) were preconditioned on a high-fat diet for 6-8 weeks
 - After acclimation, mice were dosed orally twice daily with vehicle or PL7737 (3, 10, or 20 mg/kg) for 7 days (**Figure 1**)
 - Dosing by oral gavage occurred 1 h before lights out in the AM and 6 h after lights out in the PM
 - A 7-day recovery period followed to assess weight regain
 - Body weights were assessed daily, and blood pressure was continuously recorded
 - Body weight was measured 1 h before lights out

Figure 1. Study Design for 2-Week Mouse Study

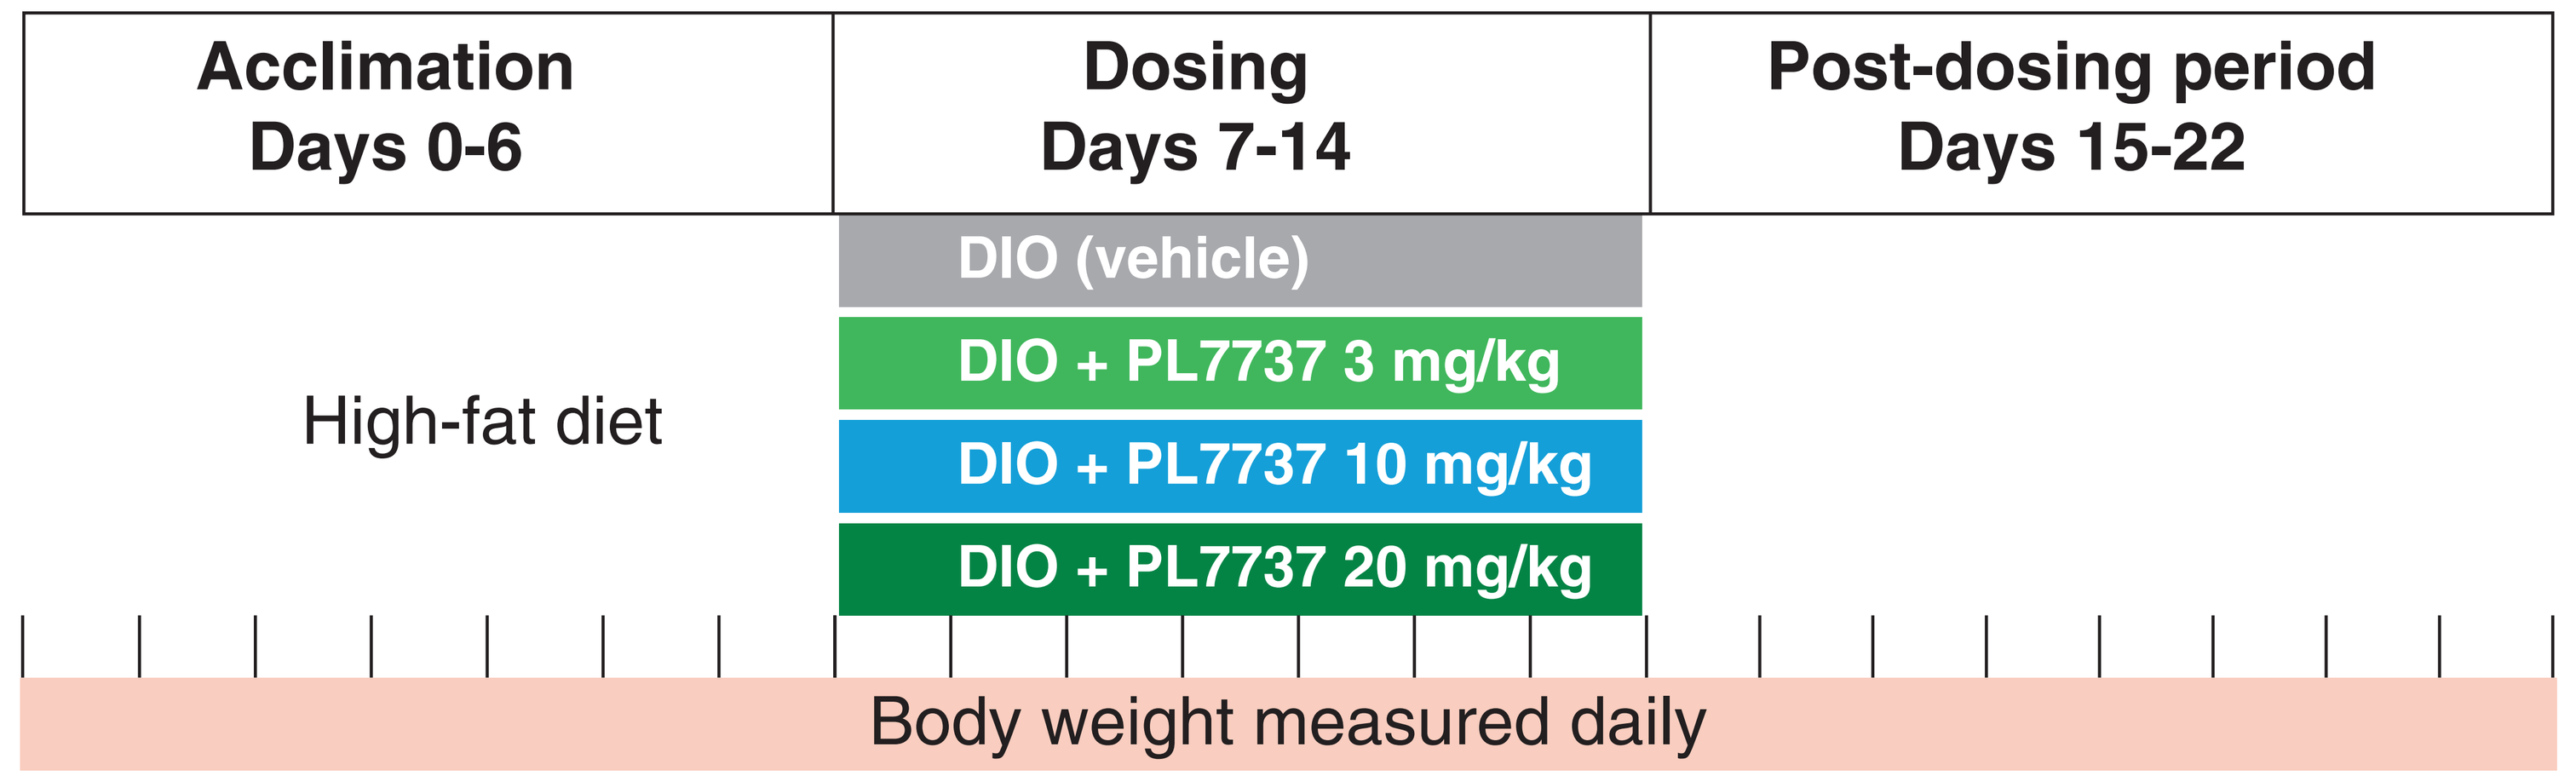
Acclimation
Days 0-6
Dosing
Days 7-14
Post-dosing period
Days 15-22
DIO (vehicle)
DIO + PL7737 3 mg/kg
DIO + PL7737 10 mg/kg
DIO + PL7737 20 mg/kg
High-fat diet
Body weight measured daily

Mouse Study 2: Single-Week Body Weight Study

- Male C57BL/6 DIO mice (n=9), aged 17 weeks, were preconditioned on a high-fat diet for 11 weeks
- After acclimation to a reverse light cycle and handling, mice were dosed with PL7737 (3, 10, or 30 mg/kg) or vehicle
 - Dosing occurred once per day 1 h before lights out on days 0-3
- Body weight was measured daily from day –3 to day 3 twice per day 1 h before lights out and 6 h after lights out

Rat Pharmacokinetic and Toxicology Analyses

- PL7737 pharmacokinetics
 - Male Sprague-Dawley (SD) rats weighing 250±20 g were implanted with a telemetry device and allowed to recover for ≥1 week
 - Rats were dosed with PL7737 at 10 or 30 mg/kg SC, 30 mg/kg PO, or 1 mg/kg IV
 - Plasma PK samples were collected and sent to Frontage for analysis
- Toxicology (hERG and mutagenicity)
 - Standard Ames test was performed per industry standard on a dose-response curve of PL7737 starting with 100 µM to assess the mutagenic potential of PL7737[3]
 - Ames test, also known as standard bacterial reverse mutation assay, was performed using histidine-dependent Salmonella tester strains TA98, TA100, TA1535, and TA1537
 - This test was performed both with and without S9 liver extract in order to account for metabolic activation
 - Dose-response curve of PL7737 starting at 1 µM was tested for hERG activation in cell-based functional assays in Eurofins Discovery Services CardiacProfiler™ panel using the IonWorks Quattro electrophysiological platform
 - PL7737 was also tested in hERG Radioligand Binding assay at a concentration of 10 µM
- 28-day non-GLP toxicology study in rats
 - PL7737 was given at 25 mg/kg orally twice daily for a period of 28 days and assessed for the possible toxic effects in both male and female SD rats (n=5 each sex)
 - Points assessed at the end of the study included: body weight gain, food and water consumption, clinical chemistry, hematology, complete blood count (CBC), blood coagulation, external and internal gross observation, organ weight, and histopathological examination

RESULTS

- **In Mouse Study 1**, PL7737-treated animals showed rapid weight loss, which was statistically significant for the 20 mg/kg group (**Figure 2**)
 - Oral PL7737 did not have an effect on systolic blood pressure acutely or chronically at any dose evaluated (**Figure 3**)

Figure 2. Body Weight Change in DIO Mice (Vehicle Subtracted) Before and After Dosing

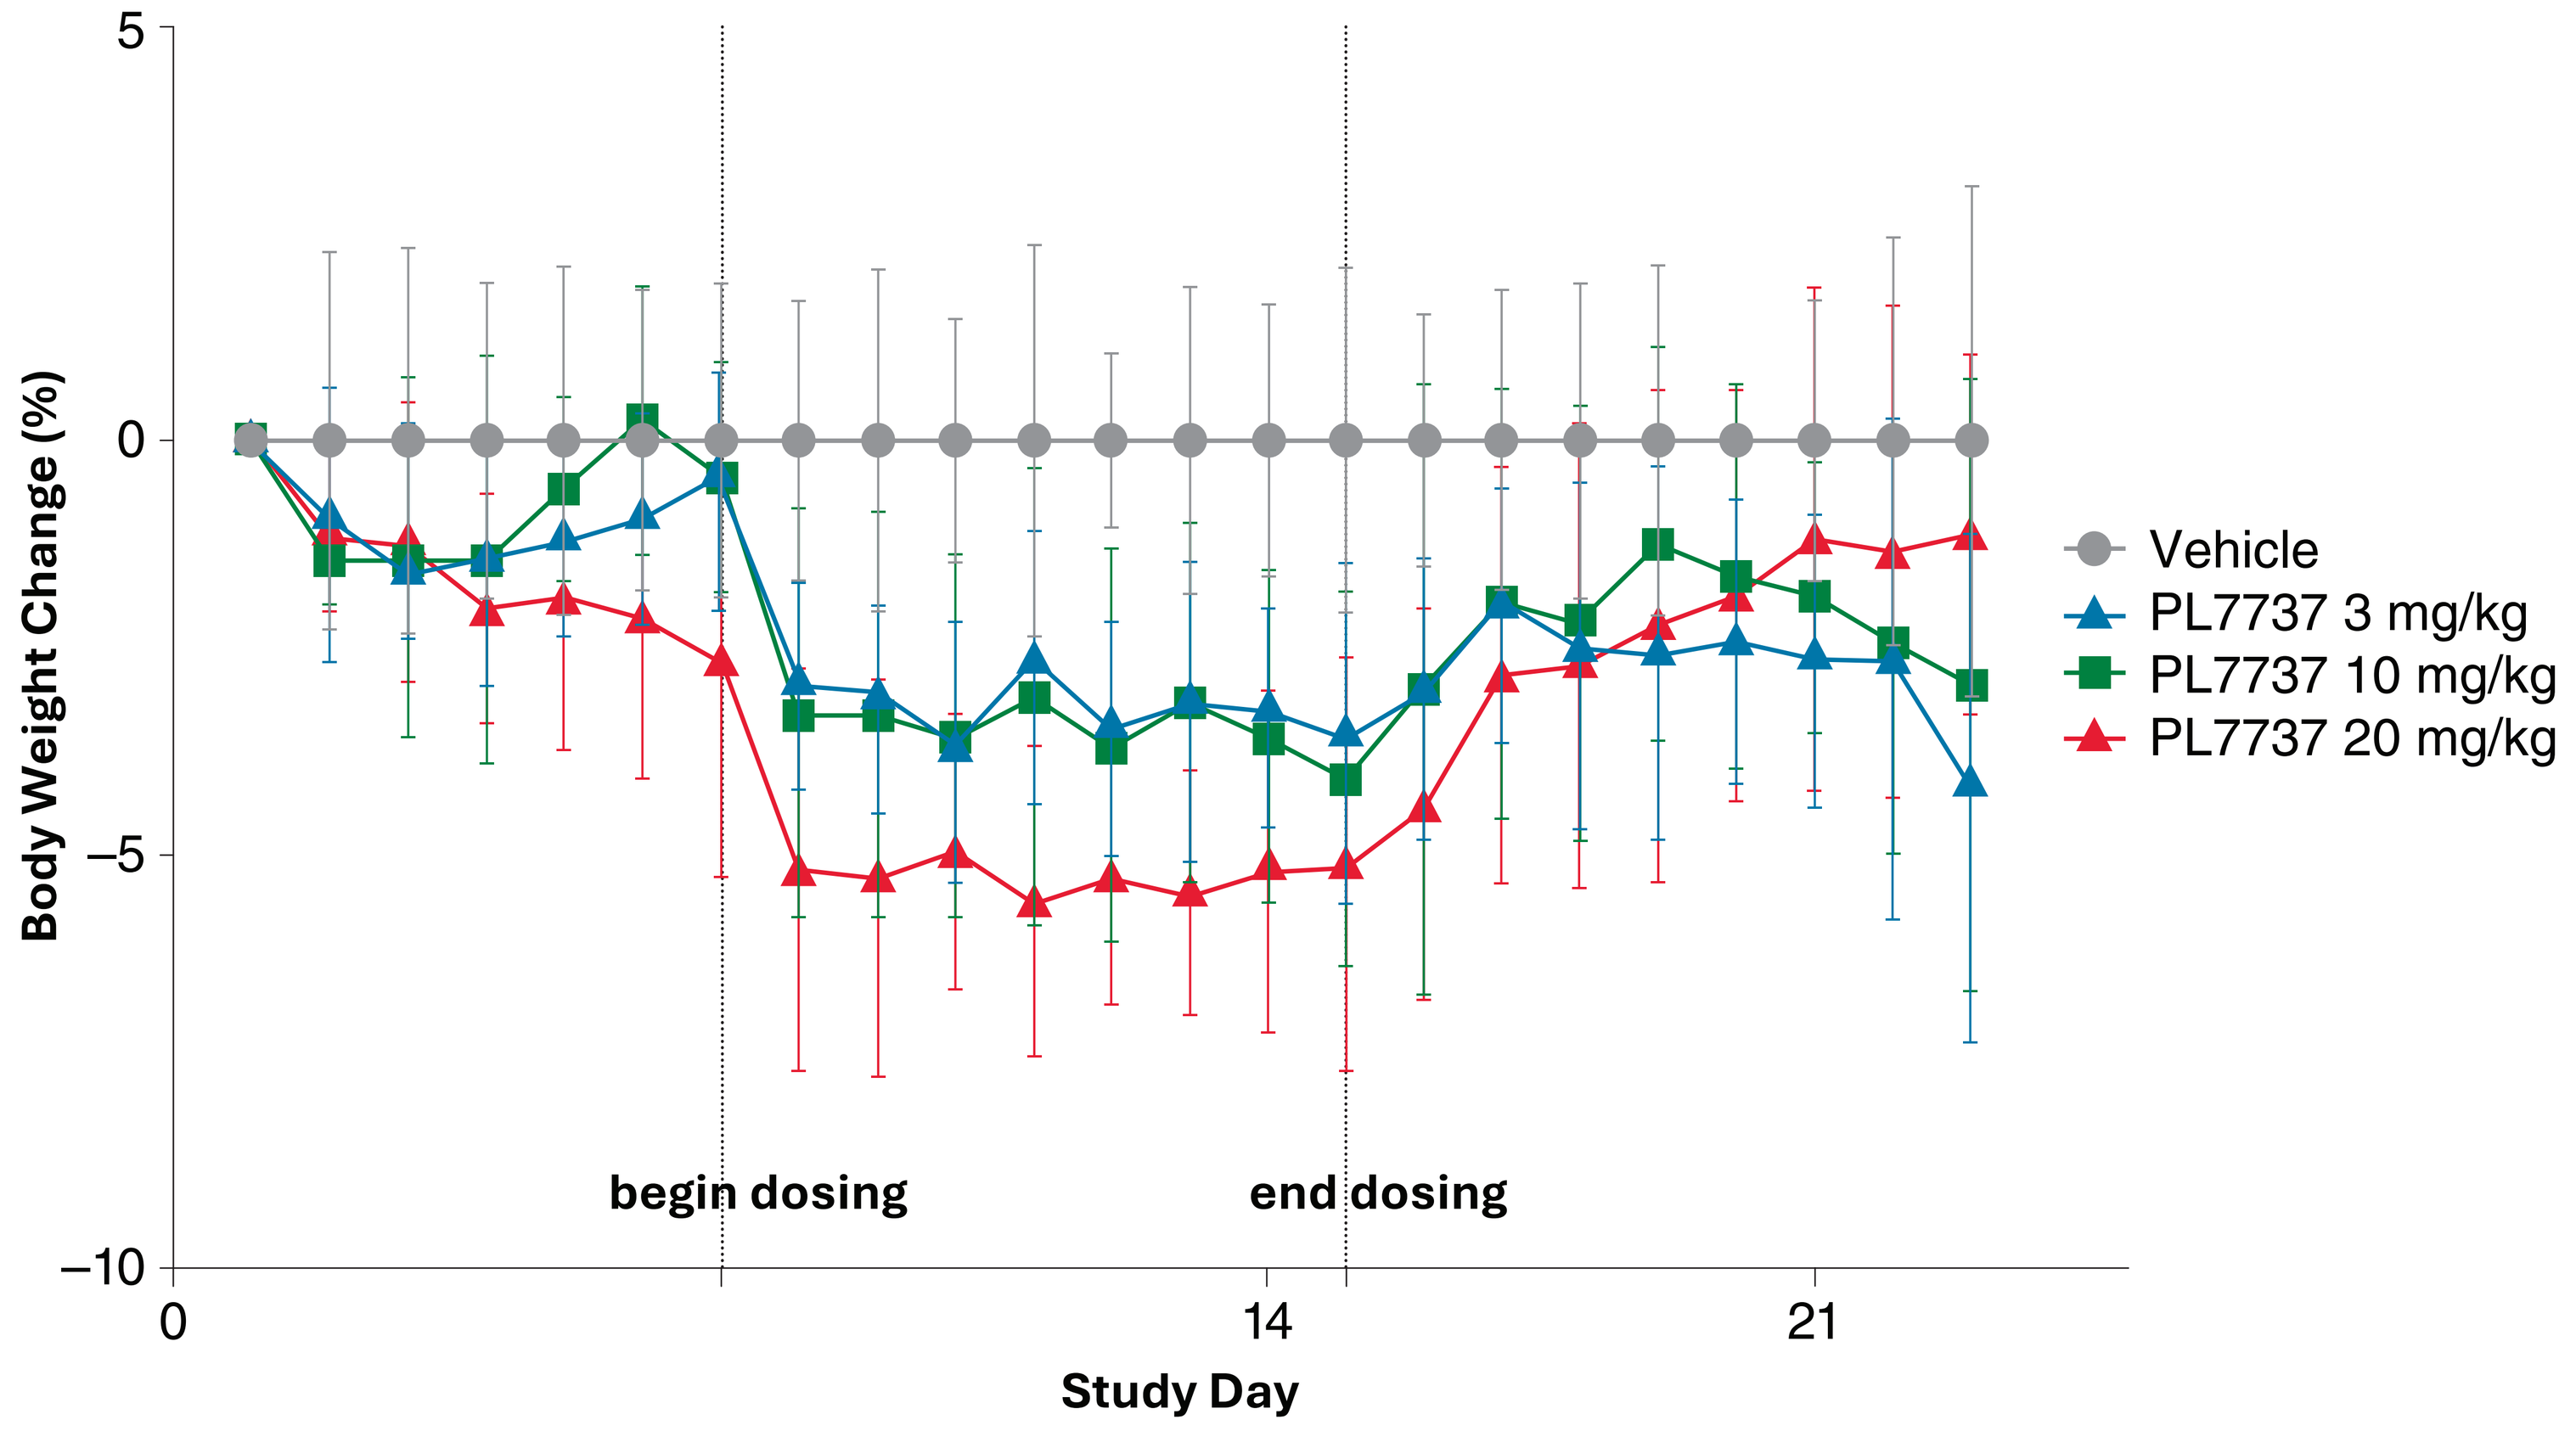
Body Weight Change (%)
5
0
−5
−10
0
14
21
Study Day
begin dosing
end dosing
Vehicle
PL7737 3 mg/kg
PL7737 10 mg/kg
PL7737 20 mg/kg

Figure 3. Systolic Blood Pressure Not Affected in DIO Mice with Telemetry

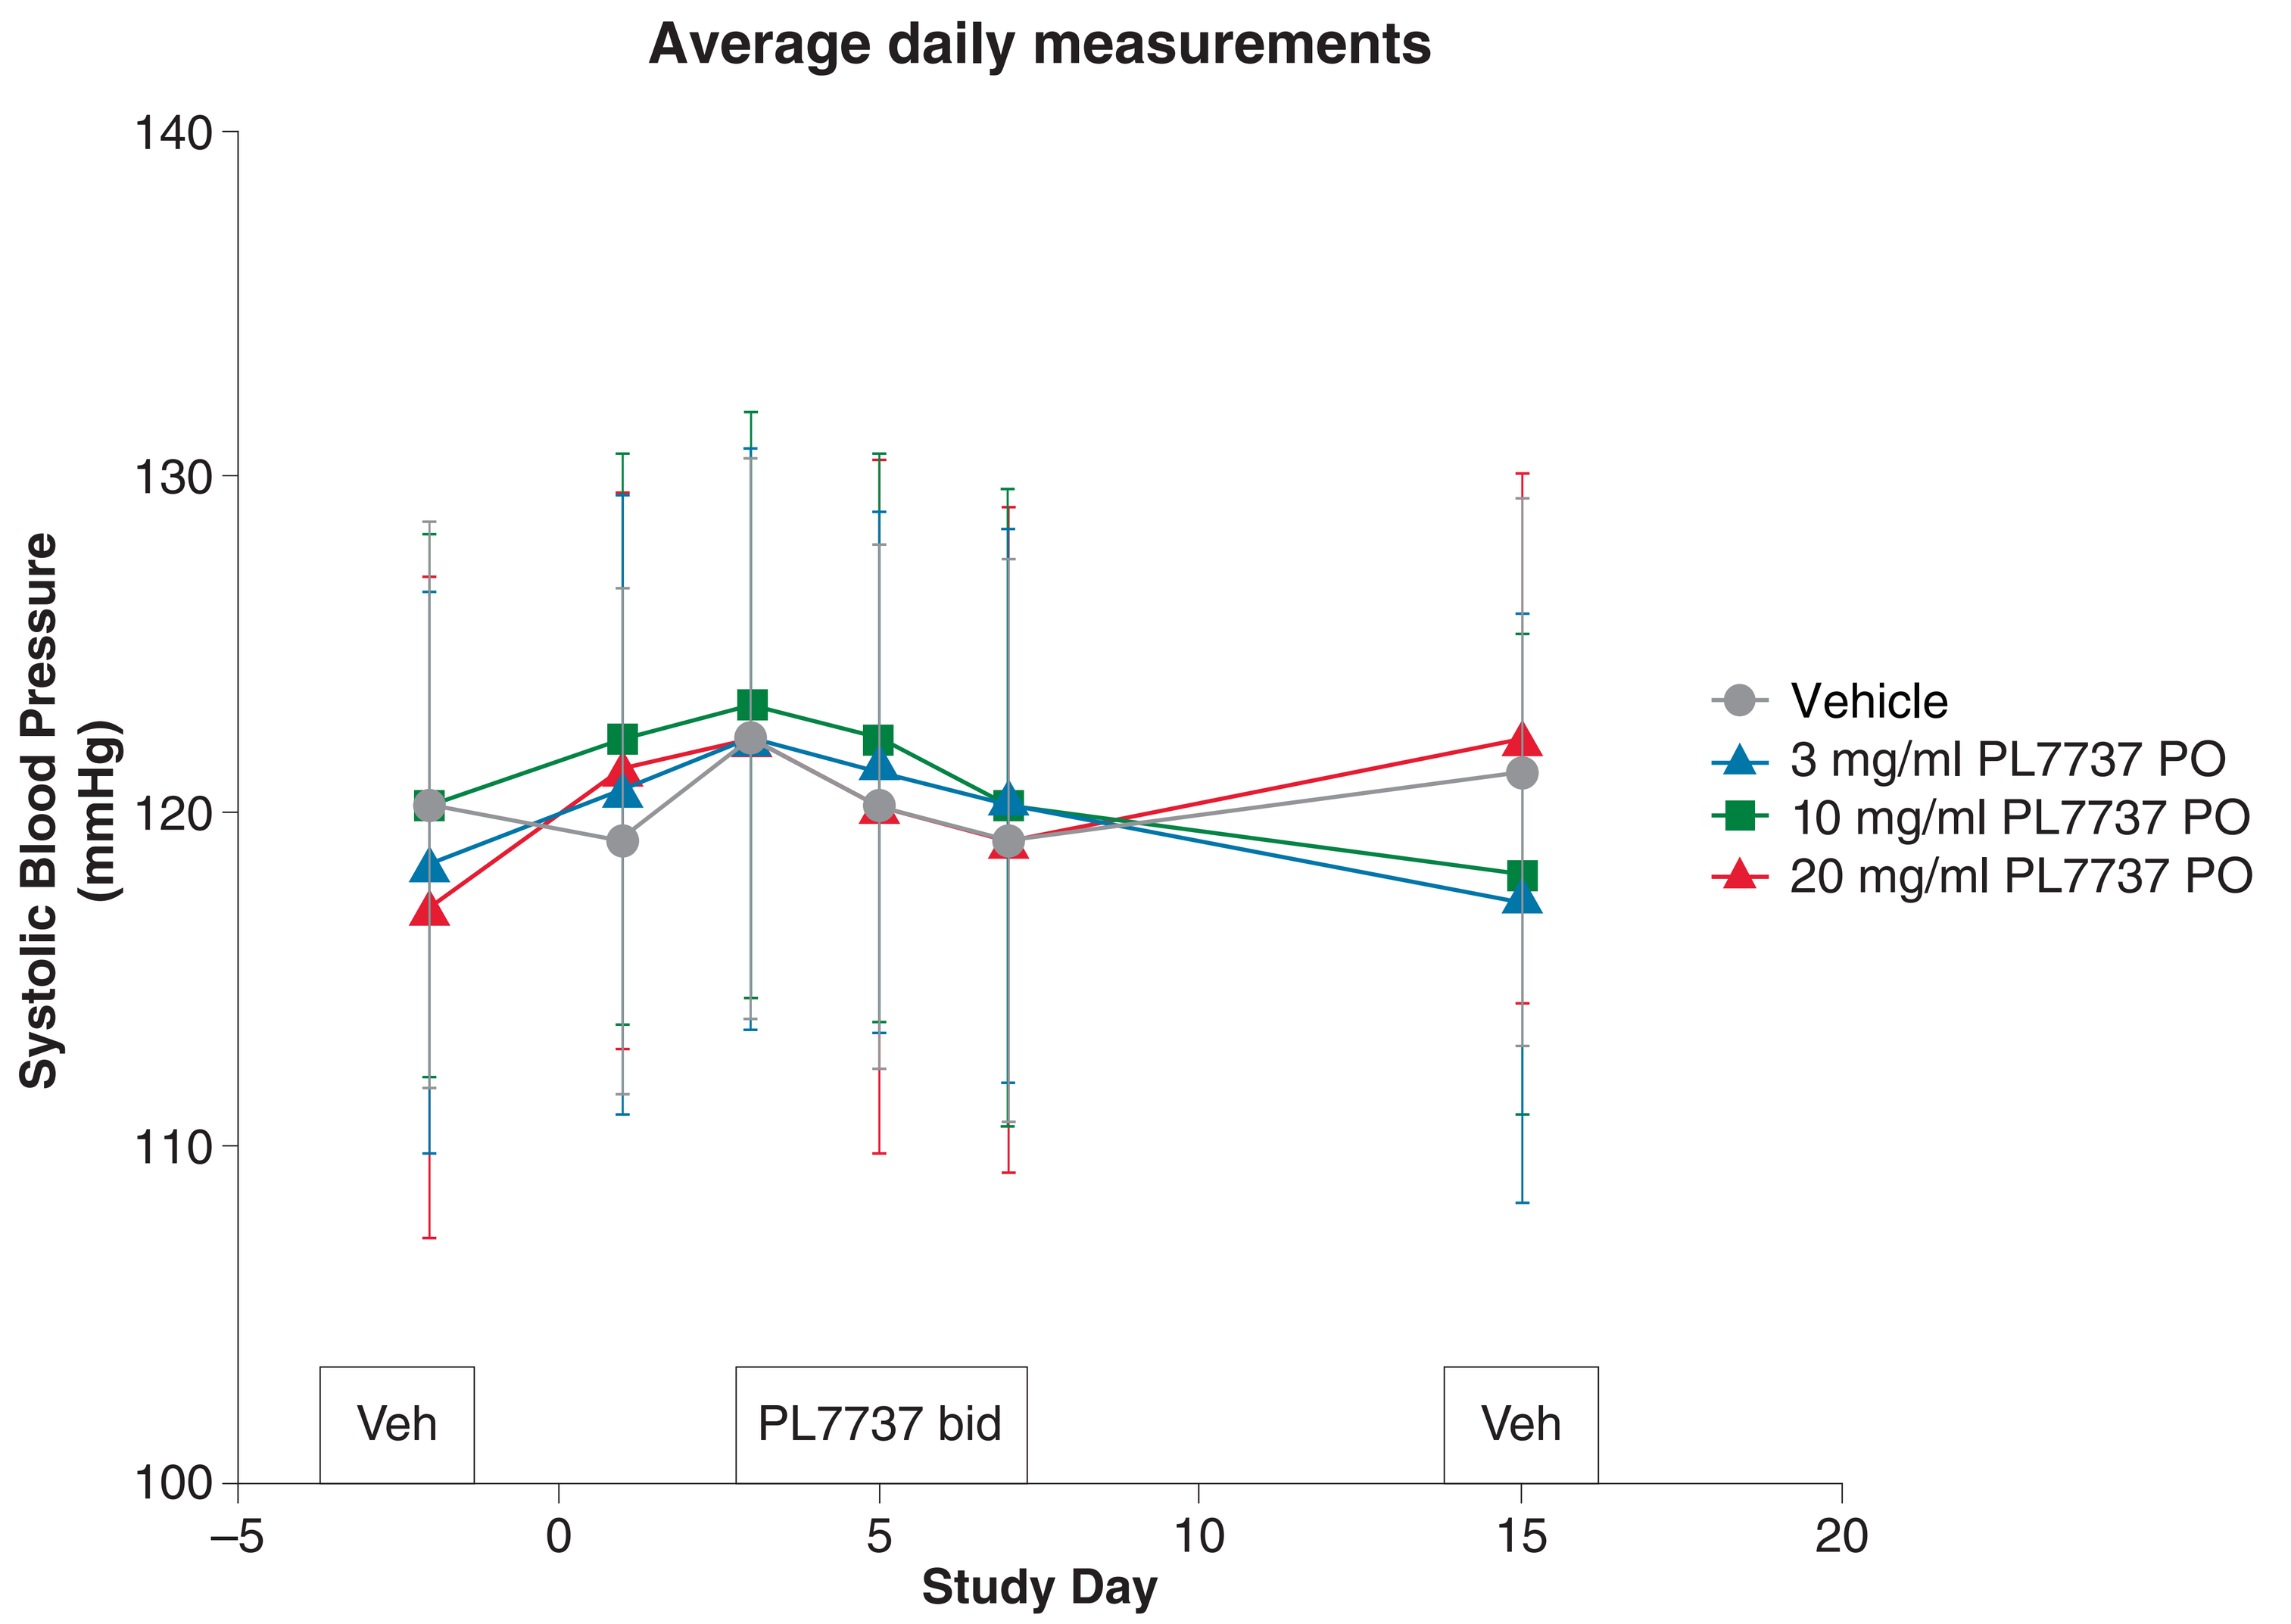
Average daily measurements
Systolic Blood Pressure (mmHg)
140
130
120
110
100
−5
0
5
10
15
20
Study Day
Veh
PL7737 bid
Veh
Vehicle
3 mg/ml PL7737 PO
10 mg/ml PL7737 PO
20 mg/ml PL7737 PO

- **In Mouse Study 2**, oral PL7737 reduced body weight in DIO mice in a dose-dependent manner compared to vehicle control
 - The 30 mg/kg BID dose resulted in a significant 5% (vehicle subtracted) reduction in body weight (P<0.05) (**Figure 4**)

Figure 4. Body Weight Change in DIO Mice (Vehicle Subtracted)

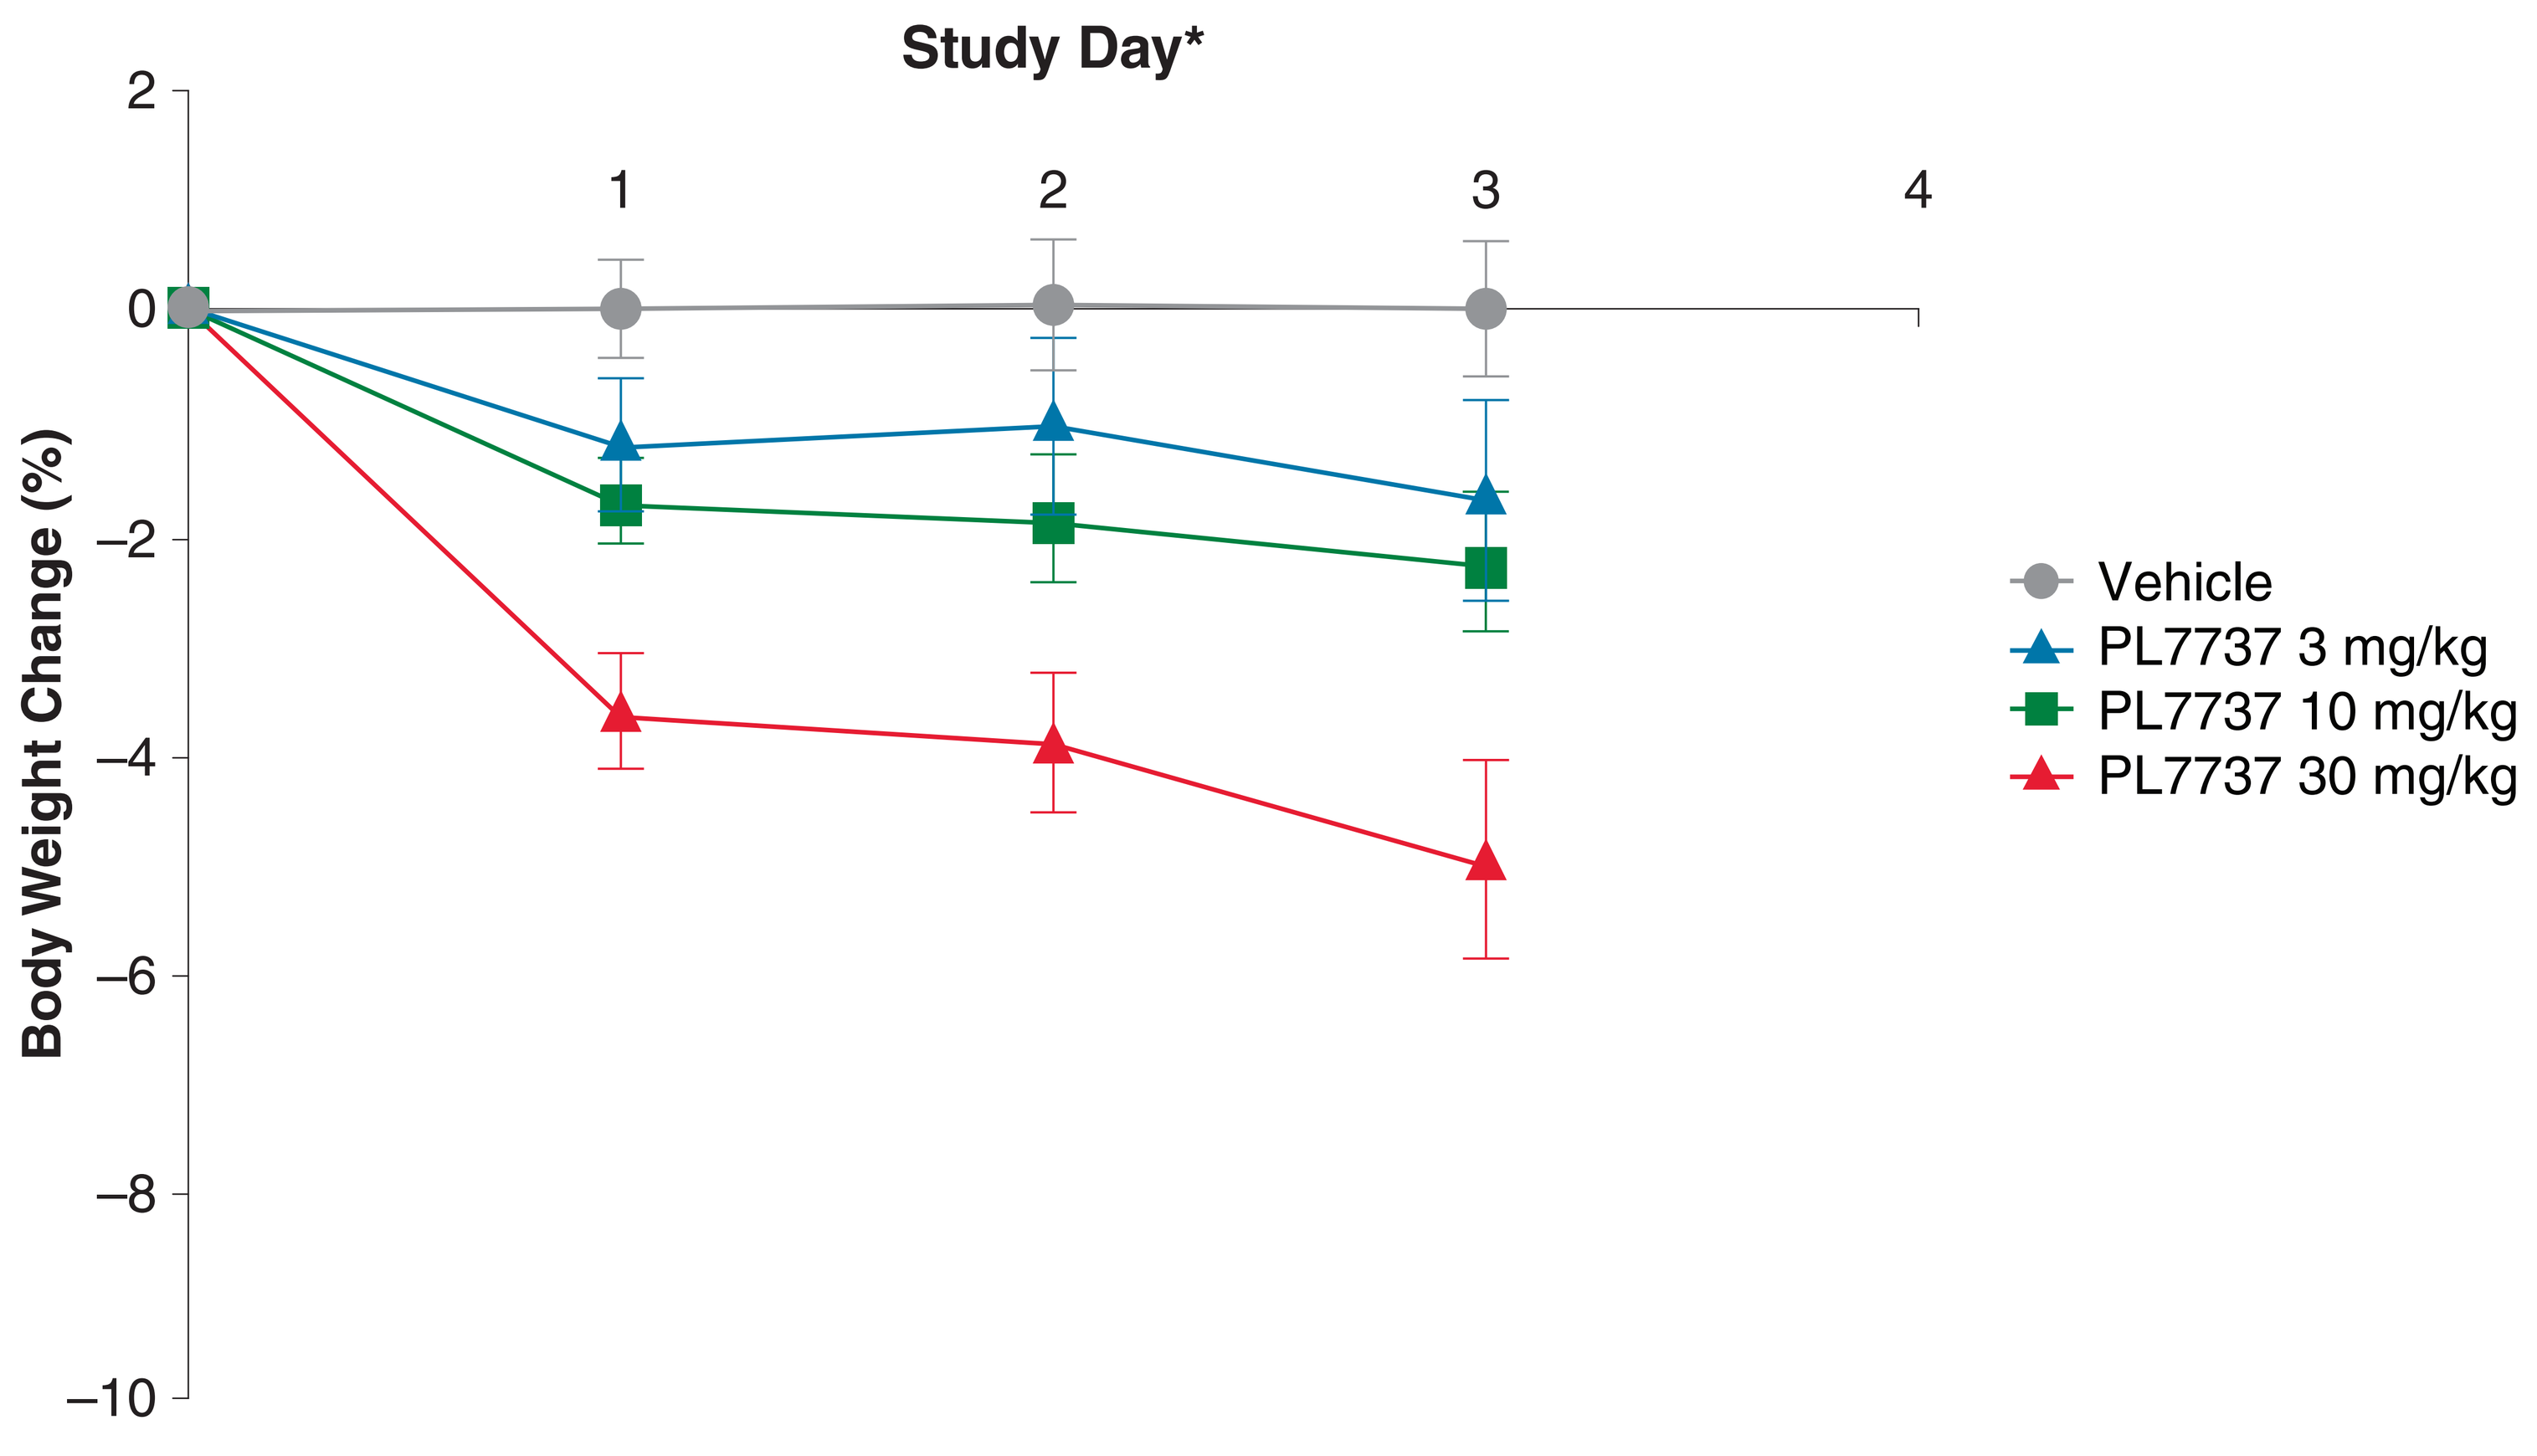
Study Day*
1
2
3
4
Body Weight Change (%)
2
0
−2
−4
−6
−8
−10
Vehicle
PL7737 3 mg/kg
PL7737 10 mg/kg
PL7737 30 mg/kg

*Mice were dosed on days 0, 1, and 2.

- **Rat pharmacology and safety studies**
 - PL7737 had an oral bioavailability of ≈50% in rats with a half-life >3 hours
 - There were no effects in hERG (>33 µM) or Ames mutagenicity assays
 - The 28-day non-GLP toxicity study showed no evidence of toxicity

CONCLUSIONS

- Long-term management of weight loss will require multiple safe and effective mechanisms
- PL7737 is a potent MC4R agonist that can cause significant weight loss when delivered orally
 - Preclinical studies indicate that oral PL7737 does not affect blood pressure or prolong the QT_c segment
 - The emerging preclinical efficacy and toxicology profile supports the clinical development of PL7737 as an oral treatment for obesity
- Investigational new drug-enabling studies have been initiated and phase 1 safety studies are planned

Funding: This study was funded by Palatin Technologies, Inc. (Monmouth Junction, New Jersey).

Acknowledgments: Medical writing support was provided by Hannah Riley Knight, PhD, from Citrus Health Group, Inc. (Chicago, Illinois), and was funded by Palatin Technologies, Inc. (Monmouth Junction, New Jersey).

Disclosures: **Carl Spana, John Dodd,** and **Lakmal W. Boteju** are employees of Palatin Technologies, Inc.

References: 1. Singh RK, et al. *C R Biol*. 2017;340(2):87-108. 2. Orphan Drug Designations and Approvals. https://www.accessdata.fda.gov/scripts/opdlisting/oopd/detailedIndex.cfm?cfgridkey=1065924. 3. Maron DM, Ames BN. *Mutat Res*. 1983;113(3-4):173-215.